BOLT BIOTHERAPEUTICS

Dectin-2, a Novel Target for Tumor Macrophage Reprogramming in Cancer Immunotherapy

Justin A. Kenkel[1,2], Po Y. Ho[1], Sameera Kongara[2], Karla A. Henning[1], Cindy L. Kreder[1], Jess L. Nolin[1], Steven J. Chapin[1], Marcin Kowanetz[1], Michael N. Alonso[1,2], Shelley E. Ackerman[1,3], Edgar G. Engleman[2], and David Dornan[1]

[1]Bolt Biotherapeutics, Redwood City, CA, USA; [2]Stanford University School of Medicine, Department of Pathology, Stanford, CA, USA; [3]Stanford University Schools of Medicine and Engineering, Department of Bioengineering, Stanford, CA, USA

ABSTRACT 862

INTRODUCTION

Tumor-associated macrophages (TAMs) are an abundant immune cell population in most cancers that support tumor progression through their immunosuppressive effects. We discovered that TAMs express the pattern recognition receptor Dectin-2 *(Clec4n/CLEC6A)*, an activating C-type lectin receptor (CLR) that binds to high-mannose glycans on fungi and other microbes and induces protective immune responses against infectious disease. Dectin-2 is selectively expressed by myeloid cells, and upon ligation, mediates enhanced phagocytosis, antigen processing and presentation, and pro-inflammatory cytokine production. Given these properties, we evaluated the therapeutic potential of targeting Dectin-2 using naturally derived ligands. We also generated human Dectin-2-targeted agonistic antibodies capable of robustly activating immunosuppressive "M2" or TAM-like macrophages.

Dectin-2 agonism activates TAMs and elicits anti-tumor immune response

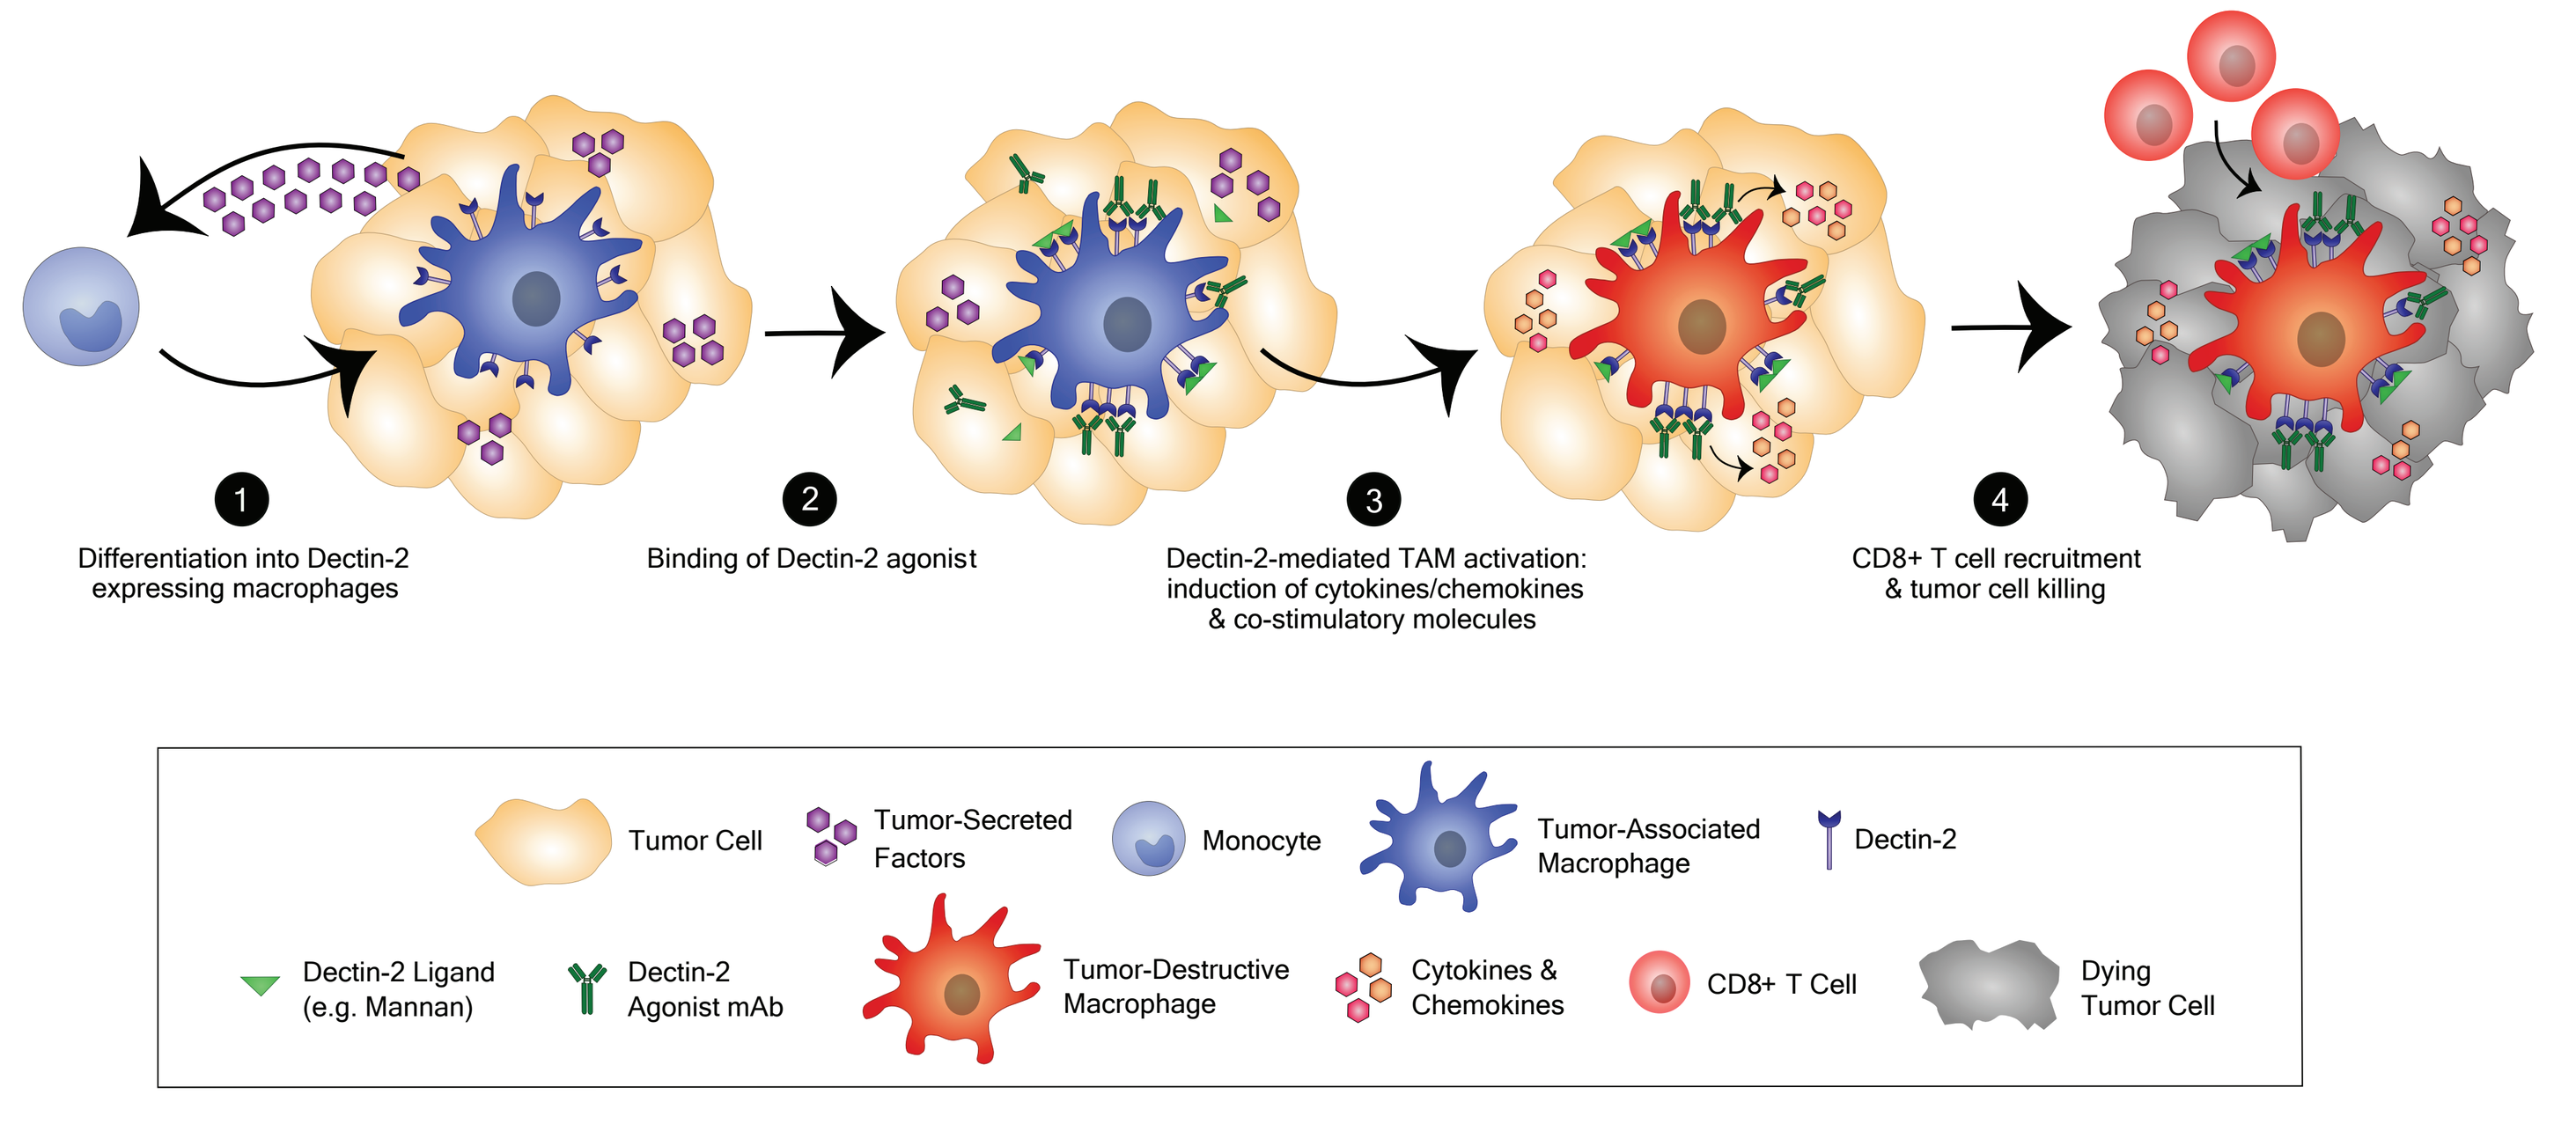
1 Differentiation into Dectin-2 expressing macrophages
2 Binding of Dectin-2 agonist
3 Dectin-2-mediated TAM activation: induction of cytokines/chemokines & co-stimulatory molecules
4 CD8+ T cell recruitment & tumor cell killing
Tumor Cell
Tumor-Secreted Factors
Monocyte
Tumor-Associated Macrophage
Dectin-2
Dectin-2 Ligand (e.g. Mannan)
Dectin-2 Agonist mAb
Tumor-Destructive Macrophage
Cytokines & Chemokines
CD8+ T Cell
Dying Tumor Cell

