TLR7/8 immune stimulating antibody conjugates elicit robust myeloid activation and durable anti-tumor immunity

ABSTRACT
1559

Shelley E. Ackerman[1,2], Joseph C. Gonzalez[1], Josh D. Gregorio[1], Jason C. Paik[3], Felix J. Hartmann[3], Justin A. Kenkel[1,3], Arthur Lee[1], Angela Luo[1], Cecelia I. Pearson[1], Murray L. Nguyen[1], Benjamin Ackerman[4], Lauren Y. Sheu[3], Richard P. Laura[1], Steven J. Chapin[1], Brian S. Safina[1], Sean C. Bendall[3], David Dornan[1], Edgar G. Engleman[3], Michael N. Alonso[1,3]

[1]Bolt Biotherapeutics, [2]Stanford University Department of Bioengineering, [3]Stanford University Department of Pathology, [4]Johns Hopkins University Department of Biostatistics

ABSTRACT

Recent studies indicate that local delivery of immunostimulatory adjuvants can activate tumor resident antigen presenting cells (APCs), driving uptake, processing and presentation of tumor neoantigens to T cells that mediate anti-tumor immunity. To overcome challenges associated with intratumoral delivery of such adjuvants, we developed a novel class of systemically delivered TLR immune-stimulating antibody conjugates (ISACs) that comprise a TLR7/8 agonist conjugated to tumor-targeting monoclonal antibodies. *In vitro* co-cultures of human cancer cell lines and leukocytes revealed that ISACs potently activate primary myeloid APCs, leading to increased co-stimulatory molecule expression and secretion of pro-inflammatory cytokines. Surprisingly, ISACs also induced DC differentiation from monocytes, as measured by changes in cellular morphology and DC associated surface markers. Finally, we demonstrated *in vivo* efficacy in xenograft and syngeneic tumor models in which ISAC treatment led to tumor clearance and development of immunologic memory. These results provide a strong rationale for this technology as a platform for cancer immunotherapy.

IMMUNE STIMULATING ANTIBODY CONJUGATES

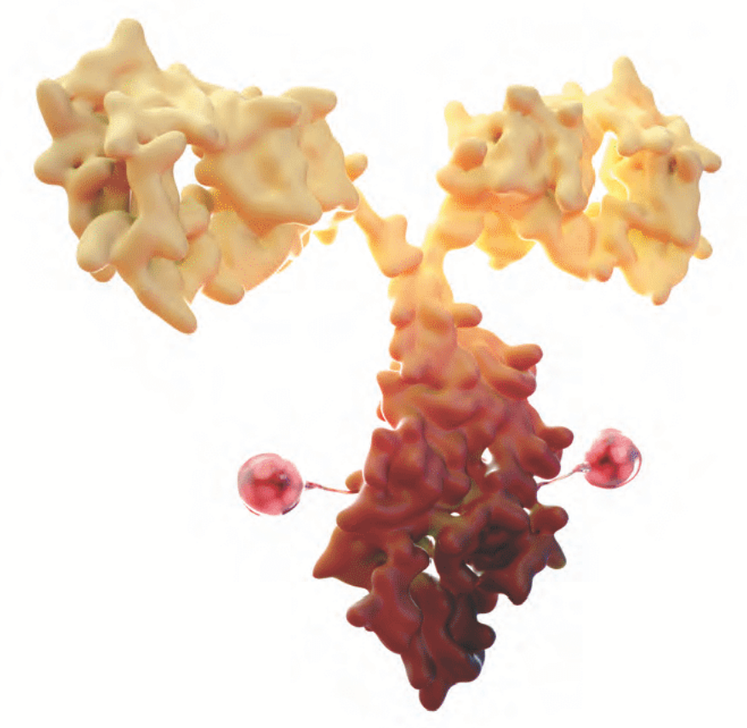

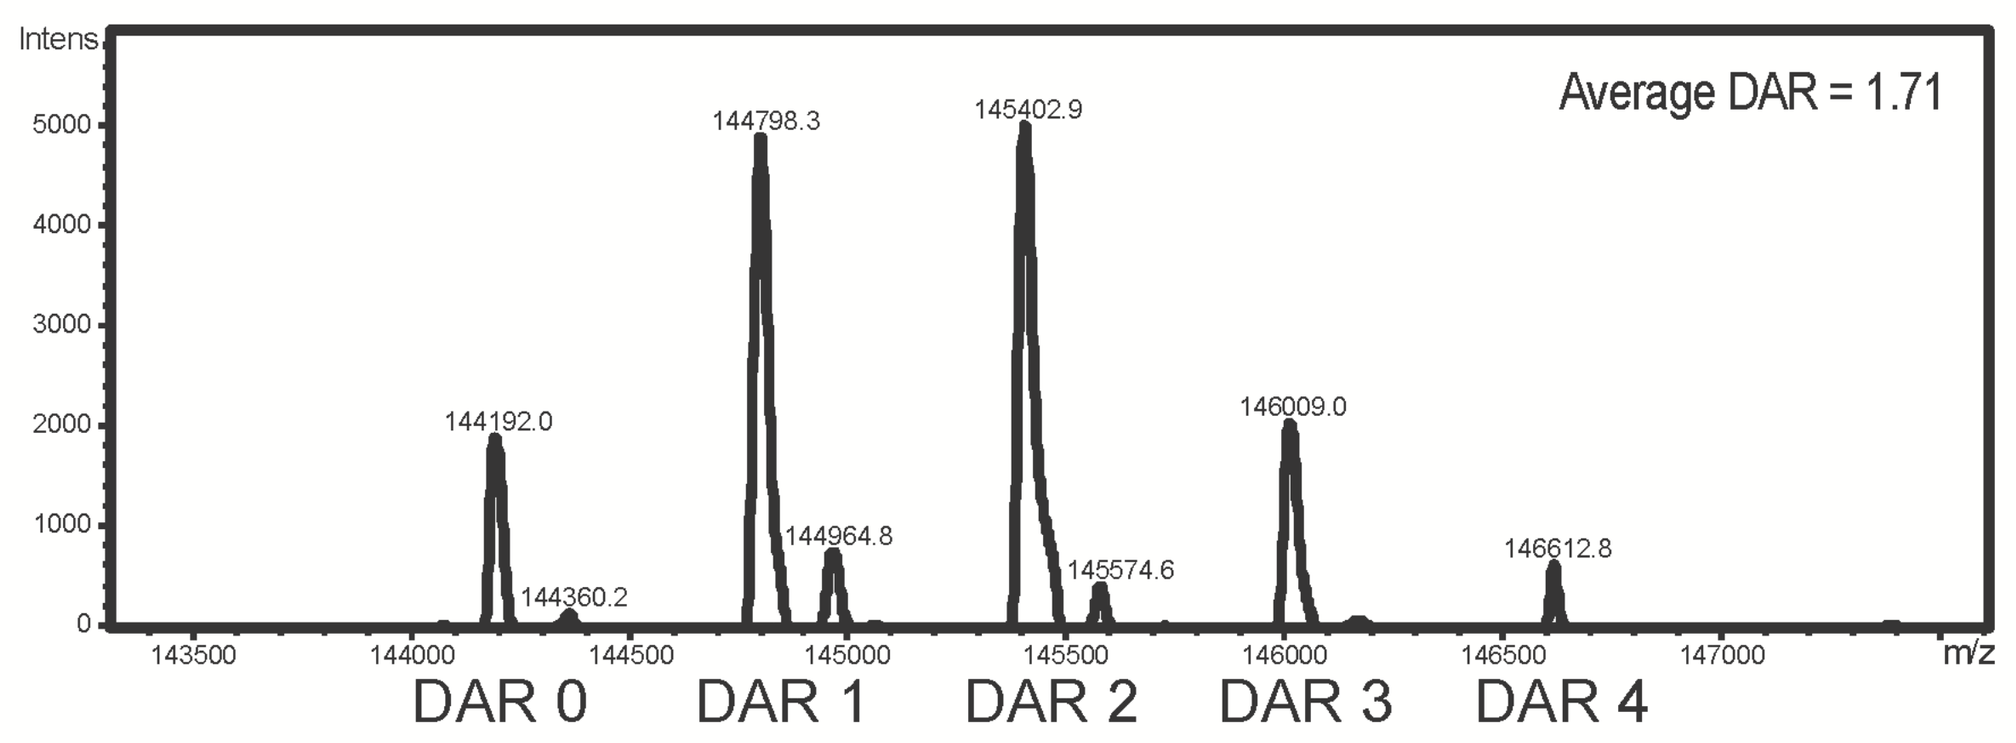
Intens.
Average DAR = 1.71
144192.0
144360.2
144798.3
144964.8
145402.9
145574.6
146009.0
146612.8
m/z
DAR 0
DAR 1
DAR 2
DAR 3
DAR 4

