BDC-3042: A Dectin-2 Agonistic Antibody for Tumor-Associated Macrophage-Directed Immunotherapy

Justin A. Kenkel, Rishali K. Gadkari, Jess L. Nolin, Fang Xiao, Po Y. Ho, Cindy L. Kreder, Laughing Bear Torrez, David T. Omstead, Katelynn A. McEachin, Jason Ptacek, Lu Xu, Karla A. Henning, Steven J. Chapin, David Dornan, Michael N. Alonso, Shelley E. Ackerman

Bolt Biotherapeutics, Redwood City, CA, USA

ABSTRACT
1348

INTRODUCTION

Tumor-associated macrophages (TAMs) generally support tumor progression through their immunosuppressive effects on the tumor microenvironment (TME) and are the predominant immune cell population in most cancers. TAMs express the pattern recognition receptor Dectin-2 (*CLEC6A*), an activating C-type lectin receptor (CLR) that binds to high-mannose glycans on fungi and other microbes and induces protective immune responses against infectious disease. Dectin-2 ligation mediates enhanced phagocytosis, antigen processing and presentation, and proinflammatory cytokine production. Agonism of Dectin-2 on TAMs using naturally derived ligands drives potent anti-tumor immunity in syngeneic mouse tumor models. Given these findings, we developed a human Dectin-2-targeted agonistic antibody, BDC-3042, which is capable of robustly activating TAMs as a novel approach to myeloid-directed immunotherapy.

Dectin-2 agonism activates TAMs and elicits anti-tumor immune response

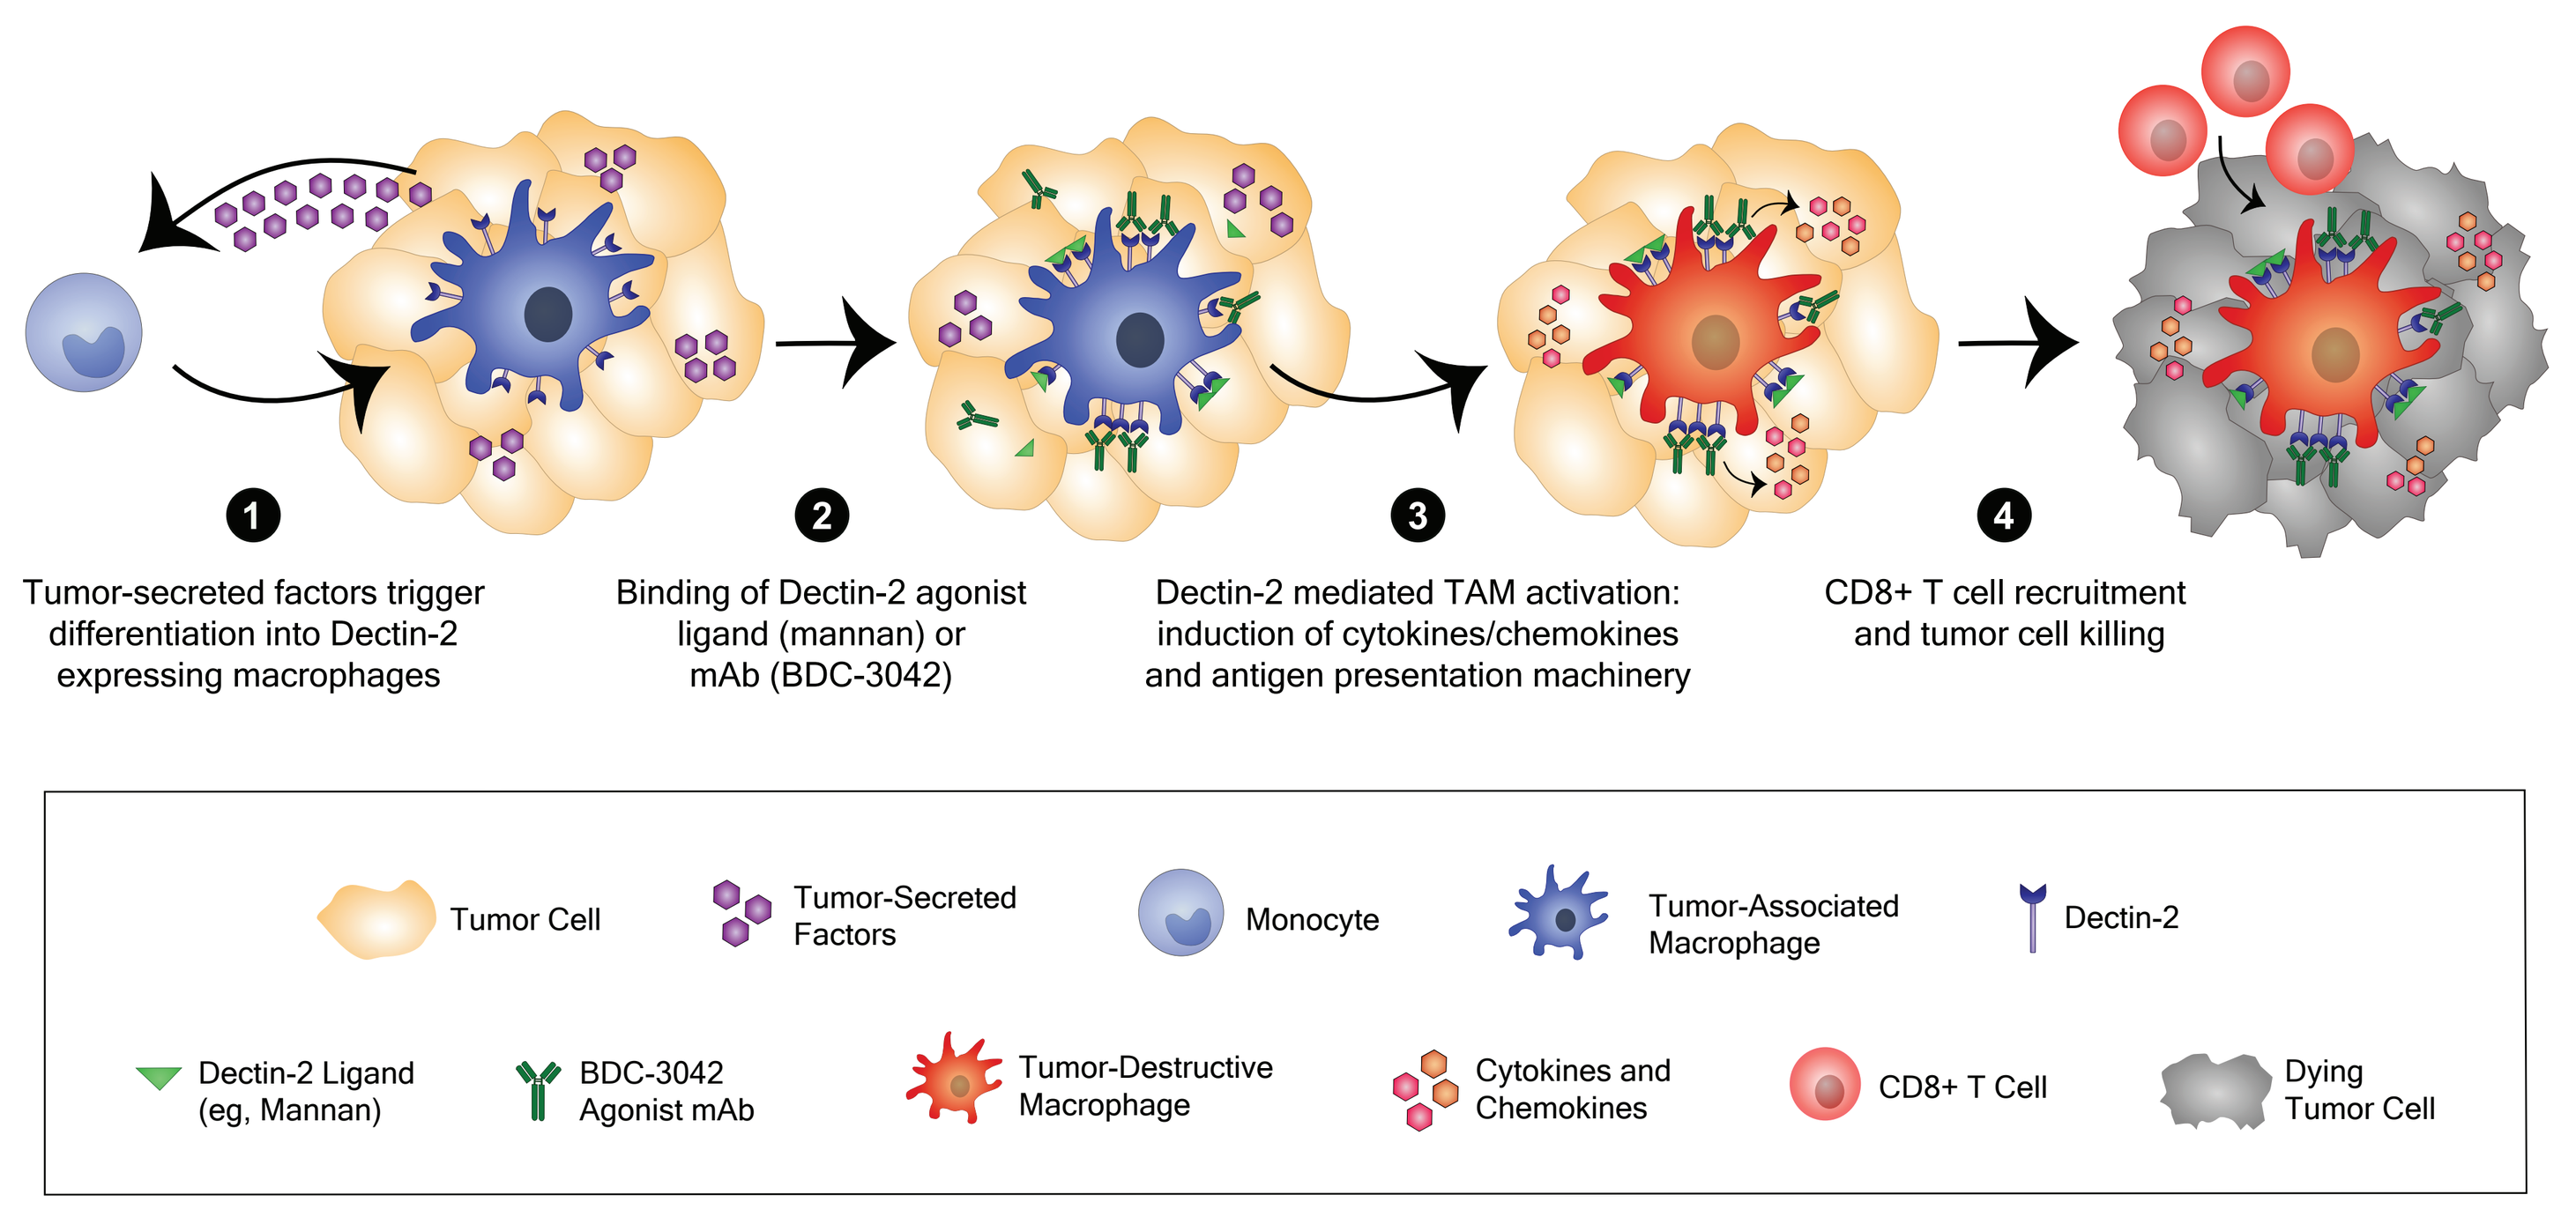
1
Tumor-secreted factors trigger differentiation into Dectin-2 expressing macrophages
2
Binding of Dectin-2 agonist ligand (mannan) or mAb (BDC-3042)
3
Dectin-2 mediated TAM activation: induction of cytokines/chemokines and antigen presentation machinery
4
CD8+ T cell recruitment and tumor cell killing
Tumor Cell
Tumor-Secreted Factors
Monocyte
Tumor-Associated Macrophage
Dectin-2
Dectin-2 Ligand (eg, Mannan)
BDC-3042 Agonist mAb
Tumor-Destructive Macrophage
Cytokines and Chemokines
CD8+ T Cell
Dying Tumor Cell

