BDC-2034: Discovery of a CEA-targeting Immune-Stimulating Antibody Conjugate (ISAC) for Solid Tumors

William G. Mallet, Laughing Bear Torrez Dulgeroff, Lisa K. Blum, Cecelia I. Pearson, Rishali Gadkari, Romas Kudirka, Jess L. Nolin, Andrew Luo, Angela Luo, Puneet Anand, Ganapathy Sarma, Karla A. Henning, Shelley E. Ackerman, Steven J. Chapin, Liz Bogaert, Michael N. Alonso, Marcin Kowanetz, Brian S. Safina, David Dornan

Bolt Biotherapeutics, 900 Chesapeake Drive, Redwood City, CA, USA

ABSTRACT 784

BACKGROUND

CEA (CEACAM5) is a well-validated cell-surface antigen that is highly expressed in multiple solid tumors. Upon systemic delivery, Bolt Biotherapeutics' pioneering immune-stimulating antibody conjugates (ISACs) use proprietary TLR7/8 dual agonists to activate tumor-infiltrating myeloid cells and initiate a broad innate and adaptive anti-tumor immune response.[1] The favorable properties of CEA, including robust cell surface expression, low internalization rate, and limited normal tissue expression, suggest that the antigen may be a suitable ISAC target. We are evaluating the CEA-targeting ISAC, BDC-2034, as a novel approach to treat CEA-expressing cancers.

Boltbody™ ISAC mechanism targets the innate immune system

Spreads to Adaptive Immune System for Optimal Anti-tumor Response

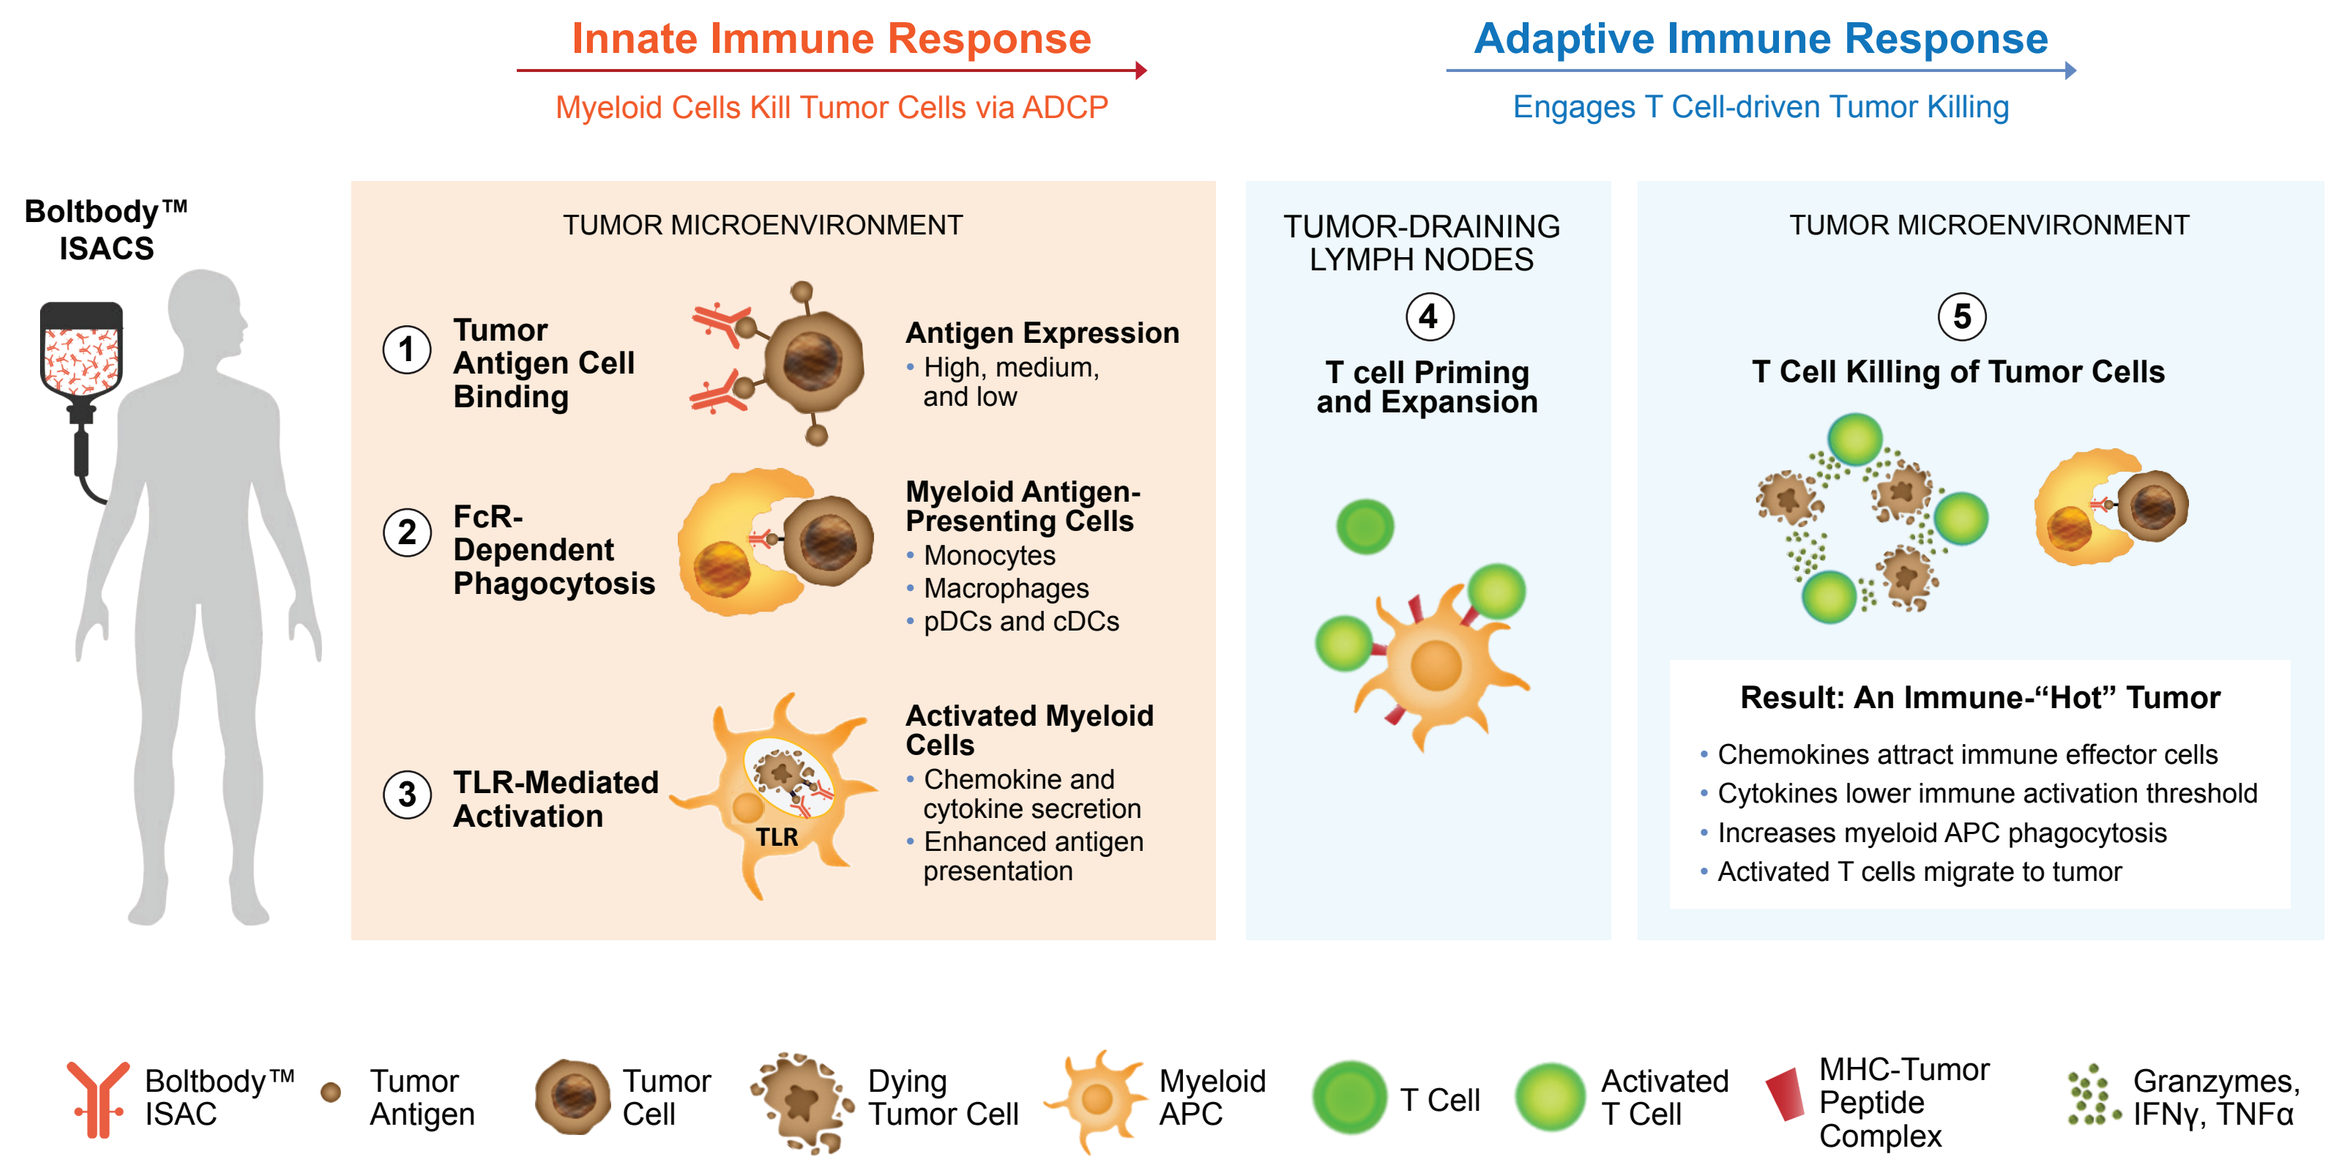
Innate Immune Response
Myeloid Cells Kill Tumor Cells via ADCP
Adaptive Immune Response
Engages T Cell-driven Tumor Killing
Boltbody™ ISACS
TUMOR MICROENVIRONMENT
1 Tumor Antigen Cell Binding
Antigen Expression
• High, medium, and low
2 FcR-Dependent Phagocytosis
Myeloid Antigen-Presenting Cells
• Monocytes
• Macrophages
• pDCs and cDCs
3 TLR-Mediated Activation
TLR
Activated Myeloid Cells
• Chemokine and cytokine secretion
• Enhanced antigen presentation
TUMOR-DRAINING LYMPH NODES
4 T cell Priming and Expansion
TUMOR MICROENVIRONMENT
5 T Cell Killing of Tumor Cells
Result: An Immune-"Hot" Tumor
• Chemokines attract immune effector cells
• Cytokines lower immune activation threshold
• Increases myeloid APC phagocytosis
• Activated T cells migrate to tumor
Boltbody™ ISAC
Tumor Antigen
Tumor Cell
Dying Tumor Cell
Myeloid APC
T Cell
Activated T Cell
MHC-Tumor Peptide Complex
Granzymes, IFNγ, TNFα