Figure 1: Schematic of proposed mechanism of action driving Dectin-2-mediated anti-tumor activity.

RESULTS

Dectin-2 gene expression is elevated across tumor types

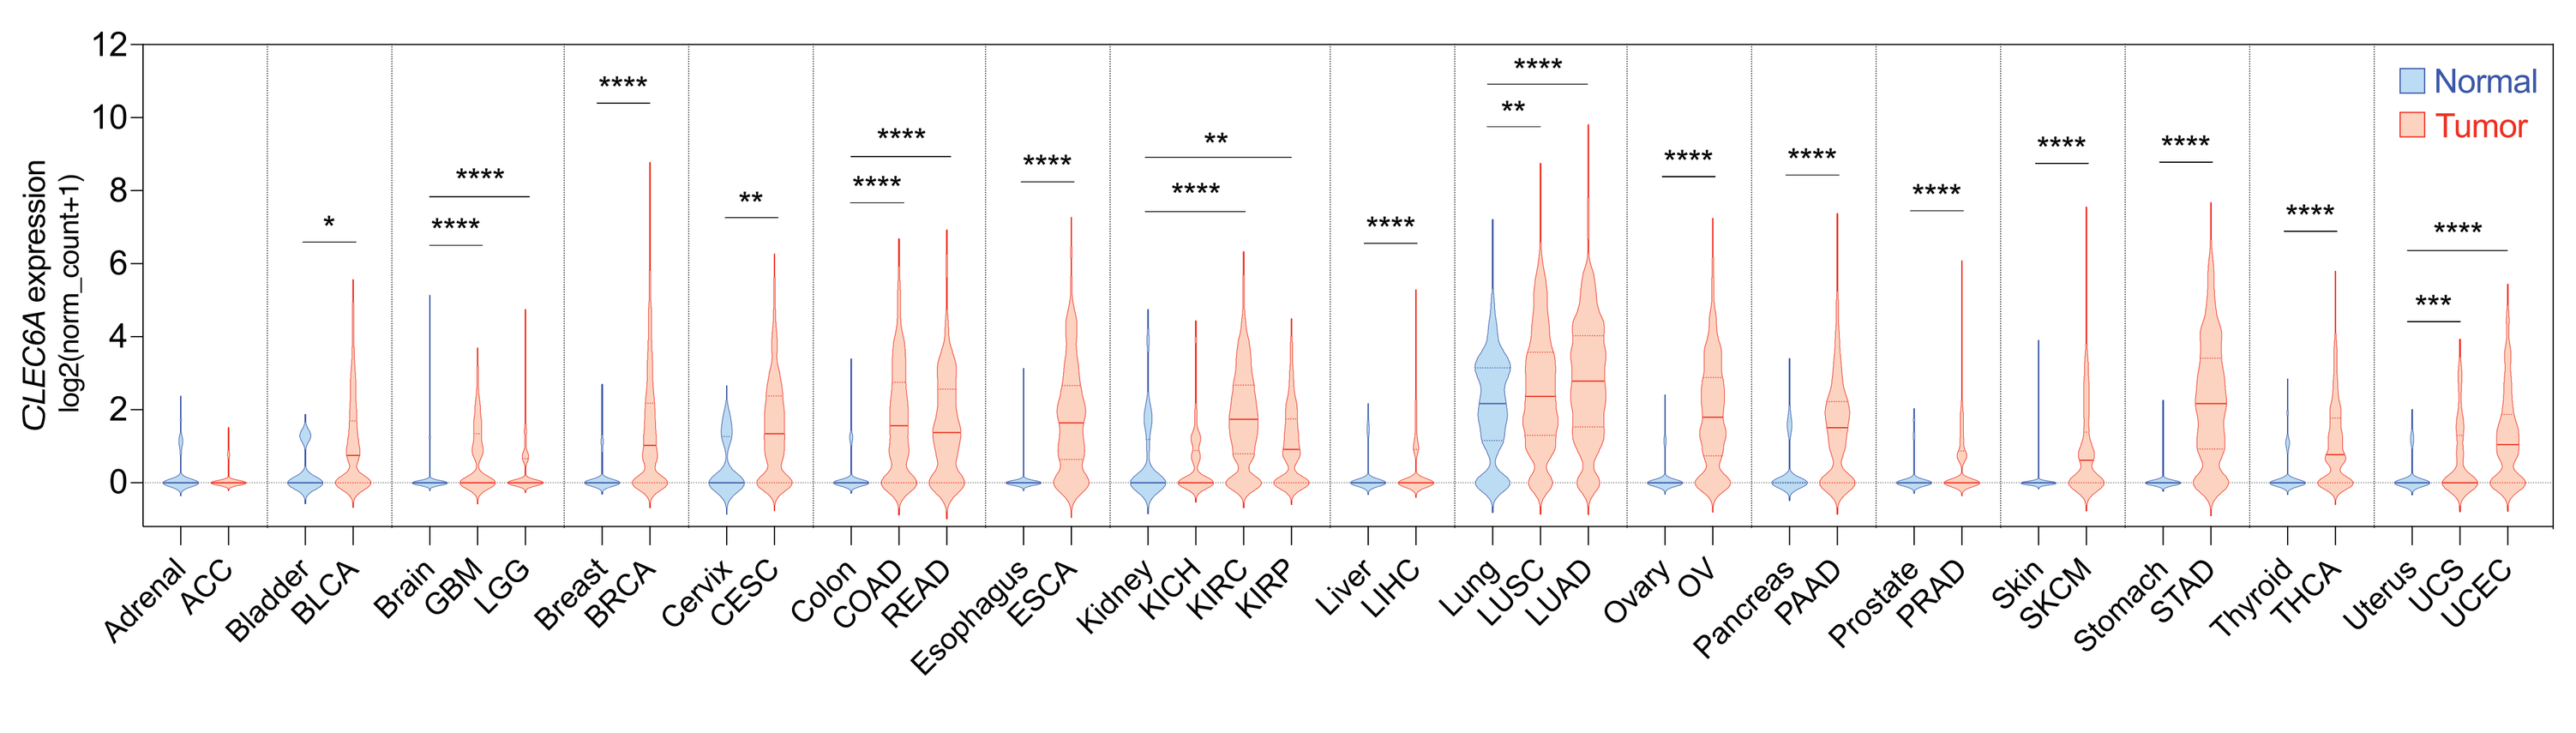
CLEC6A expression log2(norm_count+1)
Normal
Tumor
Adrenal ACC Bladder BLCA Brain GBM LGG Breast BRCA Cervix CESC Colon COAD READ Esophagus ESCA Kidney KICH KIRC KIRP Liver LIHC Lung LUSC LUAD Ovary OV Pancreas PAAD Prostate PRAD Skin SKCM Stomach STAD Thyroid THCA Uterus UCS UCEC

Figure 2: Dectin-2 gene expression is elevated in tumors but low in most normal tissues. Dectin-2/*CLEC6A* mRNA expression in human tissue samples from the TCGA (tumor) and GTEX (normal) datasets. TCGA study abbreviations are shown for the tumor subtypes. Data were processed using a uniform bioinformatic pipeline and obtained from UCSC Xena (xena.ucsc.edu). Data shown as median with interquartile range. Statistics were calculated by Mann-Whitney U test; *$P \le 0.05$, **$P \le 0.01$, ***$P \le 0.001$, ****$P \le 0.0001$.