Figure 1: Schematic of proposed mechanism of action driving Dectin-2 mediated anti-tumor activity.

Dectin-2 gene expression is elevated across tumor types

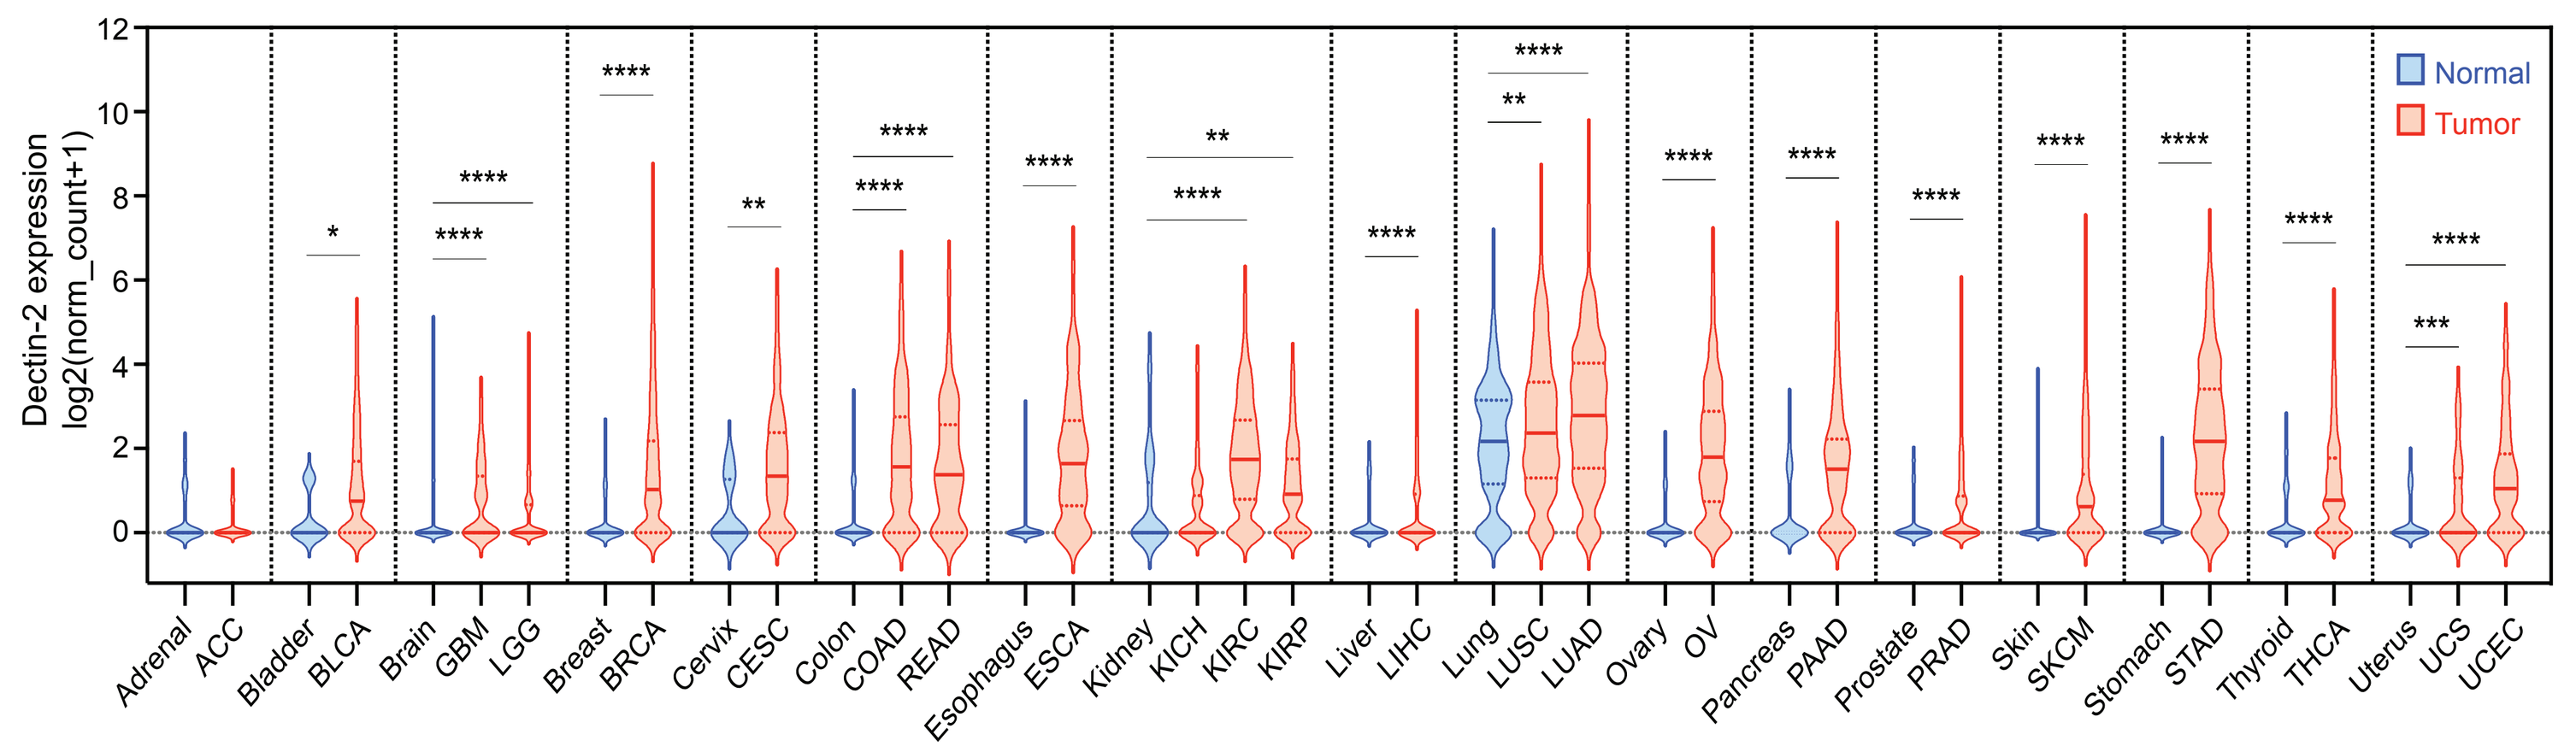
Dectin-2 expression log2(norm_count+1)
Normal
Tumor
Adrenal ACC Bladder BLCA Brain GBM LGG Breast BRCA Cervix CESC Colon COAD READ Esophagus ESCA Kidney KICH KIRC KIRP Liver LIHC Lung LUSC LUAD Ovary OV Pancreas PAAD Prostate PRAD Skin SKCM Stomach STAD Thyroid THCA Uterus UCS UCEC

Figure 2: Dectin-2 gene expression is elevated in tumors but low in most normal tissues. Dectin-2/*CLEC6A* mRNA expression in human tissue samples from the TCGA (tumor) and GTEX (normal) datasets (accessed Oct. 2019). TCGA study abbreviations are shown for the tumor subtypes. TCGA and GTEX data were processed using a uniform bioinformatic pipeline and obtained from UCSC Xena (xena.ucsc.edu). Median and interquartile range are shown on the violin plots. Statistics were calculated by Mann-Whitney U test; *p≤0.05, **p≤0.01, ***p≤0.001, ****p≤0.0001.

