A Phase 1/2 Study of a First-in-Human Immune-Stimulating Antibody Conjugate (ISAC) BDC-1001 in Patients with Advanced HER2-Expressing Solid Tumors (NCT04278144)

ABSTRACT
2538

Bob T. Li,[1] Mark D. Pegram,[2] Keun-Wook Lee,[3] Manish R. Sharma,[4] Jeeyun Lee,[5] Alexander Spira,[6] Glenn J. Hanna,[7] Yoon-Koo Kang,[8] Drew Rasco,[9] Kathleen Moore,[10] Benjamin A. Weinberg,[11] Michael N. Alonso,[12] Jason Ptacek,[12] Ming Yin,[12] Coya Tapia,[12] Lu Xu,[12] Edith A. Perez,[12] Ecaterina E. Dumbrava[13]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Stanford University, Stanford, CA, USA; [3]Seoul National University College of Medicine, Seoul National University Bundang Hospital, Seongnam, South Korea; [4]START-Midwest, Grand Rapids, MI, USA; [5]Samsung Medical Center, Seoul, South Korea; [6]Virginia Cancer Specialists, US Oncology Research and NEXT Oncology Virginia, Fairfax, VA, USA; [7]Dana-Farber Cancer Institute, Boston, MA, USA; [8]Asan Medical Center, Seoul, South Korea; [9]START, San Antonio, TX, USA; [10]Stephenson Cancer Center, Oklahoma City, OK, USA; [11]Georgetown Lombardi Comprehensive Cancer Center, Washington, DC, USA; [12]Bolt Biotherapeutics, Redwood City, CA, USA; [13]The University of Texas MD Anderson Cancer Center, Houston, TX, USA

INTRODUCTION

Novel, First-in-Class Boltbody™ Immune-Stimulating Antibody Conjugate (ISAC)

Molecular Structure

- BDC-1001 consists of
 - Trastuzumab biosimilar
 - Payload: TLR7/8 agonist
 - Linker: non-cleavable
- BDC-1001 linker-payload is cell membrane-impermeable

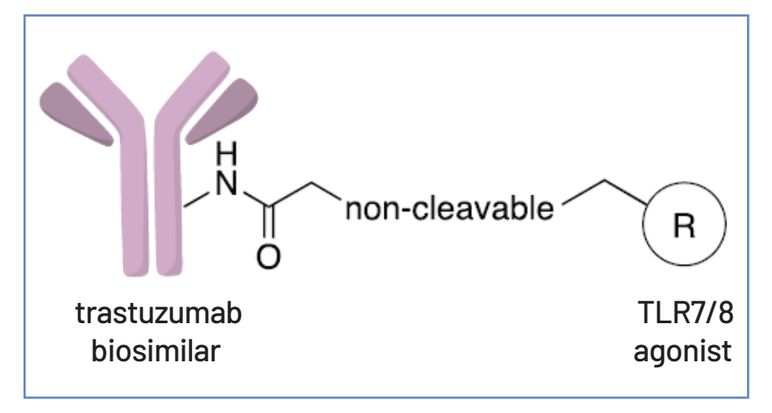

non-cleavable
R
trastuzumab biosimilar
TLR7/8 agonist

Proposed Mechanism of Action (MOA)

- Intravenous administration
- Local activation of the innate immune system
- Generates a durable tumor-targeted adaptive immune response

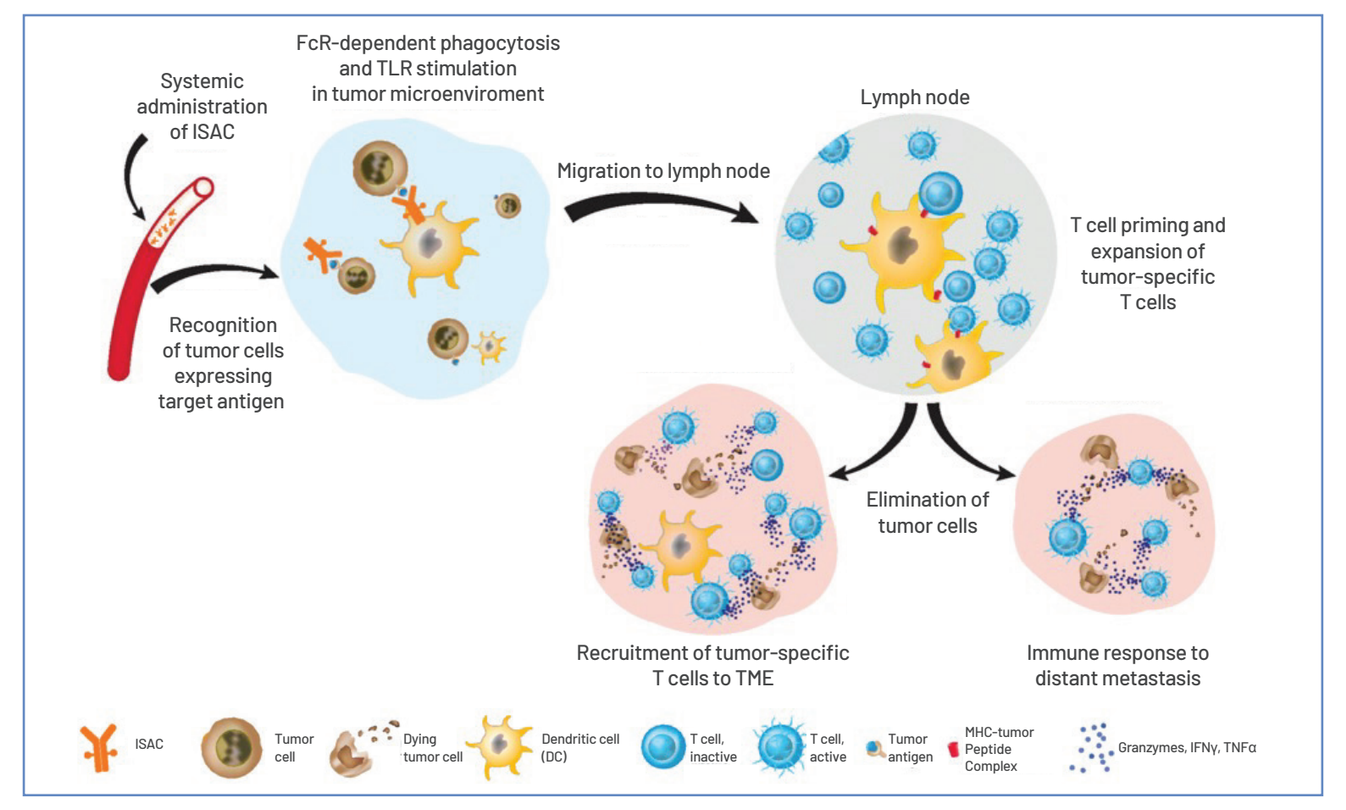

Systemic administration of ISAC
FcR-dependent phagocytosis and TLR stimulation in tumor microenviroment
Migration to lymph node
Lymph node
T cell priming and expansion of tumor-specific T cells
Recognition of tumor cells expressing target antigen
Elimination of tumor cells
Recruitment of tumor-specific T cells to TME
Immune response to distant metastasis
ISAC
Tumor cell
Dying tumor cell
Dendritic cell (DC)
T cell, inactive
T cell, active
Tumor antigen
MHC-tumor Peptide Complex
Granzymes, IFNγ, TNFα

Ackerman SE, et al. *Nature Cancer*. 2021;2(1):18-33.

METHODS

Single Agent and Combination with Nivolumab

Key Eligibility

- HER2-expressing solid tumors:
 - HER2 IHC3+ or gene amplified by ISH or NGS (HER2+)
 - HER2 IHC2+ and no gene amplification (HER2-low)
- Prior anti-HER2 and/or checkpoint inhibitor therapy allowed

Primary Objectives

- Safety and tolerability; recommended phase 2 dose (RP2D) selection

Exploratory Objectives

- Pharmacokinetics, preliminary anti-tumor activity, plasma and tissue biomarkers to explore proof of mechanism

