PD-L1-targeted ISAC Combines Myeloid Cell Activation, Immune Checkpoint Inhibition and ADCP to Improve Anti-tumor Efficacy Over Anti-PD-L1 Antibodies in Preclinical Models

Justin A. Kenkel, Rishali Gadkari, Karla A. Henning, Romas Kudirka, William G. Mallet, Po Y. Ho, Ganapathy Sarma, Steven J. Chapin, Liz Bogaert, Jennifer E. Melrose, Lisa K. Blum, Matthew Zhou, Suprit Deol, Cindy Kreder, Yuyi Shen, Puneet Anand, Arthur Lee, Hai Li, Shelley E. Ackerman, Brian S. Safina, David Dornan, Michael N. Alonso, Marcin Kowanetz

Bolt Biotherapeutics, 900 Chesapeake Drive, Redwood City, CA, USA

ABSTRACT
782

INTRODUCTION

Immune-stimulating antibody conjugates (ISACs) consist of tumor-targeting antibodies conjugated to immune stimulants and are designed to activate the innate and adaptive immune systems against tumor cells following systemic administration. PD-L1 is an immune checkpoint that regulates anti-tumor T cell responses and is expressed on tumor cells as well as tumor-infiltrating immune cells across many tumor types. We evaluated therapeutic properties of PD-L1-targeted TLR7/8 ISAC and preclinically demonstrated its potential as a multifunctional therapeutic that may improve efficacy of PD-L1/PD-1 inhibition by combining three mechanisms of action into a single molecule: TLR-mediated myeloid cell activation, T cell activation through immune checkpoint inhibition as well as antibody-dependent cellular phagocytosis (ADCP).

ISACs consist of tumor-targeting antibodies conjugated to immune stimulants

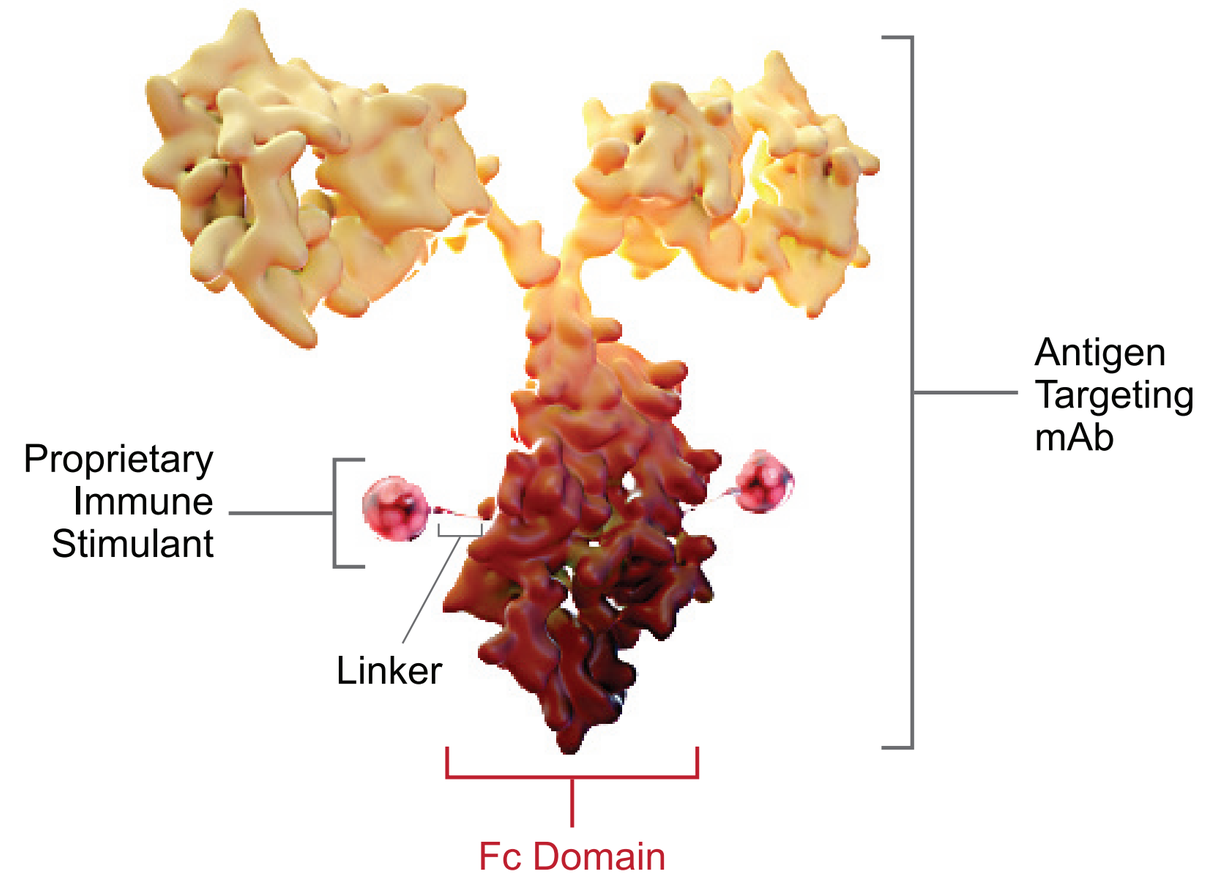
Proprietary Immune Stimulant
Linker
Fc Domain
Antigen Targeting mAb

Immune-stimulating antibody conjugates (ISACs)[1]:
- Systemically delivered, tumor-targeting therapeutics
- Antibody against a tumor antigen directs Boltbody™ ISACs to the tumor
- Proprietary immune stimulant activates myeloid antigen-presenting cells
- Myeloid cells kill tumor cells, create a "hot" tumor microenvironment, and initiate an innate and adaptive anti-tumor immune response

PD-L1 ISAC:
- PD-L1 ISAC consist of PD-L1 targeting antibody conjugated to TLR7/8 agonist

Clinical experience with ISACs:
- BDC-1001[2] (HER2 ISAC) is currently being evaluated in a Phase 1/2 clinical study

Proposed mechanism of action: PD-L1 ISAC can act through PD-L1 expressed on tumor and immune cells

PD-L1 ISAC combines three mechanisms to activate innate and adaptive immune system and induce anti-tumor response:

1. TLR-mediated myeloid cell activation leading to innate and then adaptive immunity
2. T cell activation through immune checkpoint inhibition
3. Direct tumor cell killing by ADCP