RESULTS

BDC-3042 binds to and activates Dectin-2 expressing human macrophages

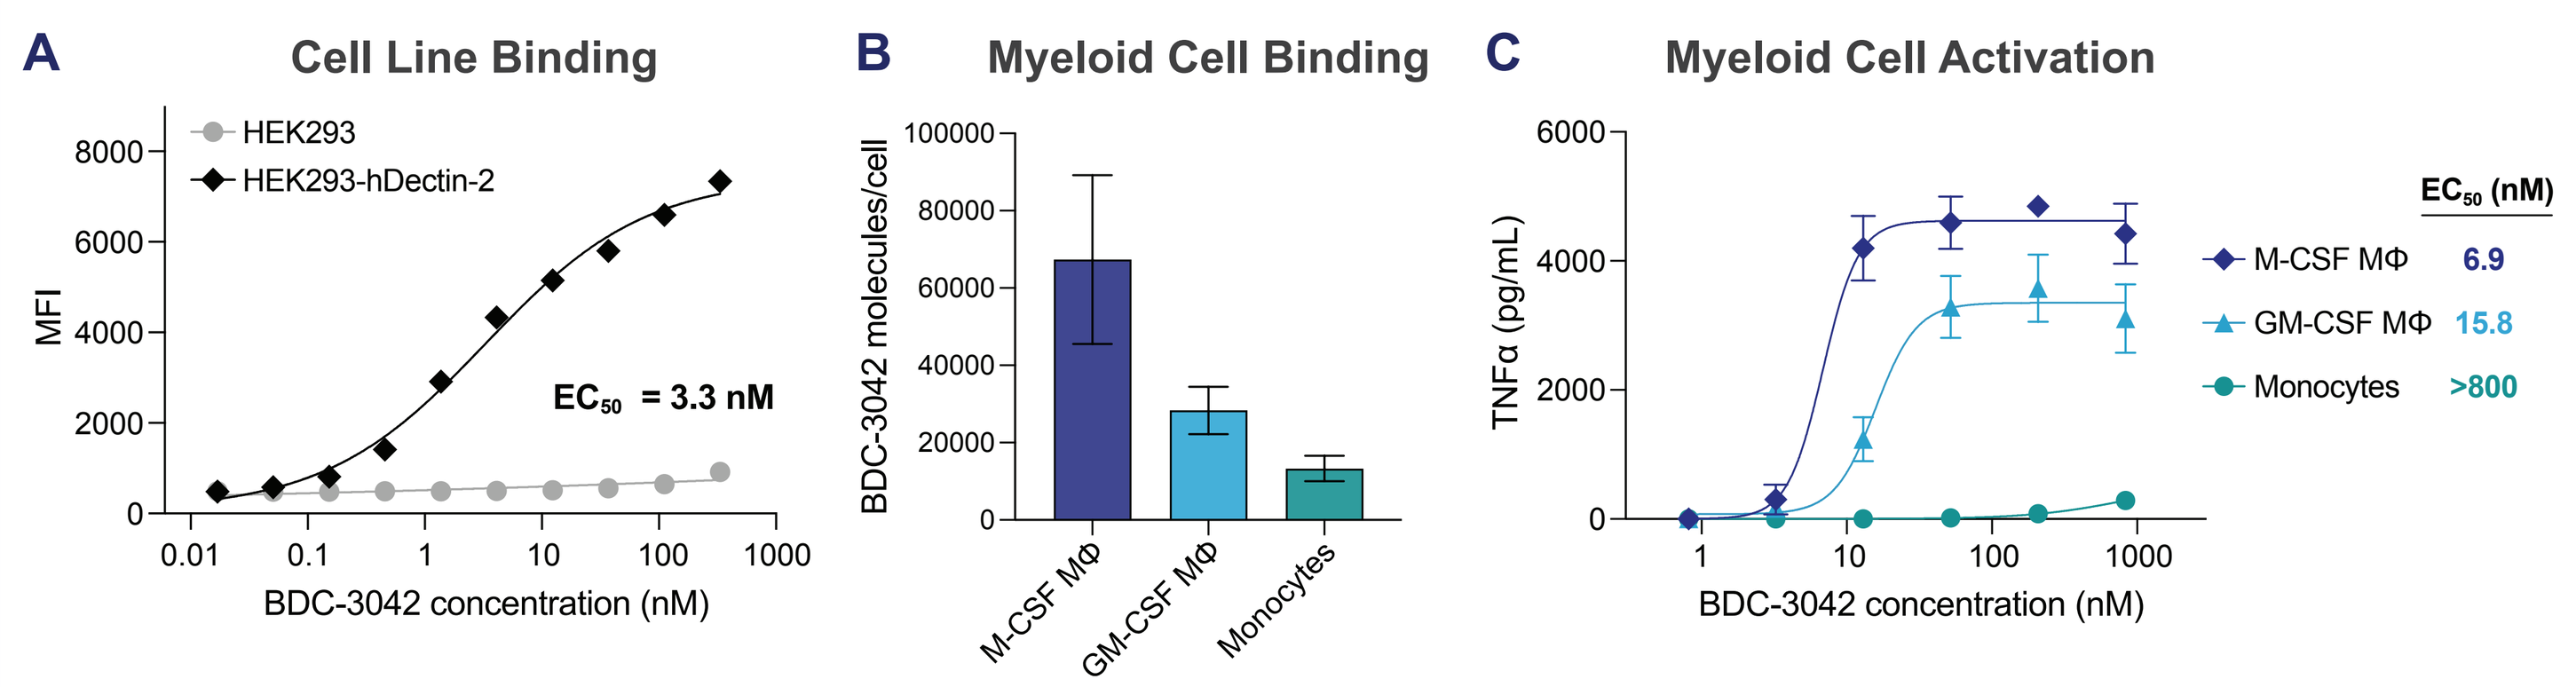
A Cell Line Binding
HEK293
HEK293-hDectin-2
EC50 = 3.3 nM
MFI
BDC-3042 concentration (nM)
B Myeloid Cell Binding
BDC-3042 molecules/cell
M-CSF MΦ
GM-CSF MΦ
Monocytes
C Myeloid Cell Activation
TNFα (pg/mL)
EC50 (nM)
M-CSF MΦ 6.9
GM-CSF MΦ 15.8
Monocytes >800
BDC-3042 concentration (nM)

Figure 3: BDC-3042 binds to Dectin-2 and activates in vitro-generated macrophages. (A) BDC-3042 binds to cells expressing Dectin-2 with single-digit nM EC_{50}s, while minimal binding is detected with HEK293T cells lacking Dectin-2 expression. **(B, C)** Fresh human monocytes or monocyte-derived macrophages generated with M-CSF or GM-CSF were assessed for **(B)** number of BDC-3042 molecules bound per cell, as measured by bead-based quantification (n=3) and **(C)** cytokine secretion following overnight stimulation with BDC-3042 (n=5-12).