Dectin-2 is selectively expressed by TAMs in a range of human tumors

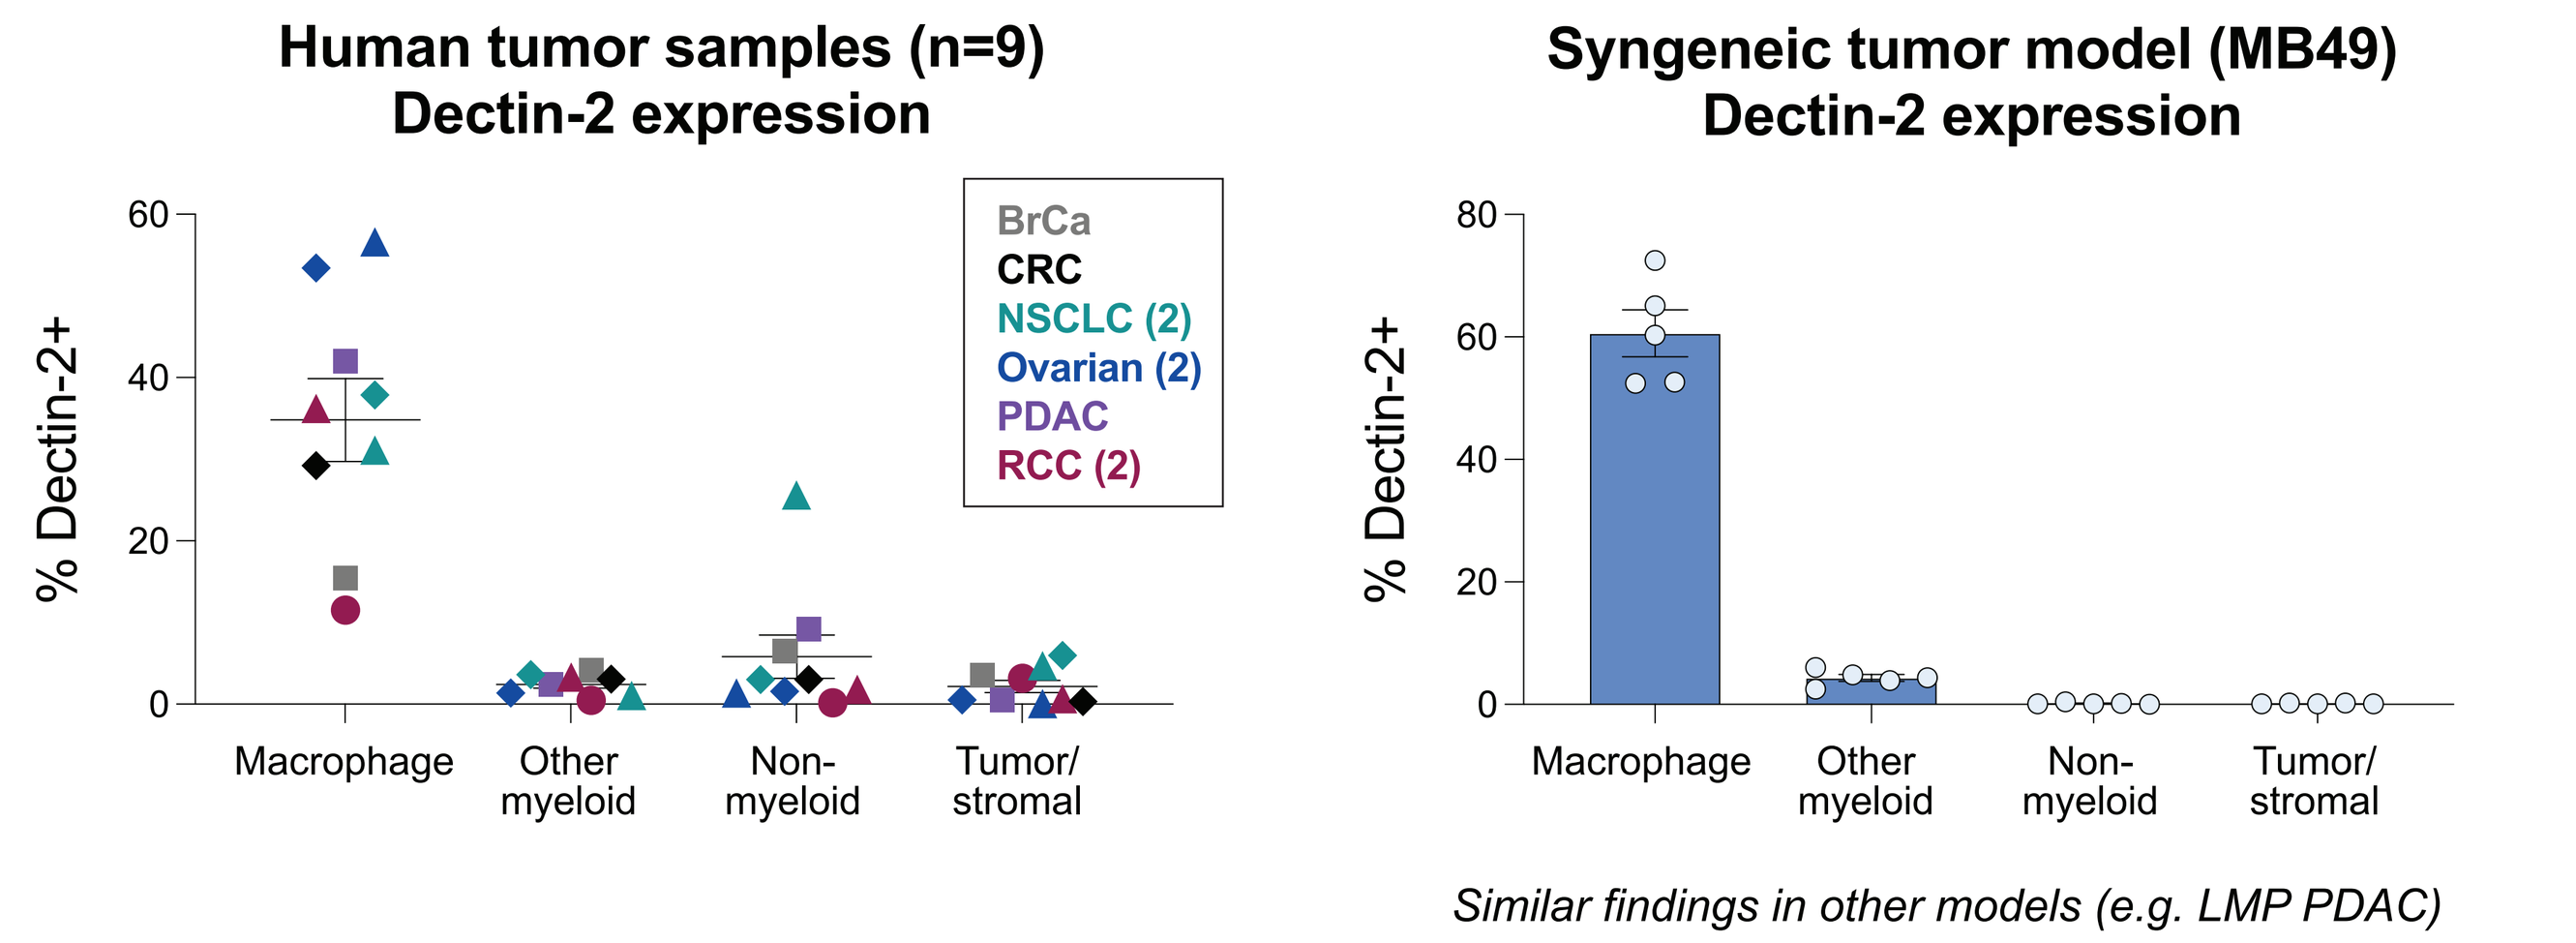
Human tumor samples (n=9)
Dectin-2 expression
Syngeneic tumor model (MB49)
Dectin-2 expression
% Dectin-2+
BrCa
CRC
NSCLC (2)
Ovarian (2)
PDAC
RCC (2)
Macrophage
Other myeloid
Non-myeloid
Tumor/ stromal
Similar findings in other models (e.g. LMP PDAC)

Figure 3: Dectin-2 is expressed by TAMs in primary human tumor samples. (Left) Human tumors were processed into single-cell suspensions and analyzed by flow cytometry. TAMs were defined as viable CD45+CD11b+CD14+HLA-DR+ cells. (Right) Tumors were harvested from MB49- or LMP-bearing mice and analyzed as single-cell suspensions by flow cytometry for Dectin-2 expression.