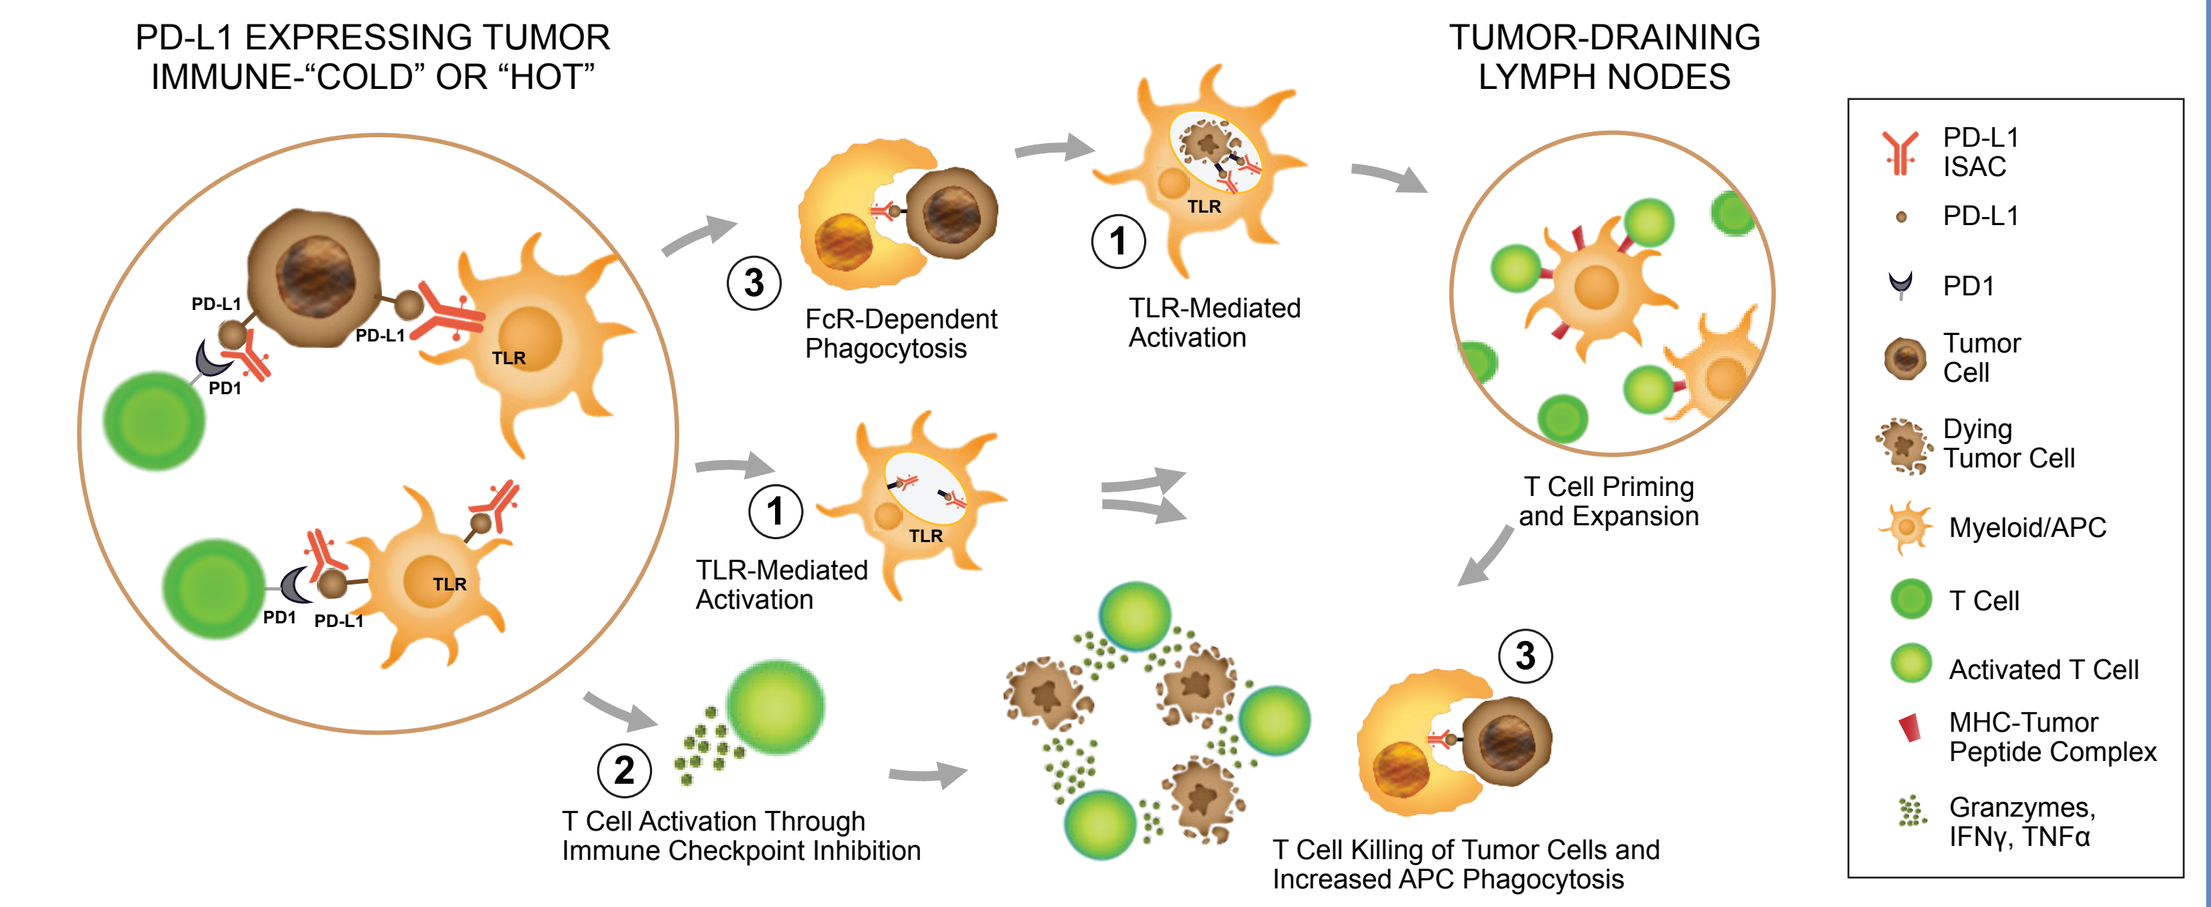
PD-L1 EXPRESSING TUMOR IMMUNE-"COLD" OR "HOT"
TUMOR-DRAINING LYMPH NODES
FcR-Dependent Phagocytosis
TLR-Mediated Activation
T Cell Priming and Expansion
T Cell Activation Through Immune Checkpoint Inhibition
T Cell Killing of Tumor Cells and Increased APC Phagocytosis
PD-L1 ISAC
PD-L1
PD1
Tumor Cell
Dying Tumor Cell
Myeloid/APC
T Cell
Activated T Cell
MHC-Tumor Peptide Complex
Granzymes, IFNγ, TNFα

REFERENCES

1. Ackerman SE, et al. *Nature Cancer*. 2021;2:18–33; 2. Sharma M, et al. ASCO 2021. Abstract 2549; 3. Herbst R, et al. *Nature*. 2014;515; (doi:10.1038/nature14011); 4. Huang R, et al. Modern Pathology. 2021;34:252–263. (https://doi.org/10.1038/s41379-020-00664-y).

RESULTS

PD-L1 is an immune checkpoint molecule expressed on tumor and immune cells

PD-L1 expression in selected tumor types

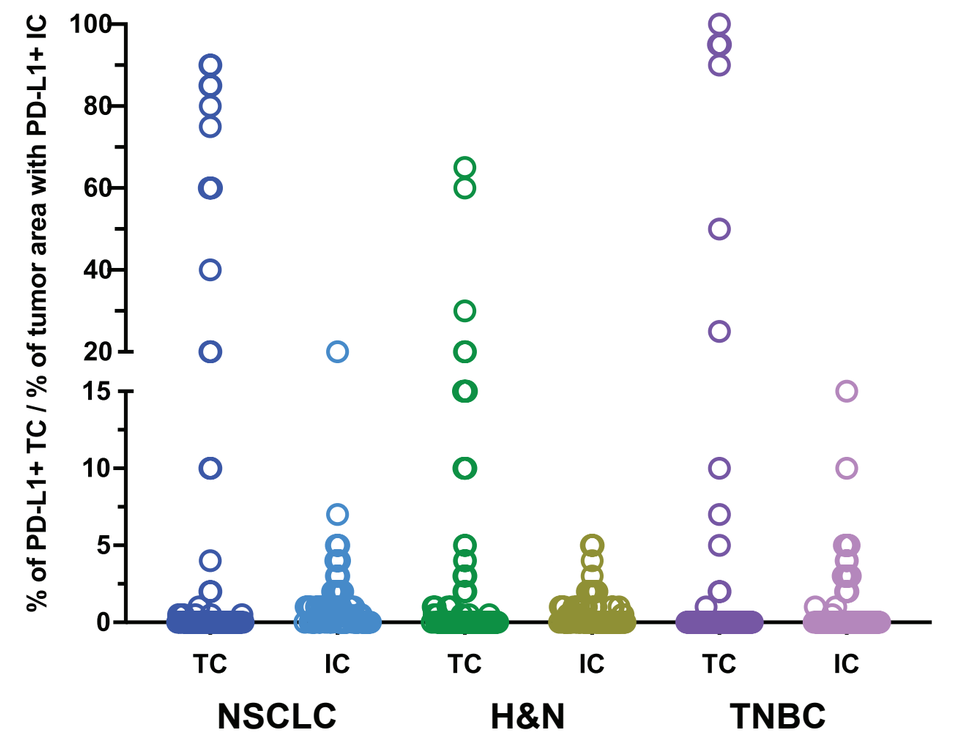

Representative images of PD-L1 expression on tumor and immune cells

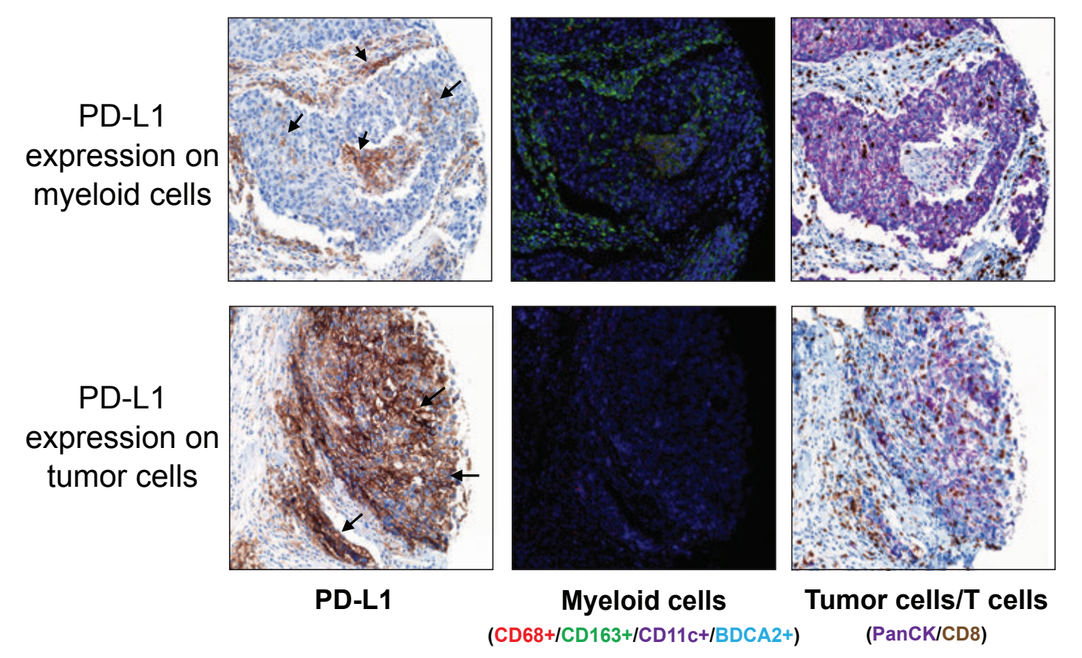
PD-L1 expression on myeloid cells
PD-L1 expression on tumor cells
PD-L1
Myeloid cells
Tumor cells/T cells