Dose-escalation Schema

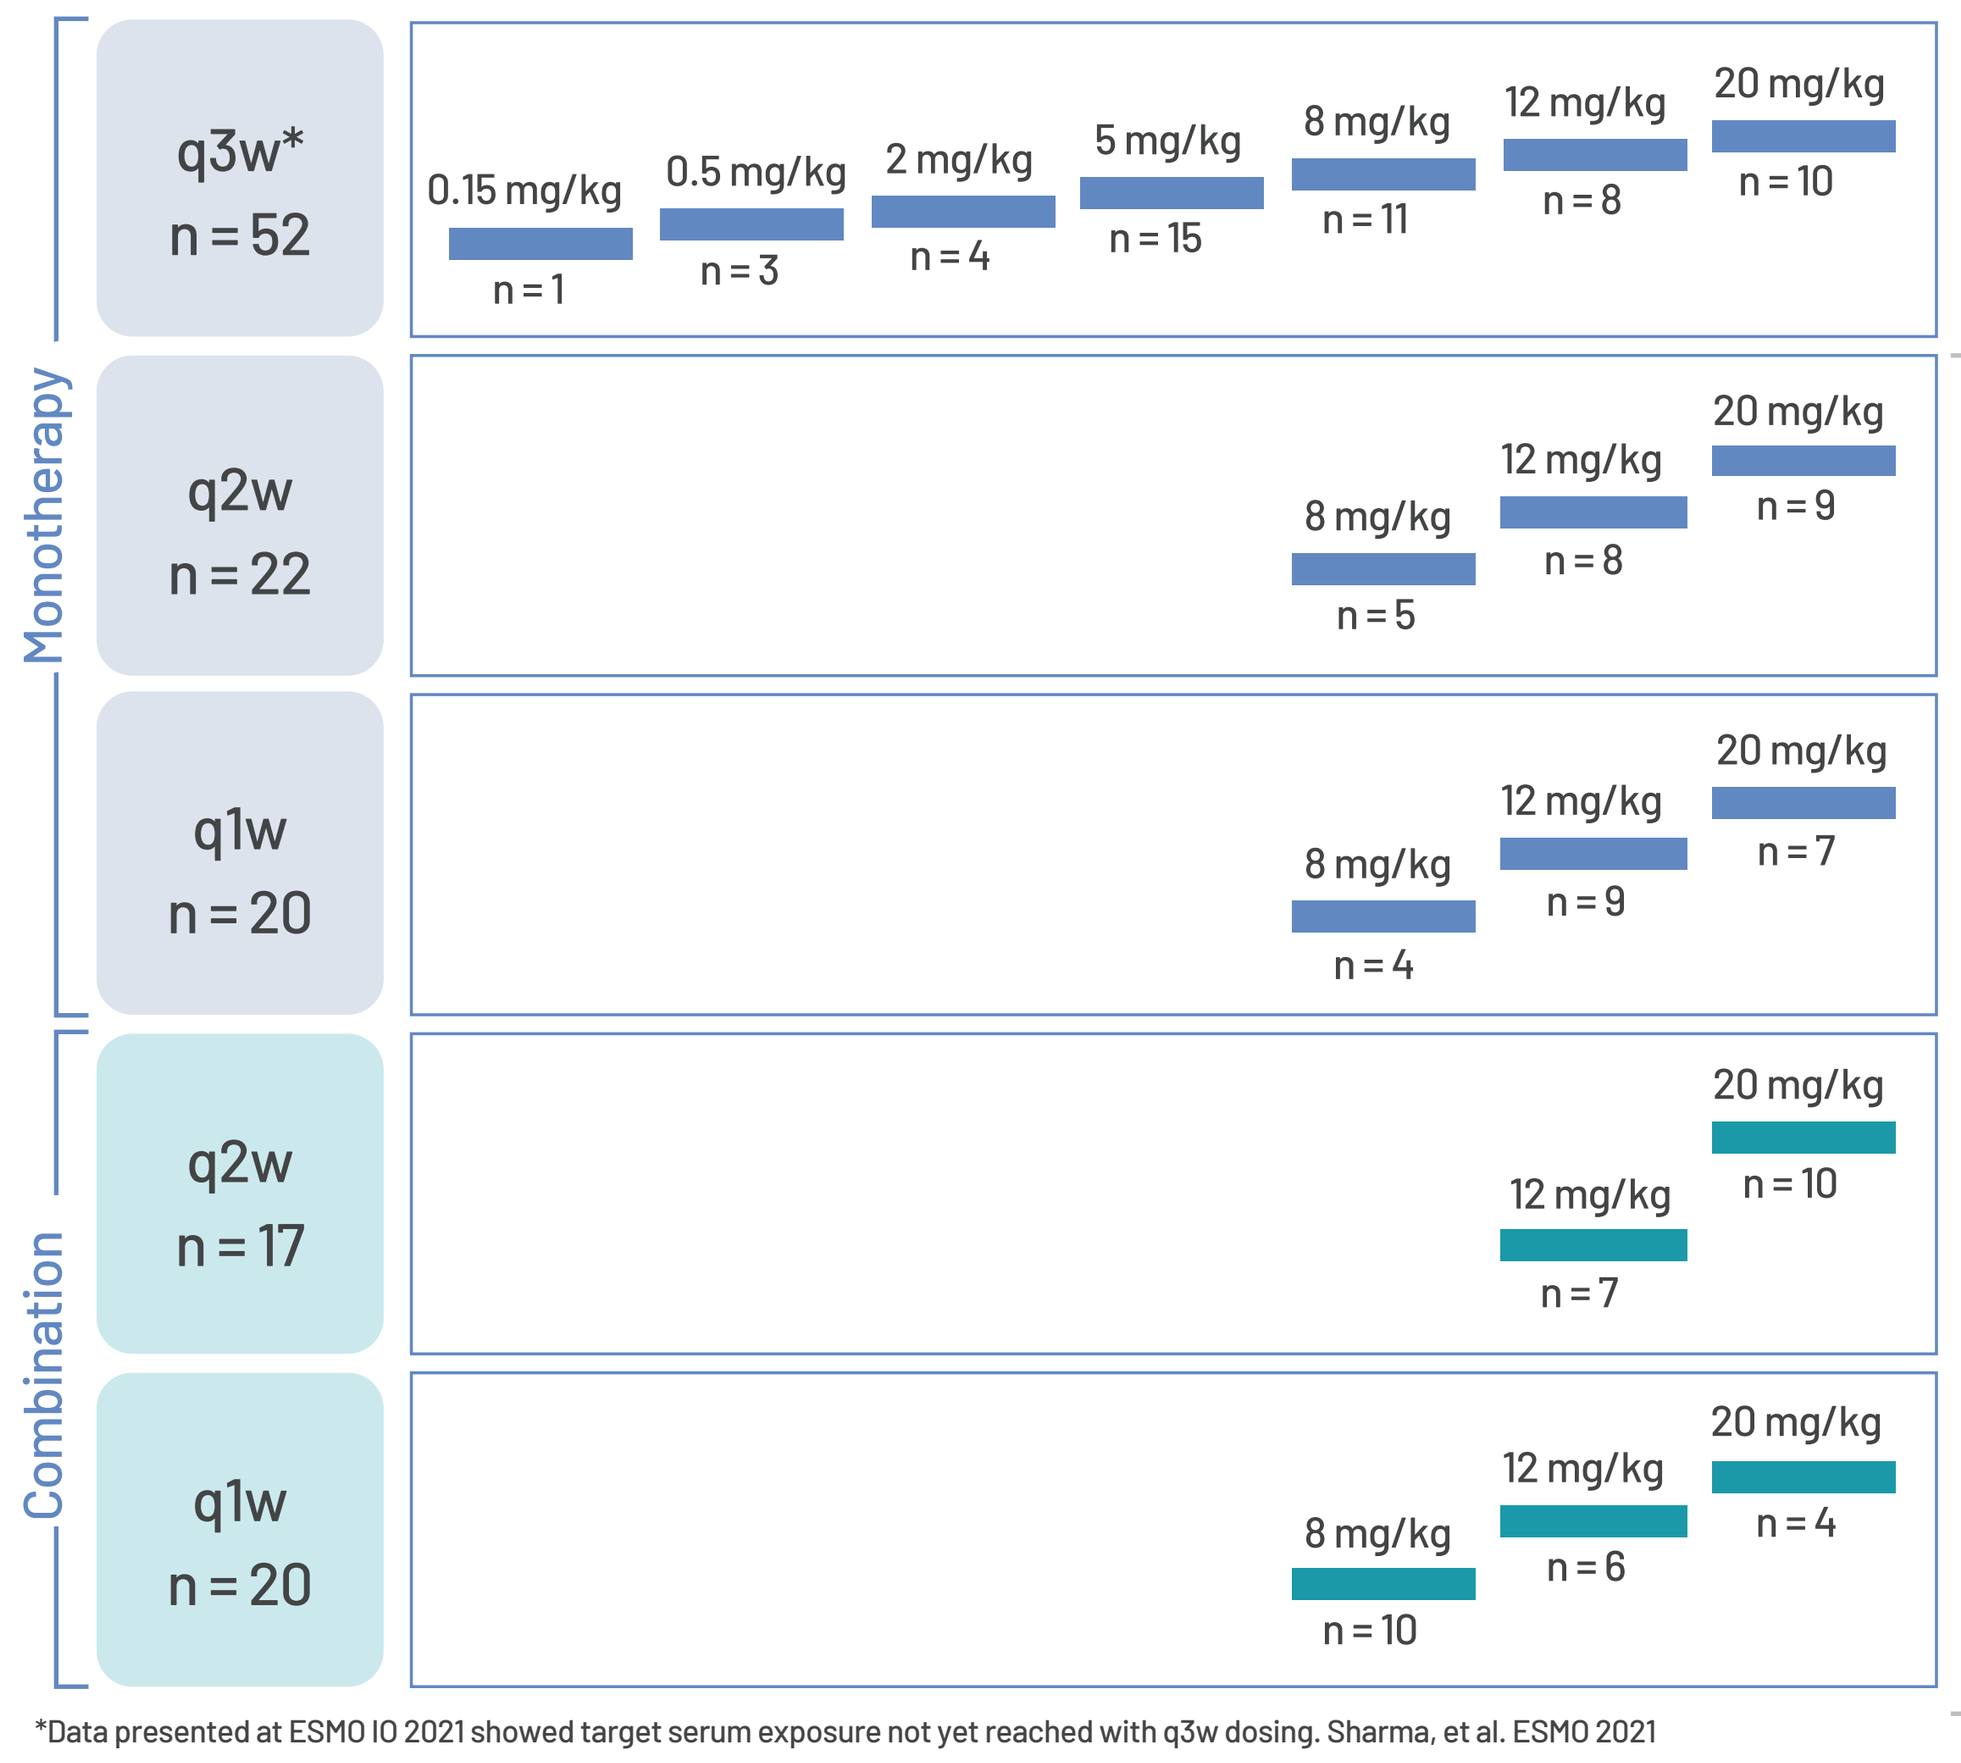

Monotherapy
q3w* n = 52
0.15 mg/kg n = 1
0.5 mg/kg n = 3
2 mg/kg n = 4
5 mg/kg n = 15
8 mg/kg n = 11
12 mg/kg n = 8
20 mg/kg n = 10
q2w n = 22
8 mg/kg n = 5
12 mg/kg n = 8
20 mg/kg n = 9
q1w n = 20
8 mg/kg n = 4
12 mg/kg n = 9
20 mg/kg n = 7
Combination
q2w n = 17
12 mg/kg n = 7
20 mg/kg n = 10
q1w n = 20
8 mg/kg n = 10
12 mg/kg n = 6
20 mg/kg n = 4
Enrollment After December 2021

*Data presented at ESMO IO 2021 showed target serum exposure not yet reached with q3w dosing. Sharma, et al. ESMO 2021

- Preclinical data demonstrate that a target exposure of ≥ 10 μg/mL in serum was required for optimal efficacy
- Protocol amendment (December 2021) added q2w and q1w cohorts of BDC-1001 as a single agent and in combination with nivolumab to achieve target safe exposure not reached with q3w
- Dose escalation now completed (Feb 2023)

RESULTS

Demographics and Baseline Characteristics

Heterogenous and Heavily Pretreated Patient Population with 16 Different Tumor Types