PROPOSED MECHANISM OF ACTION

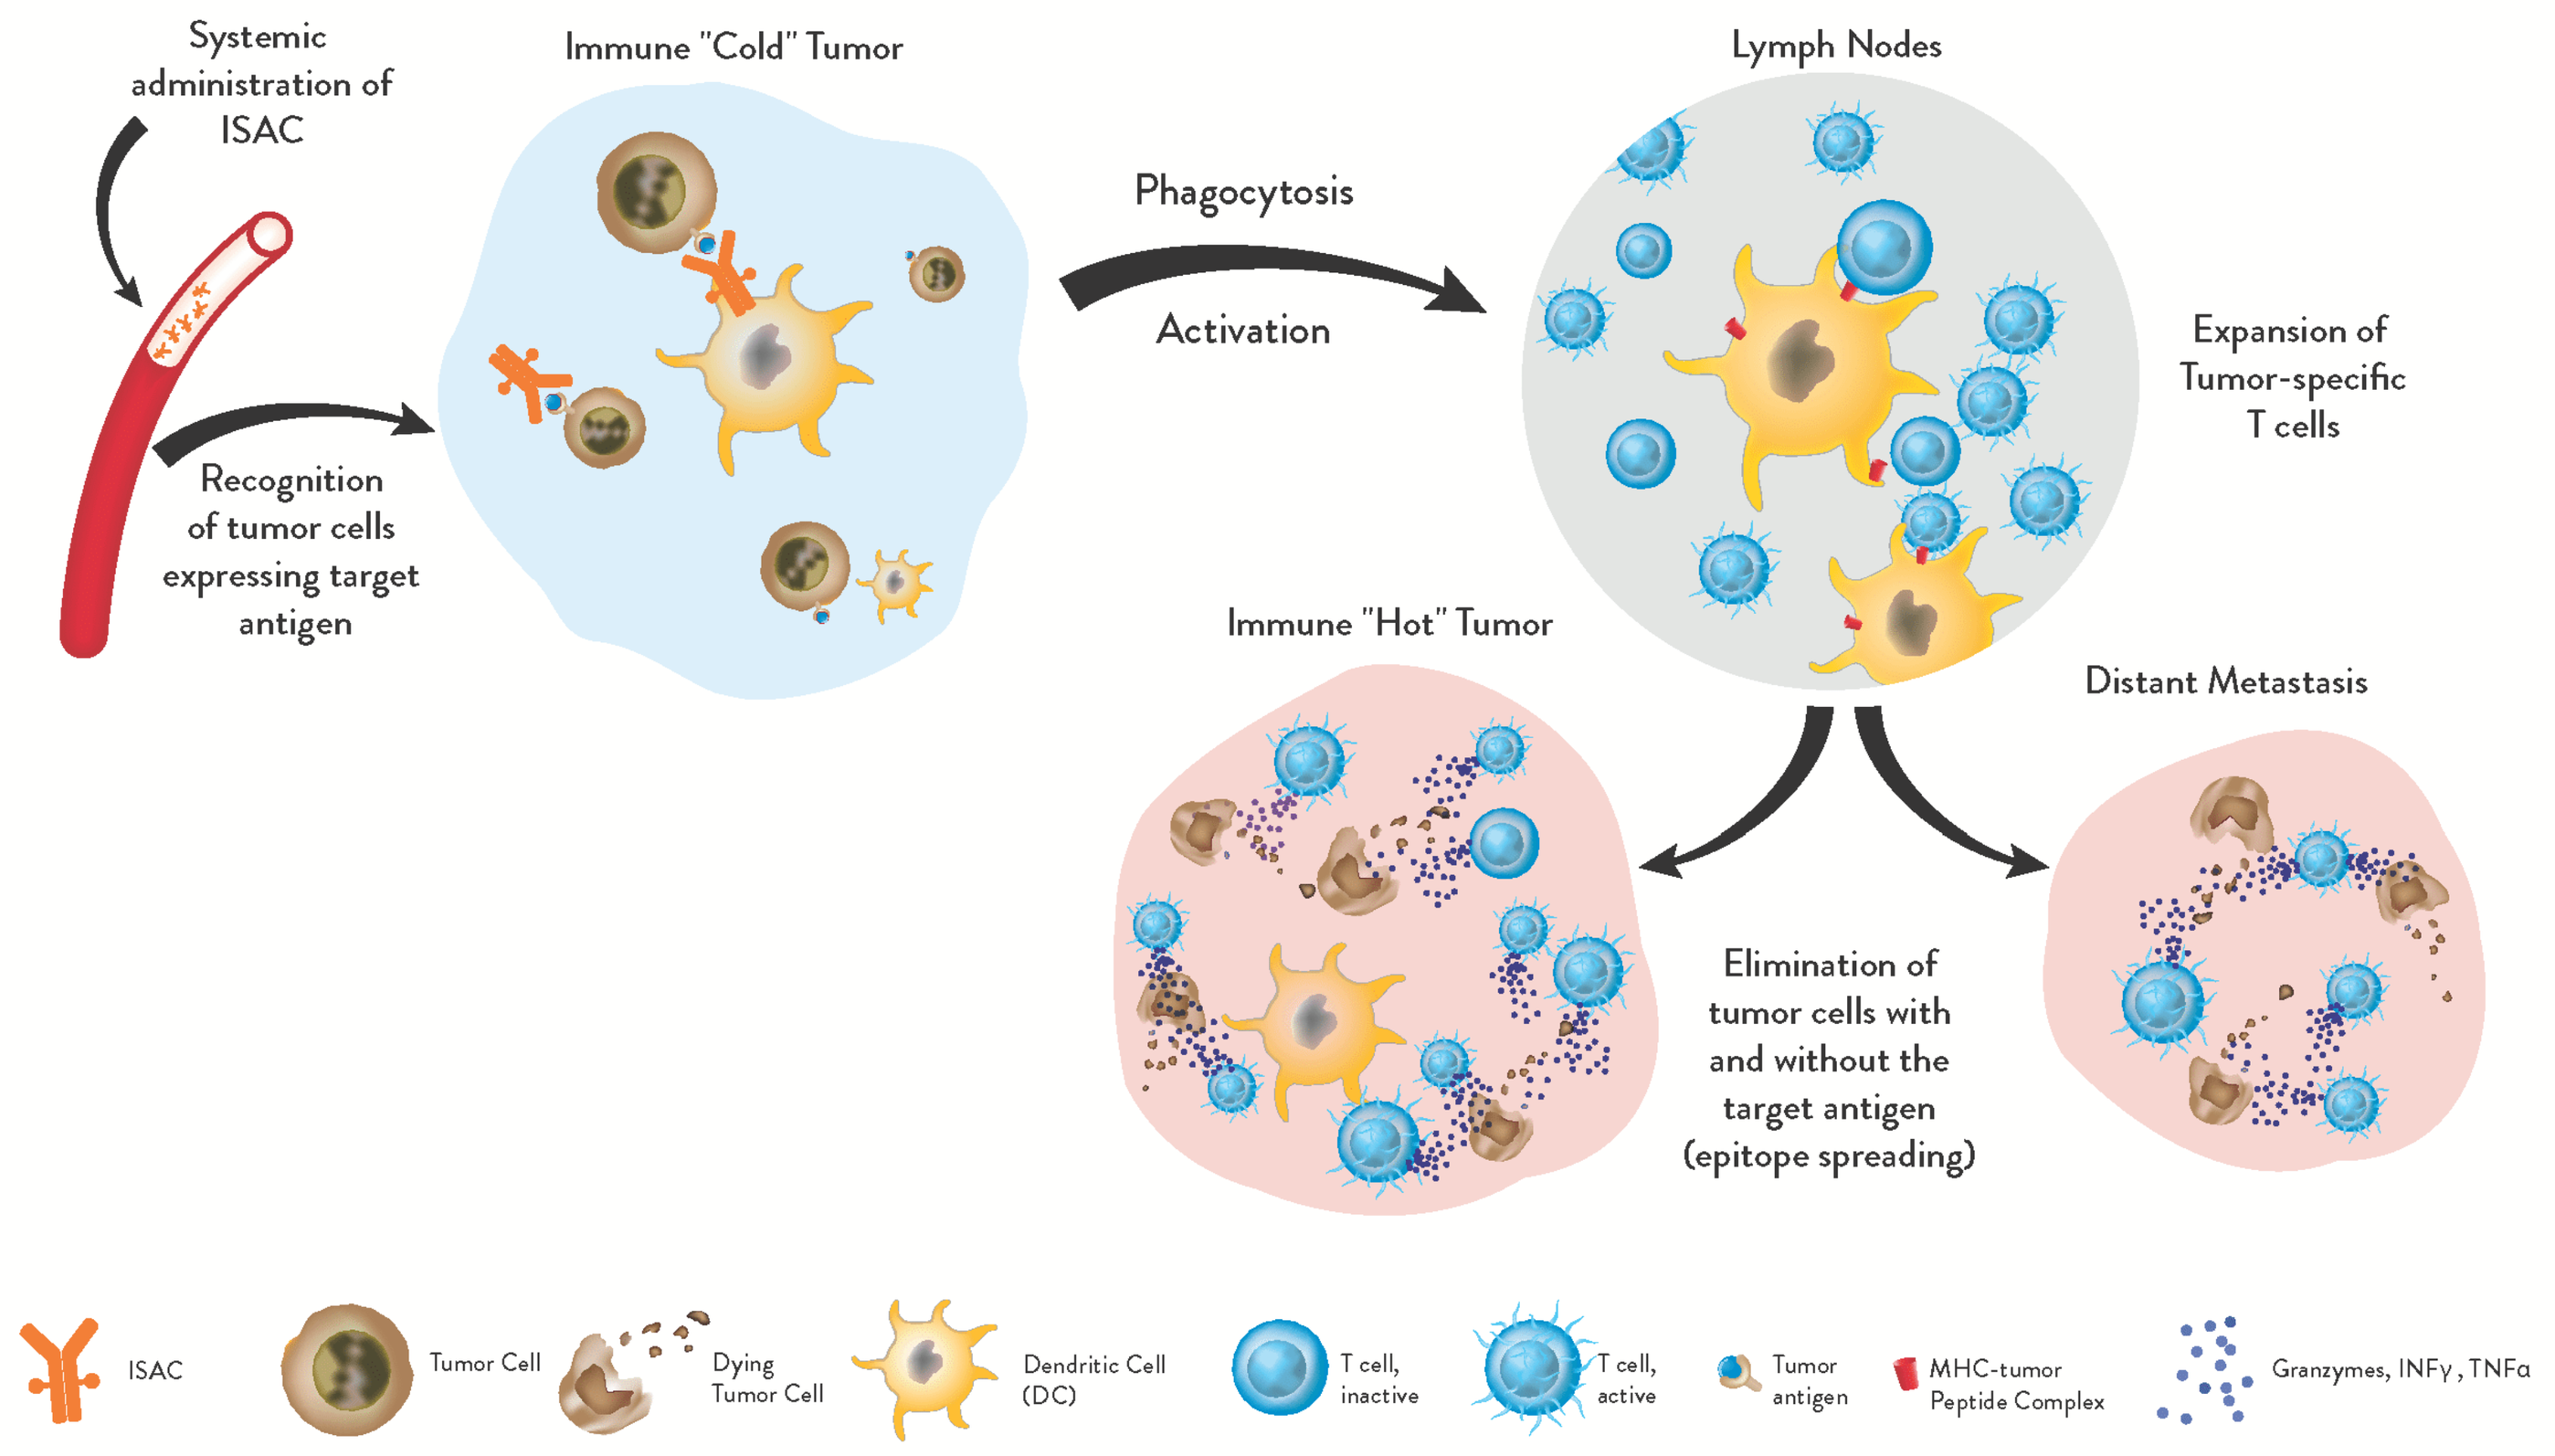
Systemic administration of ISAC
Recognition of tumor cells expressing target antigen
Immune "Cold" Tumor
Phagocytosis
Activation
Lymph Nodes
Expansion of Tumor-specific T cells
Immune "Hot" Tumor
Distant Metastasis
Elimination of tumor cells with and without the target antigen (epitope spreading)
ISAC
Tumor Cell
Dying Tumor Cell
Dendritic Cell (DC)
T cell, inactive
T cell, active
Tumor antigen
MHC-tumor Peptide Complex
Granzymes, INFγ, TNFα