- Human non-small cell lung cancer (NSCLC), head and neck (H&N) and triple negative breast cancer (TNBC) microarray was stained with the PD-L1 IHC (22c3), a CD8/PanCK dual IHC, and a custom quadruplex IF assay for identification of myeloid APC sub-populations. TC – tumor cells; IC – immune cells
- Consistent with prior literature[3,4], PD-L1 expression was detected on tumor and immune cells across tumor types
- Representative images of PD-L1 expression on TC and IC in TNBC are shown

Proprietary anti-hPD-L1 mAbs are characterized by a specific binding to hPD-L1 on cell surface

Strong binding to 293-hPD-L1 cells / **No/Poor binding to PD-L1-negative cells**

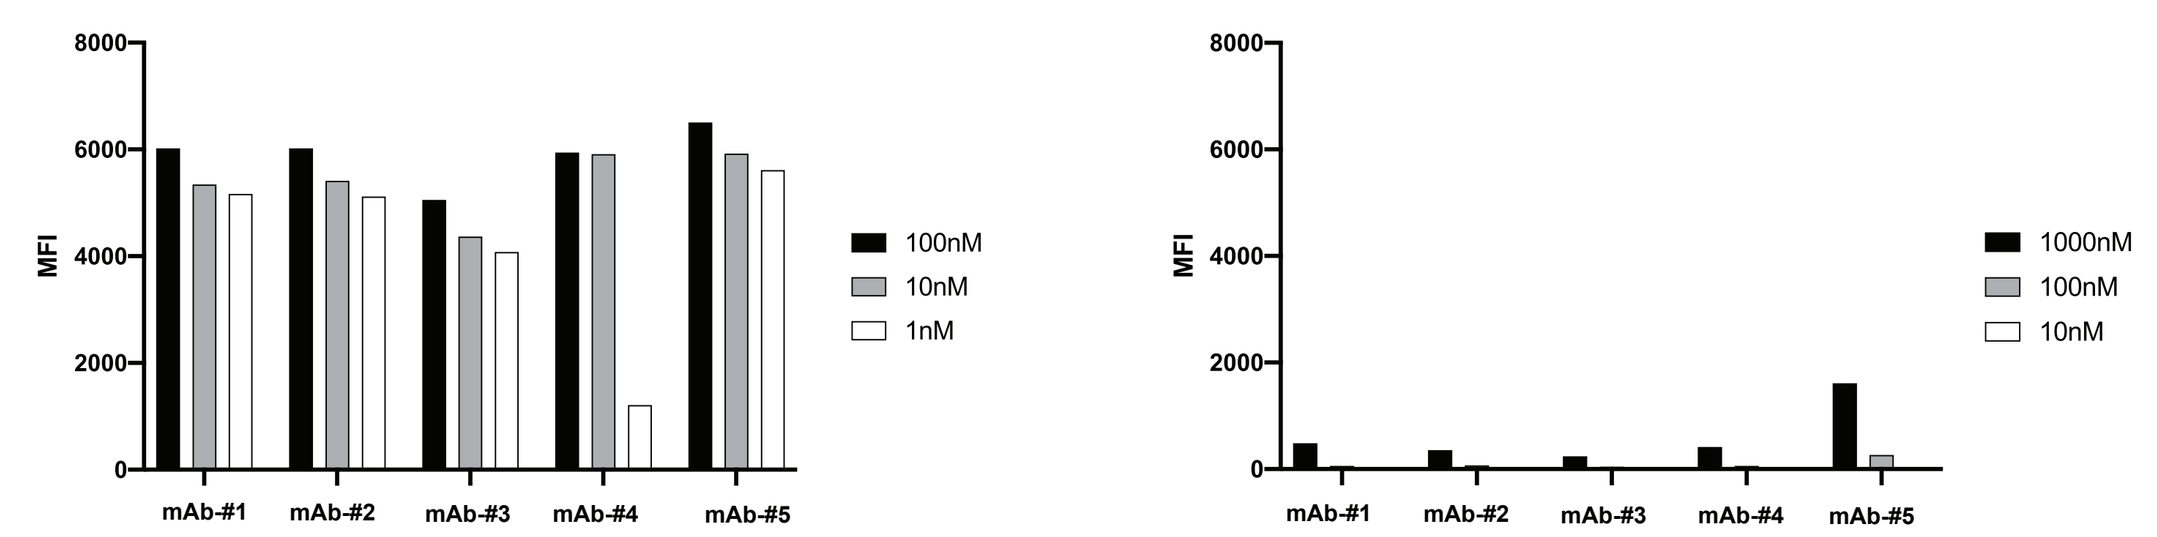

- Candidate anti-PD-L1 antibodies were incubated at 4°C with HEK-293 transduced with human PD-L1 or parental (no PD-L1 expression) HEK-293 cells. After unbound antibodies were removed by buffer wash, bound antibodies were detected using a fluorescent secondary antibody. Level of binding ("MFI") was determined by conventional flow cytometry.

Anti-hPD-L1 mAbs are potent PD-L1/PD1 blockers in vitro and in vivo

PD-L1/PD-1 Checkpoint Blockade (in vitro)

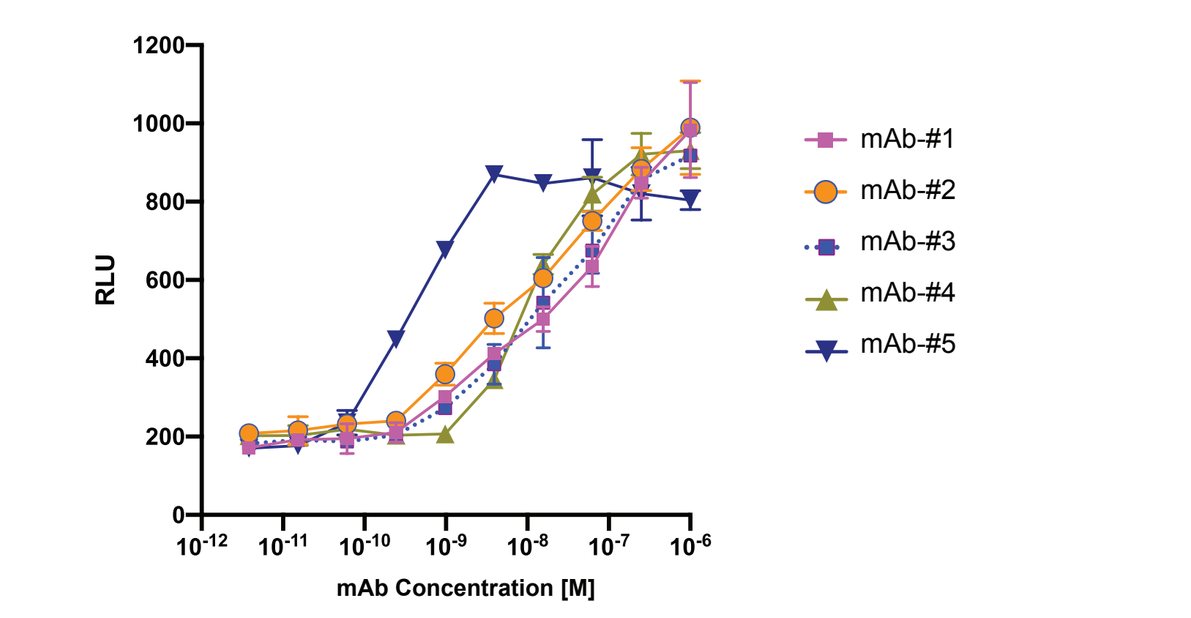

- Potential of anti-PD-L1 mAbs to block PD-L1/PD1 interaction was assessed by a bioluminescent cell-based assay, according to the vendor's protocol (Promega)