Majority of Patients Had HER2+ Tumors and Prior Anti-HER2 Therapy

		BDC-1001 Monotherapy				BDC-1001 + Nivolumab			All Patients
		q3w n = 52	q2w n = 22	q1w n = 20	Total n = 94	q2w n = 17	q1w n = 20	Total n = 37	Total n = 131
Median age, years (range)		64.0 (30, 84)	62.5 (42, 80)	63.0 (33, 85)	64.0 (30, 85)	65.0 (34, 71)	55.0 (31, 81)	57.0 (31, 81)	62.0 (30, 85)
Sex, n (%):	Female	33 (63.5)	12 (54.5)	11 (55.0)	56 (59.6)	13 (76.5)	14 (70.0)	27 (73.0)	83 (63.4)
	Male	19 (36.5)	10 (45.5)	9 (45.0)	38 (40.4)	4 (23.5)	6 (30.0)	10 (27.0)	48 (36.6)
ECOG, n (%):	0	16 (30.8)	5 (22.7)	8 (40.0)	29 (30.9)	7 (41.2)	10 (50.0)	17 (45.9)	46 (35.1)
	1	36 (69.2)	17 (77.3)	12 (60.0)	65 (69.1)	10 (58.8)	10 (50.0)	20 (54.1)	85 (64.9)
Prior lines of systemic treatment, median (range)		4 (0, 12)	3 (1, 11)	4 (1, 9)	4 (0, 12)	5 (1, 10)	5 (2, 13)	5 (1, 13)	4 (0, 13)
Prior anti-HER2 therapy, n (%)		43 (82.7)	8 (36.4)	11 (55.0)	62 (66.0)	12 (70.6)	16 (80.0)	28 (75.7)	90 (68.7)
Prior immune therapy, n (%)		16 (30.8)	5 (22.7)	8 (40.0)	29 (30.9)	4 (23.5)	5 (25.0)	9 (24.3)	38 (29.0)
HER2 categories from screening, n (%):	HER2+ (IHC3+ or gene amplification)	51 (98.1)	18 (81.8)	16 (80.0)	85 (90.4)	15 (88.2)	18 (90.0)	33 (89.2)	118 (90.1)
	HER2 low (IHC2+ and no gene amplification)	1 (1.9)	4 (18.2)	4 (20.0)	9 (9.6)	2 (11.8)	2 (10.0)	4 (10.8)	13 (9.9)
Tumor types, n (%):	Colorectal	10 (19.2)	10 (45.5)	4 (20.0)	24 (25.5)	3 (17.6)	7 (35.0)	10 (27.0)	34 (26.0)
	Gastroesophageal	16 (30.8)	4 (18.2)	4 (20.0)	24 (25.5)	2 (11.8)	2 (10.0)	4 (10.8)	28 (21.4)
	Breast	9 (17.3)	1 (4.5)	5 (25.0)	15 (16.0)	2 (11.8)	8 (40.0)	10 (27.0)	25 (19.1)
	Endometrial	6 (11.5)	0 (0.0)	1 (5.0)	7 (7.4)	2 (11.8)	1 (5.0)	3 (8.1)	10 (7.6)
	Others*	11 (21.2)	7 (31.8)	6 (30.0)	24 (25.5)	8 (47.0)	2 (10.0)	10 (27.0)	34 (26.0)

*Other tumor types include (monotherapy and combination combined): n=6 ovary, n=5 salivary gland, n=4 cervix, n=4 lung, n=4 pancreatic, n=2 biliary tract, n=2 skin, n=2 small intestine, and one each of head and neck, intestinal ampulla, liver, prostate, and urinary bladder.

Safety

BDC-1001 was Well Tolerated Up to 20 mg/kg q1w Monotherapy and in Combination with Nivolumab

- BDC-1001 has a wide therapeutic window, up to 20 mg/kg q1w with maximum-tolerated dose (MTD) not reached
 - One DLT of supraventricular tachycardia (grade 3) at 8 mg/kg BDC-1001 q1w in combination with nivolumab
 - One grade 4 and no grade 5 drug-related AEs
- Most frequent (29.0%) drug-related AEs were low grade (grade 1 and grade 2) infusion-related reactions (IRRs)
- One drug-related cytokine release syndrome (grade 1) at 12 mg/kg BDC-1001 q1w
- Left ventricular ejection fraction (LVEF) decrease
 - 6 patients with ejection fraction decrease (grade 2 [n=4], grade 3 [n=2])
 - 4 received BDC-1001 q1w
 - Monotherapy: 1 patient at 12 mg/kg, 2 at 20 mg/kg; combination: 1 at 8 mg/kg + nivolumab
 - 2 received BDC-1001 q3w or q2w
 - Monotherapy: 1 patient at 5 mg/kg q3w and 1 at 8 mg/kg q2w
- 2 patients discontinued therapy due to LVEF decrease
 - 5 mg/kg BDC-1001 q3w, 8 mg/kg BDC-1001 q2w

Safety graded by CTCAE v5; AE, adverse event; DLT, dose limiting toxicity

Details of Safety Profile of BDC-1001 Monotherapy and in Combination with Nivolumab

Summary of Treatment-related TEAEs

	BDC-1001 Monotherapy				BDC-1001 + Nivolumab					
	Treatment-related TEAEs				BDC-1001 Treatment-related TEAEs			BDC-1001 + Nivolumab Treatment-related TEAEs		
	q3w n = 52	q2w n = 22	q1w n = 20	Total n = 94	q2w n = 17	q1w n = 20	Total n = 37	q2w n = 17	q1w n = 20	Total n = 37
All grades (%)	30 (57.7)	11 (50.0)	17 (85.0)	58 (61.7)	11 (64.7)	14 (70.0)	25 (67.6)	5 (29.4)	12 (60.0)	17 (45.9)
Grade ≥ 3 (%)	5 (9.6)	1 (4.5)	1 (5.0)	7 (7.4)	0	2 (10.0)	2 (5.4)	0	1 (5.0)	1 (2.7)
Serious adverse events (%)	3 (5.8)	0	0	3 (3.2)	1 (5.9)	1 (5.0)	2 (5.4)	0	1 (5.0)	1 (2.7)
Leading to treatment discontinuation	3 (5.8)	1 (4.5)	0	4 (4.3)	0	1 (5.0)	1 (2.7)	0	1 (5.0)	1 (2.7)
Leading to treatment interruption	5 (9.6)	2 (9.1)	2 (10.0)	9 (9.6)	1 (5.9)	1 (5.0)	2 (5.4)	0	1 (5.0)	1 (2.7)
Leading to death	0	0	0	0	0	0	0	0	0	0

Safety graded by CTCAE v5; TEAE, treatment-emergent adverse event
Definition of treatment-related TEAEs = an AE considered as related to with unknown/missing relationship to study drug

Data cut-off: March 24, 2023

Pharmacokinetics of BDC-1001

Serum Target Exposure > 10 μg/mL Achieved with q2w and q1w Dosing, Not with q3w

- Population mean clearance 1.6 L/day and terminal $T_{½}$ of 4.3 days
 - Faster clearance at dose levels lower than 5 mg/kg exhibits TMDD (target-mediated drug disposition)
- Two-fold accumulation observed in median C_{min} with q2w and q1w dosing
 - At 20 mg/kg q2w, C_{min} increase from first dose to steady state: ~ 12 to 29 μg/mL
 - At 20 mg/kg q1w, C_{min} increase from first dose to steady state: ~ 34 to 68 μg/mL
 - Virtually no accumulation was observed with q3w dosing
- Presence of nivolumab did not impact PK of BDC-1001
- Low incidence of BDC-1001 ADA formation (4.2%) with no impact on PK, safety, or efficacy