HUMAN *IN VITRO* EXPERIMENTAL DESIGN

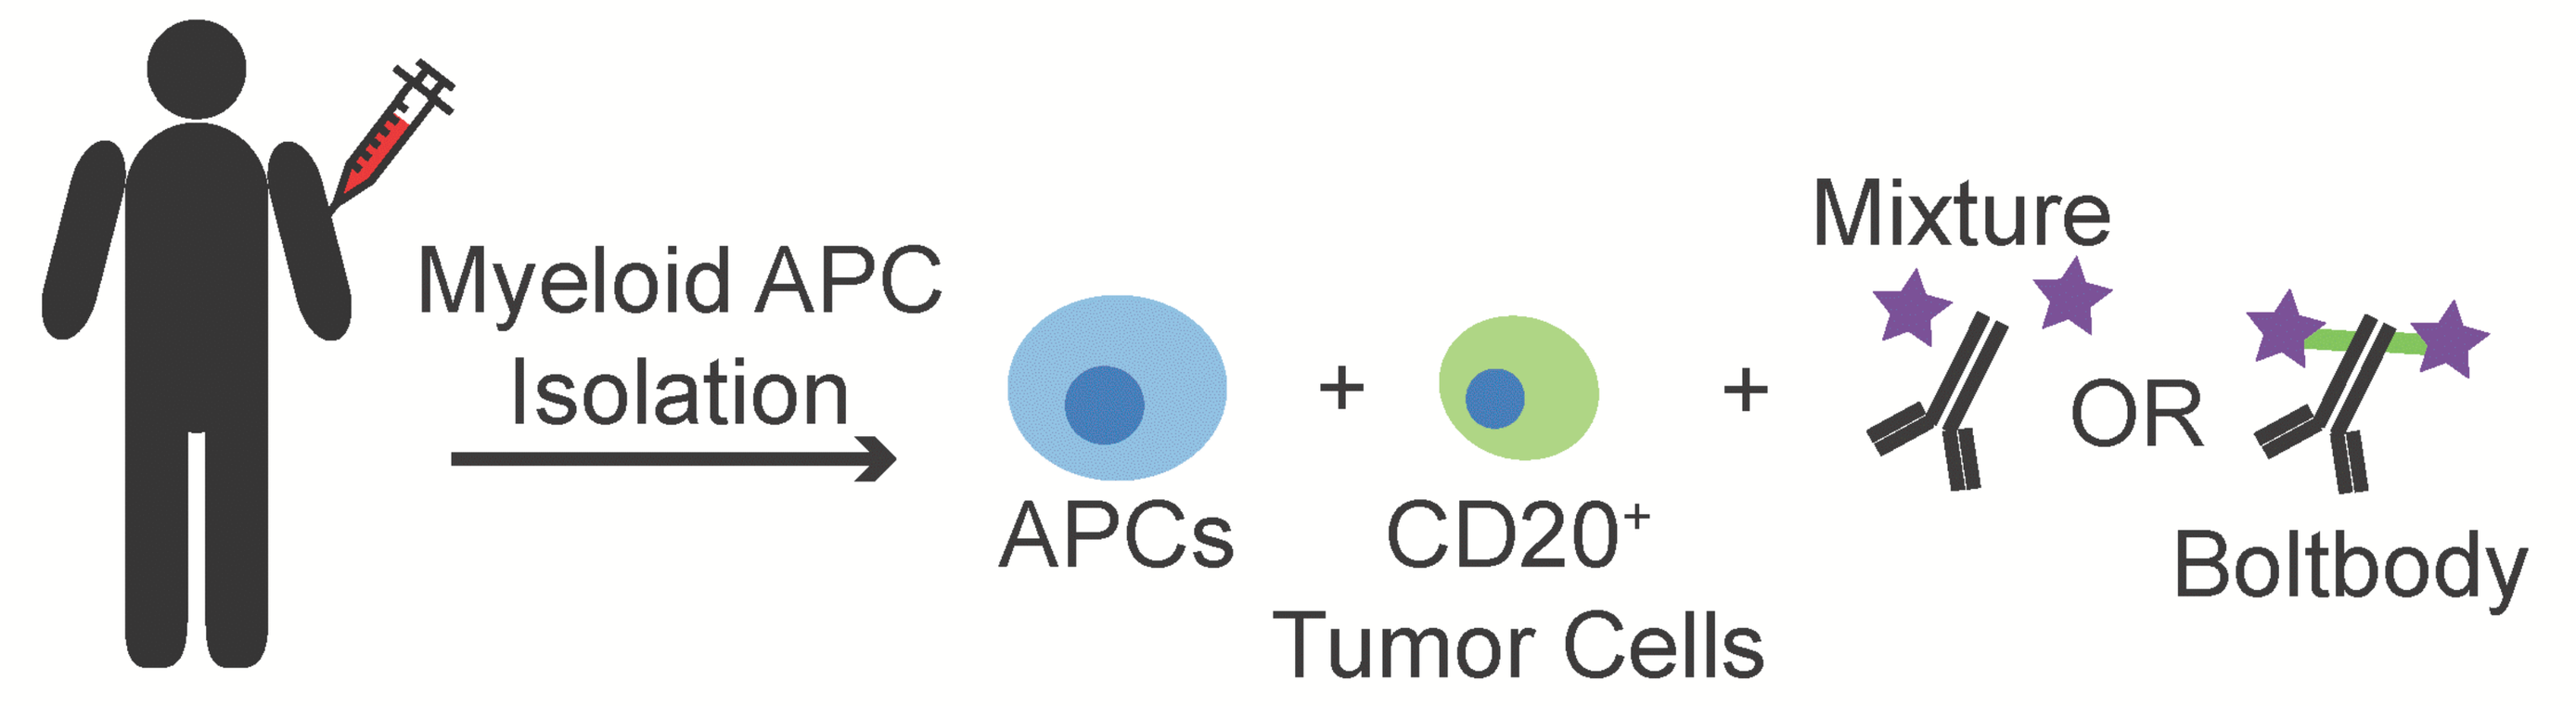
Myeloid APC Isolation
APCs
+
CD20+ Tumor Cells
+
Mixture
OR
Boltbody

Figure 1: Assessment of ISAC Activity. Myeloid APCs were isolated from healthy donor via negative selection and co-cultured with CFSE-labeled CD20+ tumor cells. After 18 hours, cells were assessed for cellular activation and differentiation via flow cytometry and ELISA (Figures 2-4).