PD-L1/PD-1 Checkpoint Blockade (in vivo)

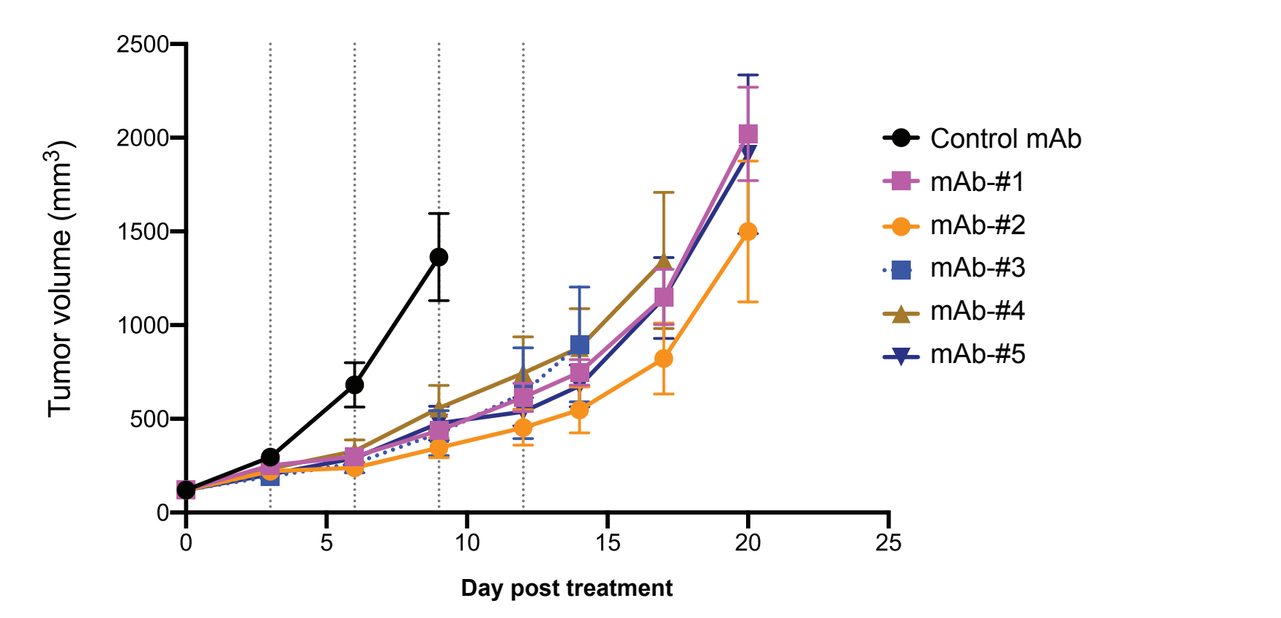

- Syngeneic MC38-hPD-L1 tumor growth in Black 6 mice following treatment with the indicated anti-PD-L1 mAbs at 5 mg/kg, IP, Q3Dx5, n=6 per group
- All tested anti-PD-L1 mAbs showed an anti-tumor growth inhibition

Anti-hPD-L1 mAbs induce potent ADCP in vitro

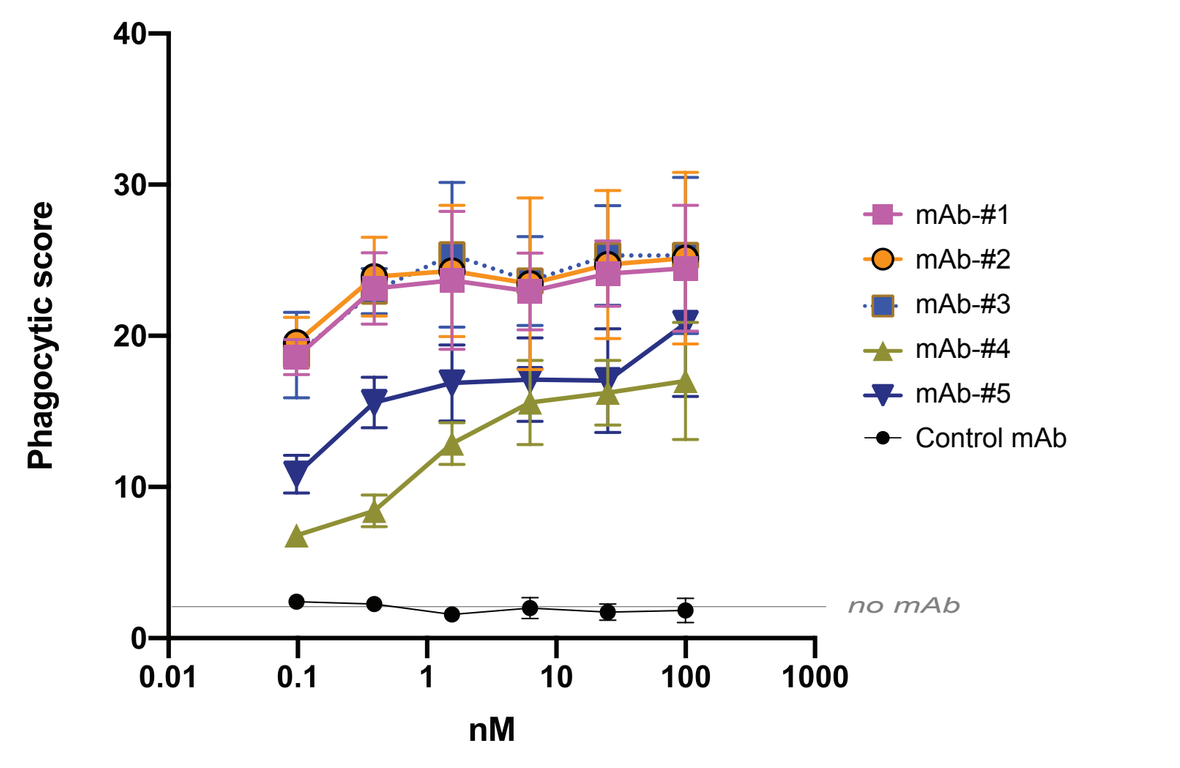

- Cell Tracker Green (CTG) labelled RKO cells were opsonized with indicated mAbs at the indicated concentrations
- Unbound mAbs were washed out, and the opsonized RKO cells were cocultured with M-CSF differentiated macrophages from healthy human donors at 1:1 E:T ratio
- Following the incubation, cells were stained with CD206 (macrophage specific marker) and analyzed by flow cytometry for CTG+CD206+ macrophages to assess level of phagocytosis
- Phagocytic score was calculated as the geometric mean fluorescence intensity (gMFI) of the CTG+CD206+ macrophages x% of CTG+CD206+ macrophages/10,000

Anti-hPD-L1 ISAC activates cDCs co-cultured with tumors expressing PD-L1 at physiological levels