Dectin-2 agonism activates murine TAMs in vitro and in vivo

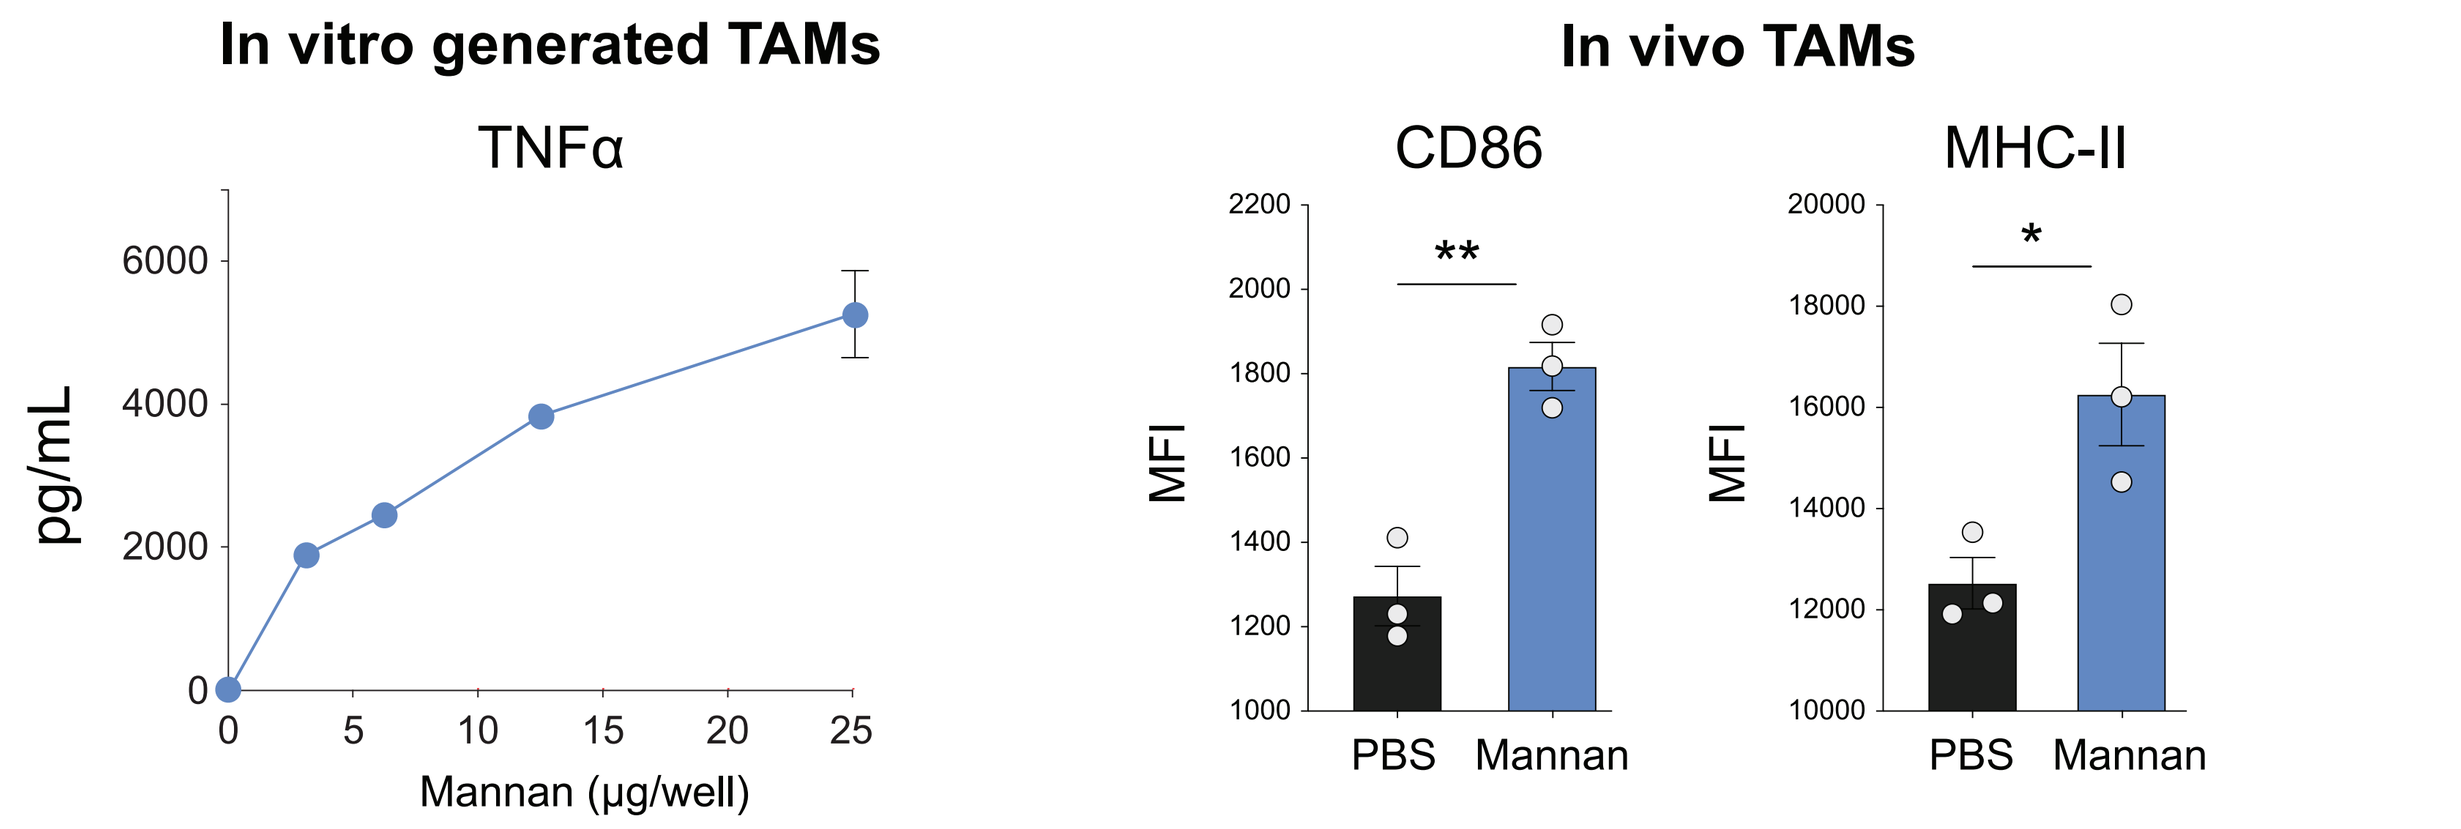
In vitro generated TAMs
TNFα
pg/mL
Mannan (μg/well)
In vivo TAMs
CD86
MHC-II
MFI
PBS
Mannan

Figure 4: Mannan, a natural ligand for Dectin-2, activates TAMs. (Left) TNFα secretion by in vitro-generated murine TAMs stimulated overnight with plate-bound *S. cerevisiae* mannan. (Right) Murine TAMs from LMP PDAC tumors were analyzed by flow cytometry for activation markers 6 hours following treatment with systemically administered mannan or PBS control. Data are shown as mean with SEM (n=3 mice per group).

Dectin-2 agonism mediates tumor regression in syngeneic models

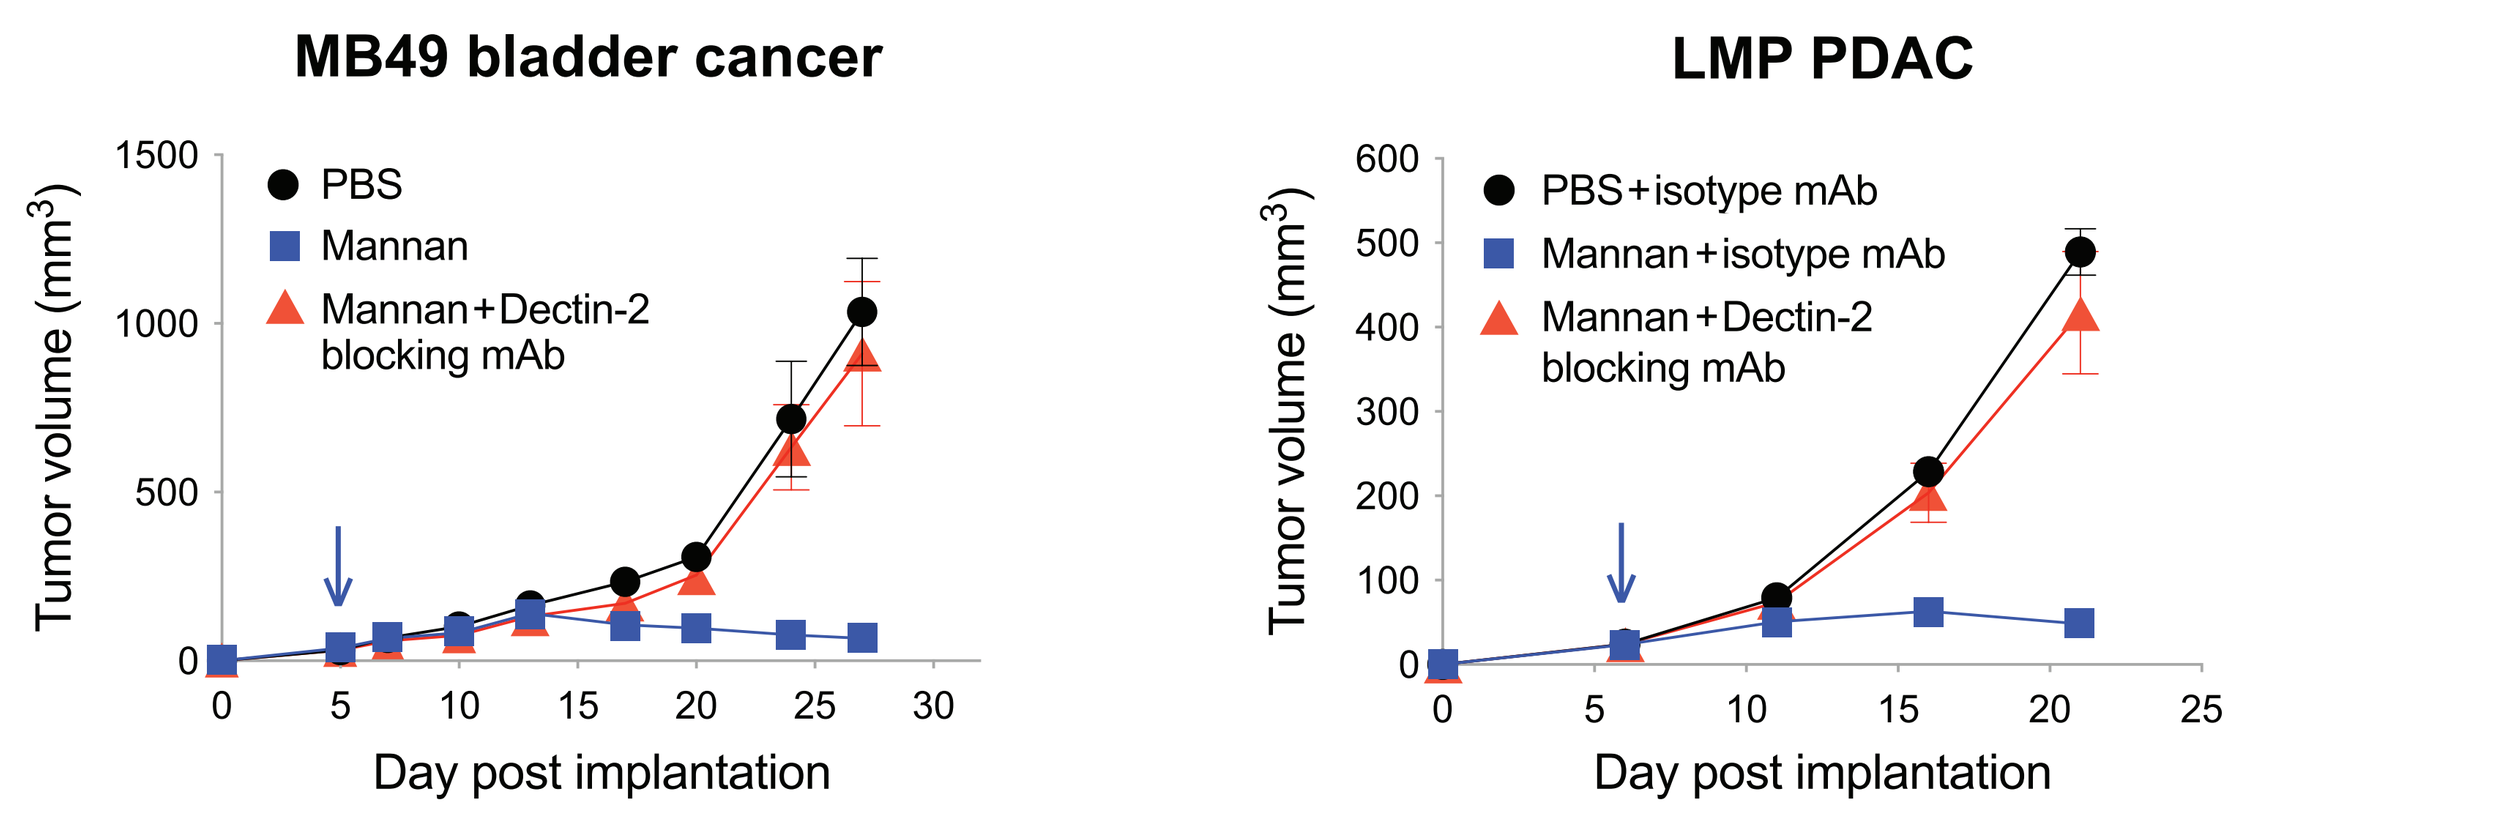
MB49 bladder cancer
PBS
Mannan
Mannan+Dectin-2 blocking mAb
LMP PDAC
PBS+isotype mAb
Mannan+isotype mAb
Mannan+Dectin-2 blocking mAb
Tumor volume (mm³)
Day post implantation