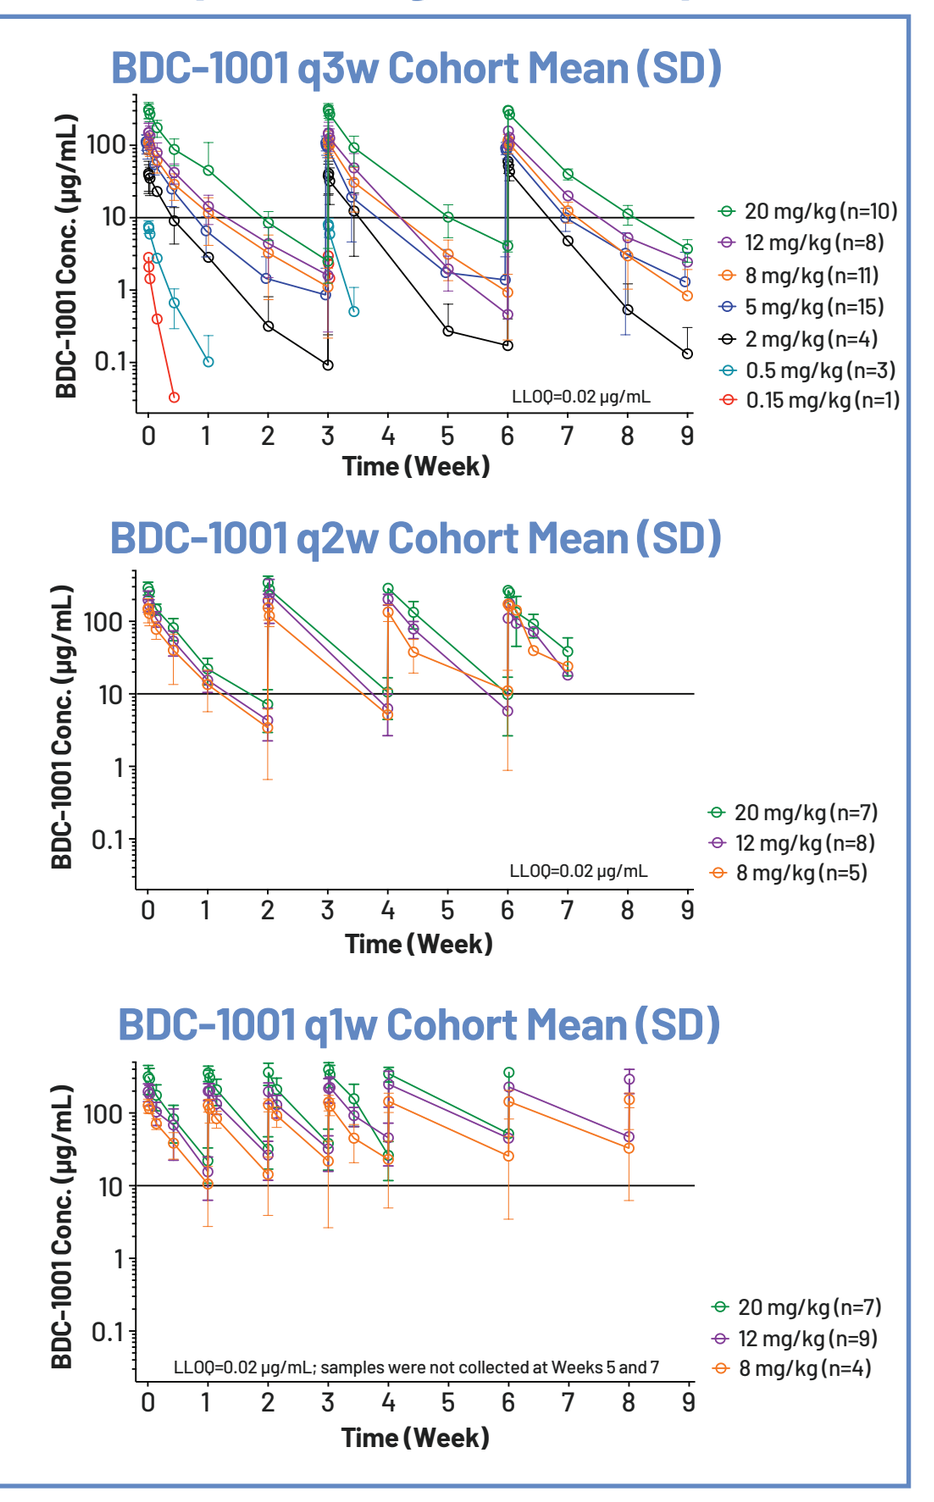

BDC-1001 q3w Cohort Mean (SD)
BDC-1001 Conc. (μg/mL)
Time (Week)
LLOQ=0.02 μg/mL
20 mg/kg (n=10)
12 mg/kg (n=8)
8 mg/kg (n=11)
5 mg/kg (n=15)
2 mg/kg (n=4)
0.5 mg/kg (n=3)
0.15 mg/kg (n=1)
BDC-1001 q2w Cohort Mean (SD)
20 mg/kg (n=7)
12 mg/kg (n=8)
8 mg/kg (n=5)
BDC-1001 q1w Cohort Mean (SD)
LLOQ=0.02 μg/mL; samples were not collected at Weeks 5 and 7
20 mg/kg (n=7)
12 mg/kg (n=9)
8 mg/kg (n=4)

ADA, anti-drug antibody; SD, standard deviation
Note: q1w samples were not collected at Weeks 5 and 7

Efficacy

Most Clinically Meaningful Efficacy Observed at 20 mg/kg q2w (RP2D)

- Six patients had PRs
 - 3 colorectal, 1 ovarian, 1 biliary, 1 salivary
 - 4 at 20 mg/kg q2w (2 mono, 2 combo)
 - 3 were MSS (mono or combo) and 1 was MSI (mono)
 - 1 at 12 mg/kg q1w (combo) in MSS tumor
 - 1 at 5 mg/kg q3w (mono) in MSS tumor
- Twelve patients had SD ≥ 24 weeks
 - 4 colorectal, 1 melanoma, 1 endometrial, 2 gastric, 1 salivary gland, 2 cervical, 1 ovarian
 - 3 of 12 at 20 mg/kg q2w with colorectal, salivary gland, and ovarian cancer
- Tumor shrinkage observed in a variety of tumor types including biliary, breast, cervical, colorectal, endometrial, gastric, lung, salivary, skin (melanoma), and ovarian cancer

Clinical Efficacy in All Patients with HER2+ Tumors Treated with 20 mg/kg q2w (RP2D)

BDC-1001 Monotherapy or in Combination with Nivolumab

		BDC-1001 20 mg/kg (n = 7) 4 Tumor Types	BDC-1001 20 mg/kg + Nivolumab (n = 8)** 5 Tumor Types	All (n = 15) 7 Tumor Types
Response assessment, n (%):	PR	2* (29%)	2 (25%)	4 (27%)
	SD	3 (43%)	4 (50%)	7 (47%)
	PD	2 (29%)	1 (13%)	3 (20%)
	Not evaluable	0	1 (13%)	1 (7%)
Overall response rate, n (%)		2 (29%)	2 (25%)	4 (27%)
Disease control rate ≥ 6 weeks, n (%)		5 (71%)	6 (75%)	11 (73%)
Disease control rate ≥ 24 weeks, n (%)		3 (43%)	4 (50%)	7 (47%)
Tumor shrinkage, n (%)		4 (57%)	5 (63%)	9 (60%)

*One PR confirmed post March 24, 2023 data cutoff; **One non-evaluable patient included
4 HER2-low tumors (2 each from BDC-1001 monotherapy and in combination) are excluded.

Data cut-off: March 24, 2023

Clinical Efficacy in All Patients with HER2+ Tumors Was Greater with 20 mg/kg Compared to 12 mg/kg q2w

Data fairly comparable for BDC-1001 Monotherapy or in Combination with Nivolumab

		BDC-1001		BDC-1001 + Nivolumab	
		12 mg/kg (n = 7) 5 Tumor Types	20 mg/kg (n = 7) 4 Tumor Types	BDC-1001 12 mg/kg (n = 7) 6 Tumor Types	BDC-1001 20 mg/kg (n = 8)** 5 Tumor Types
Response assessment, n (%):	PR	0	2* (29%)	0	2 (25%)
	SD	1 (14%)	3 (43%)	2 (29%)	4 (50%)
	PD	4 (57%)	2 (29%)	4 (57%)	1 (13%)
	Not evaluable	2 (29%)	0	1 (14%)	1 (13%)
Overall response rate, n (%)		0	2 (29%)	0	2 (25%)
Disease control rate ≥ 6 weeks, n (%)		1 (14%)	5 (71%)	2 (29%)	6 (75%)
Disease control rate ≥ 24 weeks, n (%)		1 (14%)	3 (43%)	0	4 (50%)
Tumor shrinkage, n (%)		1 (14%)	4 (57%)	2 (29%)	5 (63%)

* One PR confirmed post March 24, 2023 data cutoff; **One non-evaluable patient included
5 HER2-low tumors are excluded (1 at 12 mg/kg and 4 at 20 mg/kg)

Data cut-off: March 24, 2023

Meaningful Anti-tumor Activity in Evaluable Heterogeneous HER2+ Tumor Population at 20 mg/kg q2w (RP2D)

BDC-1001 Monotherapy and Combination with Nivolumab

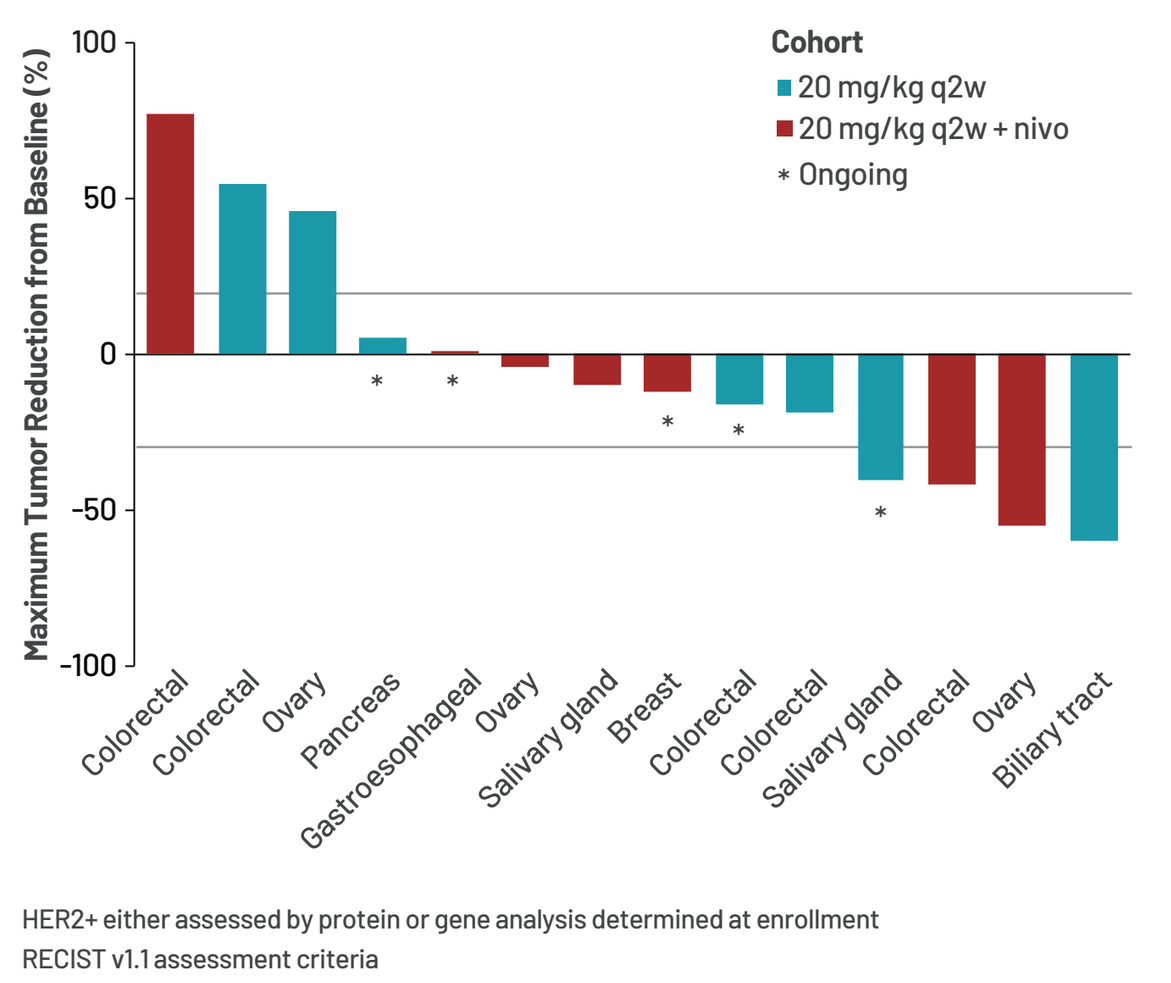

Maximum Tumor Reduction from Baseline (%)
Cohort
20 mg/kg q2w
20 mg/kg q2w + nivo
* Ongoing
Colorectal
Colorectal
Ovary
Pancreas
Gastroesophageal
Ovary
Salivary gland
Breast
Colorectal
Colorectal
Salivary gland
Colorectal
Ovary
Biliary tract