BDC-2034: Structure and critical properties

CEA-targeting mAb CEA1

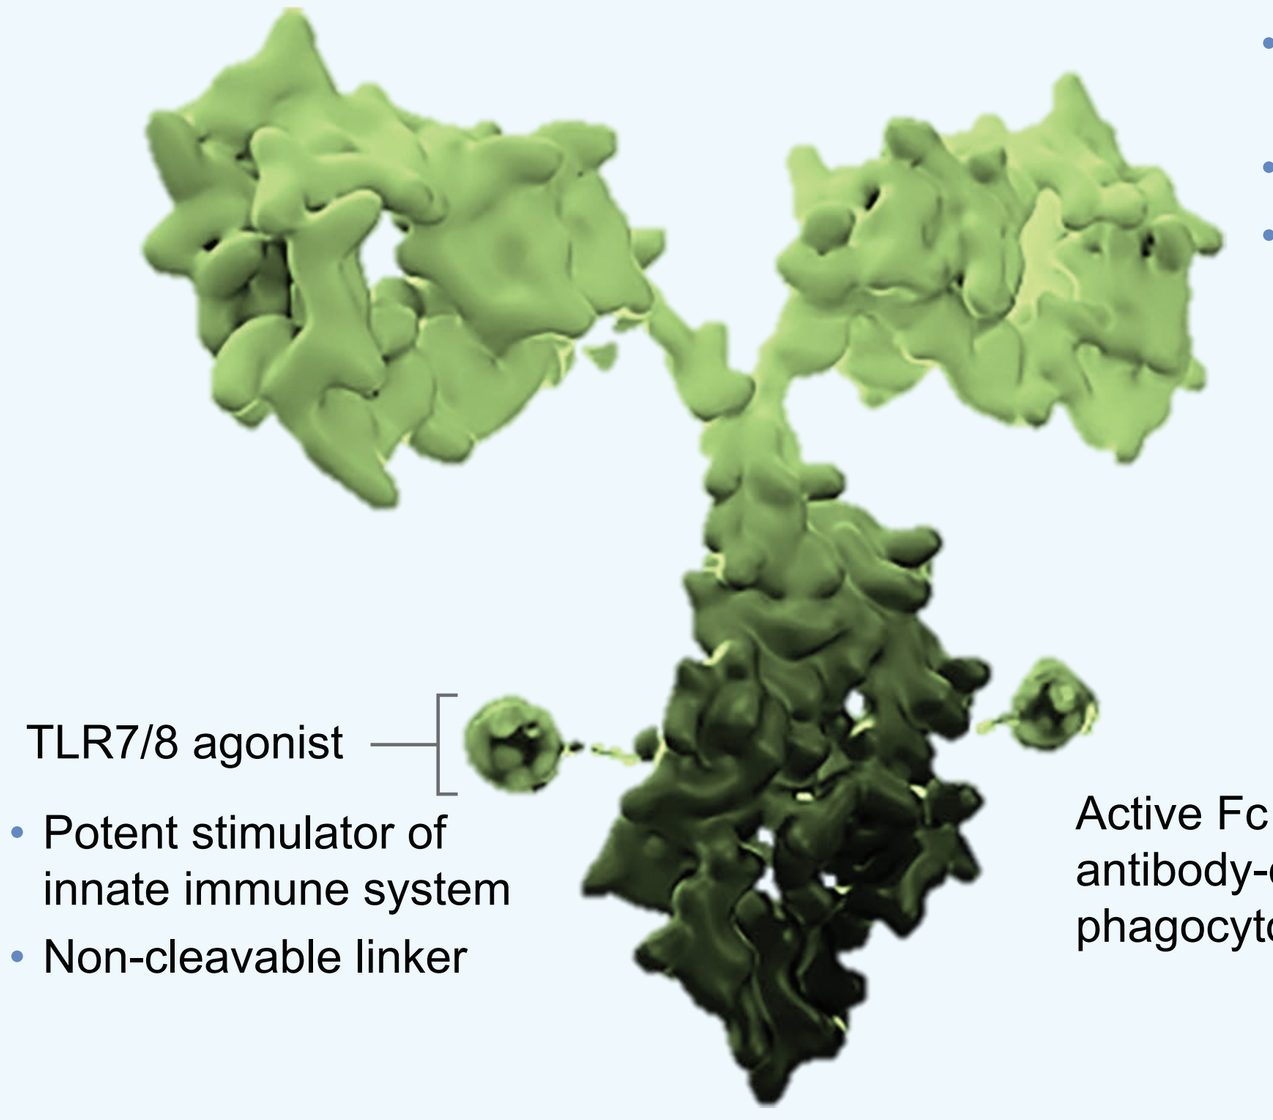

CEA1 key attributes for ISAC:
- Binds to cell-surface tumor antigen
- Monovalent K_D=18 nM
- Slowly internalizing: 60% of CEA1 remains on cell surface 5 hours after in vitro binding (two cell lines)

TLR7/8 agonist
- Potent stimulator of innate immune system
- Non-cleavable linker

Active Fc region capable of antibody-dependent cellular phagocytosis (ADCP)

1. Ackerman S, Pearson C, Gregorio J, et al. Immune-stimulating antibody conjugates elicit robust myeloid activation and durable antitumor immunity. *Nature Cancer*. 2021;2:18-33. https://doi.org/10.1038/s43018-020-00136-x

RESULTS

CEA is highly expressed in colorectal cancer and other solid tumors

CRC tumors have low CD8 T cell infiltration but moderate/high myeloid immune cell content