Figure 5: Mannan elicits tumor regression in a Dectin-2-dependent manner. MB49 (Left) or LMP (Right) tumor-bearing mice were treated systemically every 2 days with *S. cerevisiae* mannan (12.5 mg/kg iv) with or without co-administration of Dectin-2 blocking antibody (5-10 mg/kg ip). Data are shown as mean with SEM and n=4-5 mice per group; day of treatment initiation is marked with a blue arrow.

Anti-tumor efficacy requires Dectin-2 signaling components

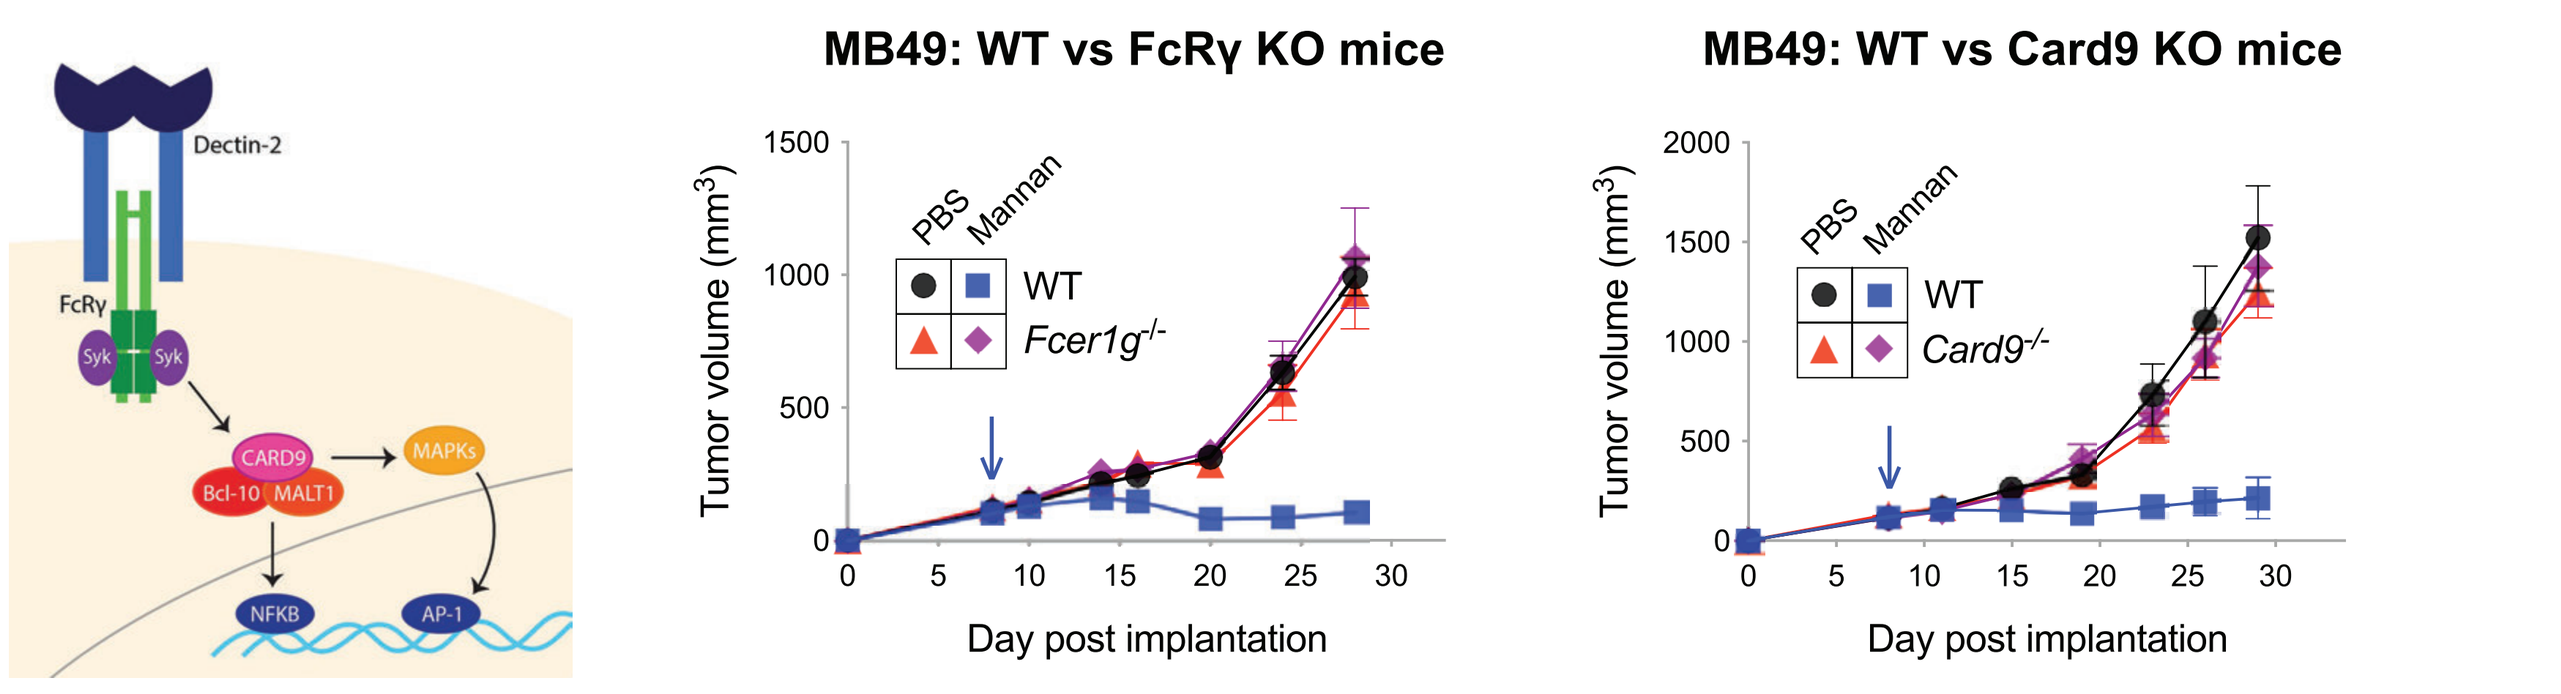
Dectin-2
FcRγ
Syk
CARD9
Bcl-10
MALT1
MAPKs
NFKB
AP-1
MB49: WT vs FcRγ KO mice
MB49: WT vs Card9 KO mice
PBS
Mannan
WT
Fcer1g-/-
Card9-/-
Tumor volume (mm³)
Day post implantation

Figure 6: Mannan efficacy requires Dectin-2 signaling components. MB49 tumor-bearing WT or KO (FcRγ or Card9) mice were treated systemically every 2 days with 10 mg/kg mannan. Data are shown as mean with SEM and n=5-6 mice per group; day of treatment initiation is marked with a blue arrow.