HER2+ either assessed by protein or gene analysis determined at enrollment
RECIST v1.1 assessment criteria

Monotherapy (n=7)

- 29% achieved PR
- 43% had disease control ≥24w
- 57% achieved tumor shrinkage
 - Tumor types: colorectal, salivary gland, and biliary tract

Combination with Nivolumab (n=7)

- 29% achieved PR
- 57% had disease control ≥24w
- 71% achieved tumor shrinkage
 - Tumor types: breast, colorectal, ovary, and salivary gland

Data cut-off: March 24, 2023

Clinical Activity: 6 PRs and 12 Long-lasting SDs (≥ 24 Weeks) Observed in 8 Tumor Types, Particularly in 20 mg/kg q2w Dose Cohorts

Best Response	Site of Primary Tumor, HER2 Status	Duration of Disease Control (PR or SD) in Wks	Prior Lines of Therapy	Prior Anti-HER2 Therapy	Prior Checkpoint Inhibitor	MSS/MSI	Dose Cohort
Partial Response	Colorectal, HER2+	84	4	No	Yes	MSS	5 mg/kg q3w
	Biliary tract cancer, HER2+	**36**	**2**	**No**	**No**	**MSS**	**20 mg/kg q2w**
	Salivary gland, HER2+	**48+**	**2**	**No**	**No**	**MSI**	**20 mg/kg q2w**
	Ovarian cancer, HER2+	**24**	**12**	**Yes**	**No**	**MSS**	**20 mg/kg q2w + nivolumab**
	Colorectal, HER2+	**48**	**5**	**Yes**	**No**	**MSS**	**20 mg/kg q2w + nivolumab**
	Colorectal, HER2+	12+	5	Yes	No	MSS	12 mg/kg q1w + nivolumab
Durable Stable Disease	Endometrial cancer, HER2+	36	3	Yes	No	No data	2 mg q3w
	Cervical cancer, HER2+	60	3	Yes	No	No data	5 mg/kg q3w
	Melanoma, HER2+	24	1	No	Yes	MSS	8 mg/kg q3w
	Colorectal, HER2+	36	11	Yes	No	MSS	20 mg/kg q3w
	Colorectal, HER2+	24+	2	No	No	MSS	8 mg/kg q2w
	Gastric cancer, HER2+	48+	2	Yes	No	No data	12 mg/kg q2w
	Colorectal, HER2+	**60+**	**2**	**No**	**No**	**MSI**	**20 mg/kg q2w**
	Salivary gland cancer, HER2+	**24**	**8**	**Yes**	**Yes**	**MSS**	**20 mg/kg q2w + nivolumab**
	Ovarian cancer, HER2+	**36**	**4**	**Yes**	**No**	**MSI**	**20 mg/kg q2w + nivolumab**
	Colorectal, HER2+	36	1	No	No	MSS	8 mg/kg q1w
	Cervical cancer, HER2+	24	5	Yes	Yes	MSS	12 mg/kg q1w
	Gastric cancer, HER2+	24	2	Yes	No	No data	12 mg/kg q1w

Bold: patients treated at RP2D

Data cut-off: March 24, 2023

Waterfall Plots for All Patients (HER2+ and HER2-Low Tumors) Treated q3w, q2w, q1w