HUMAN *IN VITRO* RESULTS

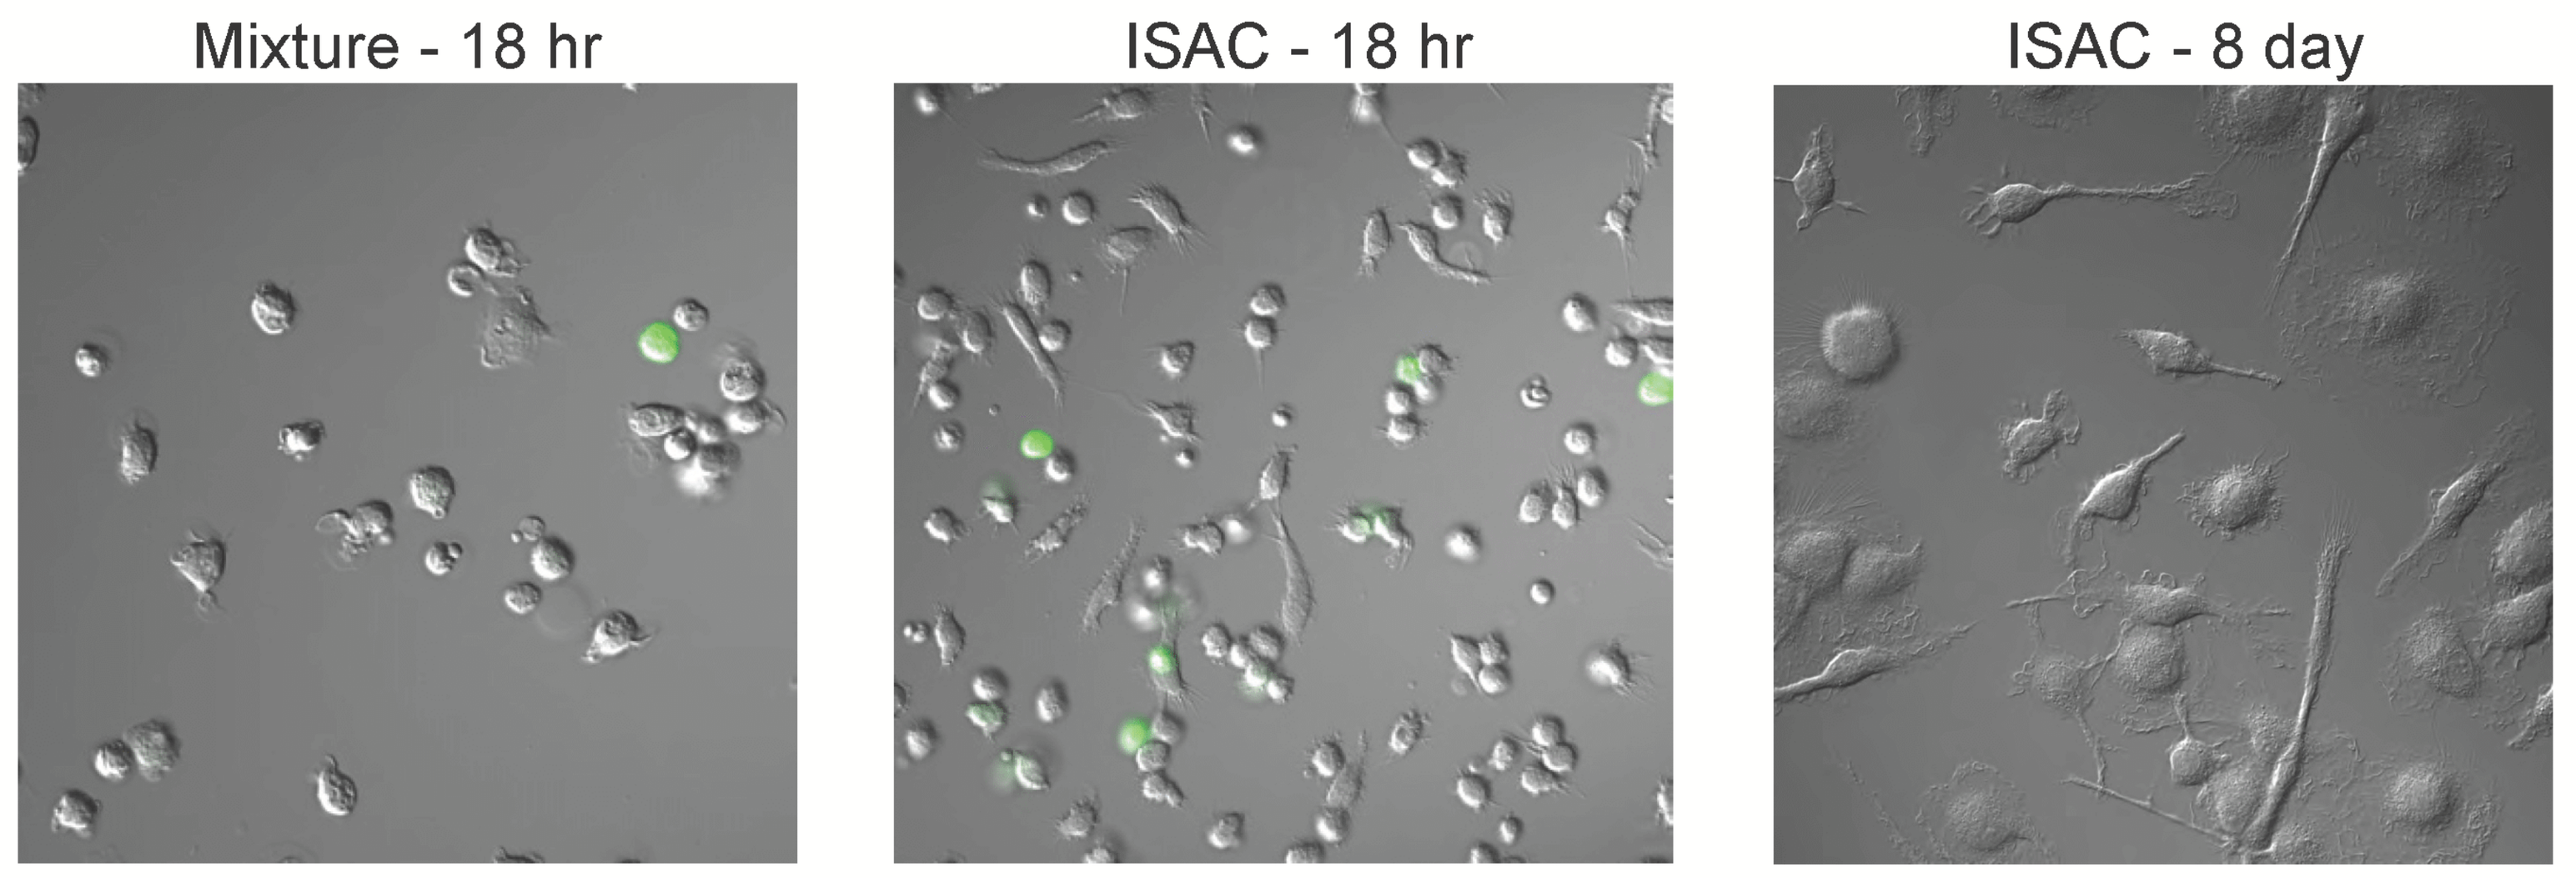
Mixture - 18 hr
ISAC - 18 hr
ISAC - 8 day

Figure 2: ISACs elicit distinct changes in cellular morphology.