Dectin-2 agonist activity is CD8 T cell dependent and elicits immunological memory

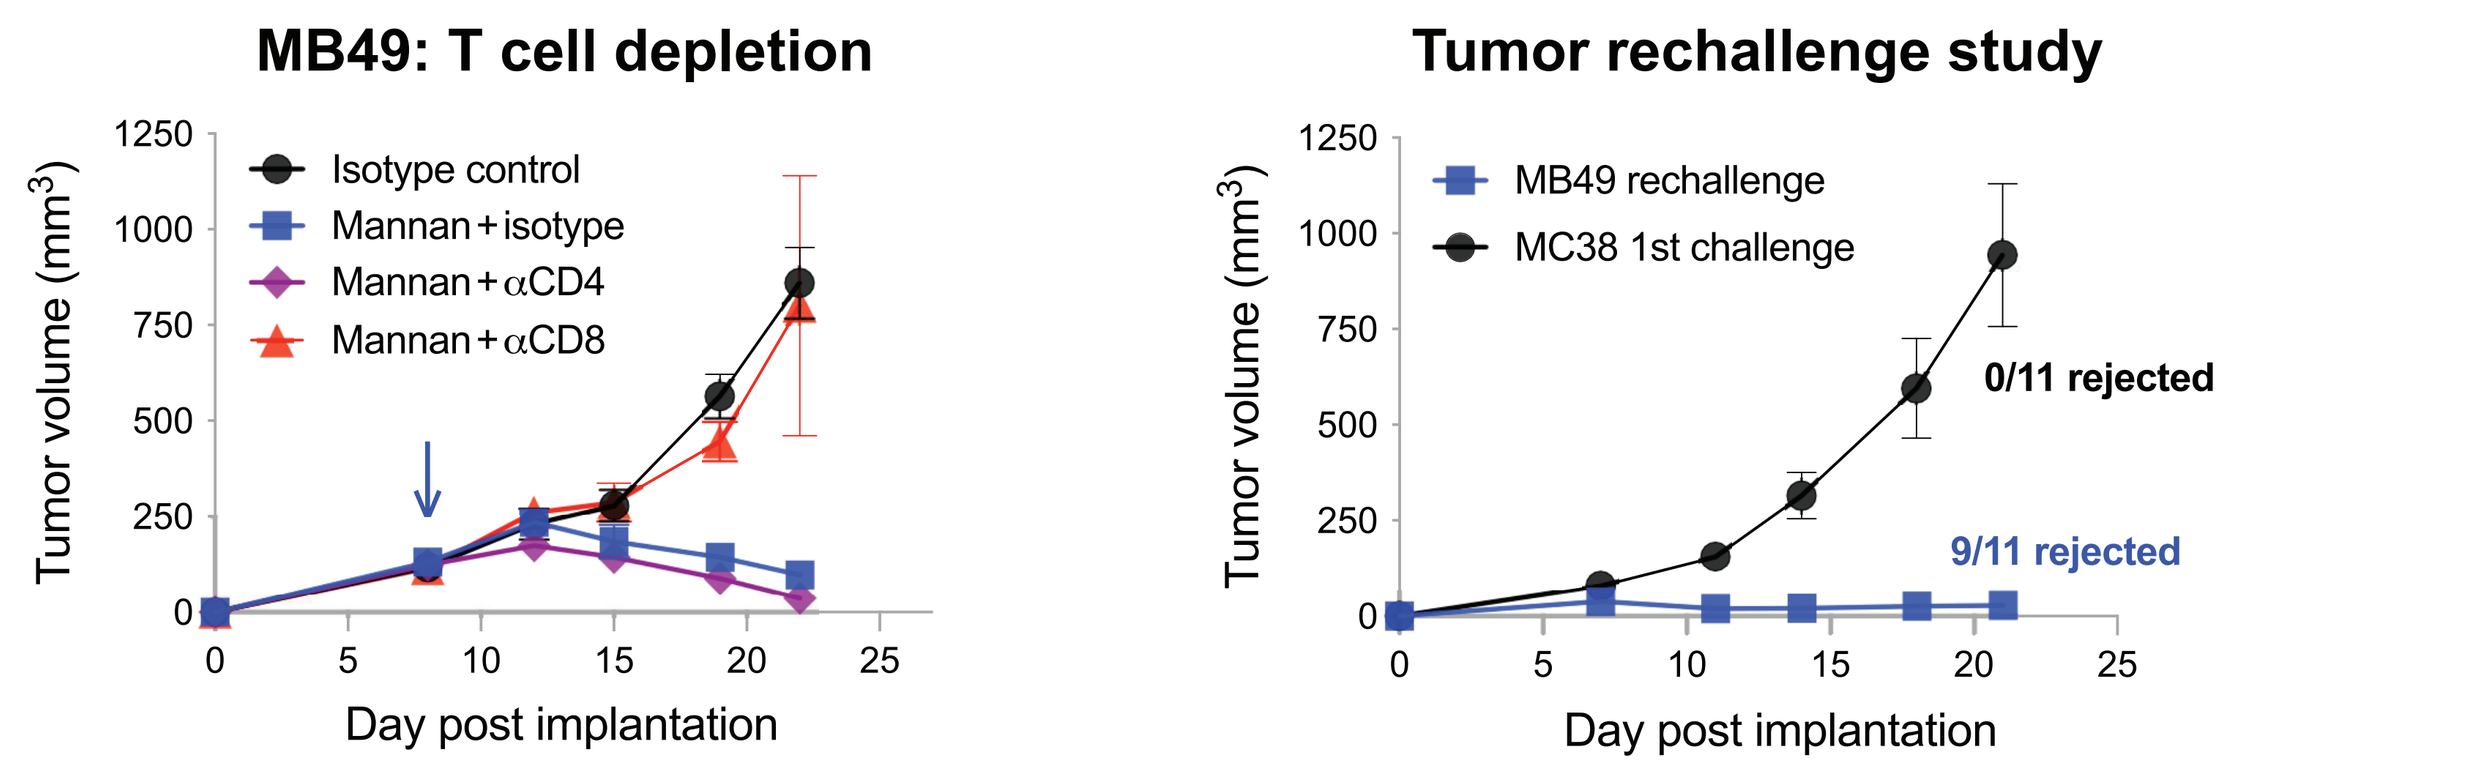
MB49: T cell depletion
Isotype control
Mannan+isotype
Mannan+αCD4
Mannan+αCD8
Tumor rechallenge study
MB49 rechallenge
MC38 1st challenge
0/11 rejected
9/11 rejected
Tumor volume (mm³)
Day post implantation

Figure 7: Dectin-2 agonist activity is CD8 T cell dependent and elicits immunological memory. (Left) MB49 tumor-bearing mice were treated systemically with mannan, and 250 ug of isotype IgG, anti-CD4, or anti-CD8 depleting antibody every 3 days (n=4-5 mice, day of treatment initiation marked with a blue arrow). (Right) Mice that experienced complete regression of MB49 tumors following mannan treatment were rechallenged with MB49 cells as well as an unrelated tumor cell line (MC38) (n=11 mice). Data are shown as mean with SEM.