BDC-3042 exhibits minimal binding to and activation of peripheral immune cells

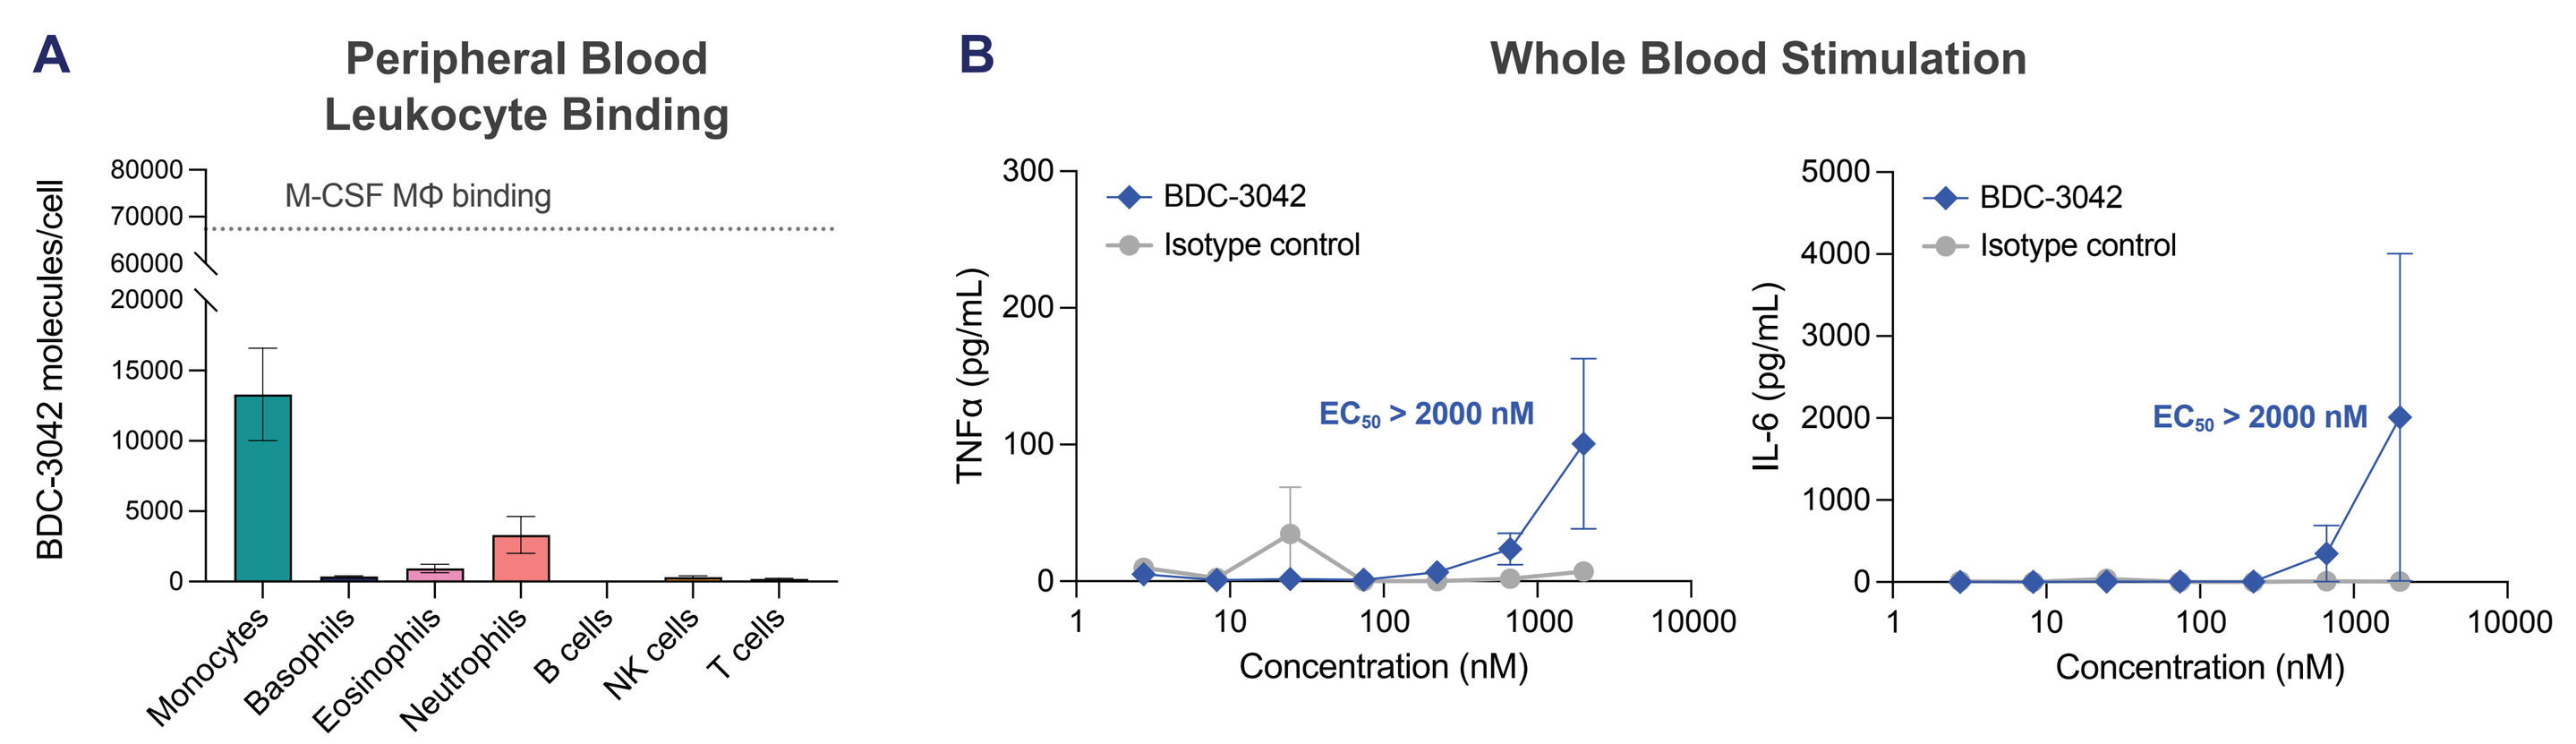
A Peripheral Blood Leukocyte Binding
M-CSF MΦ binding
BDC-3042 molecules/cell
Monocytes Basophils Eosinophils Neutrophils B cells NK cells T cells
B Whole Blood Stimulation
BDC-3042
Isotype control
EC50 > 2000 nM
TNFα (pg/mL)
IL-6 (pg/mL)
Concentration (nM)

Figure 4: BDC-3042 exhibits minimal binding to and activation of immune cells in peripheral blood. (A) Number of BDC-3042 molecules bound per cell, as measured by bead-based quantification with peripheral blood leukocytes from healthy donors (n=3). **(B)** Human whole blood from healthy donors was incubated overnight with the indicated test article, and cytokine secretion measured by cytokine bead array (n=9).

BDC-3042 activates primary human TAMs ex vivo

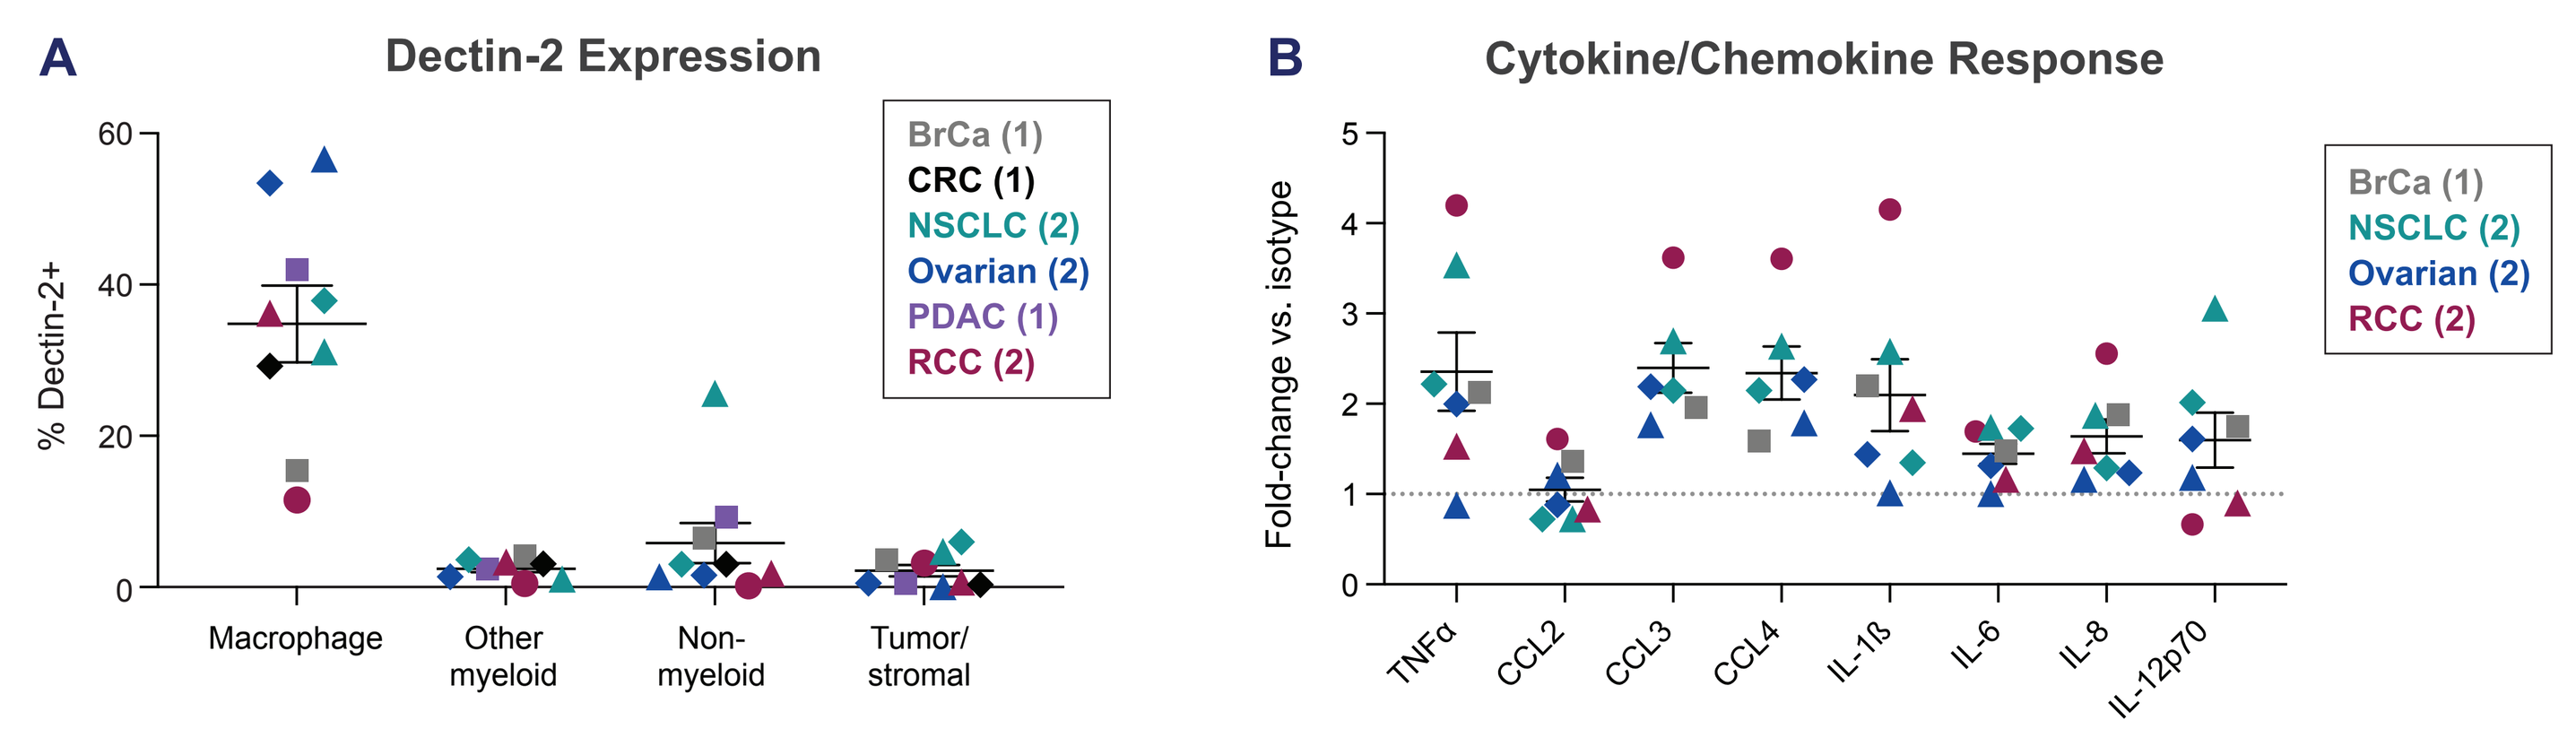
A Dectin-2 Expression
% Dectin-2+
BrCa (1)
CRC (1)
NSCLC (2)
Ovarian (2)
PDAC (1)
RCC (2)
Macrophage
Other myeloid
Non-myeloid
Tumor/ stromal
B Cytokine/Chemokine Response
Fold-change vs. isotype
BrCa (1)
NSCLC (2)
Ovarian (2)
RCC (2)
TNFα CCL2 CCL3 CCL4 IL-1ß IL-6 IL-8 IL-12p70