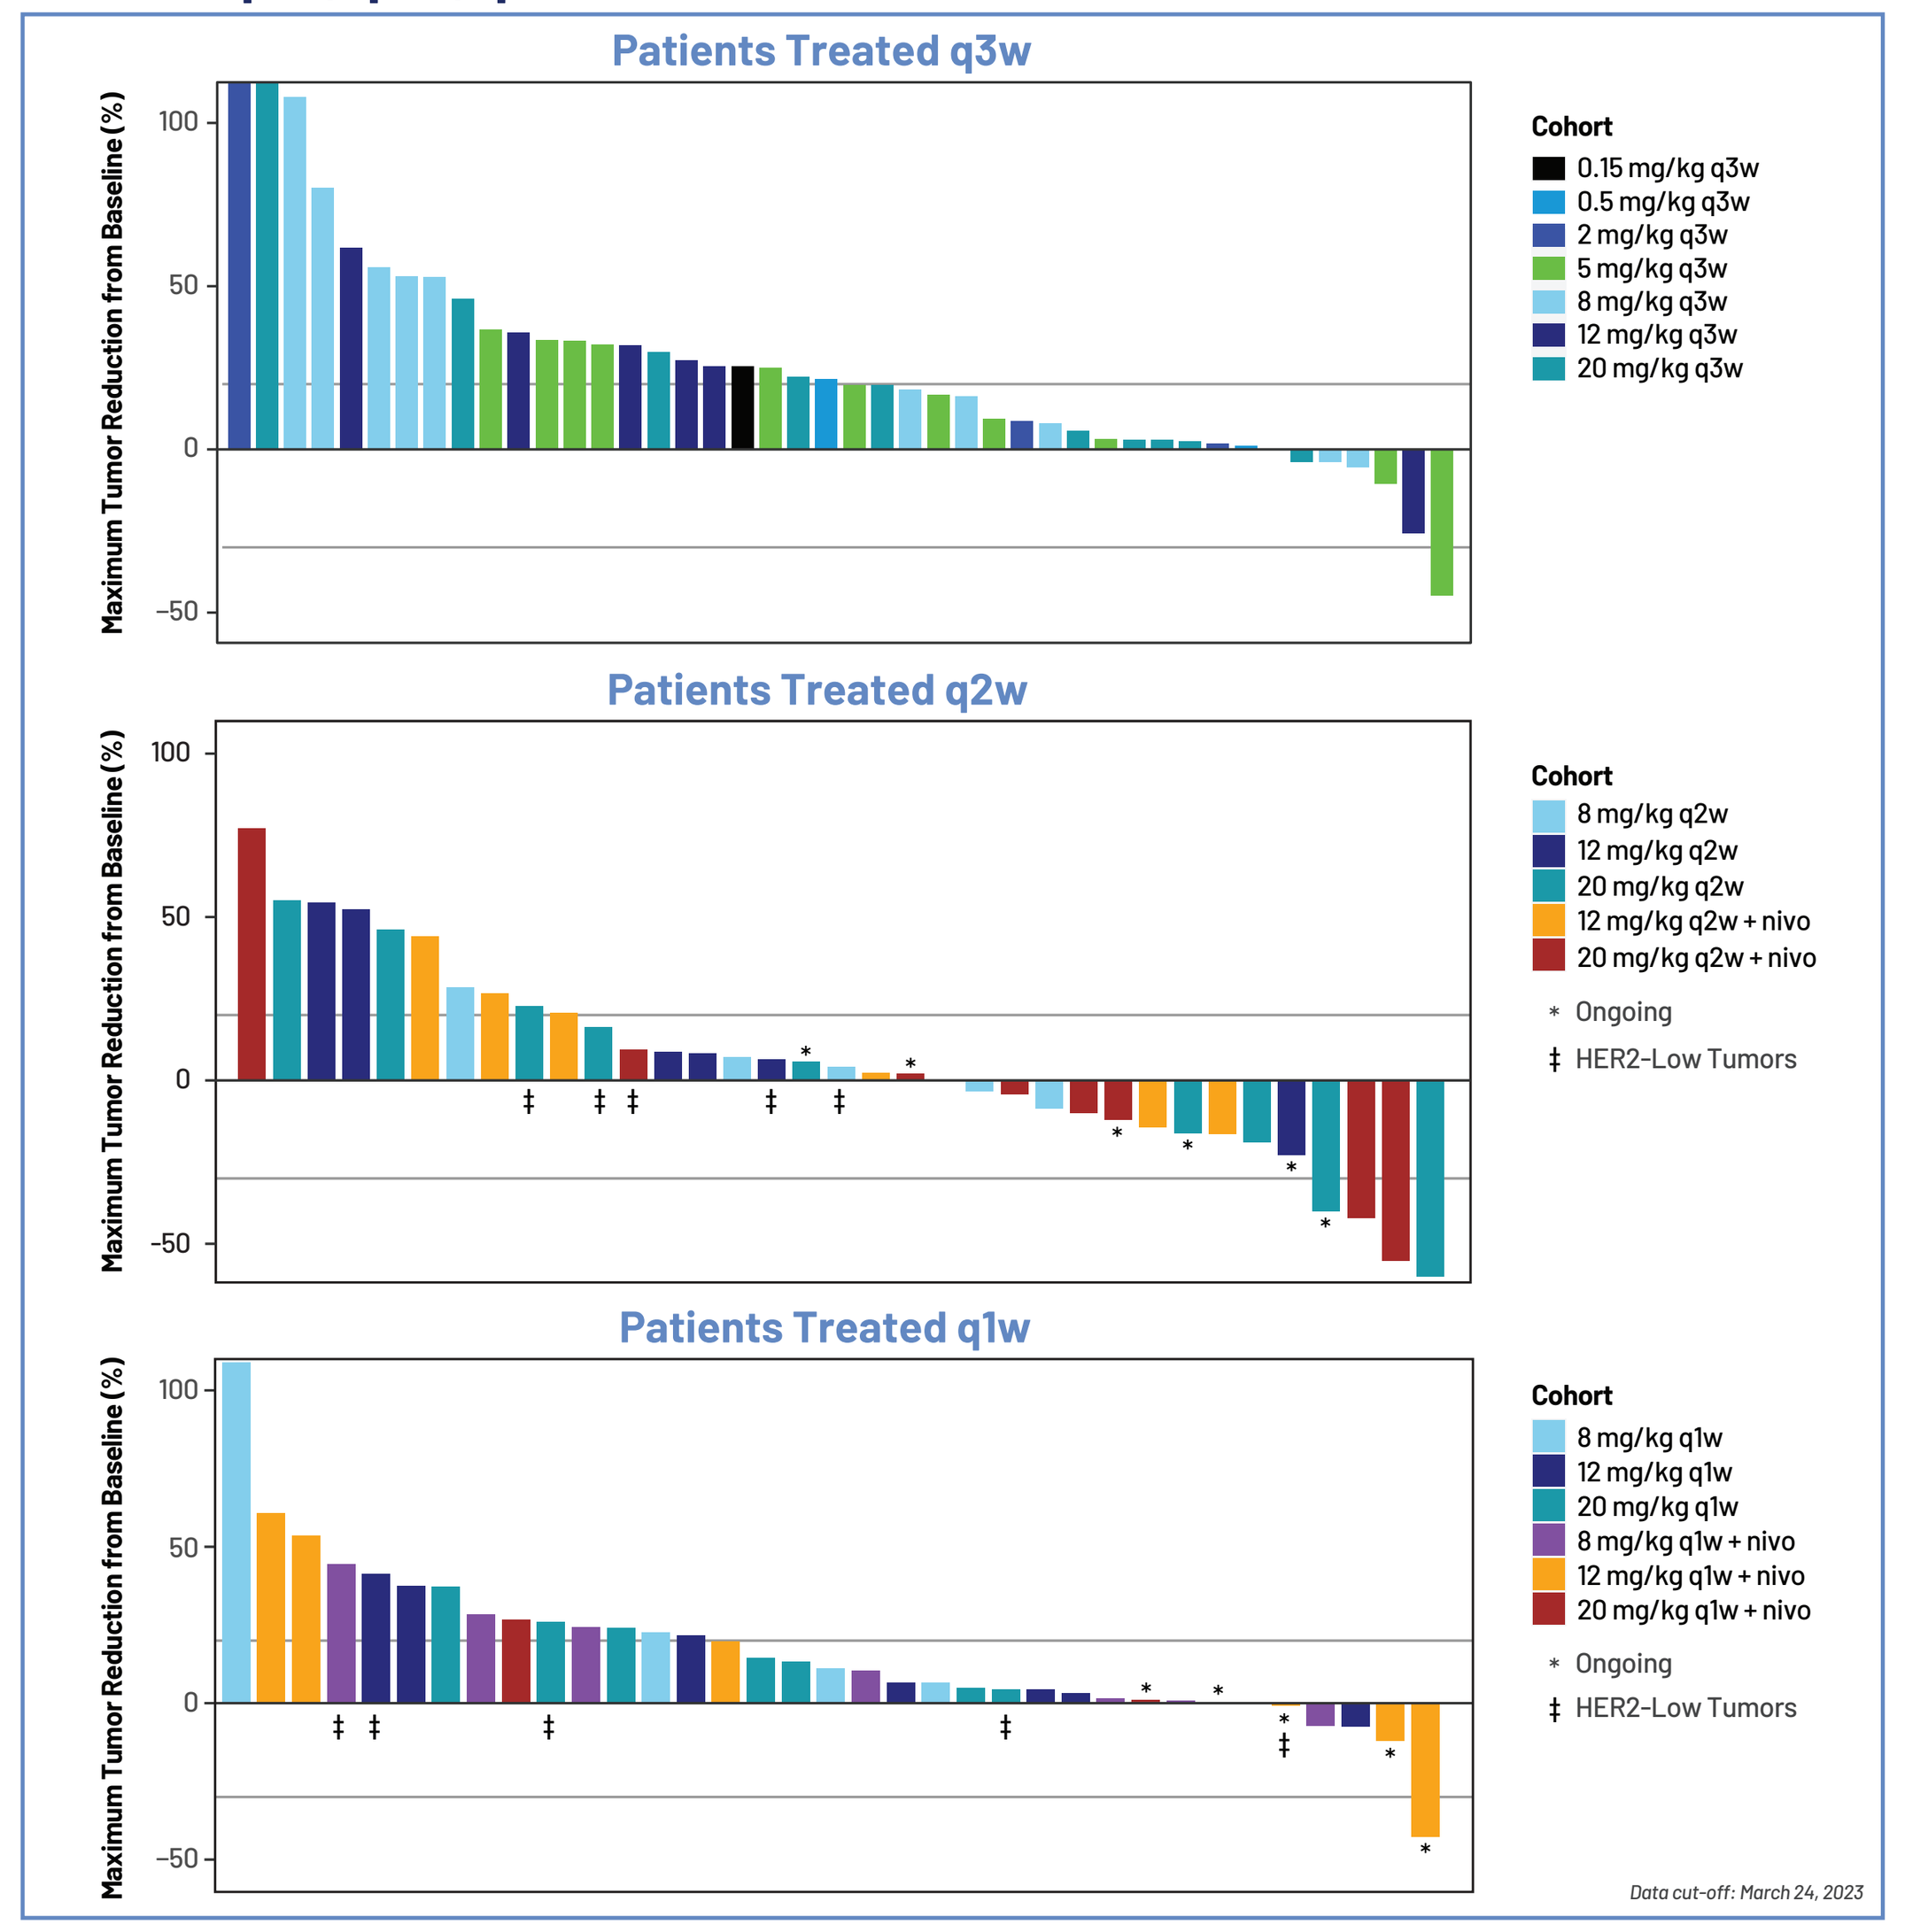

Patients Treated q3w
Maximum Tumor Reduction from Baseline (%)
Cohort
0.15 mg/kg q3w
0.5 mg/kg q3w
2 mg/kg q3w
5 mg/kg q3w
8 mg/kg q3w
12 mg/kg q3w
20 mg/kg q3w
Patients Treated q2w
Cohort
8 mg/kg q2w
12 mg/kg q2w
20 mg/kg q2w
12 mg/kg q2w + nivo
20 mg/kg q2w + nivo
* Ongoing
‡ HER2-Low Tumors
Patients Treated q1w
Cohort
8 mg/kg q1w
12 mg/kg q1w
20 mg/kg q1w
8 mg/kg q1w + nivo
12 mg/kg q1w + nivo
20 mg/kg q1w + nivo
* Ongoing
‡ HER2-Low Tumors

Data cut-off: March 24, 2023

Peak Increase in Plasma Myeloid Activation Markers at 4 Hours

Confirms MOA and Safety Profile

- Plasma samples for cytokines and chemokines obtained from all patients
- Dose-dependent peak increases in Cycle 1 were observed in multiple cytokines and chemokines*
 - Similar responses observed for MIP-1α, IFNγ, TNFα and eotaxin

Average IL-6 levels were low at all doses (< 50 pg/mL)