Agonist mAbs bind Dectin-2 and activate human macrophages

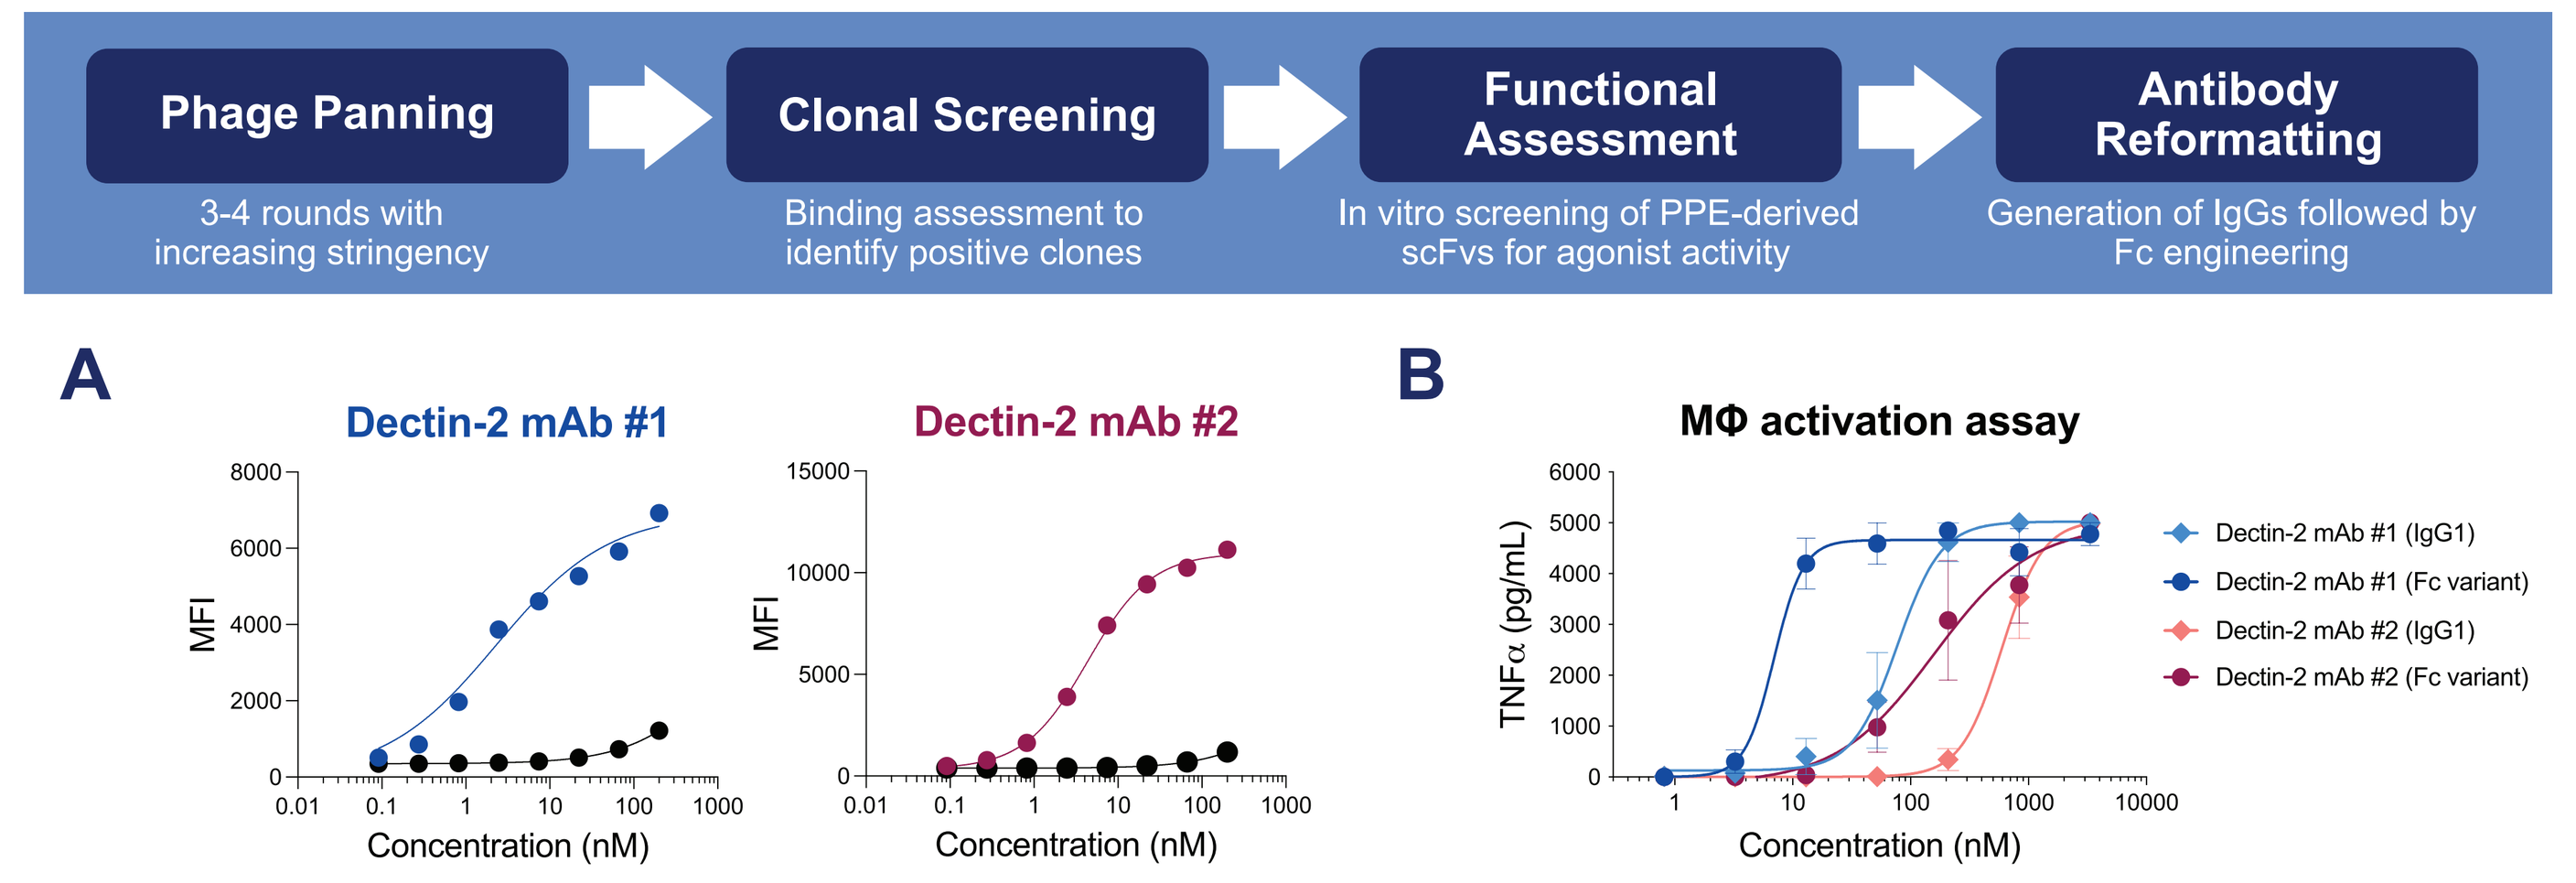
Phage Panning
3-4 rounds with increasing stringency
Clonal Screening
Binding assessment to identify positive clones
Functional Assessment
In vitro screening of PPE-derived scFvs for agonist activity
Antibody Reformatting
Generation of IgGs followed by Fc engineering
A
Dectin-2 mAb #1
Dectin-2 mAb #2
MFI
Concentration (nM)
B
MΦ activation assay
TNFα (pg/mL)
Dectin-2 mAb #1 (IgG1)
Dectin-2 mAb #1 (Fc variant)
Dectin-2 mAb #2 (IgG1)
Dectin-2 mAb #2 (Fc variant)

Figure 8: Agonist mAbs bind Dectin-2 and activate human macrophages. (A) Dectin-2 agonist mAbs bind to cells expressing Dectin-2 with single digit nM EC50s, while minimal binding is detected with HEK293T cells lacking Dectin-2 expression (black circles). **(B)** Human monocytes isolated from healthy human blood (n=5) were differentiated with M-CSF for 5 days and then stimulated overnight with indicated mAbs. Activation was measured as TNFα secretion by ELISA.

Fc-engineered agonist mAb potently activates human macrophages

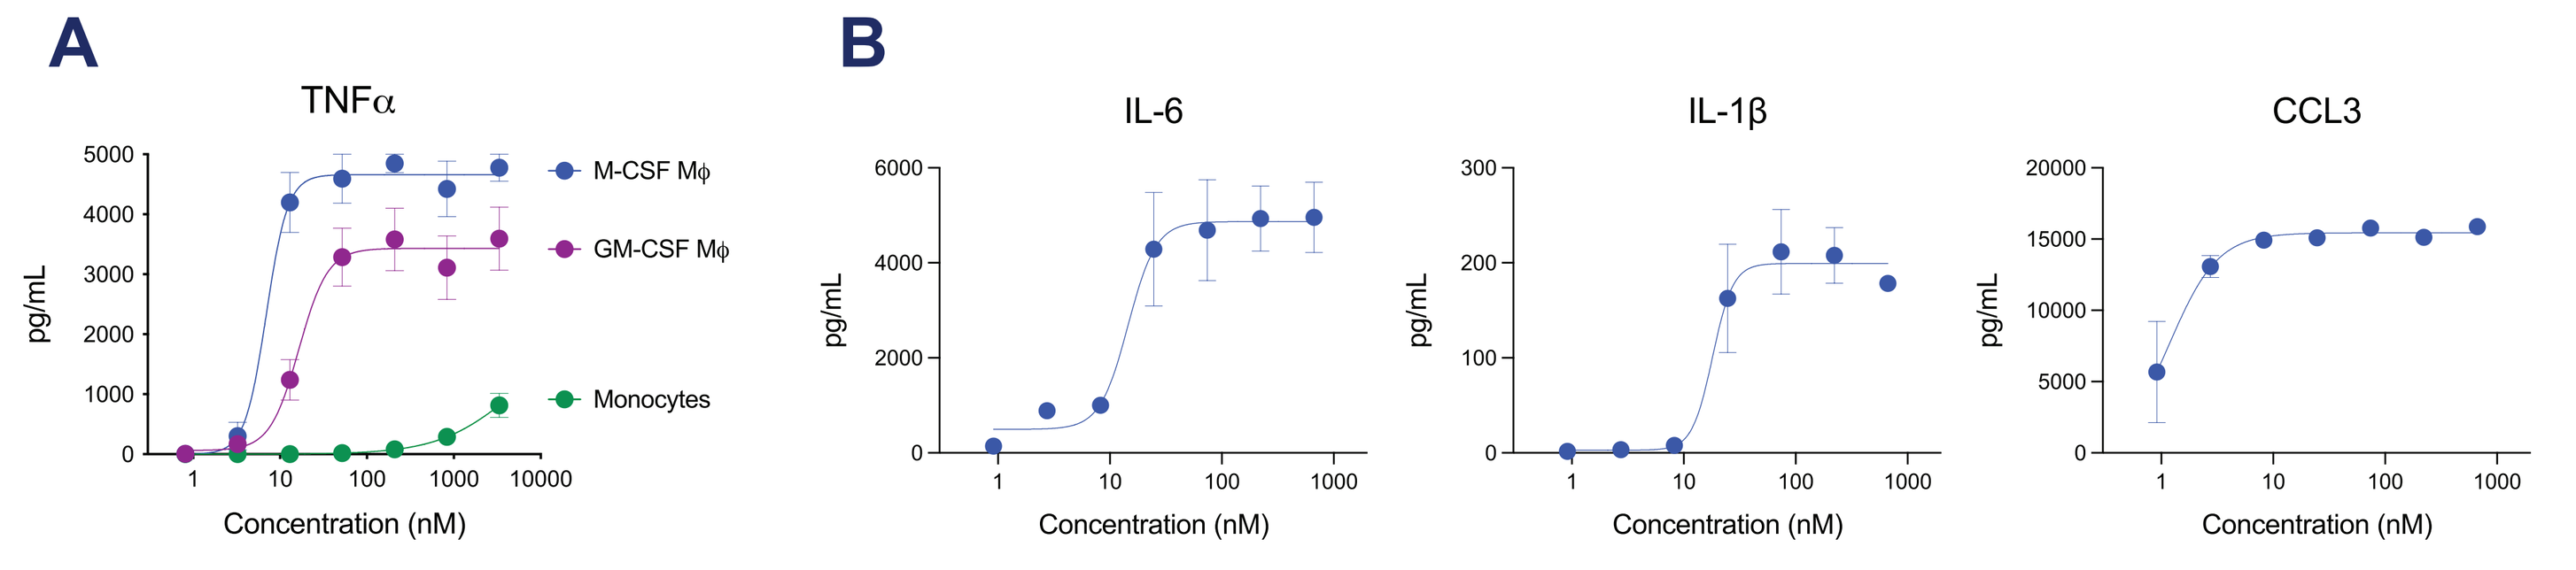
A
TNFα
M-CSF Mϕ
GM-CSF Mϕ
Monocytes
B
IL-6
IL-1β
CCL3
pg/mL
Concentration (nM)