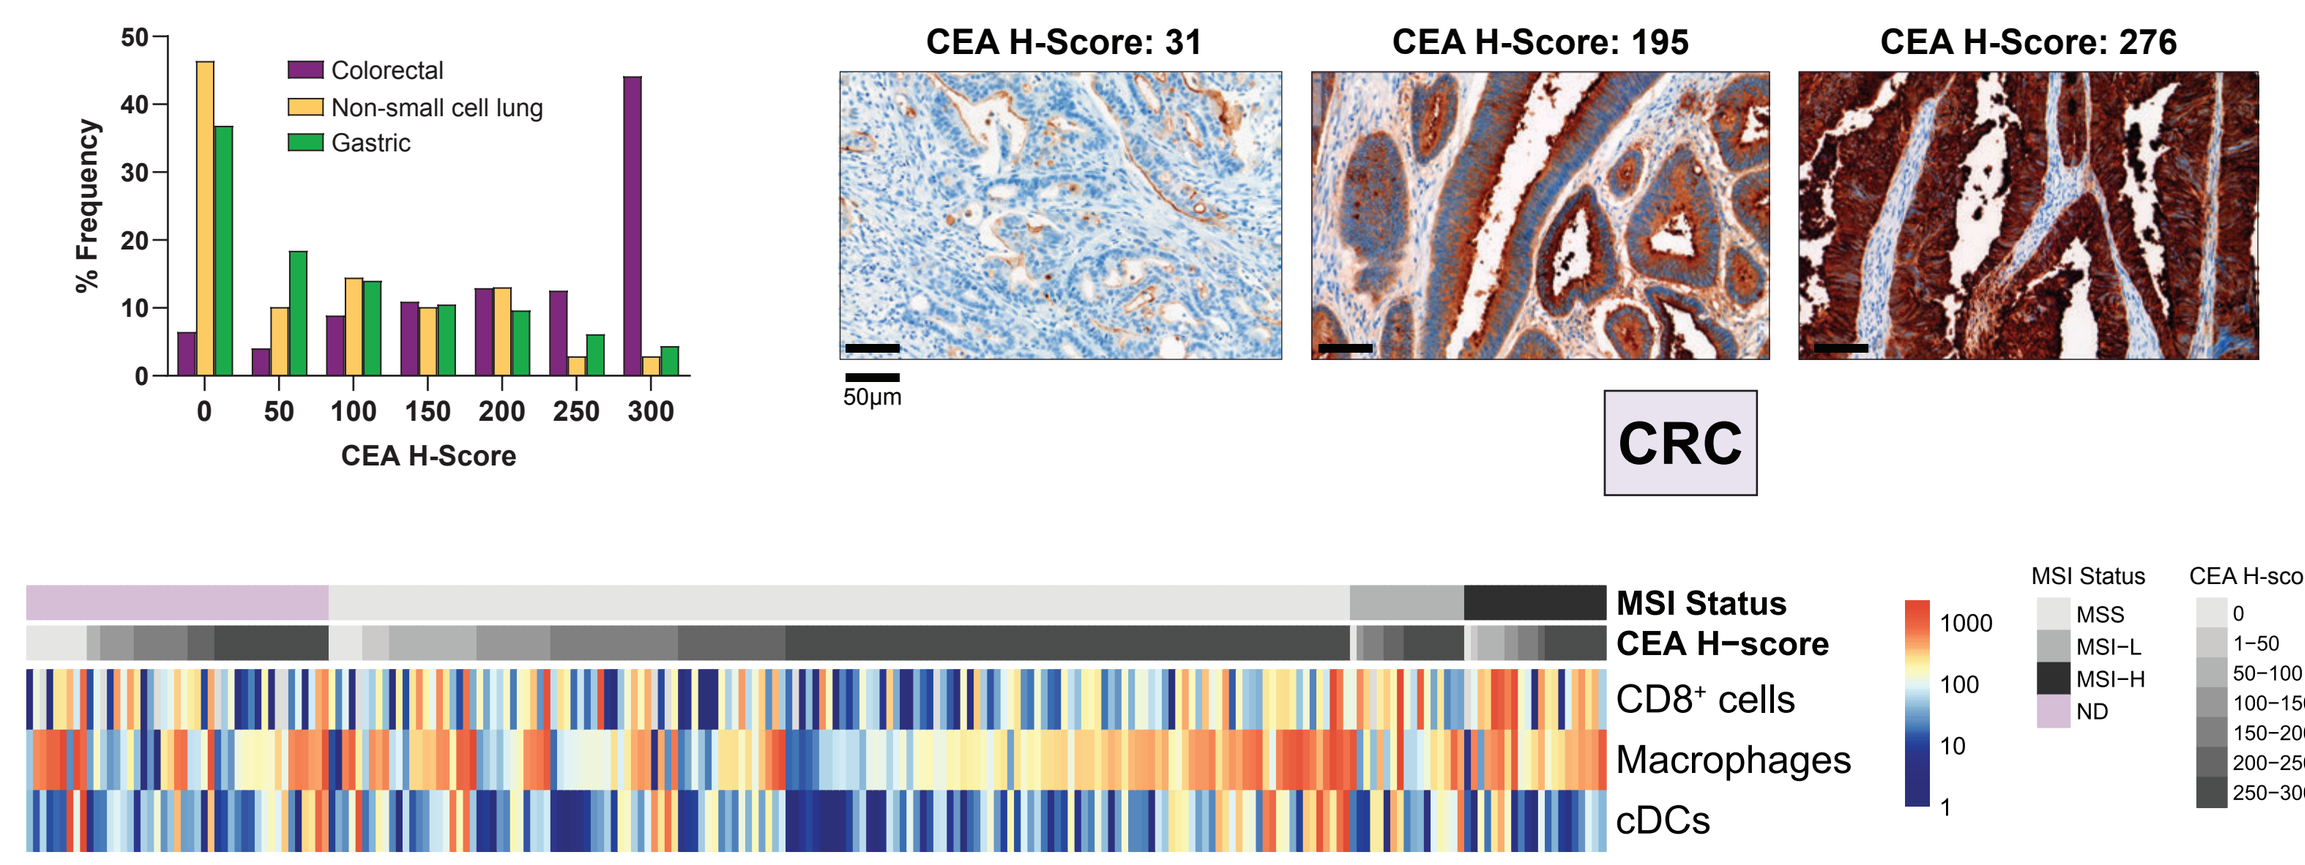
Colorectal
Non-small cell lung
Gastric
% Frequency
CEA H-Score
CEA H-Score: 31
CEA H-Score: 195
CEA H-Score: 276
50µm
CRC
MSI Status
CEA H−score
CD8⁺ cells
Macrophages
cDCs
MSI Status
MSS
MSI−L
MSI−H
ND
CEA H-score

- Human colorectal cancer (CRC) array (n=247), non-small cell lung cancer (NSCLC) array (n=69), and gastric/gastroesophageal (gastric/GEJ) cancer array (n=114) were stained with the CEA31 IHC assay (Ventana/Cell Marque). Representative images are shown from the colorectal cancer array.
- CRC array was also stained with a CD8/PanCK duplex IHC and a custom quad-plex IF assay for identification of myeloid APC sub-populations.
- Consistent with prior literature, IHC staining of TMAs showed >90% CEA+ among CRC tumors, irrespective of MSI status. For NSCLC and gastric/GEJ, >50% of tumors showed CEA+ staining.
- In CRC, myeloid immune cell infiltration was detected irrespective of MSI status, but $CD8^+$ T cell levels were low except in MSI-H tumors.

Tumor Antigen Binding: CEA1 exhibits similar affinity to human and cynomolgus monkey CEACAM5 (by ELISA)

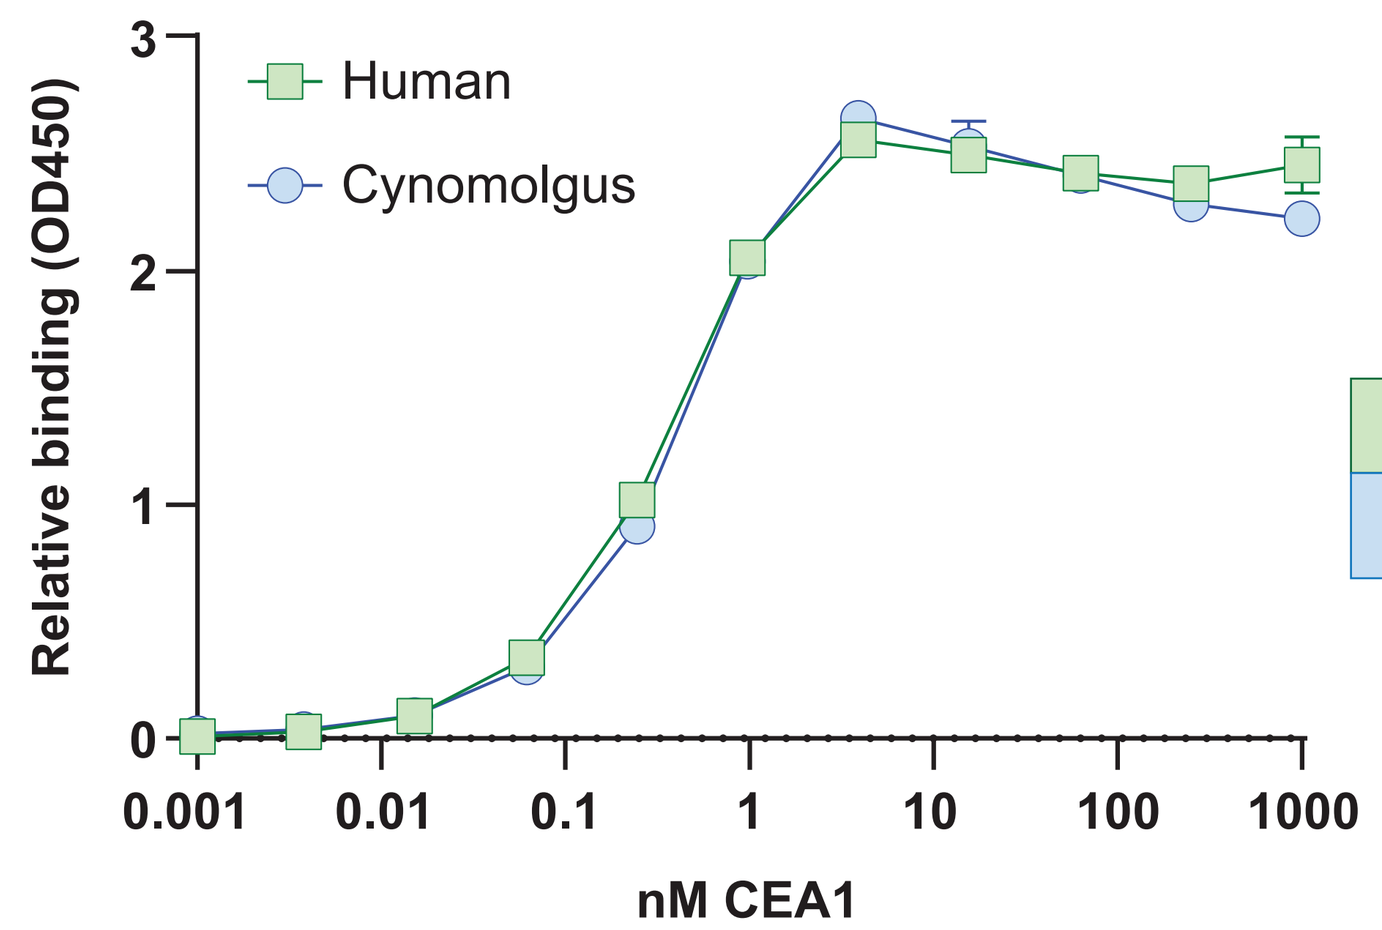
Human
Cynomolgus
Relative binding (OD450)
nM CEA1

	EC50 (nM)
Human	0.31
Cynomolgus	0.34

Tumor Antigen Binding: BDC-2034 binds differentially to surface-expressed CEA on a panel of cell lines