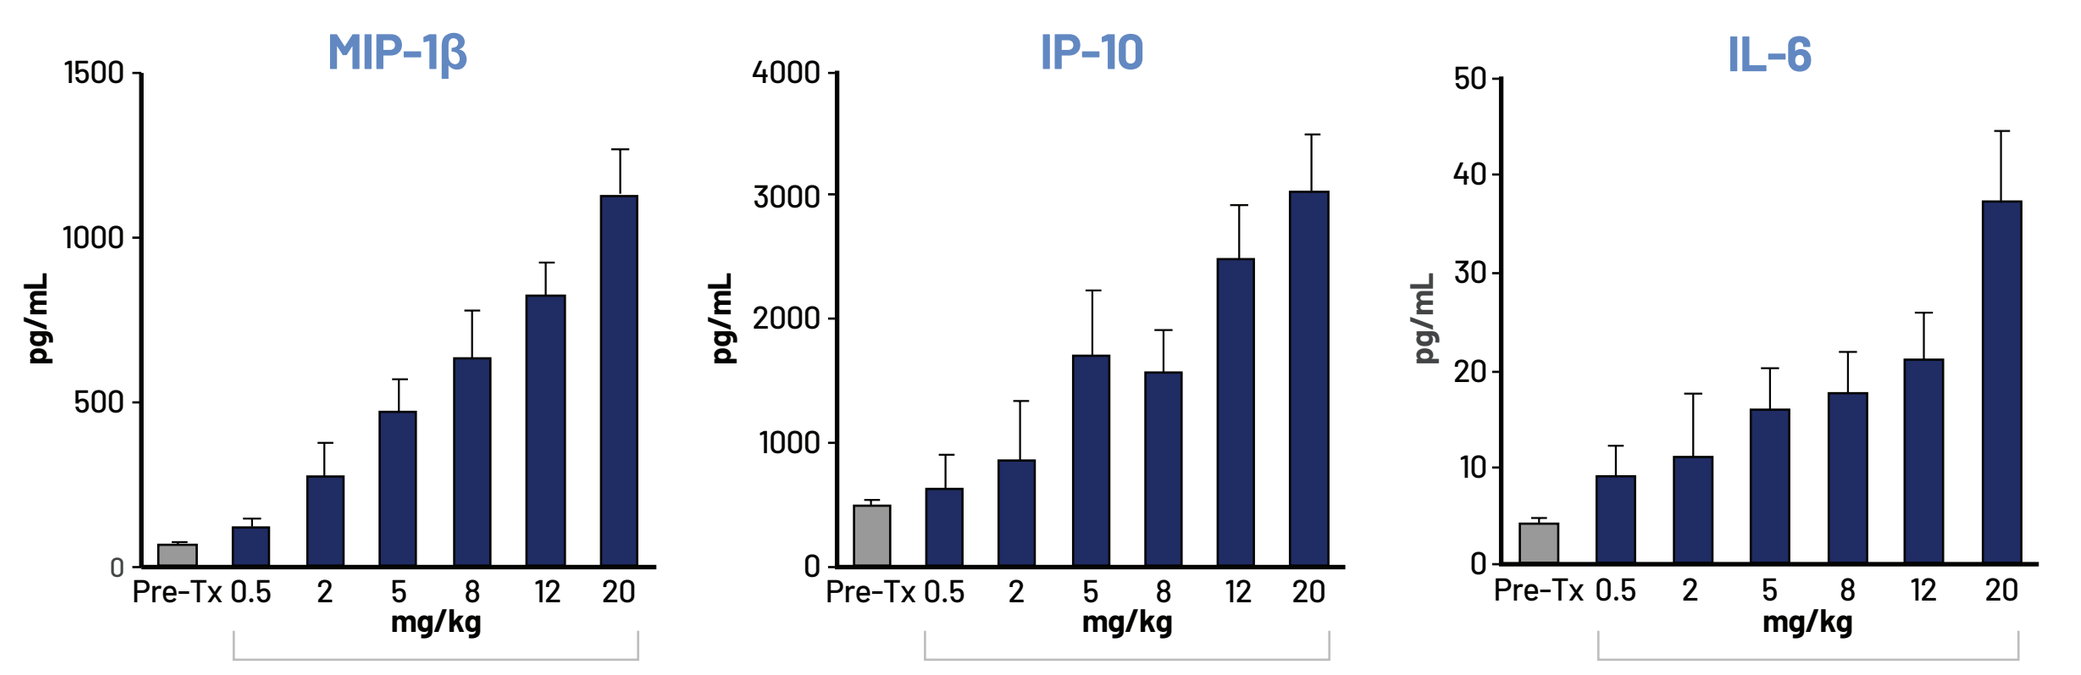

MIP-1β
IP-10
IL-6
pg/mL
Pre-Tx 0.5 2 5 8 12 20
mg/kg

*Representative graphs are shown

Data cut-off: March 24, 2023

BDC-1001 Drives Myeloid and T Cell Infiltration in HER2+ Tumors

Data from Paired Fresh Tumor Biopsies

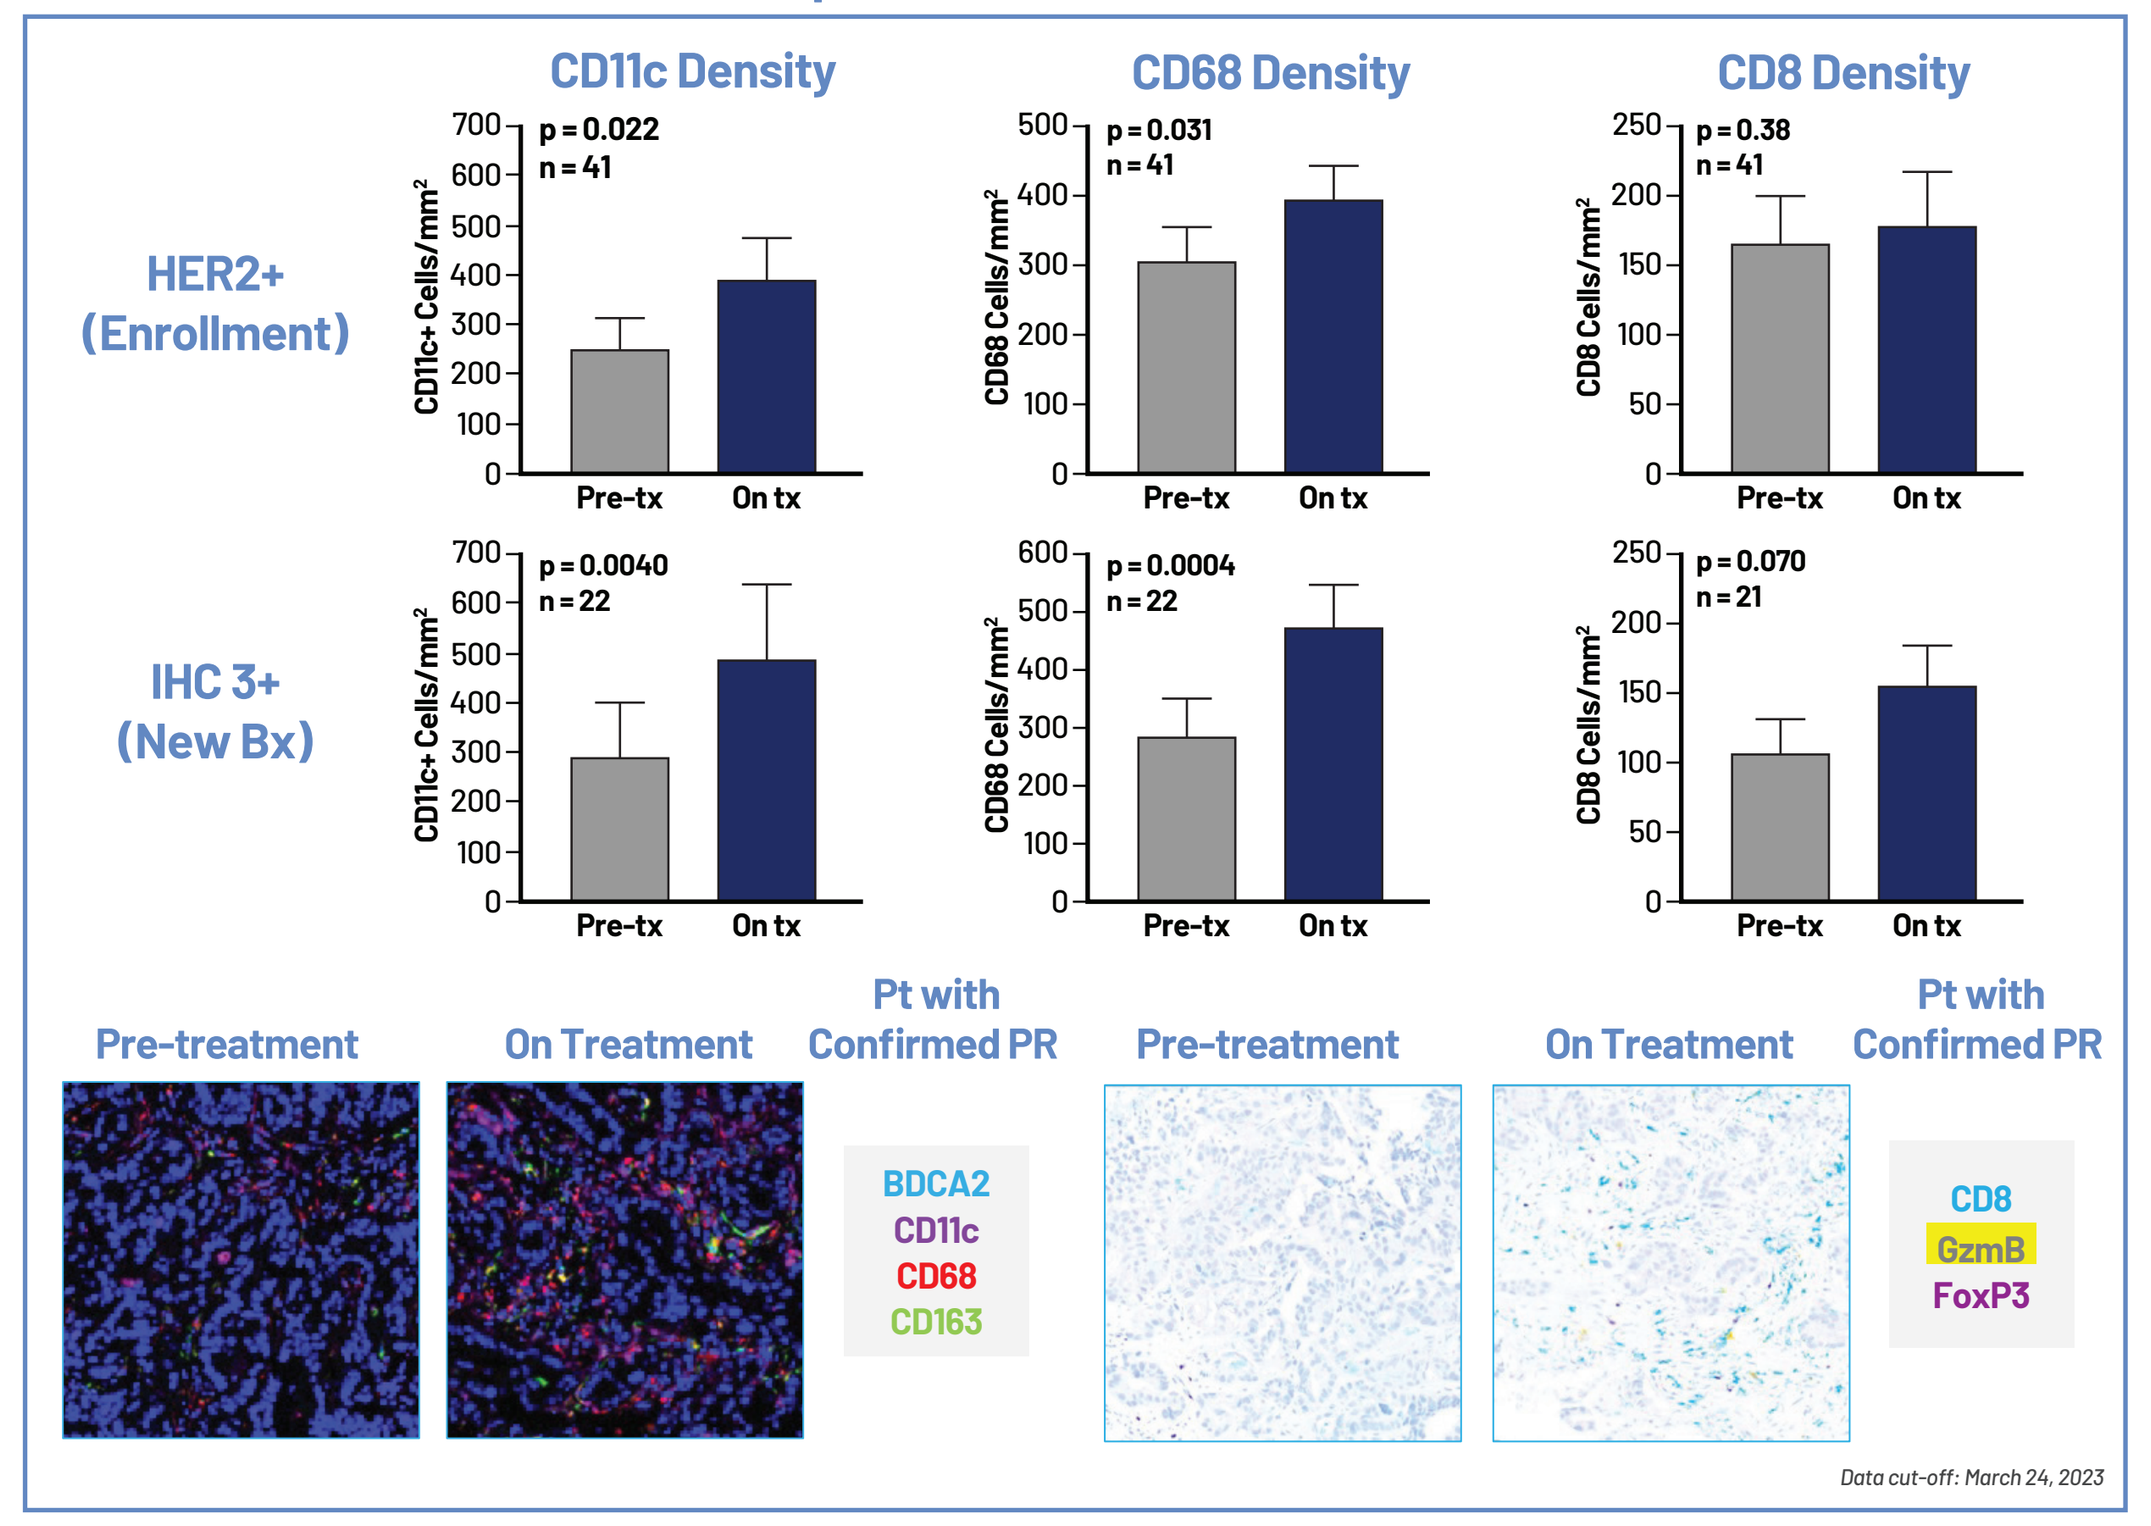

CD11c Density
CD68 Density
CD8 Density
HER2+ (Enrollment)
p = 0.022 n = 41
p = 0.031 n = 41
p = 0.38 n = 41
IHC 3+ (New Bx)
p = 0.0040 n = 22
p = 0.0004 n = 22
p = 0.070 n = 21
CD11c+ Cells/mm²
CD68 Cells/mm²
CD8 Cells/mm²
Pre-tx
On tx
Pre-treatment
On Treatment
Pt with Confirmed PR
BDCA2
CD11c
CD68
CD163
CD8
GzmB
FoxP3