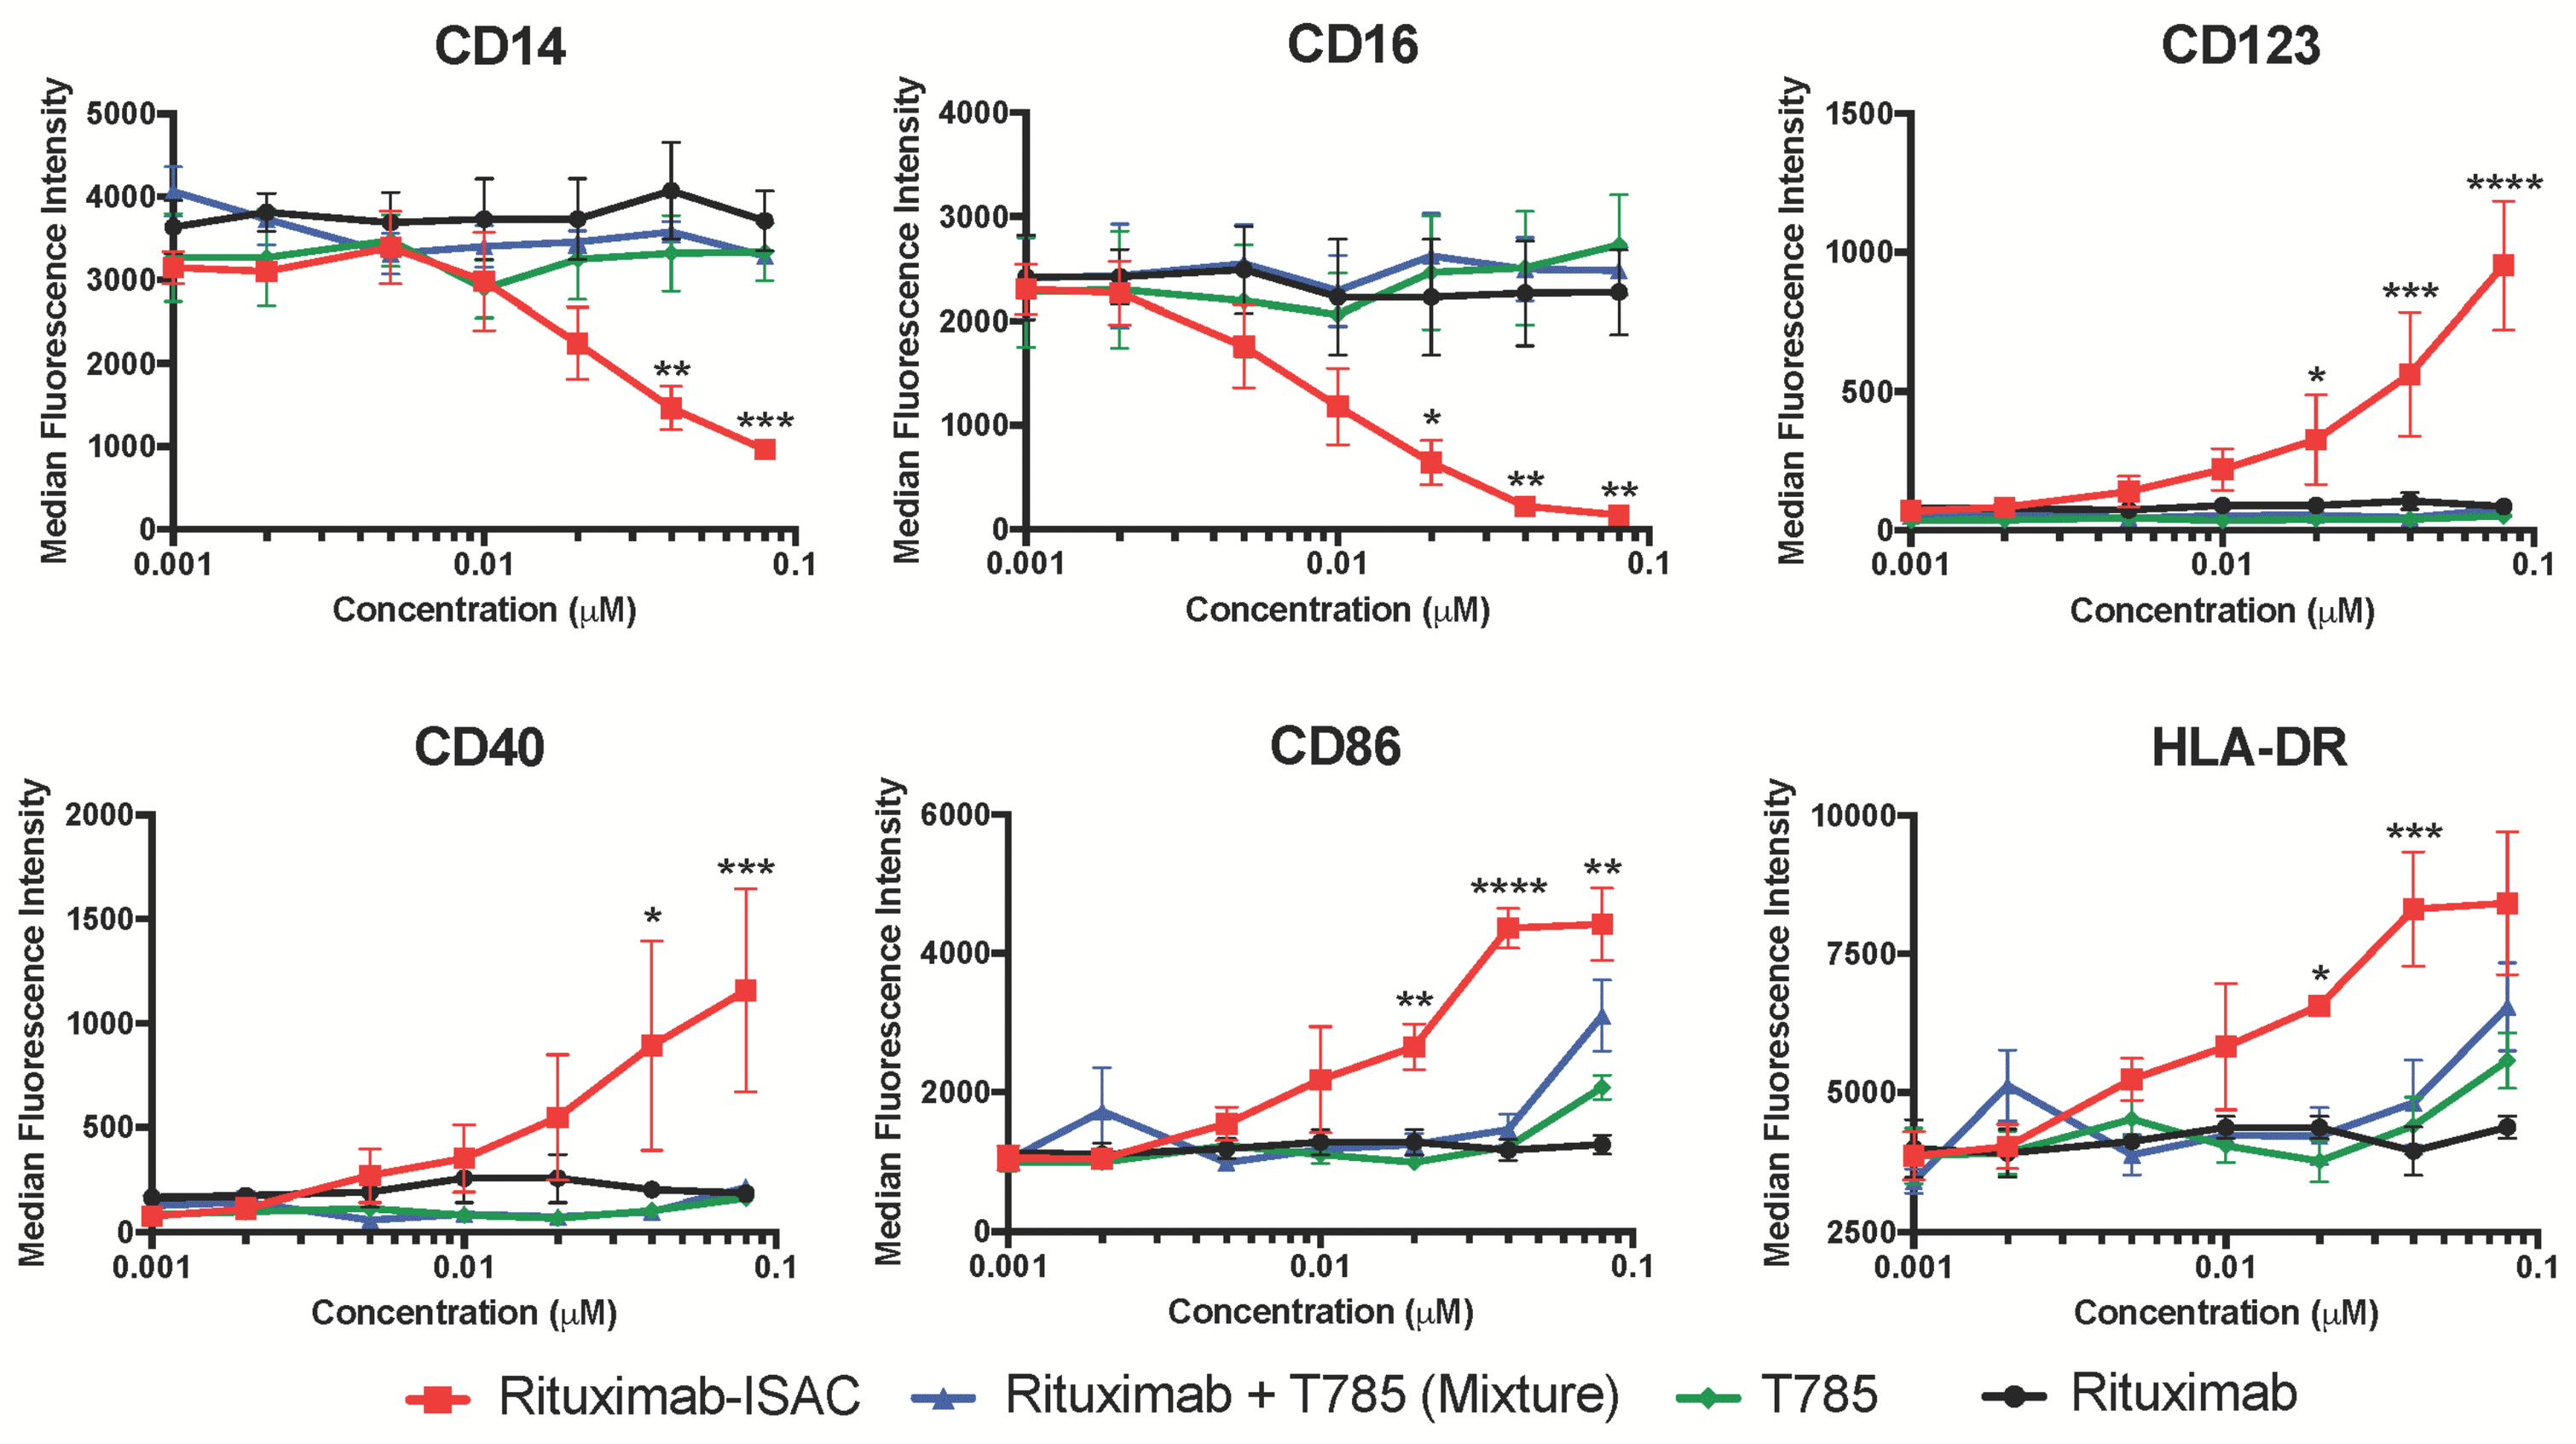
CD14
CD16
CD123
CD40
CD86
HLA-DR
Median Fluorescence Intensity
Concentration (μM)
Rituximab-ISAC
Rituximab + T785 (Mixture)
T785
Rituximab