Binding of 200nM Alexa Fluor 488™ labeled BDC-2034 to cell lines

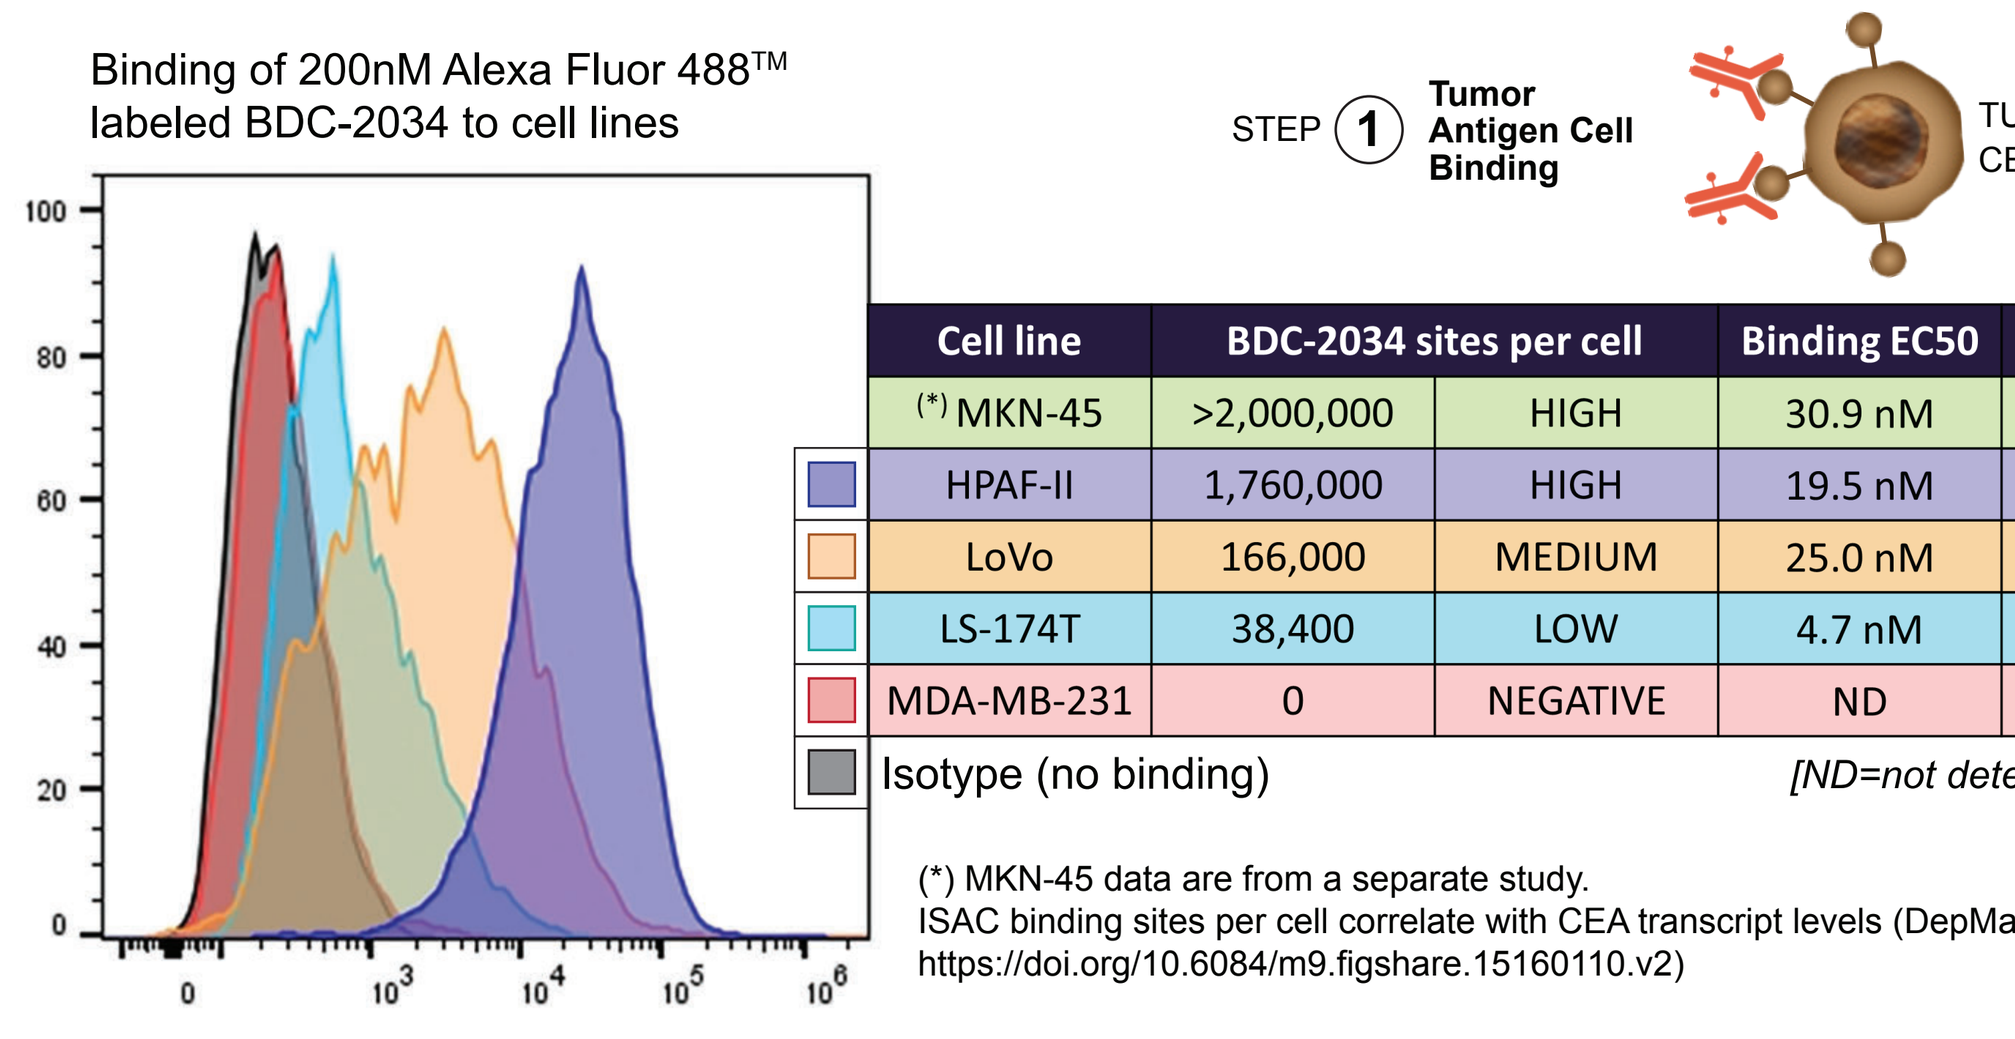
STEP 1 Tumor Antigen Cell Binding
TUMOR CELL

Cell line	BDC-2034 sites per cell		Binding EC50	IHC H-score
(*) MKN-45	>2,000,000	HIGH	30.9 nM	300
HPAF-II	1,760,000	HIGH	19.5 nM	220
LoVo	166,000	MEDIUM	25.0 nM	110
LS-174T	38,400	LOW	4.7 nM	ND
MDA-MB-231	0	NEGATIVE	ND	ND
Isotype (no binding)				

[ND=not determined]

(*) MKN-45 data are from a separate study.
ISAC binding sites per cell correlate with CEA transcript levels (DepMap, Broad (Q3 2021): https://doi.org/10.6084/m9.figshare.15160110.v2)

Phagocytosis: Lead antibody efficiently mediates ADCP

CEA1 precursor antibody (6.85 nM) induces phagocytosis of Raji/CEA cells by M-CSF differentiated, monocyte-derived macrophages (E:T=2:1)

- Activity is superior to reference anti-CEA antibody labetuzumab
- No ADCP detected with Fc-active isotype (does not bind to Raji/CEA)
- Rituximab (anti-CD20) is a positive control for Raji ADCP

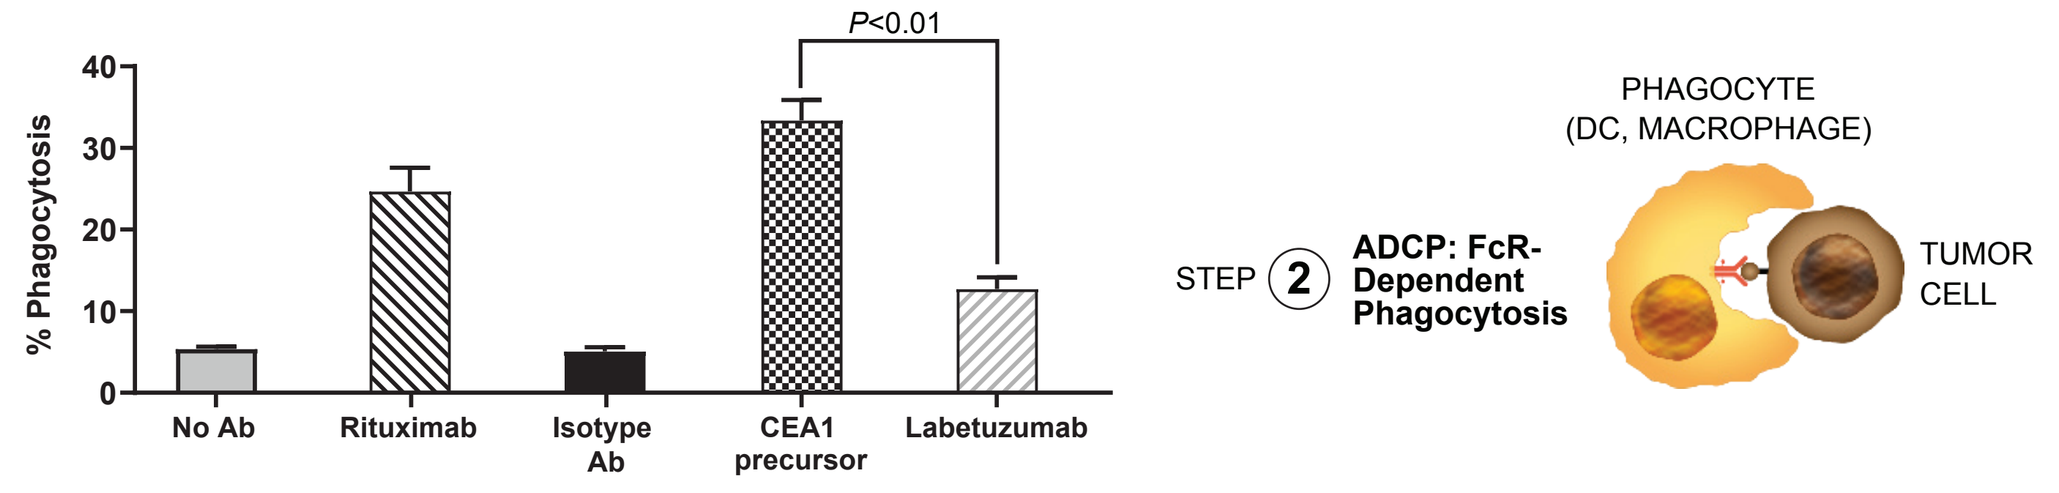
P<0.01
% Phagocytosis
No Ab
Rituximab
Isotype Ab
CEA1 precursor
Labetuzumab
STEP 2 ADCP: FcR-Dependent Phagocytosis
PHAGOCYTE (DC, MACROPHAGE)
TUMOR CELL