Figure 5: BDC-3042 activates primary human TAMs ex vivo. (A) Human tumors were processed into single-cell suspensions and analyzed by flow cytometry using a commercial Dectin-2 mAb. TAMs were defined as viable CD45+CD11b+CD14+HLA-DR+ cells. **(B)** Primary human tumor samples were processed into single-cell suspensions and cultured overnight with BDC-3042 or a non-binding isotype control antibody. Data are shown with mean and SEM; breast cancer (BrCa, n=1), colorectal cancer (CRC, n=1), non-small cell lung cancer (NSCLC, n=2), ovarian cancer (n=2), pancreatic ductal adenocarcinoma (PDAC, n=1), renal cell carcinoma (RCC, n=2).

BDC-3042 elicits dose-dependent activation of TAMs in RCC

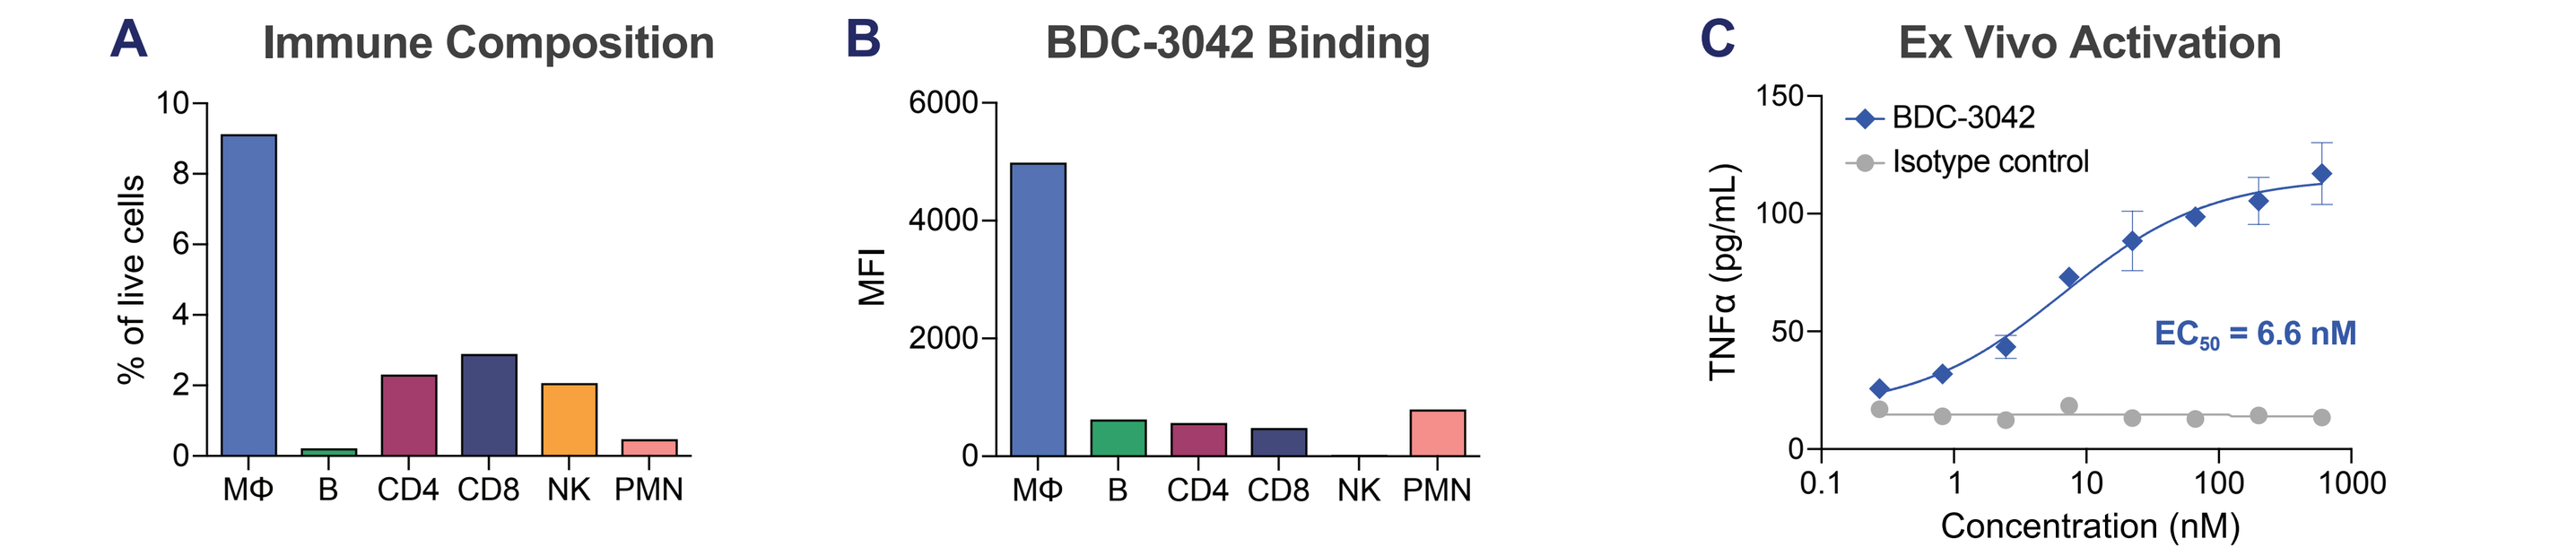
A Immune Composition
% of live cells
MΦ B CD4 CD8 NK PMN
B BDC-3042 Binding
MFI
C Ex Vivo Activation
BDC-3042
Isotype control
EC50 = 6.6 nM
TNFα (pg/mL)
Concentration (nM)

Figure 6: BDC-3042 elicits dose-dependent activation of TAMs in renal cell carcinoma (RCC). (A, B) Human RCC tumor sample was processed into single-cell suspension and analyzed by flow cytometry to assess **(A)** immune cell composition and **(B)** binding of 5 μg/mL BDC-3042 to immune cells as measured by MFI. **(C)** Cell suspension was incubated for 24 hours with BDC-3042 or isotype control mAb. TNFα secretion was measured by ELISA.