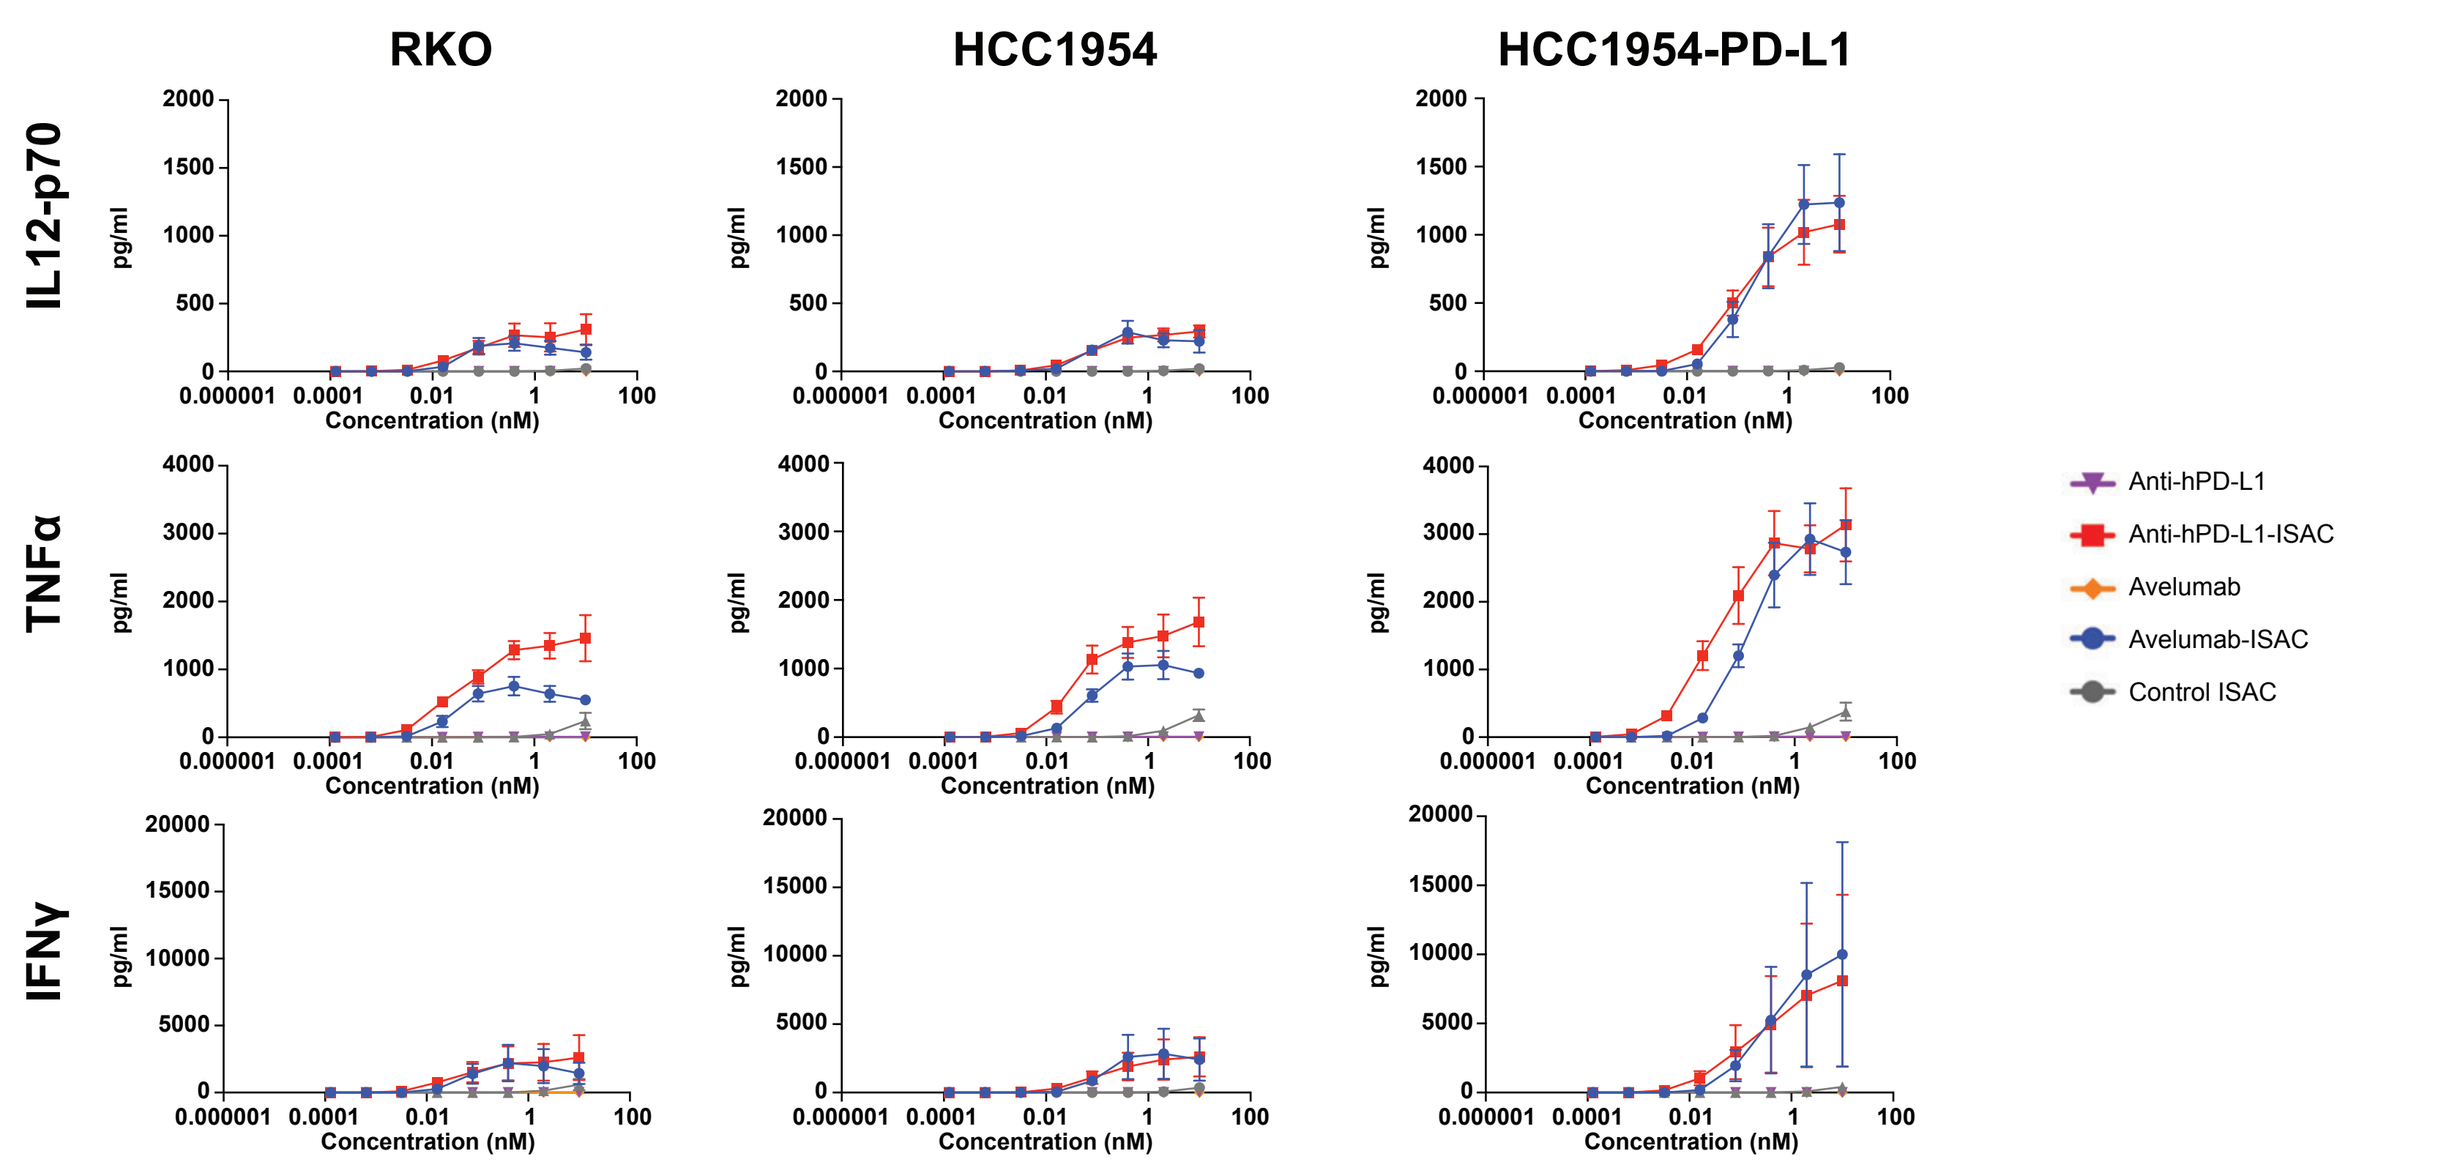
RKO
HCC1954
HCC1954-PD-L1
IL12-p70
TNFα
IFNγ
Anti-hPD-L1
Anti-hPD-L1-ISAC
Avelumab
Avelumab-ISAC
Control ISAC

- cDC-enriched primary cells were co-cultured with HCC1954-PD-L1, HCC1954 or RKO tumor cells (5:1 E:T ratio) in the presence of indicated mAbs or ISACs. Supernatants were subsequently analyzed for cytokine expression. Data are shown as mean with SEM of n=3 donors.