Phagocytosis: Medium CEA expression is sufficient for BDC-2034 induced ADCP

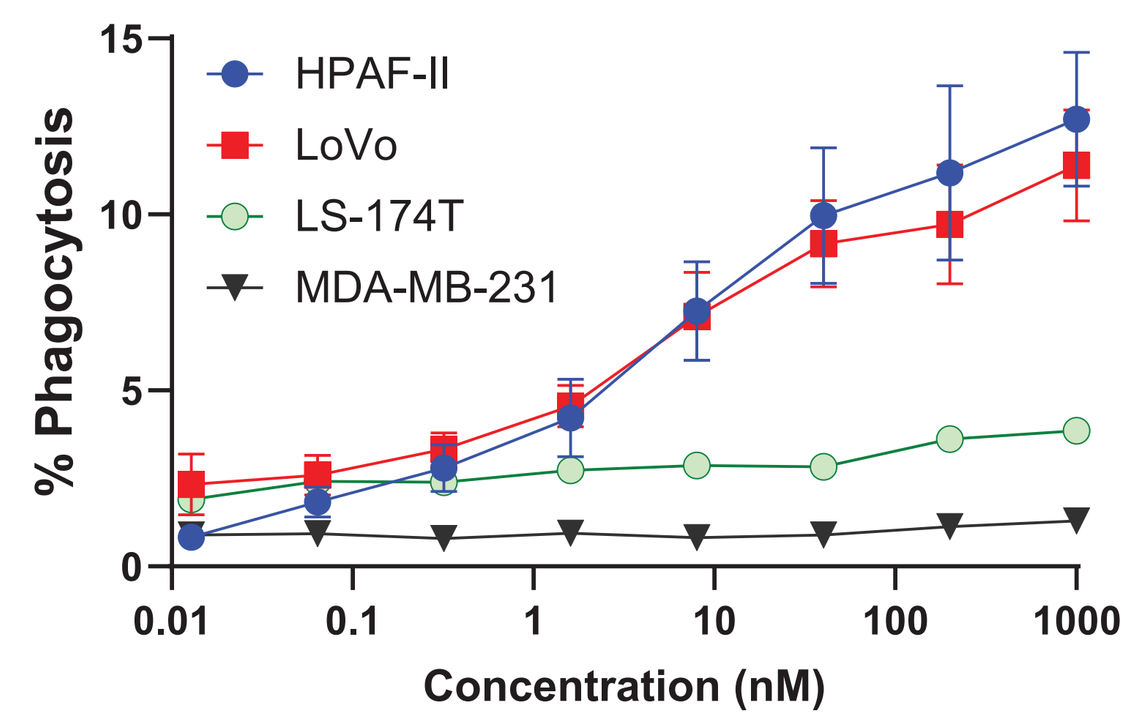
HPAF-II
LoVo
LS-174T
MDA-MB-231
% Phagocytosis
Concentration (nM)

BDC-2034 induces dose-dependent phagocytosis of CEA-high HPAF II (EC50=9.2 ± 2.3 nM) and CEA-medium LoVo (EC50=11.4 ± 3.5 nM); minimal ADCP observed for CEA-low LS-174T or CEA-negative MDA-MB-231. Cells co-cultured with M-CSF differentiated monocyte-derived macrophages; means +/- standard deviations from three donors shown.

Innate Immune Activation: BDC-2034 is superior to reference labetuzumab CEA ISAC and to CEA1 antibody

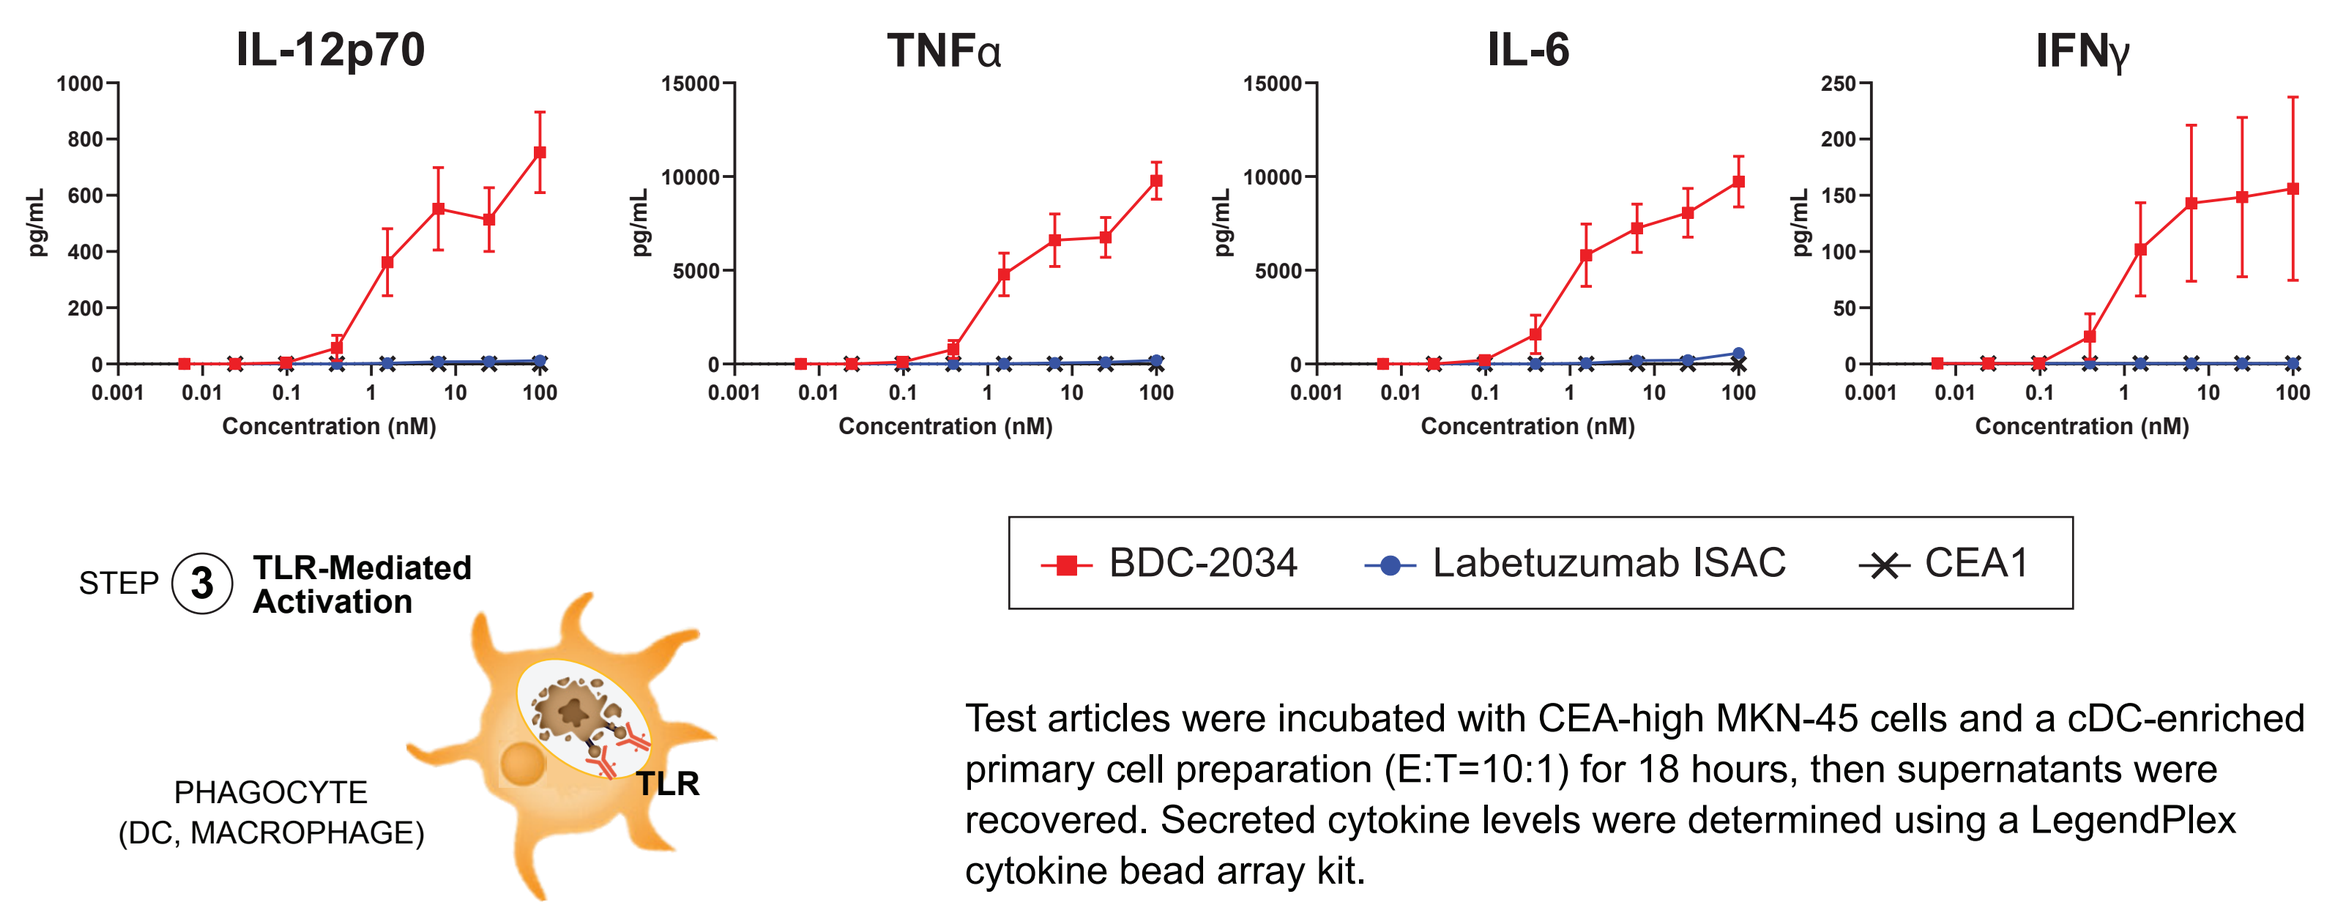
IL-12p70
TNFα
IL-6
IFNγ
pg/mL
Concentration (nM)
BDC-2034
Labetuzumab ISAC
CEA1
STEP 3 TLR-Mediated Activation
PHAGOCYTE (DC, MACROPHAGE)
TLR

Test articles were incubated with CEA-high MKN-45 cells and a cDC-enriched primary cell preparation (E:T=10:1) for 18 hours, then supernatants were recovered. Secreted cytokine levels were determined using a LegendPlex cytokine bead array kit.