Dectin-2 is expressed by macrophages in triple-negative breast cancer

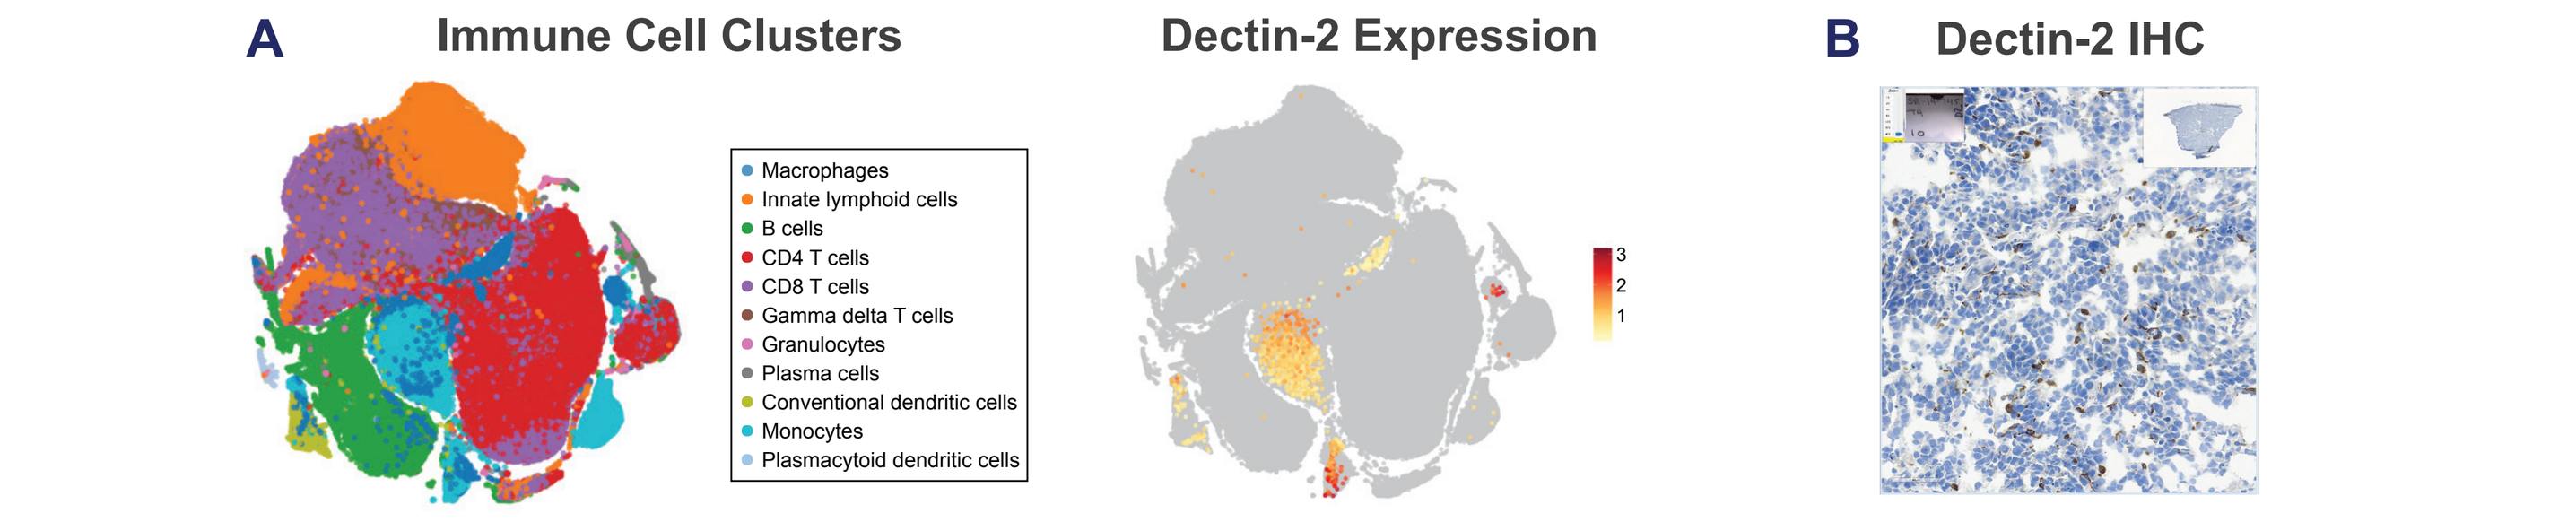
A Immune Cell Clusters
Macrophages
Innate lymphoid cells
B cells
CD4 T cells
CD8 T cells
Gamma delta T cells
Granulocytes
Plasma cells
Conventional dendritic cells
Monocytes
Plasmacytoid dendritic cells
Dectin-2 Expression
B Dectin-2 IHC

Figure 7: Dectin-2 is expressed by macrophages in triple-negative breast cancer (TNBC). (A) Single-cell RNA-Seq analysis of TNBC samples from 22 patients treated with paclitaxel or paclitaxel/atezolizumab (Zhang, et al. *Cancer Cell*, 2021; GSE169246). Immune cell clusters are visualized using t-distributed stochastic neighbor embedding (t-SNE). Dectin-2 expression is restricted to clusters of cells identified as monocytes or macrophages. **(B)** Dectin-2 staining in a frozen tissue section from human TNBC sample. Data are representative, where 8 of 9 samples assessed contained Dectin-2 positive cells.

Humanized mice for in vivo assessment of BDC-3042 anti-tumor activity

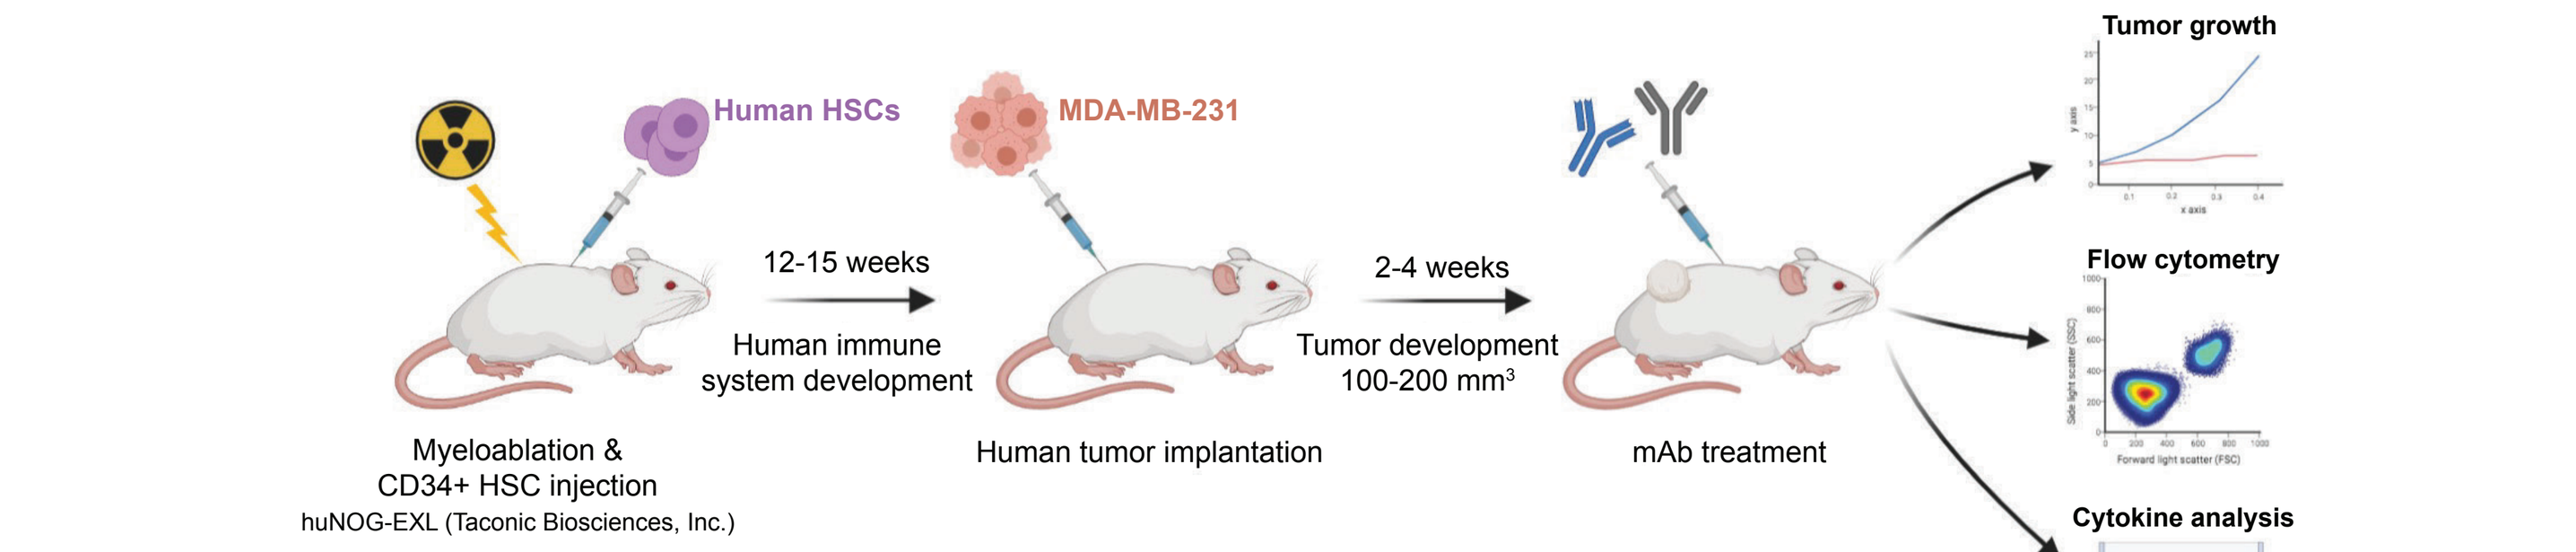
Human HSCs
MDA-MB-231
12-15 weeks
Human immune system development
2-4 weeks
Tumor development 100-200 mm³
Myeloablation & CD34+ HSC injection
huNOG-EXL (Taconic Biosciences, Inc.)
Human tumor implantation
mAb treatment
Tumor growth
Flow cytometry
Cytokine analysis
Kenkel, JA. AACR 2022.

Acknowledgments: The HIS mouse work described was performed in collaboration with Taconic Biosciences, Inc.

Figure 8: Humanized mice for in vivo assessment of BDC-3042 anti-tumor activity.