Physiological PD-L1 expression level in RKO and HCC1954 tumor cell lines co-cultured with cDC

PD-L1 expression on target tumor cell lines

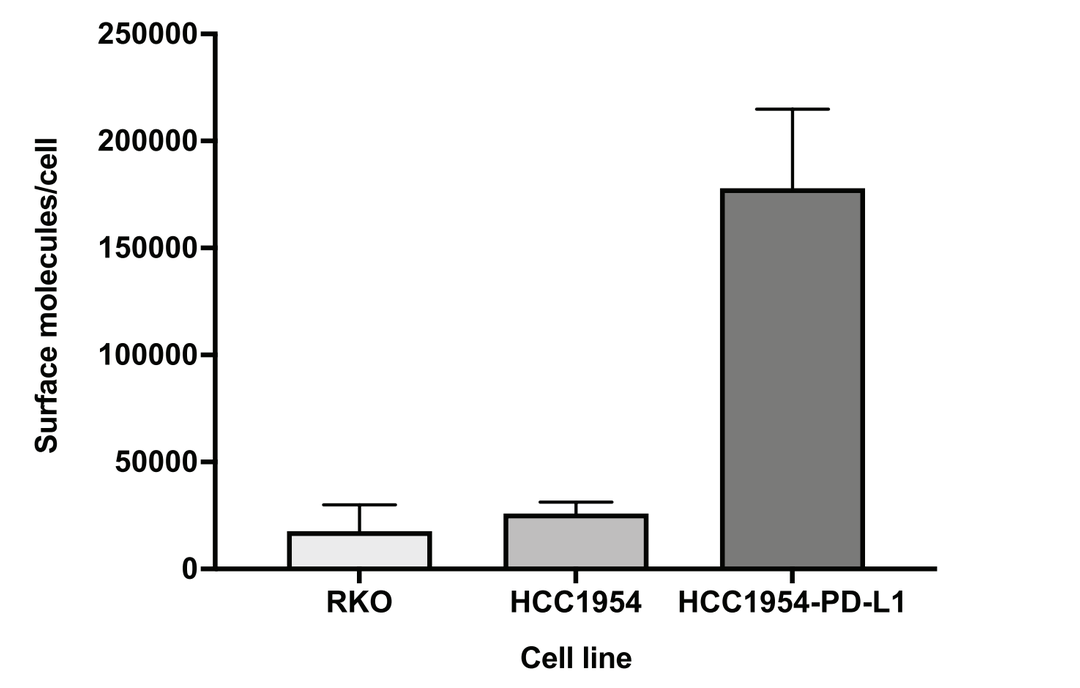

- Cell surface expression of PD-L1 in RKO, HCC1954 and PD-L1 over-expressing HCC1954-PD-L1 cells was quantified by flow cytometry using bead-based standards (Agilent QiFi kit).

PD-L1 expression by IHC

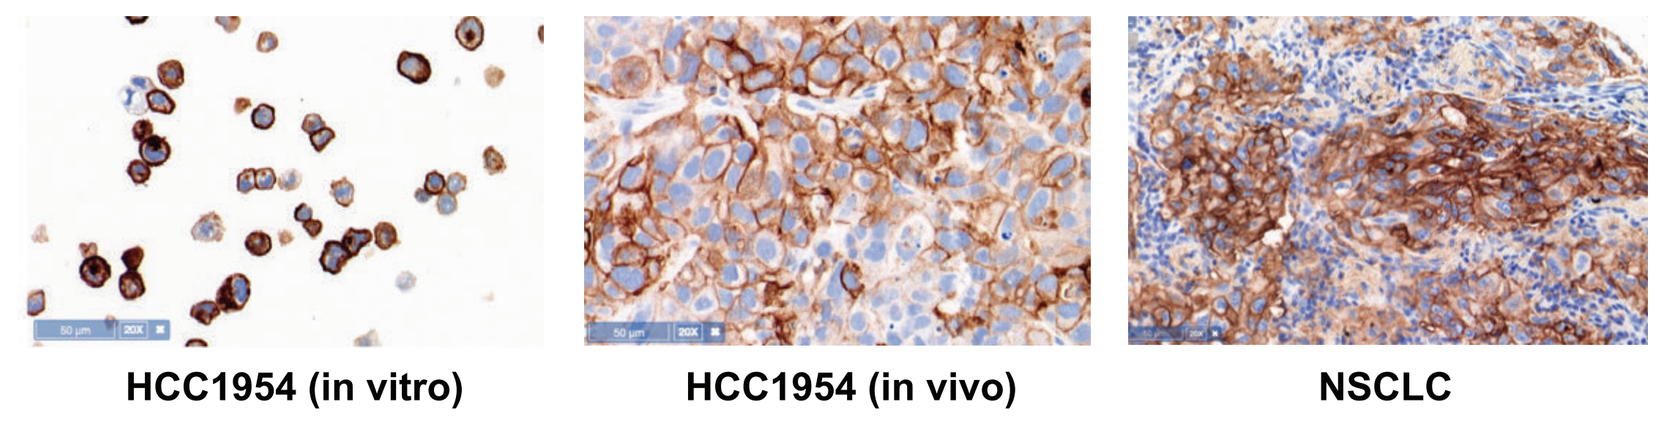
HCC1954 (in vitro)
HCC1954 (in vivo)
NSCLC

- PD-L1 expression on HCC1954 cells cultured in vitro and in vivo was evaluated with PD-L1 IHC (22c3). For comparison, an example of PD-L1 expression in a human specimen of NSCLC is shown.

Anti-mPD-L1-ISAC shows profound efficacy vs anti-PD-L1 treatment and induces immunological memory

Syngeneic MB49 tumor growth inhibition in mice treated with PD-L1 ISAC

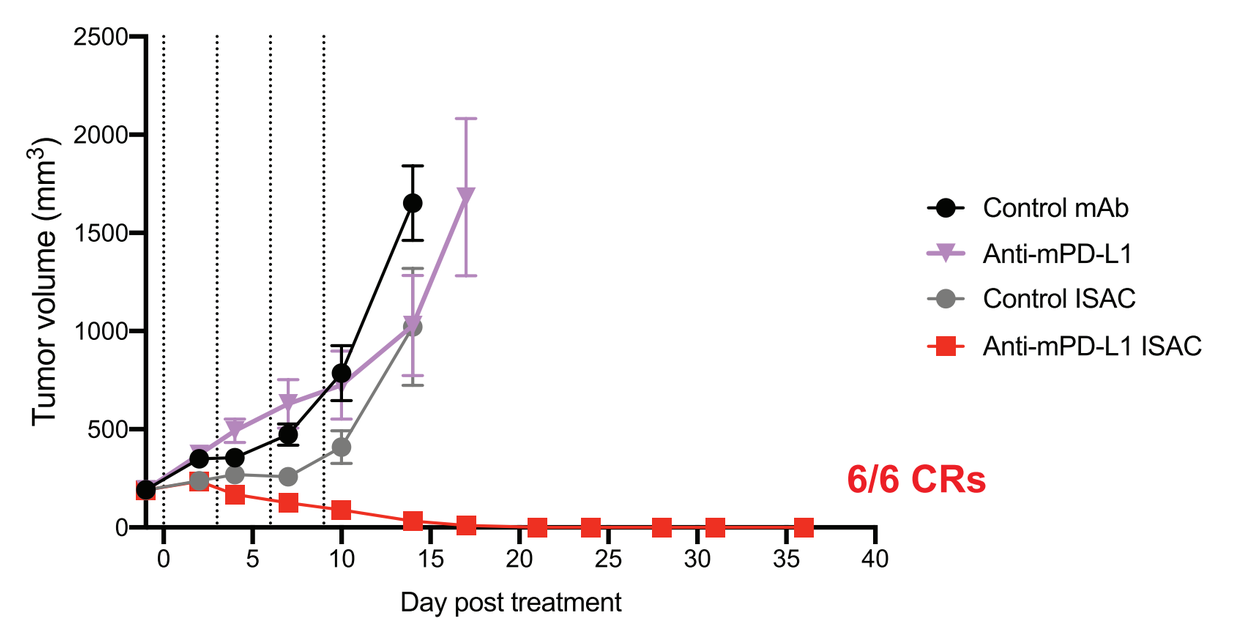
6/6 CRs

- Large (~200 mm³) syngeneic MB49 tumors were treated with the indicated mAbs or ISACs at 10 mg/kg IP, Q3Dx4 (n=6 per group)
- Treatment with PD-L1 ISAC resulted in improved anti-tumor efficacy compared to anti-PD-L1 treatment
- Treatment with anti-mPD-L1 ISAC resulted in complete responses (CRs) in all 6 animals