Figure 3: ISACs elicit myeloid activation and DC differentiation.

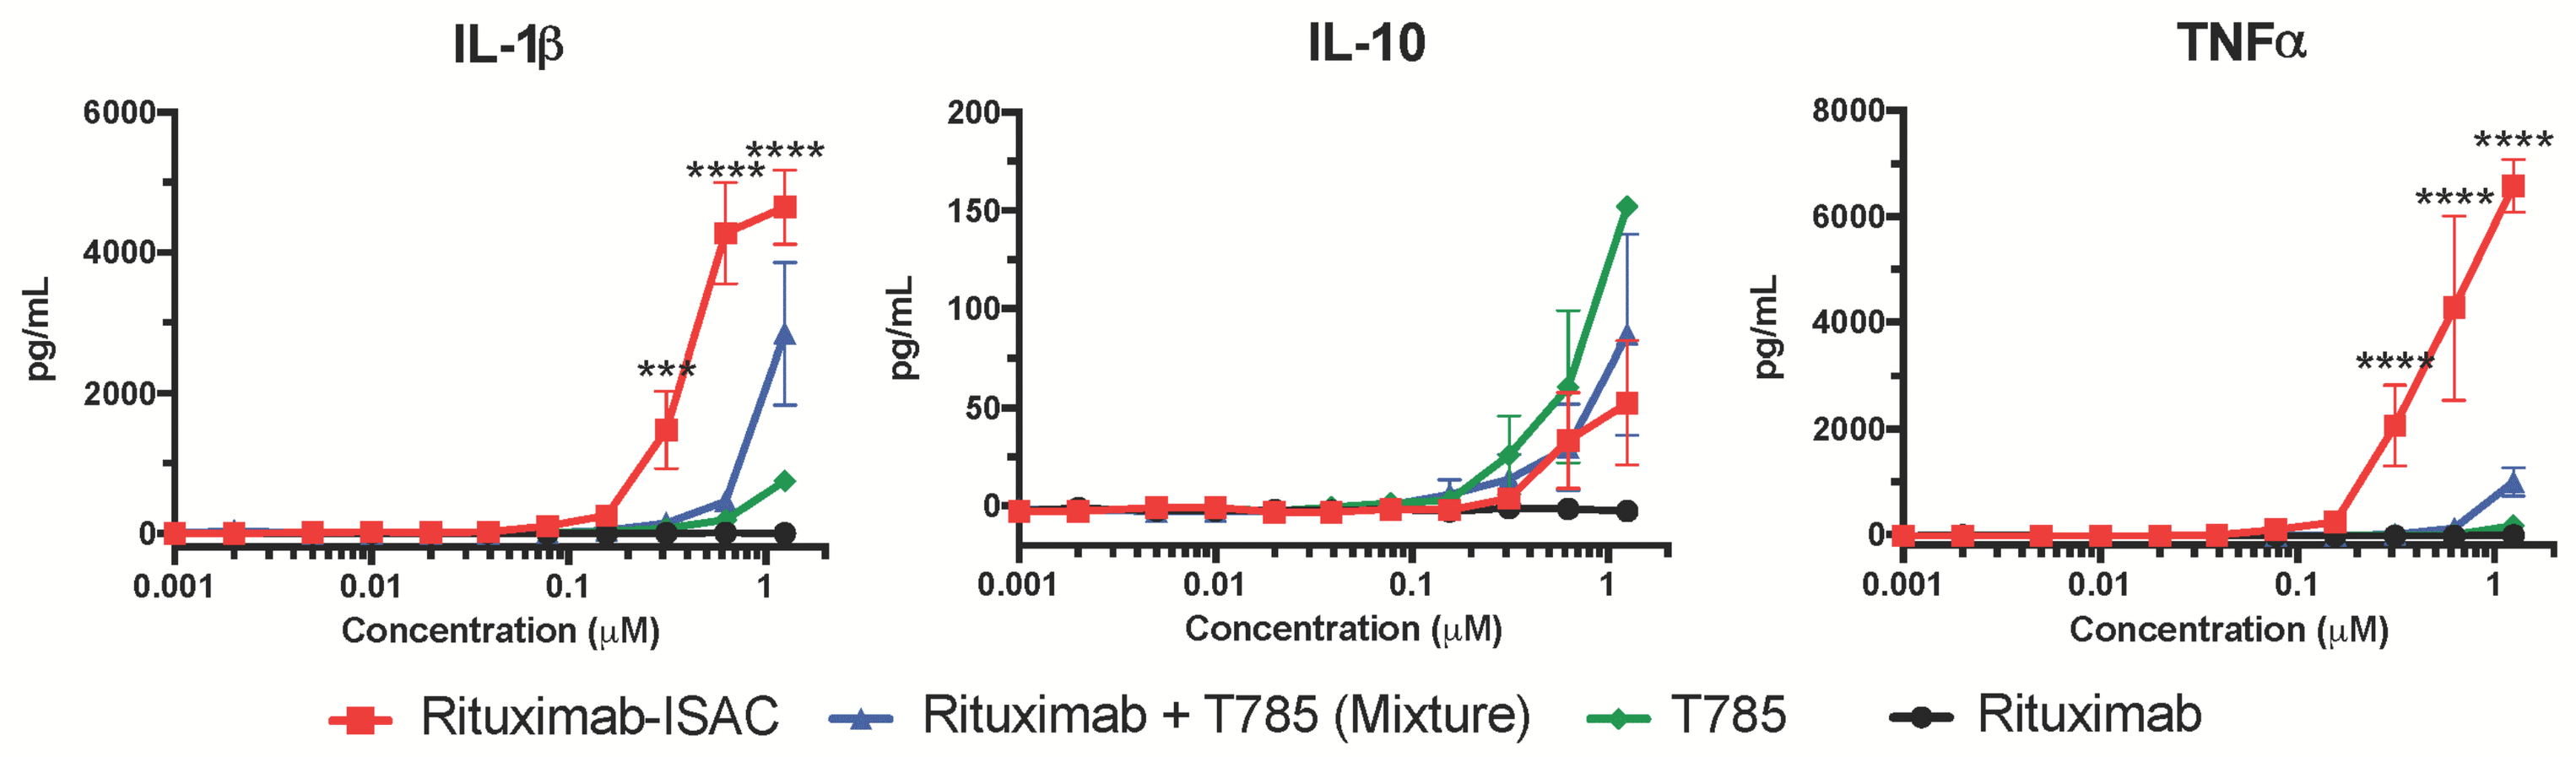
IL-1β
IL-10
TNFα
pg/mL
Concentration (μM)
Rituximab-ISAC
Rituximab + T785 (Mixture)
T785
Rituximab