Data cut-off: March 24, 2023

CT Imaging and Biomarker Data in a Patient with PR

Patient with HER2+ by NGS, MSS biliary tract carcinoma
- No prior anti-HER2 or immunotherapy

Fresh matched (pre- and on-Tx) biomarker data

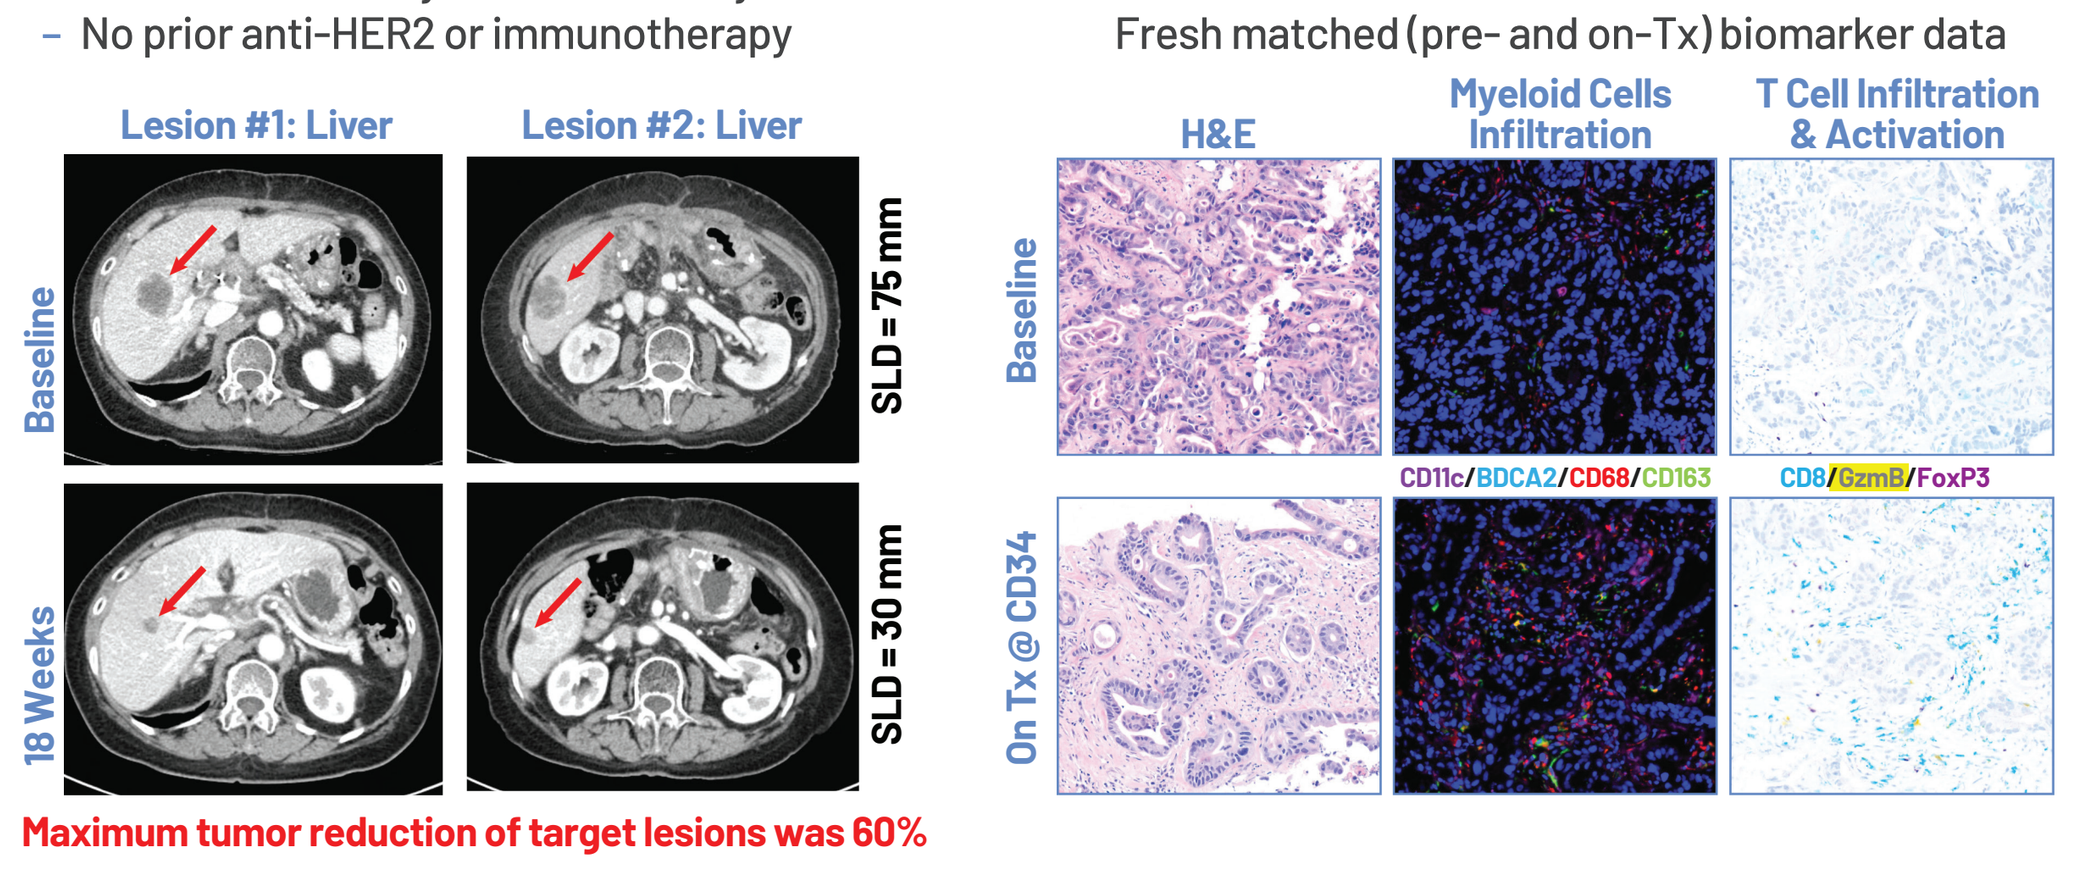

Lesion #1: Liver
Lesion #2: Liver
Baseline
18 Weeks
SLD = 75 mm
SLD = 30 mm
H&E
Myeloid Cells Infiltration
T Cell Infiltration & Activation
Baseline
On Tx @ CD34
CD11c/BDCA2/CD68/CD163
CD8/GzmB/FoxP3

Maximum tumor reduction of target lesions was 60%

Key observations:

- cDC (CD11c) increased by 16% and pDC (BDCA2) decreased by 70%
- 62% increase in M1 (CD68+CD163-) macrophage, 161% increase in monocyte-derived DCs (CD11c+CD163+), and 16% increase in cDC (CD11c+CD163-)
- 500% increase in CD8+ T cell infiltration and 400% increase in CD8+Granzyme B+ T cell activation

CT= Computerized tomography; SLD=Sum of Longest Diameter

Data cut-off: March 24, 2023

CONCLUSIONS

Results demonstrate encouraging evidence of safety, anti-tumor efficacy, and biomarker changes consistent with MoA of ISAC technology

- BDC-1001 was well-tolerated at all doses and dosing frequencies up to 20 mg/kg q1w
 - In a heterogeneous (16 different tumor types in 18 cohorts) and heavily pretreated (median 4 prior lines of systemic treatment) patient population
- Target exposure established in preclinical models achieved at higher dose and increased frequency of administration
 - C_{min} above 10 μg/mL achieved at q2w and q1w schedules
 - Improved efficacy observed with q2w compared to q3w or q1w
- Clinical activity of BDC-1001 observed alone and in combination with nivolumab, particularly in the 20 mg/kg q2w cohorts
- Pharmacodynamic responses in both plasma and tissue consistent with ISAC MOA
 - Responses of myeloid and T cell activation and infiltration not anticipated with trastuzumab treatment alone
- Selection of 20 mg/kg q2w as RP2D based on the totality of safety, efficacy, PK, and biomarkers
- Results support Phase 2 development of BDC-1001 as a single agent and in combination strategies

FUTURE DIRECTION

Two multi-arm Phase 2 studies in four HER2+ solid tumor types

- Phase 2 portion of the BDC-1001 Phase 1/2 Trial (BBI-20201001) will enroll patients with HER2+ colorectal, gastroesophageal, and endometrial cancers evaluating BDC-1001 as single agent and in combination with nivolumab*
- A new Phase 2 trial (BBI-20231001) will evaluate BDC-1001 as single agent and in combination with pertuzumab** in patients with HER2+ metastatic breast cancer previously treated with trastuzumab deruxtecan
- Tumor selection based on unmet patient need, evolving therapeutic landscape, accrued clinical data, regulatory path, and strategic collaborations
- Studies to expand globally in 2023 to include Spain, Italy, and France, in addition to the U.S. and South Korea
- Continue Phase 1 patient follow-up and conduct gene analysis of tumor samples

*Provided by BMS; **provided by Roche

ACKNOWLEDGEMENTS

- The authors would like to acknowledge the patients and their caregivers for their support, as well as the investigators and their study teams for their contributions
- This presentation is the intellectual property of Bolt Biotherapeutics, Inc. and can be found on boltbio.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.