Assessment of immunological memory in re-challenge study

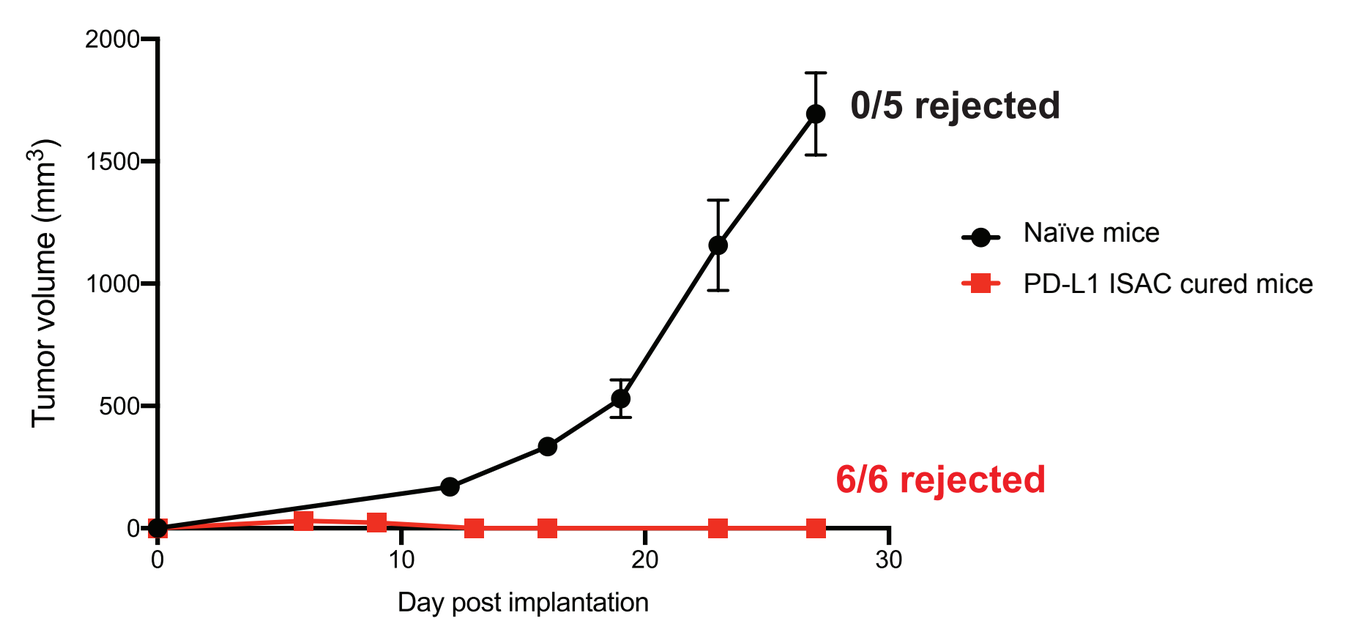
0/5 rejected
6/6 rejected
Naïve mice
PD-L1 ISAC cured mice

- All 6 mice that experienced complete regression of MB49 tumors following anti-mPD-L1 ISAC treatment were re-challenged with MB49 cells ~1 month after tumor clearance (~7 weeks post final dose). No new treatment was given after the re-challenge. None of mice developed tumors
- Naïve mice were challenged with MB49 tumors as control. Tumors from all 5 naïve mice grew without regression

mPD-L1 ISAC can induce anti-tumor response through PD-L1 on immune cells

Tumor growth inhibition of parental MB49 with mPD-L1 ISAC

Tumor growth inhibition of MB49 PD-L1 KO with mPD-L1 ISAC

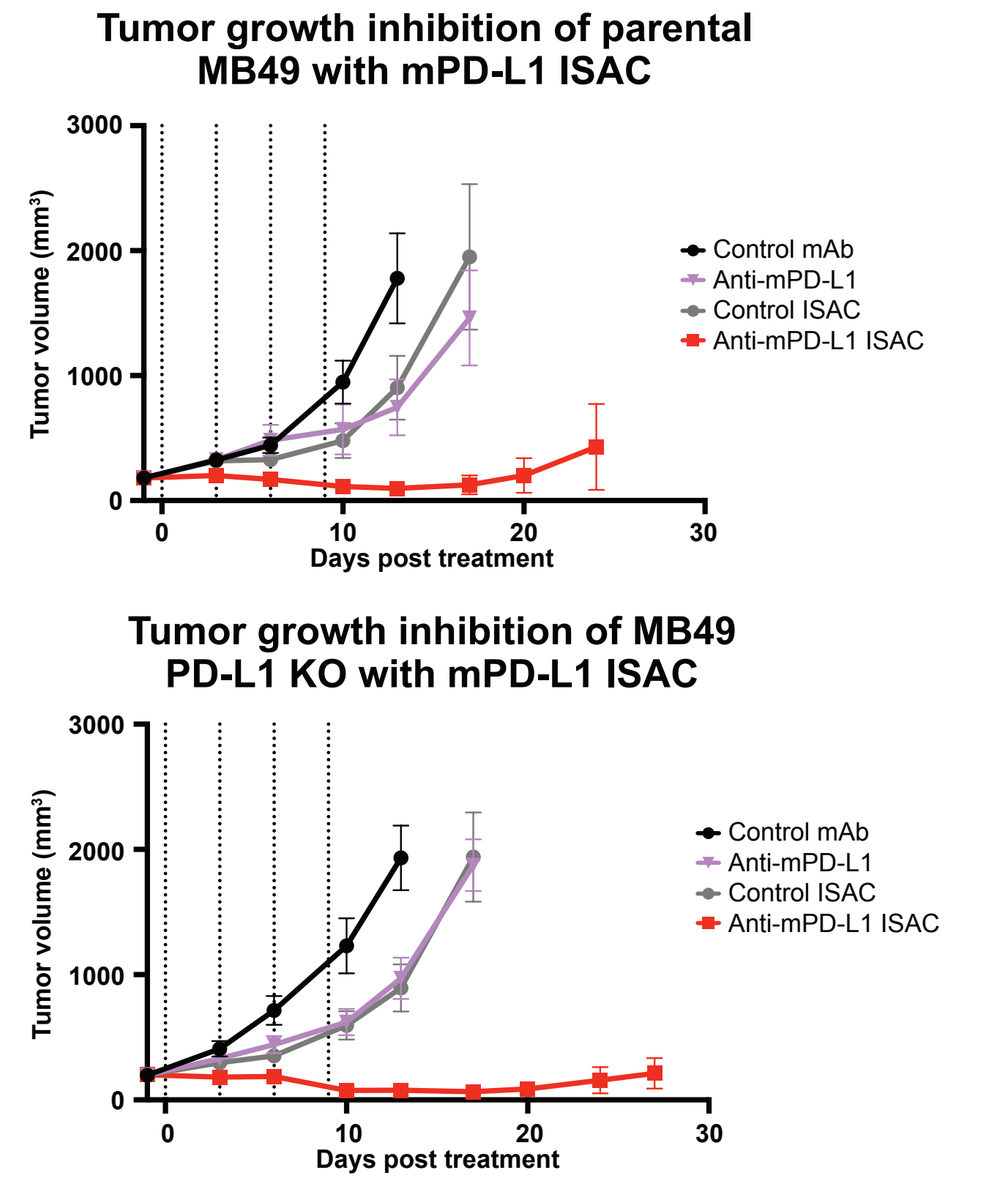

PD-L1 expression in MB49 and MB49-PD-L1 KO tumors:

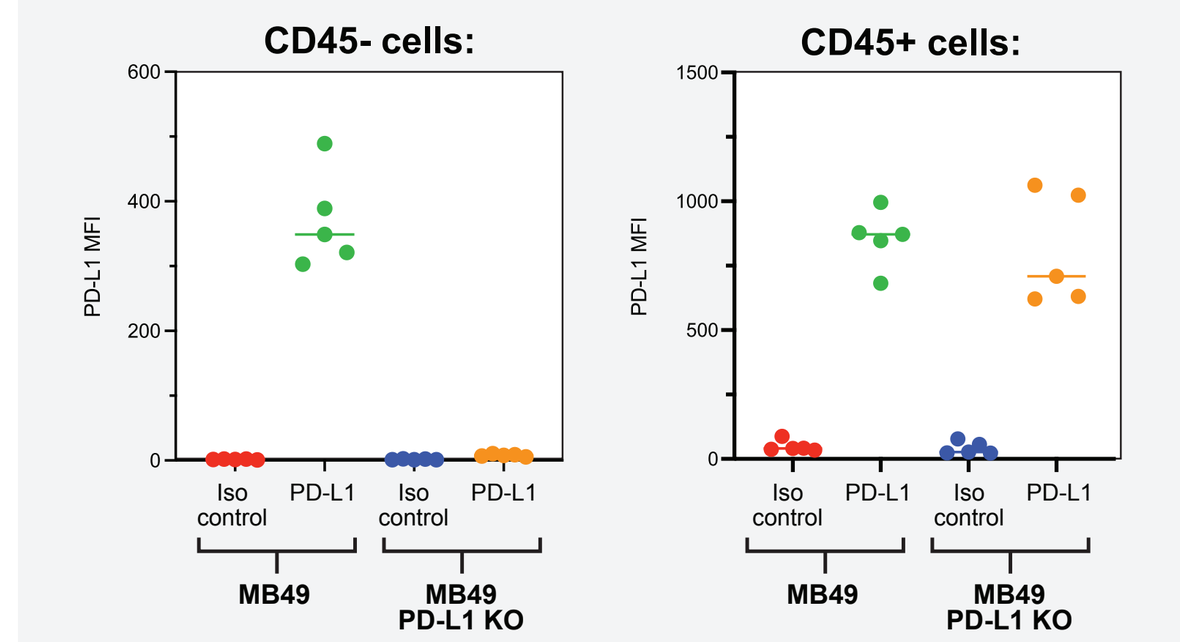
CD45- cells:
CD45+ cells:

PD-L1 expression in various cell types in MB49 tumors in vivo:

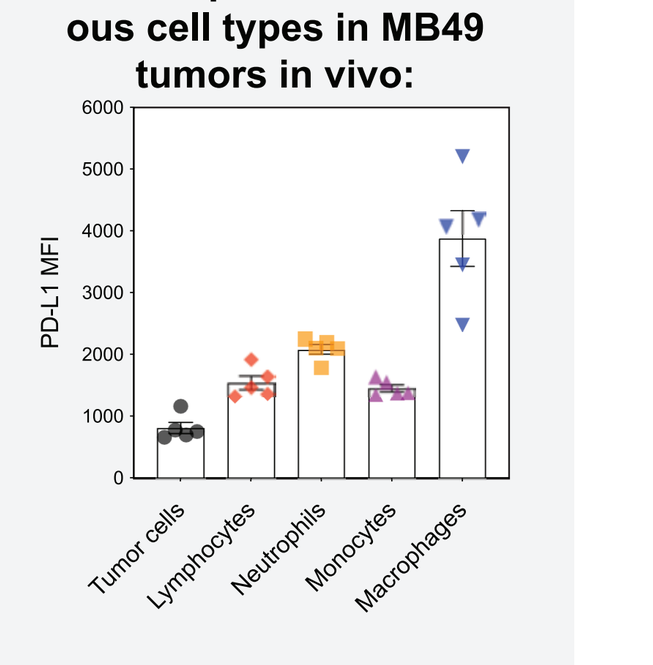

- Syngeneic MB49 tumors (parental or PD-L1 KO) were treated with the indicated mAbs or ISACs at 5 mg/kg IP, Q3Dx4
- Treatment with PD-L1 ISAC resulted in improved anti-tumor efficacy even in the absence of PD-L1 expression on tumor cells
- **Right panels:** PD-L1 expression in indicated cells in parental and PD-L1 KO MB49 tumors in vivo was evaluated with a flow cytometry

PD-L1 ISAC improves anti-tumor response over anti-PD-L1 treatment in syngeneic and xenograft tumor models expressing hPD-L1

Syngeneic MC38-hPD-L1

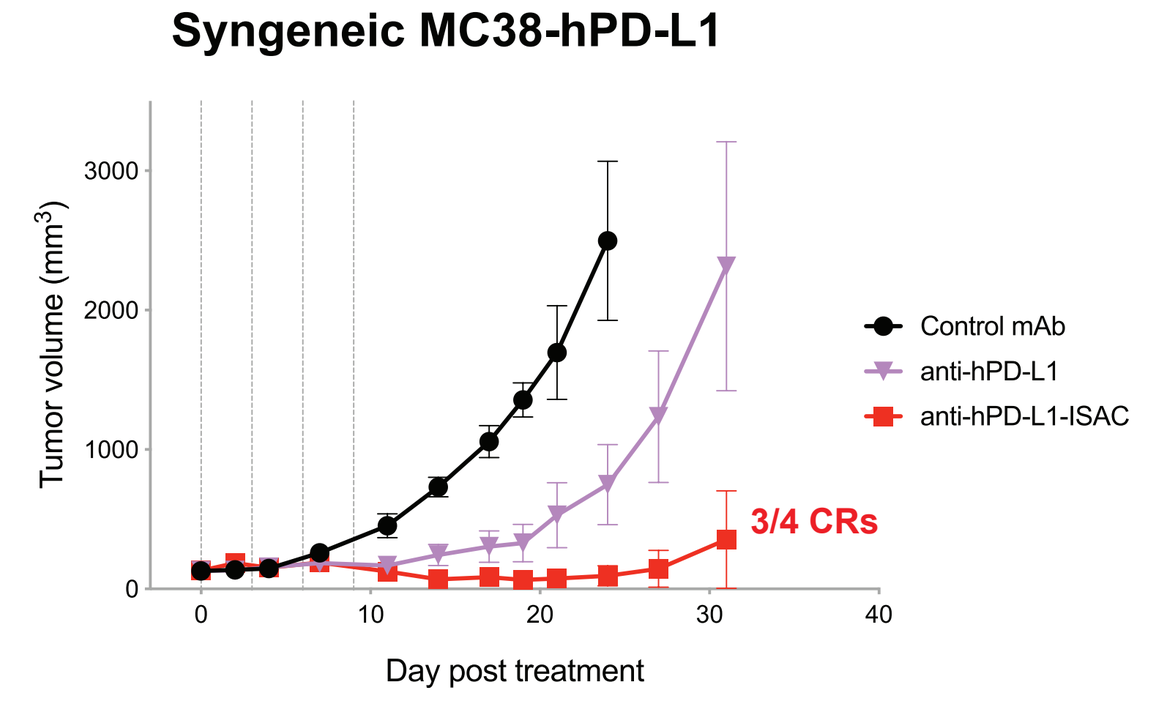
3/4 CRs

Xenograft HCC1954-hPD-L1

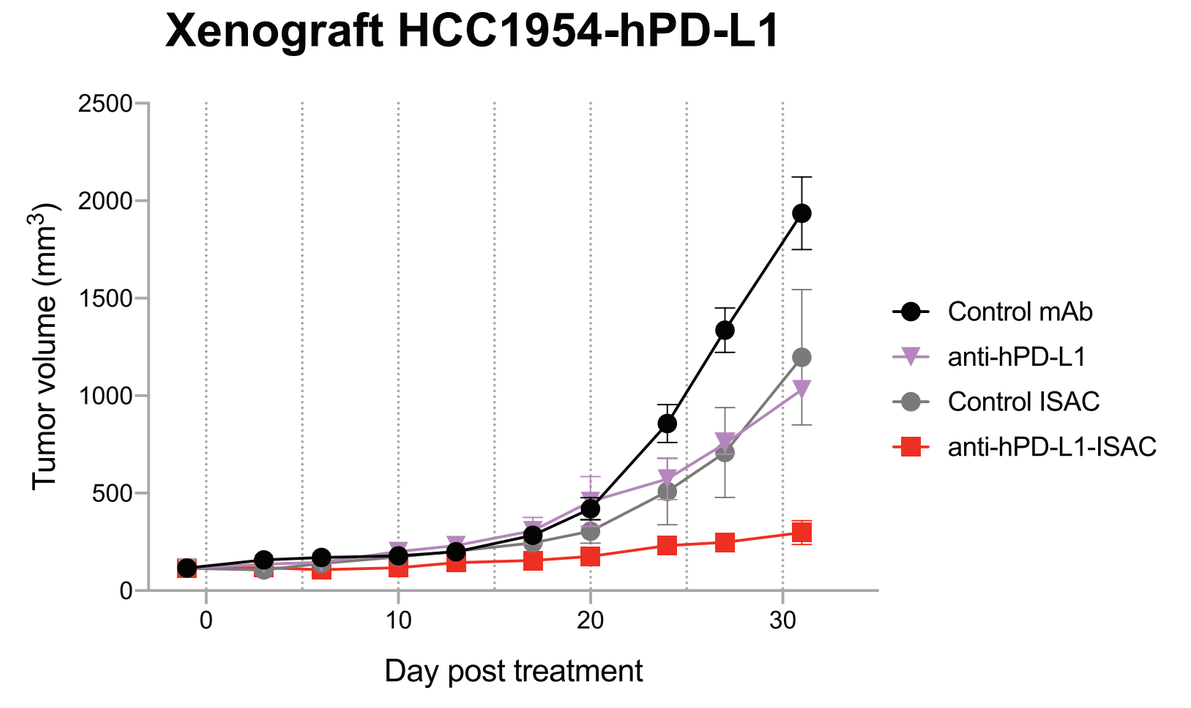

- Syngeneic MC38-hPD-L1 (left panel) or xenograft HCC1954-hPD-L1 (right panel) tumors were treated with the indicated mAbs or ISACs at 5mg/kg IP (MC38: Q3Dx4; HCC1954: Q5Dx7) (n=4-5 per group)
- Treatment with anti-hPD-L1 ISACs resulted in improved anti-tumor efficacy compared to anti-PD-L1 treatment

CONCLUSIONS

- **PD-L1-targeted ISAC is a novel multifunctional therapeutic that has improved efficacy of PD-L1/PD-1 inhibition preclinically in syngeneic and xenograft tumor models**
- **PD-L1 ISAC combines three mechanisms of action (MoA): myeloid cell activation, immune checkpoint inhibition, and ADCP and can induce its MoA through PD-L1 expressed on either tumor or immune cells**
- **PD-L1 ISACs induce robust, target-dependent activation of the immune system that leads to effective anti-tumor response that is substantially improved over the PD-L1 antibody blockade**
- **PD-L1 ISAC induces immunological memory**
- **Systemically delivered PD-L1 ISACs are well-tolerated in mice (data not shown)**
- **These data warrant continued research and development of PD-L1 ISAC with a potential to improve efficacy in patients including those with tumor types that do not responding to current immune checkpoint therapy**