Figure 9: Dectin-2 agonist mAb 1 (Fc variant) elicits proinflammatory cytokine and chemokine production by human macrophages. (A) Fresh human monocytes (n=12 donors) or monocyte-derived macrophages generated with M-CSF (n=5) or GM-CSF (n=12) were stimulated overnight with the Dectin-2 agonist mAb, followed by cytokine analysis by ELISA. **(B)** Human M-CSF macrophages (n=3) were stimulated overnight with the Dectin-2 mAb, followed by cytokine and chemokine analysis using MSD kits.

Dectin-2 agonist mAb activates primary human TAMs ex vivo

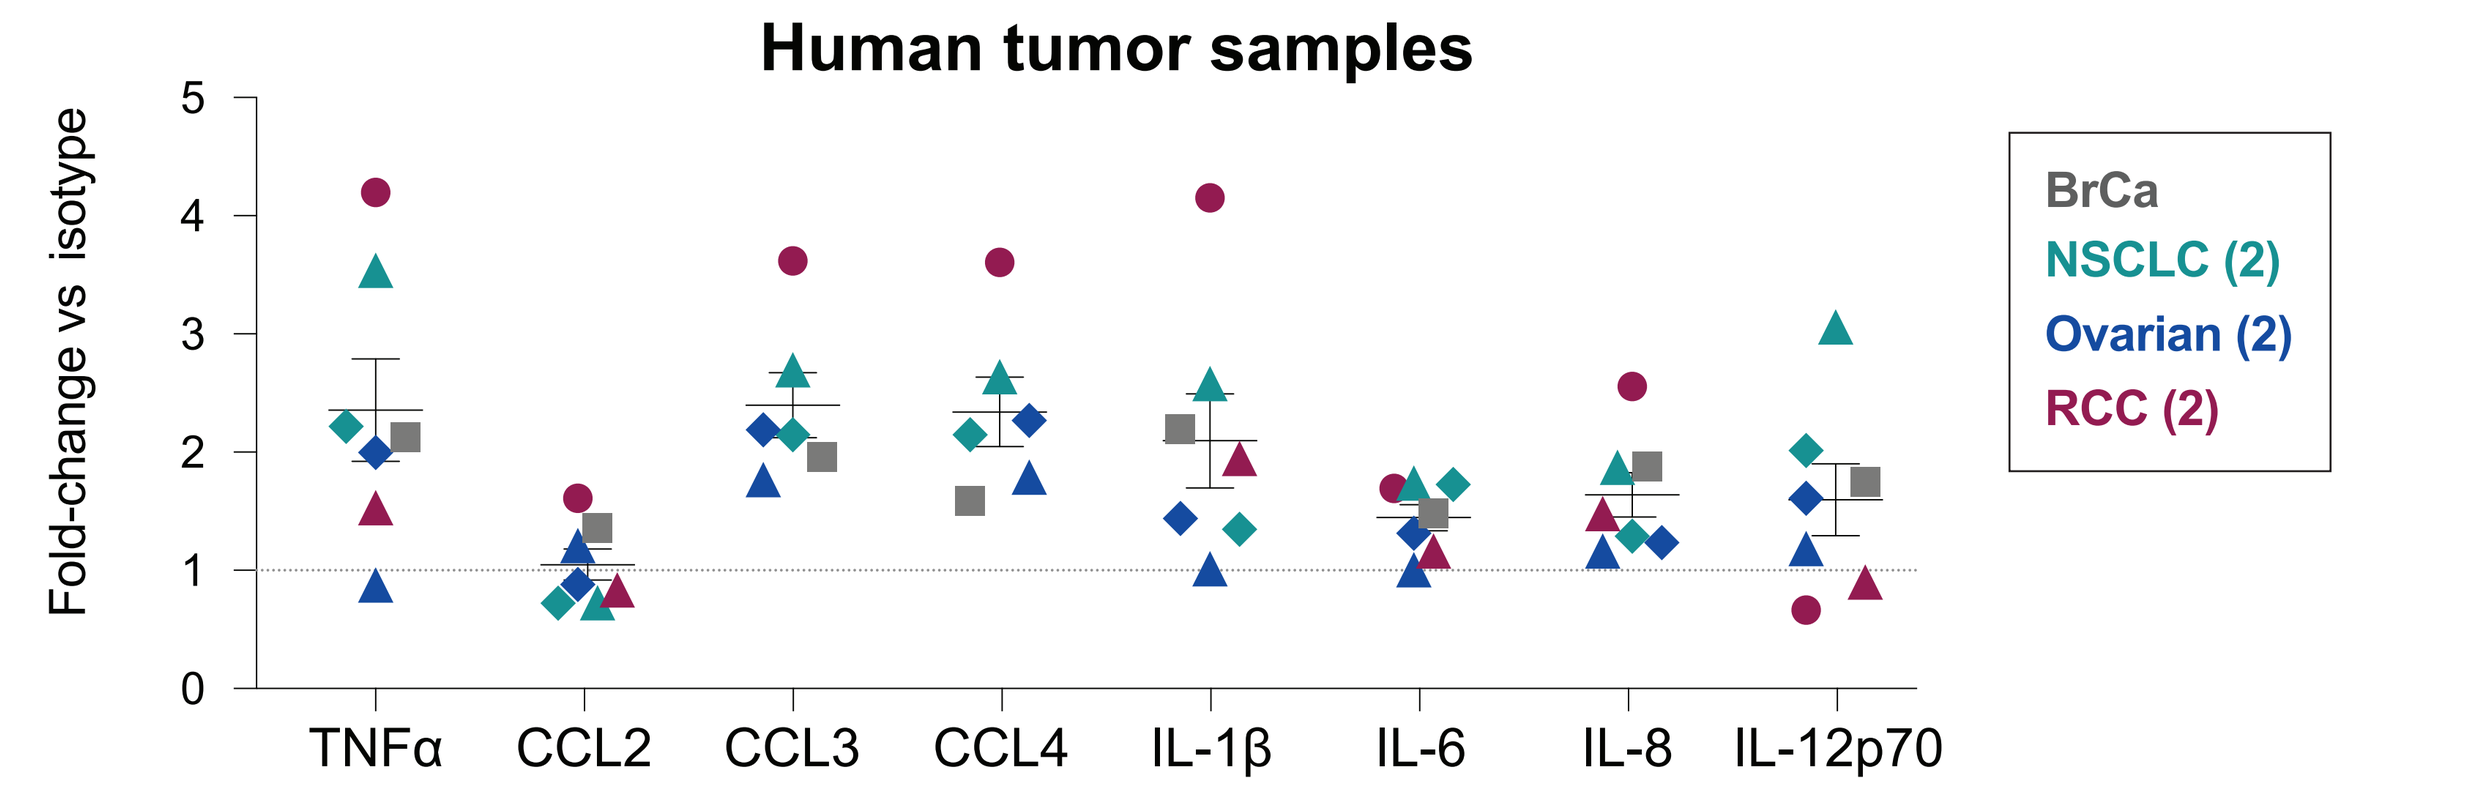
Human tumor samples
Fold-change vs isotype
BrCa
NSCLC (2)
Ovarian (2)
RCC (2)
TNFα
CCL2
CCL3
CCL4
IL-1β
IL-6
IL-8
IL-12p70

Figure 10: Dectin-2 agonist mAb 1 (Fc variant) activates primary human TAMs. Primary human tumor samples were processed into single-cell suspensions and cultured overnight with a Dectin-2 agonist antibody or non-binding isotype control antibody. Increased secretion of potent pro-inflammatory cytokines and chemokines was measured across all tumor samples tested. Data are shown as mean with SEM; Breast Cancer (BrCa, n=1), Non-Small Cell Lung Cancer (NSCLC, n=2), Ovarian (n=2), Renal Cell Carcinoma (RCC, n=2).

CONCLUSIONS

- **Dectin-2 is a novel target expressed by tumor-associated macrophages (TAMs)**
- **Agonism of Dectin-2 on TAMs elicits secretion of pro-inflammatory cytokines and chemokines to stimulate a productive anti-tumor immune response**
- **Dectin-2 agonism mediates anti-tumor efficacy in a CD8 T cell-dependent manner and elicits immunological memory**
- **Discovery and lead optimization identified a potent agonist antibody targeting Dectin-2 with an engineered Fc domain**
- **Bolt Biotherapeutics' agonist antibody has the potential to reprogram tumor-supportive macrophages into tumor-destructive macrophages as a novel anti-tumor immunotherapy**