Innate Immune Activation: Medium tumor CEA expression levels are sufficient for activation of myeloid effector cells by CEA ISAC2 (BDC-2034 linker variant)

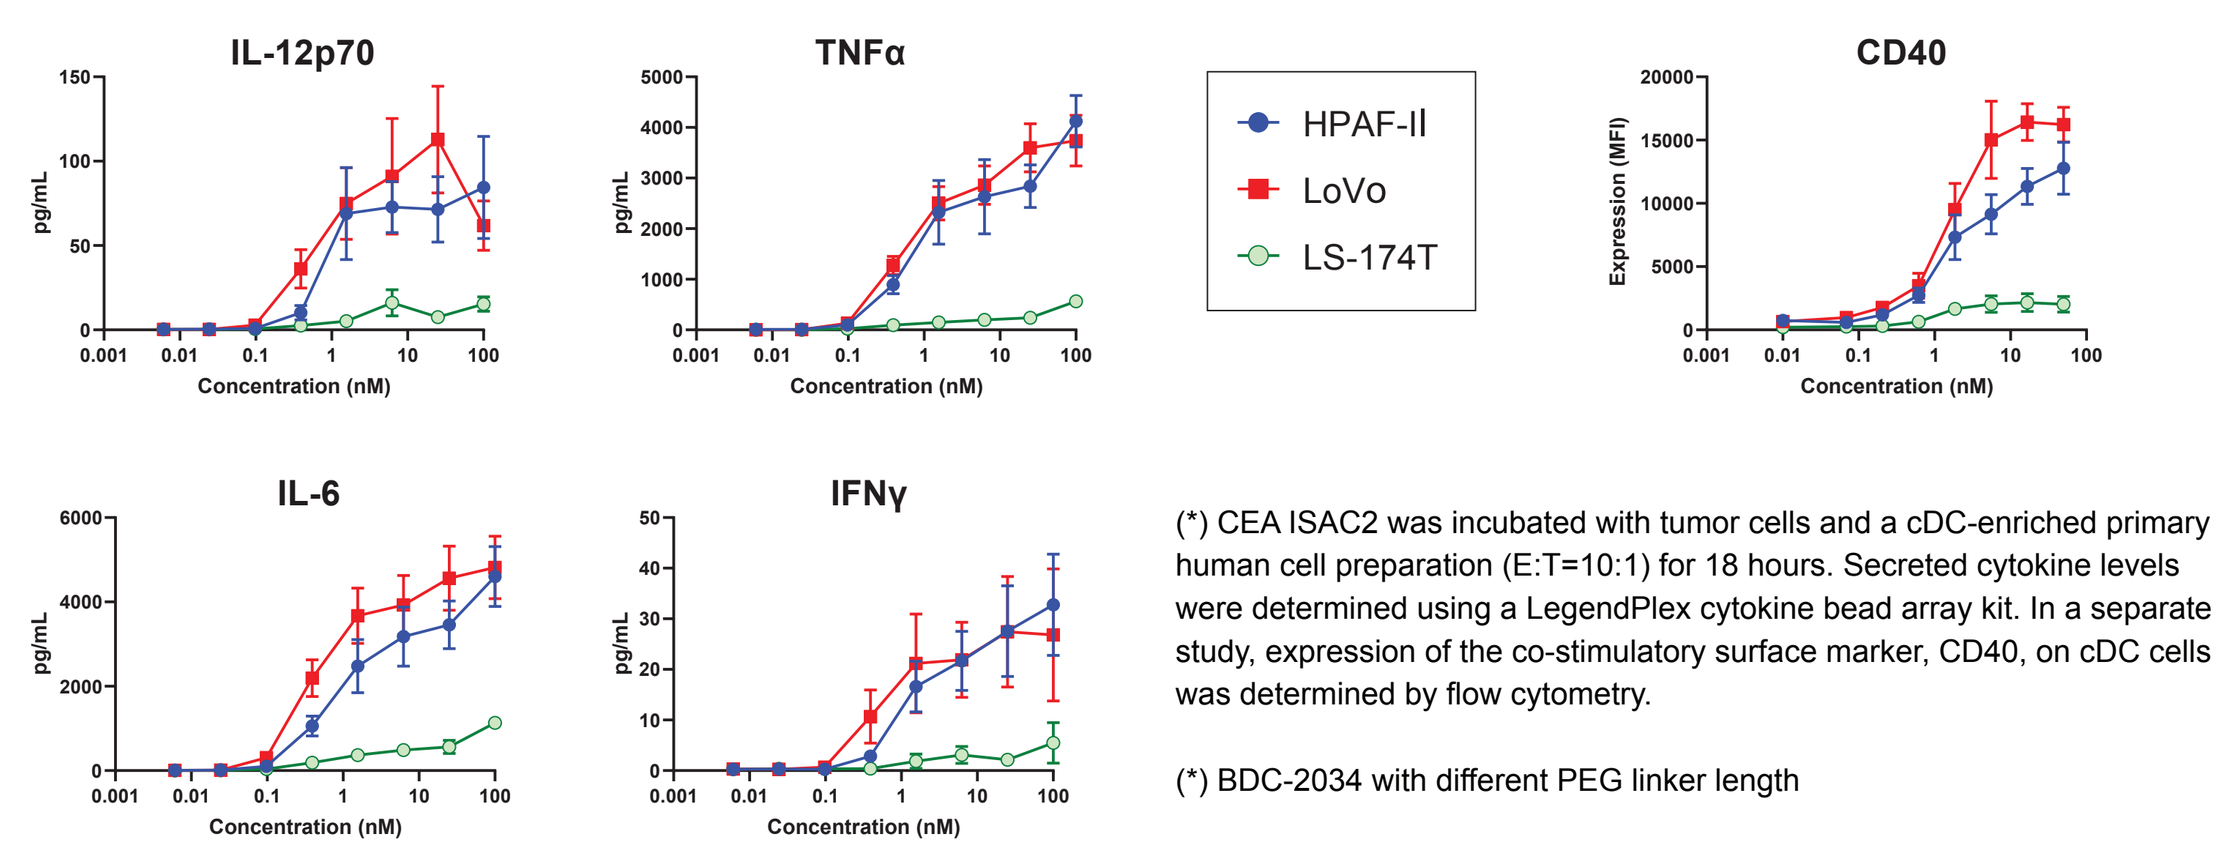
IL-12p70
TNFα
CD40
IL-6
IFNγ
HPAF-II
LoVo
LS-174T
pg/mL
Expression (MFI)
Concentration (nM)

(*) CEA ISAC2 was incubated with tumor cells and a cDC-enriched primary human cell preparation (E:T=10:1) for 18 hours. Secreted cytokine levels were determined using a LegendPlex cytokine bead array kit. In a separate study, expression of the co-stimulatory surface marker, CD40, on cDC cells was determined by flow cytometry.

(*) BDC-2034 with different PEG linker length

CEA ISAC3 is a mouse-stable surrogate for BDC-2034

ISAC3 is a CEA1 ISAC using a linker-payload (L-P) that is a close chemical analog of the BDC-2034 linker-payload
- Modified to improve stability in rodents
- Similar activity profile in TLR7/8 reporter assays and myeloid activation assays

	Serum stability: Half-life (hr)	
Species	**BDC-2034 L-P**	**CEA ISAC3 L-P**
Human	>24	>24
Cynomolgus monkey	>24	>24
Mouse	9.2 (± 0.3)	>24