BDC-3042 binds strongly to TAMs in humanized mouse model

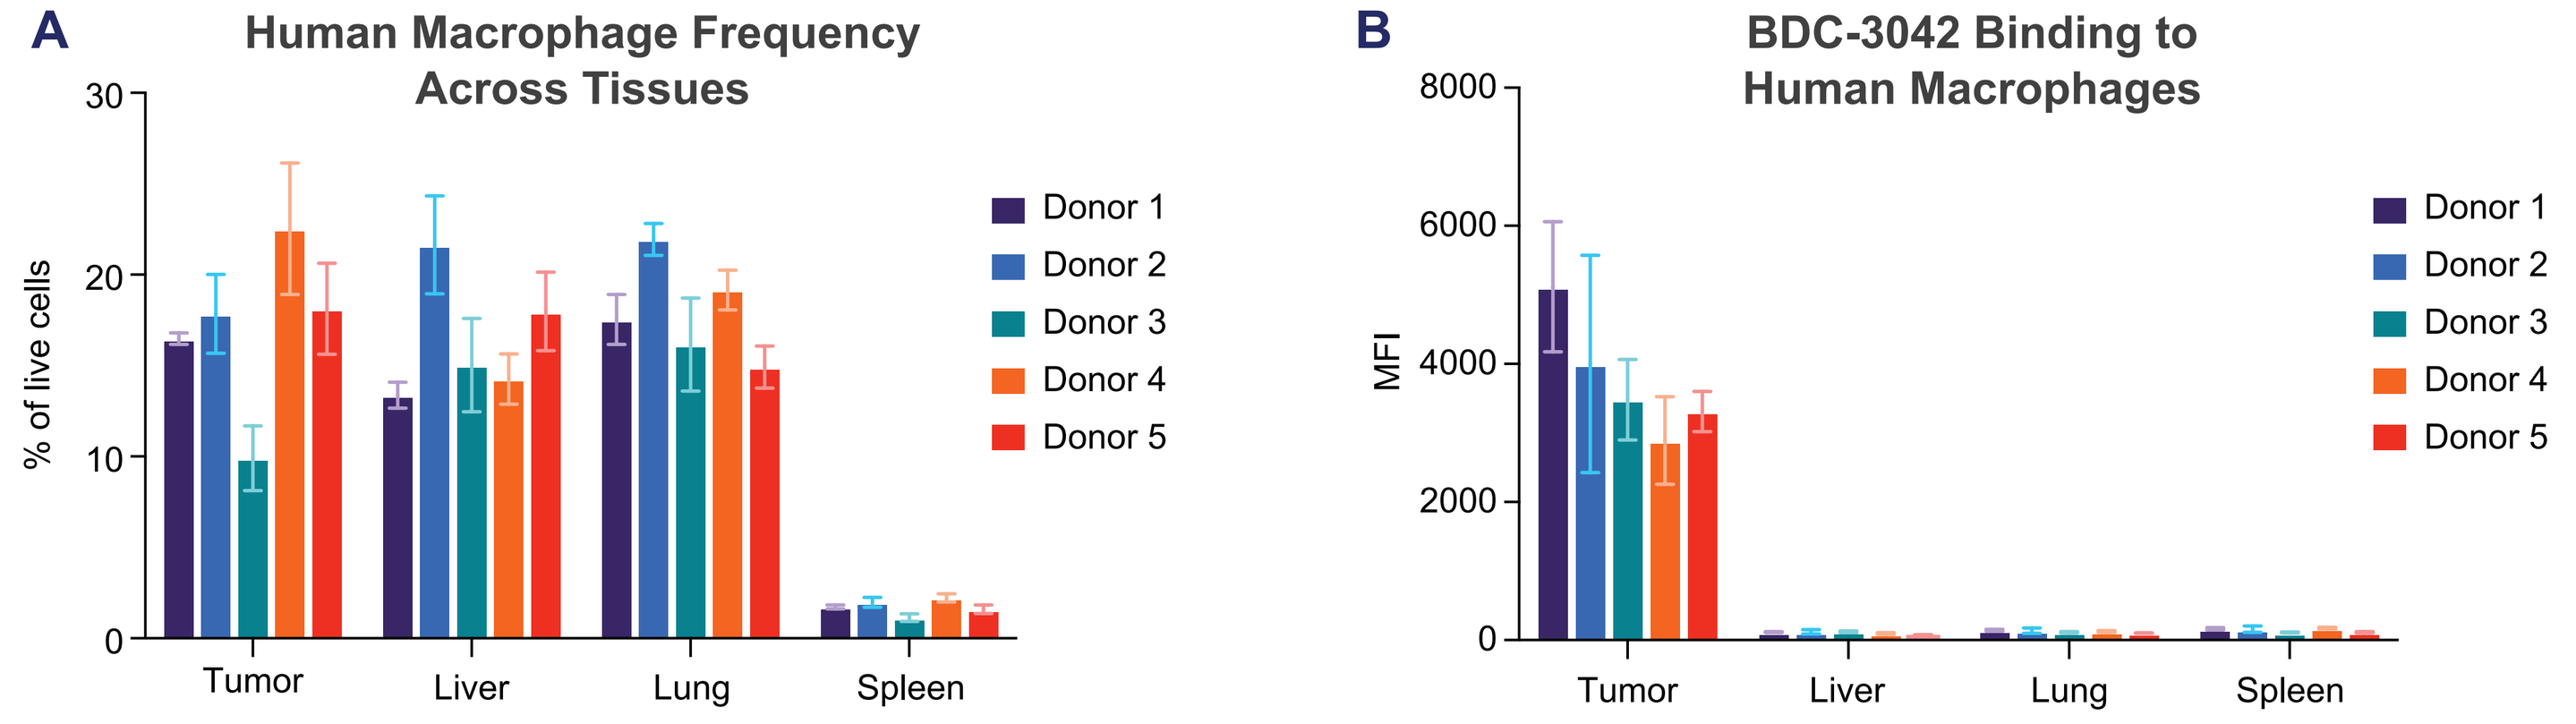
A Human Macrophage Frequency Across Tissues
% of live cells
B BDC-3042 Binding to Human Macrophages
MFI
Donor 1
Donor 2
Donor 3
Donor 4
Donor 5
Tumor Liver Lung Spleen

Figure 9: BDC-3042 binding to tissue-residing macrophages in humanized mice. (A, B) Tissues from MDA-MB-231 tumor-bearing huNOG-EXL mice generated using five unique HSC donors were analyzed by flow cytometry (n=4-5 mice/donor). **(A)** Human macrophage frequencies in the indicated tissues expressed as percentage of total live cells. **(B)** BDC-3042 binding to human macrophages recovered from the indicated tissues. Data are shown as mean with SEM.

BDC-3042 activates TAMs from MDA-MB-231 tumors in humanized mice

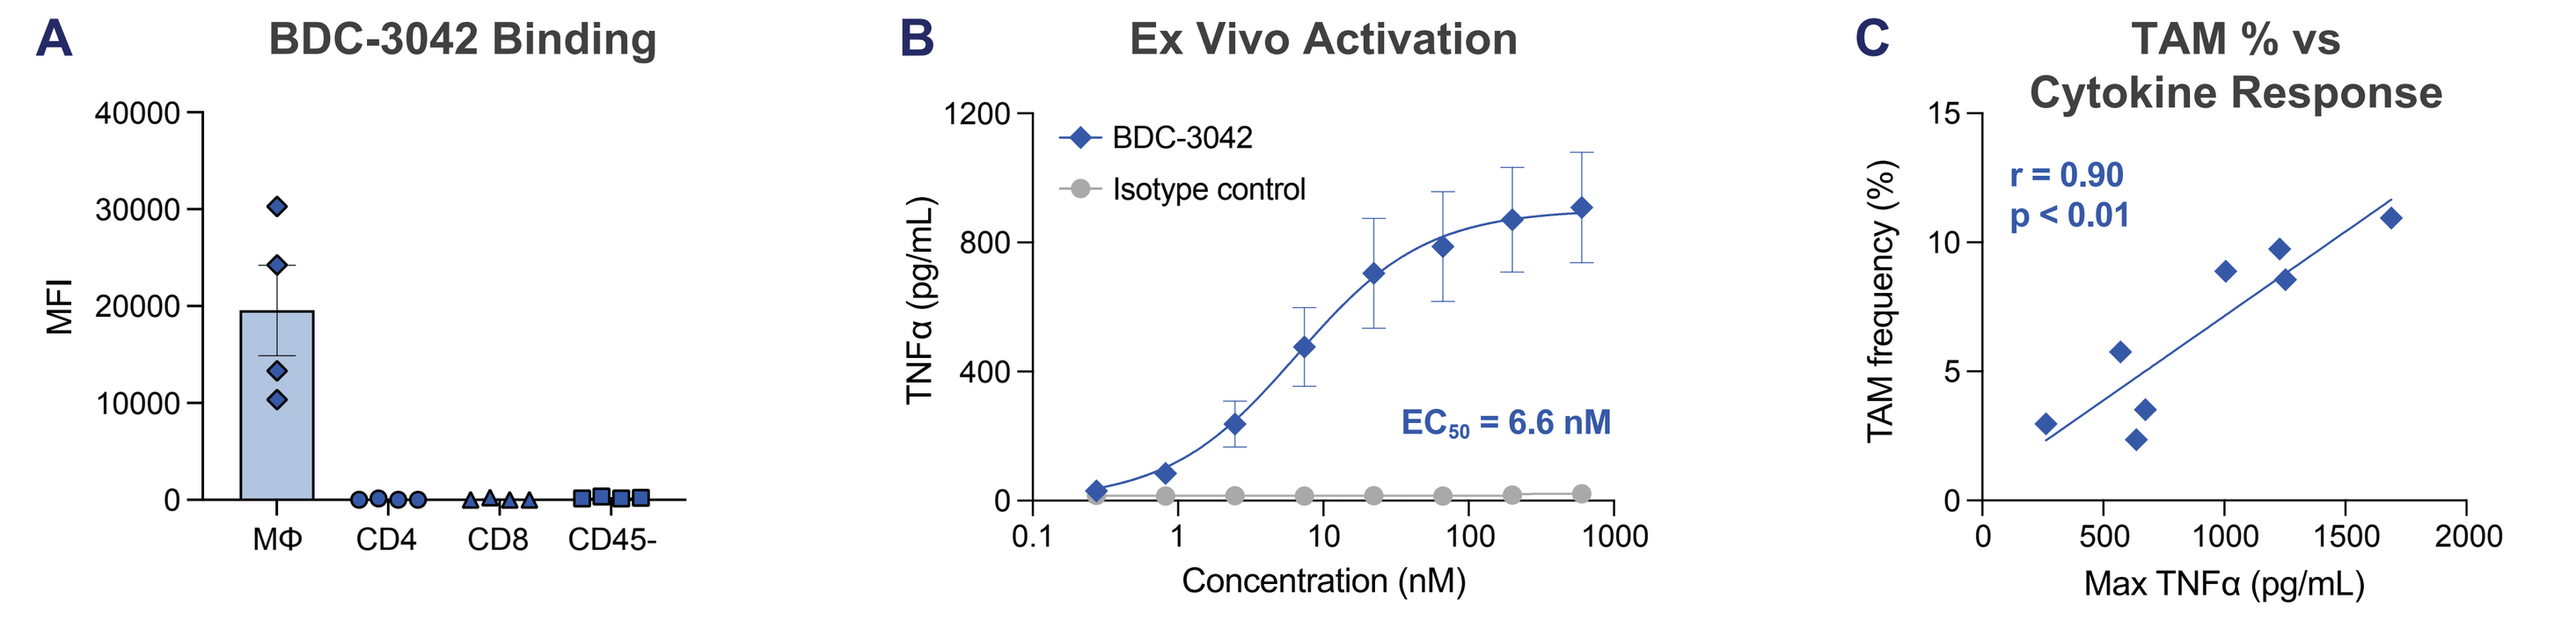
A BDC-3042 Binding
MFI
MΦ CD4 CD8 CD45-
B Ex Vivo Activation
BDC-3042
Isotype control
EC50 = 6.6 nM
TNFα (pg/mL)
Concentration (nM)
C TAM % vs Cytokine Response
r = 0.90
p < 0.01
TAM frequency (%)
Max TNFα (pg/mL)