Mouse surrogate CEA ISAC3 inhibits growth of CEA-high HPAF-II tumors

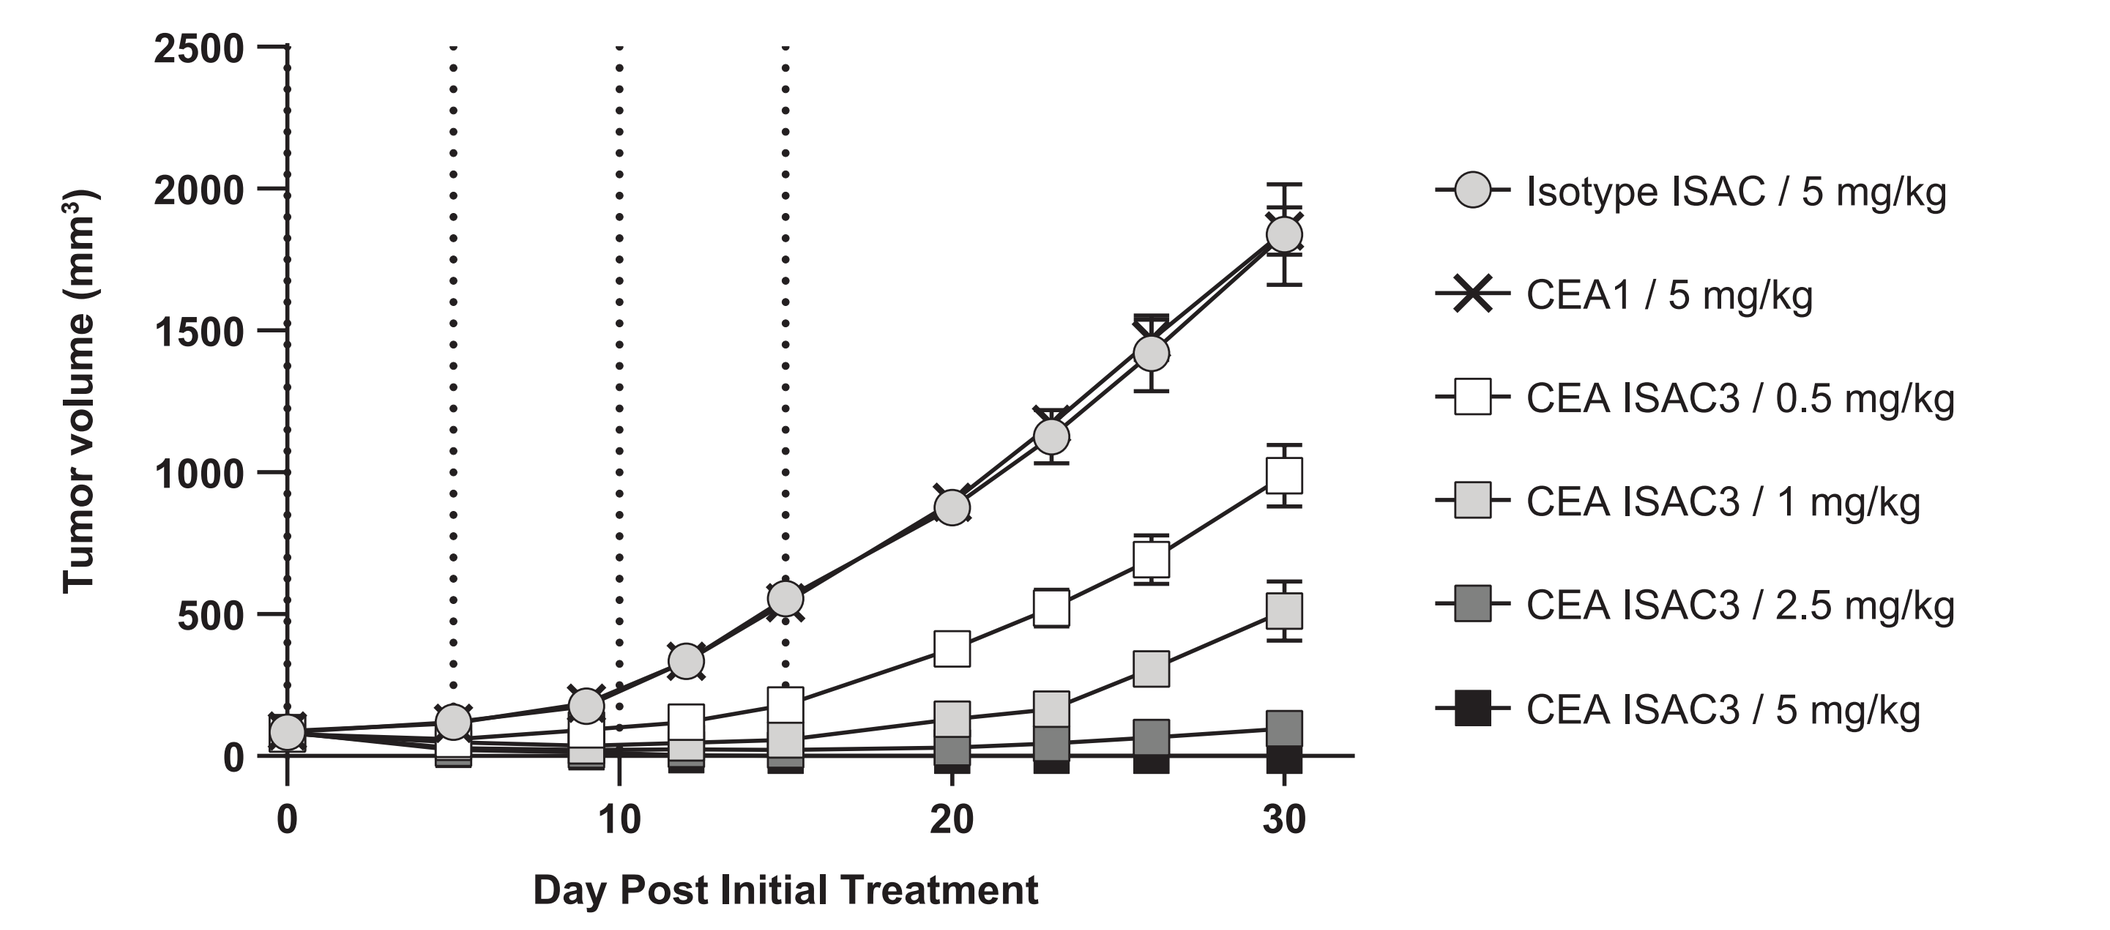
Tumor volume (mm³)
Day Post Initial Treatment
Isotype ISAC / 5 mg/kg
CEA1 / 5 mg/kg
CEA ISAC3 / 0.5 mg/kg
CEA ISAC3 / 1 mg/kg
CEA ISAC3 / 2.5 mg/kg
CEA ISAC3 / 5 mg/kg

CEA-high HPAF-II tumors ($80mm^3$) in SCID beige mice (5 mice/group)

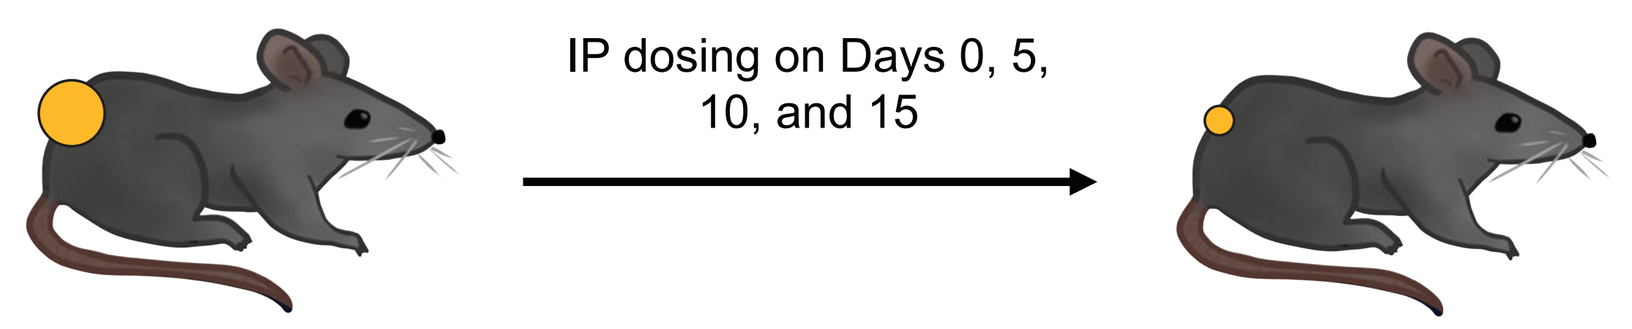
IP dosing on Days 0, 5, 10, and 15

CEA ISAC MoA study in xenograft tumor mouse model

CEA-high HPAF-II tumors ($150mm^3$) in SCID beige mice

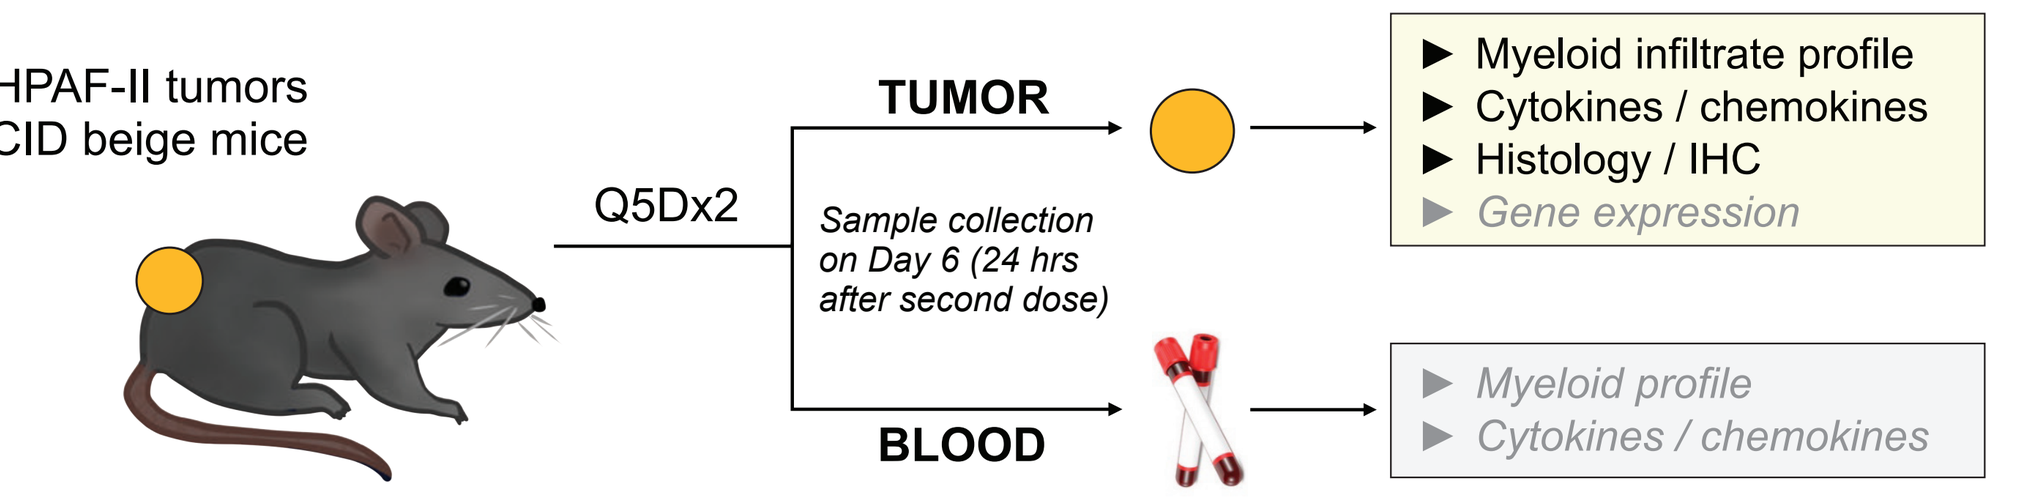
Q5Dx2
TUMOR
Sample collection on Day 6 (24 hrs after second dose)
BLOOD
Myeloid infiltrate profile
Cytokines / chemokines
Histology / IHC
Gene expression
Myeloid profile
Cytokines / chemokines

- BDC-2034 Surrogate (ISAC3): 0.05, 0.2, 1, 5 mg/kg
- Isotype ISAC, CEA1, rituximab (isotype Ab): 5 mg/kg
- N=5 per group

CEA ISAC3 promotes CEA-dependent cytokine secretion and immune infiltration in HPAF-II xenograft tumors

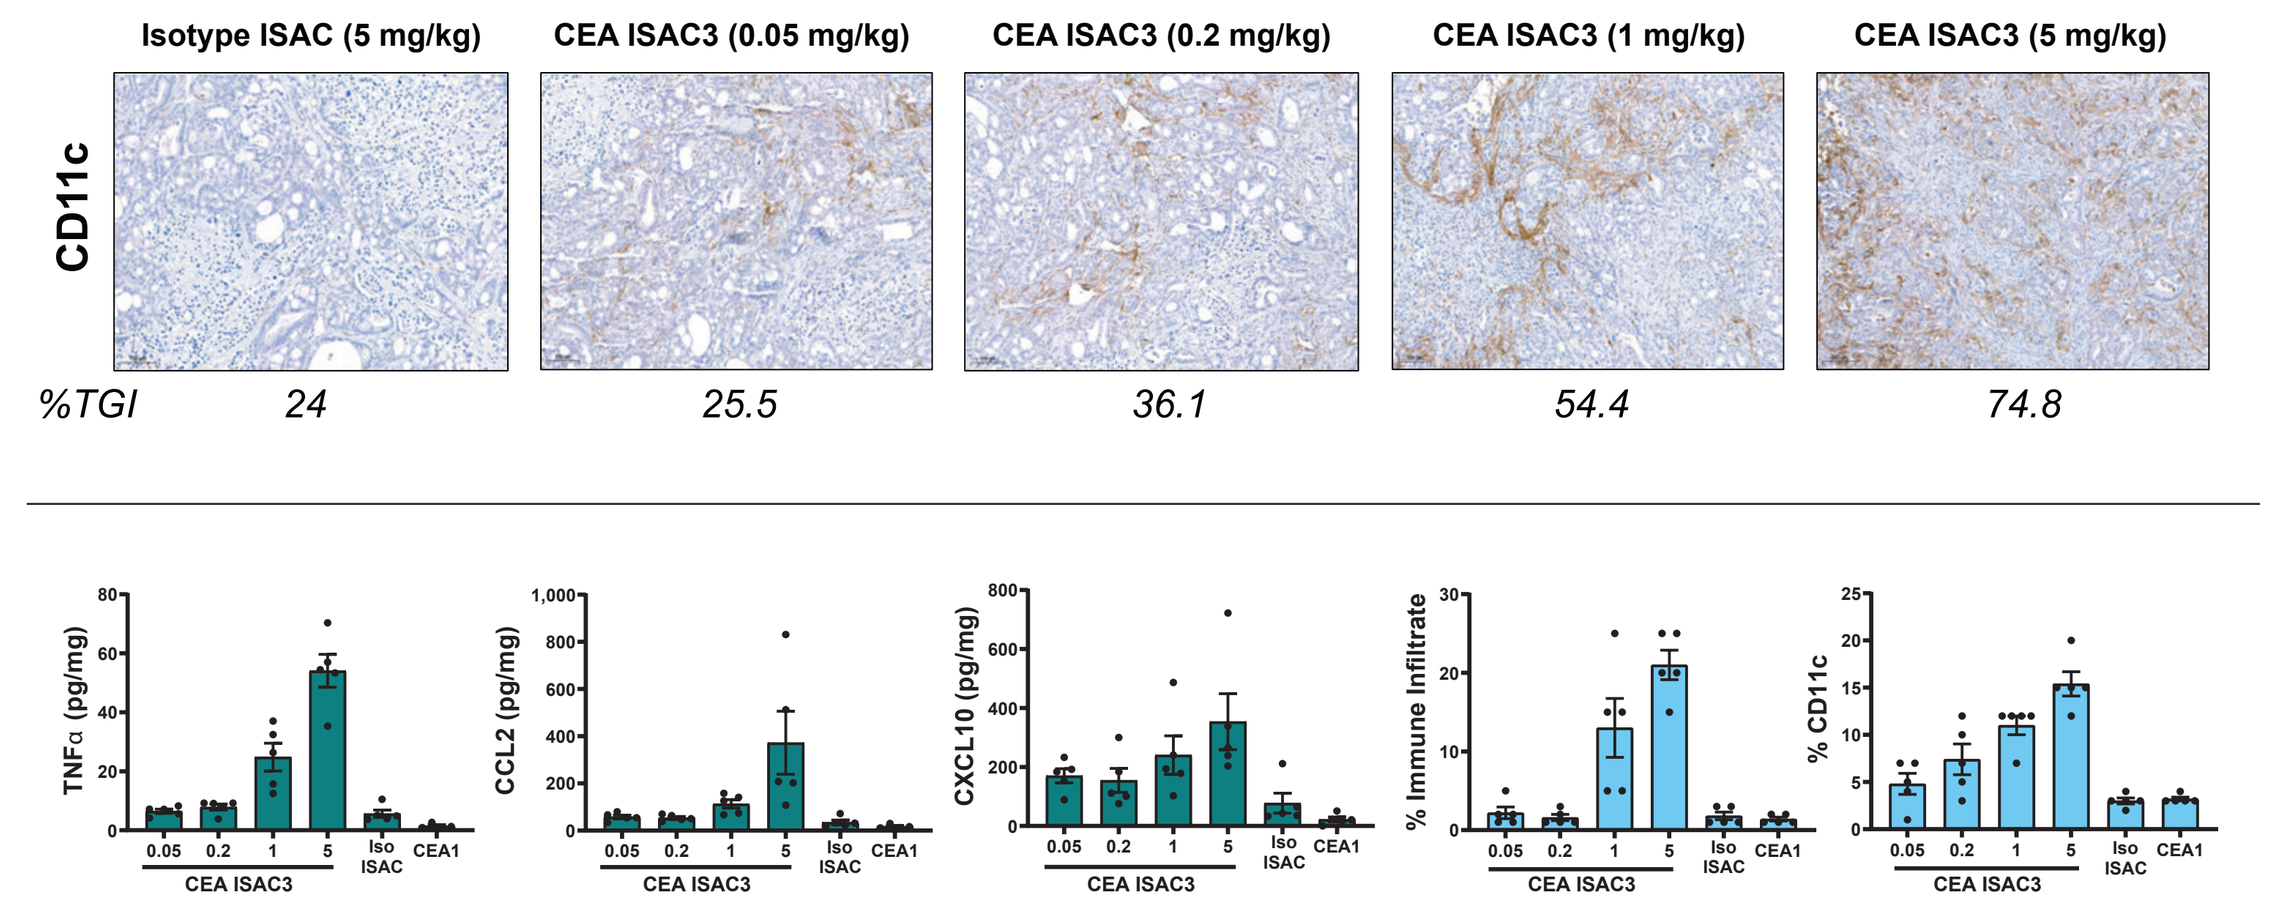
Isotype ISAC (5 mg/kg)
CEA ISAC3 (0.05 mg/kg)
CEA ISAC3 (0.2 mg/kg)
CEA ISAC3 (1 mg/kg)
CEA ISAC3 (5 mg/kg)
CD11c
%TGI
24
25.5
36.1
54.4
74.8
TNFα (pg/mg)
CCL2 (pg/mg)
CXCL10 (pg/mg)
% Immune Infiltrate
% CD11c
CEA ISAC3
Iso ISAC
CEA1

- CD11c IHC was performed as previously described,[1] and samples were scored by a pathologist
- Cytokines and chemokines were measured in tumor lysates using the MSD U-plex platform; all data are shown as mean +/- SEM
- %TGI (tumor growth inhibition) calculated relative to CEA1 on Day 6 using the formula $([TV^{CEA1}-TV^{ISAC}]/TV^{CEA1})*100$
 - TV=tumor volume

CONCLUSIONS

- CEA is expressed in solid tumors having robust innate immune infiltrates, including colorectal cancers
- High affinity for CEA, cross-reactivity with cynomolgus antigen, and potent induction of ADCP establishes the CEA1 antibody as a suitable candidate for the ISAC approach
- Conjugation of CEA1 with a TLR7/8 dual agonist payload via a non-cleavable linker generates the ISAC BDC-2034
- BDC-2034 and surrogates exhibit promising activity in pre-clinical models
 - Tumor-dependent induction of immune-stimulating cytokine secretion by primary human innate effector cells
 - Innate immune activation with CEA-medium models (CEA expression levels comparable to human cancers)
 - Anti-tumor efficacy in a xenograft model at dose levels as low as 0.5 mg/kg
 - Dose-dependent tumor recruitment of innate effector cells and induction of immune-stimulating cytokines
- Bolt Biotherapeutics' pre-clinical data support further development of BDC-2034 as a therapeutic option for patients with CEA-expressing cancers