Figure 10: BDC-3042 activates TAMs from MDA-MB-231 tumors in humanized mice. MDA-MB-231 tumors were harvested from huNOG-EXL mice and digested into single-cell suspensions (DTCs). **(A)** BDC-3042 binding to tumor-infiltrating human leukocytes was assessed by flow cytometry (n=4). **(B)** MDA-MB-231 DTCs were incubated for 18 hours with BDC-3042 or isotype control mAb, and human TNFα secretion was measured by ELISA (n=8). **(C)** Correlation of the TAM frequency in DTCs and peak TNFα secretion values. Pearson correlation coefficient and p-value are shown.

BDC-3042 mediates greater anti-tumor activity than pembrolizumab in MDA-MB-231 tumor-bearing humanized mice

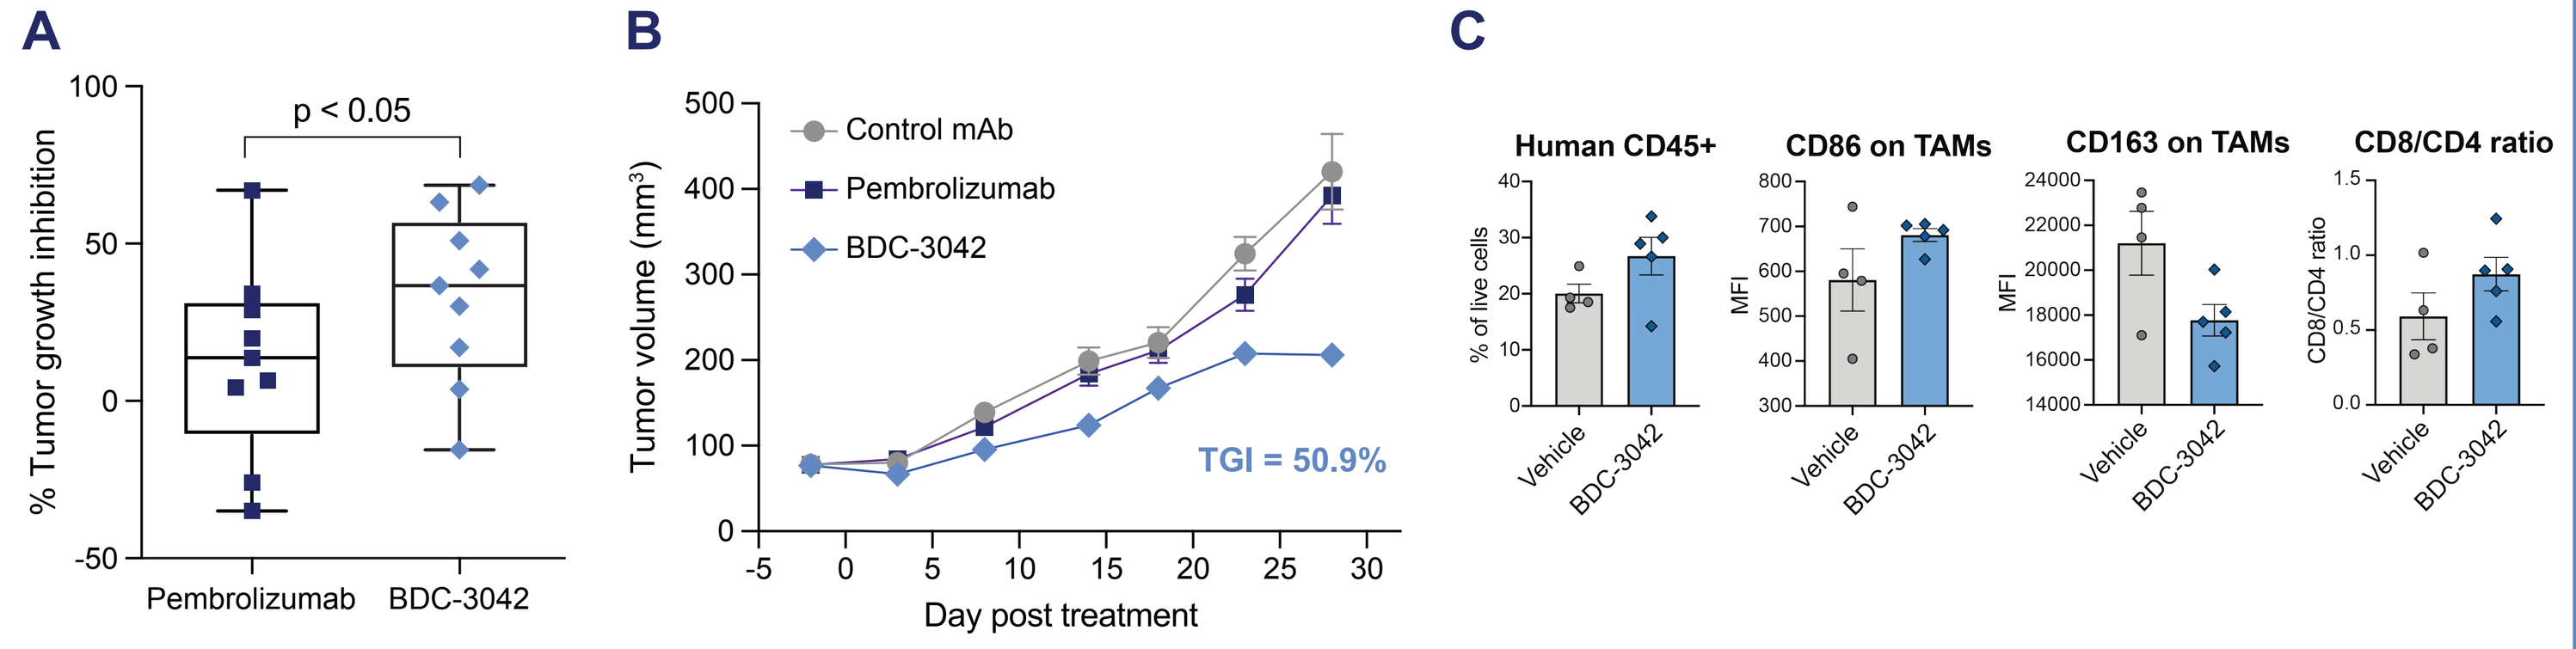
A
% Tumor growth inhibition
p < 0.05
Pembrolizumab
BDC-3042
B
Control mAb
Pembrolizumab
BDC-3042
Tumor volume (mm³)
TGI = 50.9%
Day post treatment
C
Human CD45+
CD86 on TAMs
CD163 on TAMs
CD8/CD4 ratio
Vehicle
BDC-3042

Figure 11: BDC-3042 mediates greater anti-tumor activity than pembrolizumab in MDA-MB-231 tumor-bearing humanized mice. huNOG-EXL mice were implanted with bilateral MDA-MB-231 tumors and treated with the indicated test articles to assess anti-tumor activity and pharmacodynamic responses. **(A)** Tumor growth inhibition (TGI) across 9 HSC donor cohorts for BDC-3042 or pembrolizumab as compared to an isotype control antibody. Statistics were performed by two-tailed paired t-test. **(B)** Tumor growth curves for mice from a single donor treated with BDC-3042 (1 mg/kg), isotype control mAb (5 mg/kg), or pembrolizumab (5 mg/kg) (all IP Q5Dx6). TGI was calculated relative to the isotype control mAb. **(C)** Tumors were assessed for pharmacodynamic changes by flow cytometry 48 hours following the second dose of vehicle or BDC-3042 (n=4-5 mice/group).

CONCLUSIONS

- **Dectin-2 is a novel immuno-oncology target expressed by tumor-associated macrophages (TAMs) across a range of solid tumor types**
- **BDC-3042 is an agonistic antibody targeting Dectin-2 that is designed to reprogram immunosuppressive TAMs into immunostimulatory cells that drive anti-tumor immunity**
- **BDC-3042 selectively binds to Dectin-2-expressing macrophages and induces an array of pro-inflammatory cytokines, chemokines, and antigen presentation molecules**
- **BDC-3042 exhibits minimal binding to and activation of peripheral leukocytes**
- **BDC-3042 repolarizes TAMs toward an immunostimulatory phenotype and mediates greater anti-tumor activity than pembrolizumab in tumor-bearing humanized mice**
- **Preclinical data support clinical evaluation of BDC-3042, with initiation of a Phase 1 clinical trial planned for 2023**