Figure 4: ISACs elicit proinflammatory cytokine secretion.

IN VIVO EXPERIMENTAL DESIGN

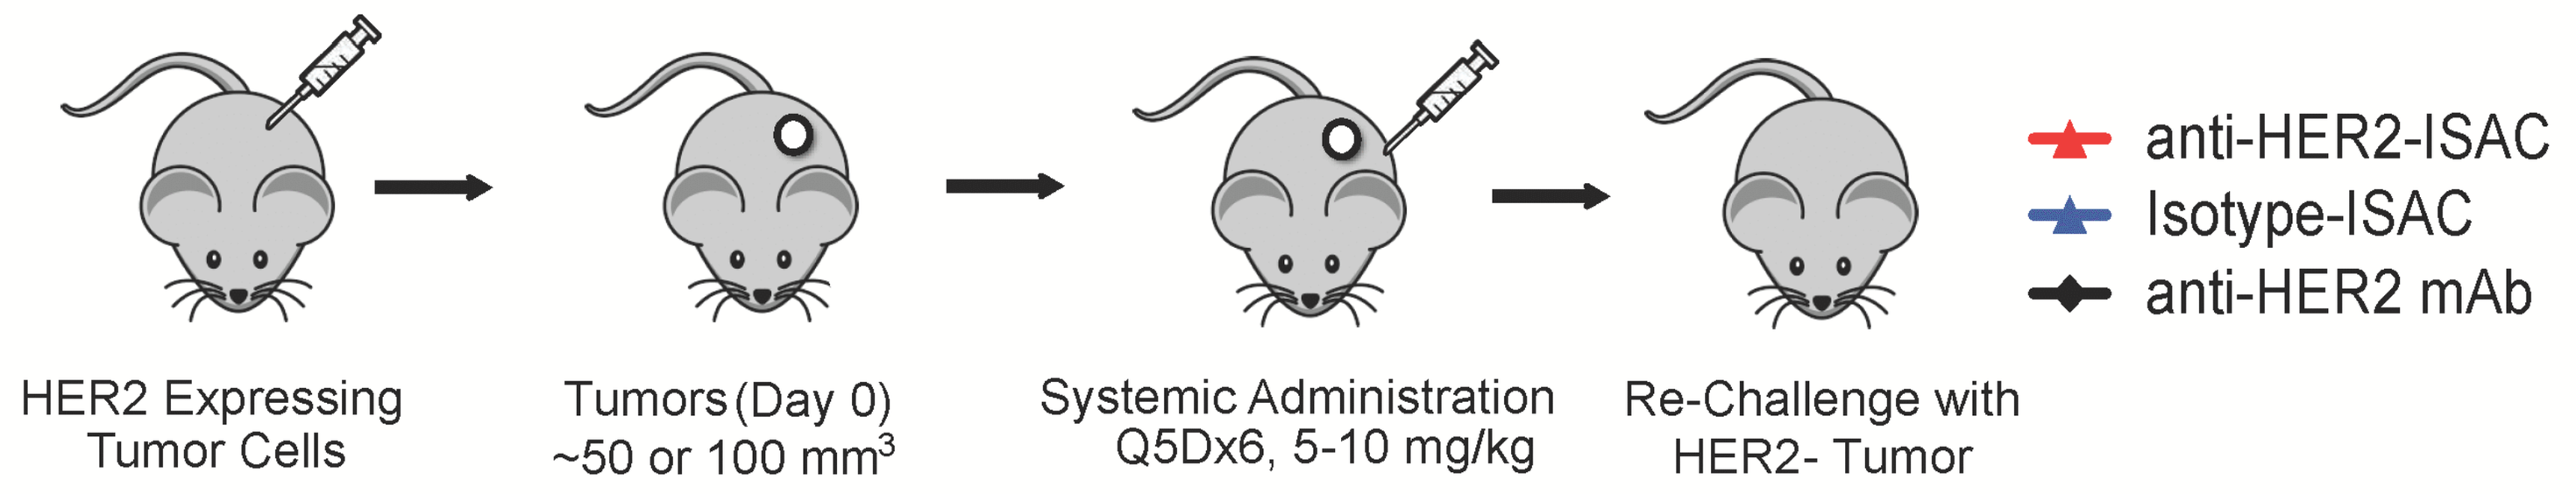
HER2 Expressing Tumor Cells
Tumors (Day 0) ~50 or 100 mm3
Systemic Administration Q5Dx6, 5-10 mg/kg
Re-Challenge with HER2- Tumor
anti-HER2-ISAC
Isotype-ISAC
anti-HER2 mAb

Figure 5: Assessment of ISAC Anti-Tumor Activity. Myeloid-mediated anti-tumor efficacy was assessed in mice that lacked B/T/NK cells with huHER2+ tumor cell lines (Q5Dx6, 5 mg/kg - Figure 6). Immunologic memory was assessed in WT mice with rHER2+ CT26 cells (Q5Dx6, 10 mg/kg) following rechallenge with parental CT26 cells (Figure 7).

IN VIVO RESULTS

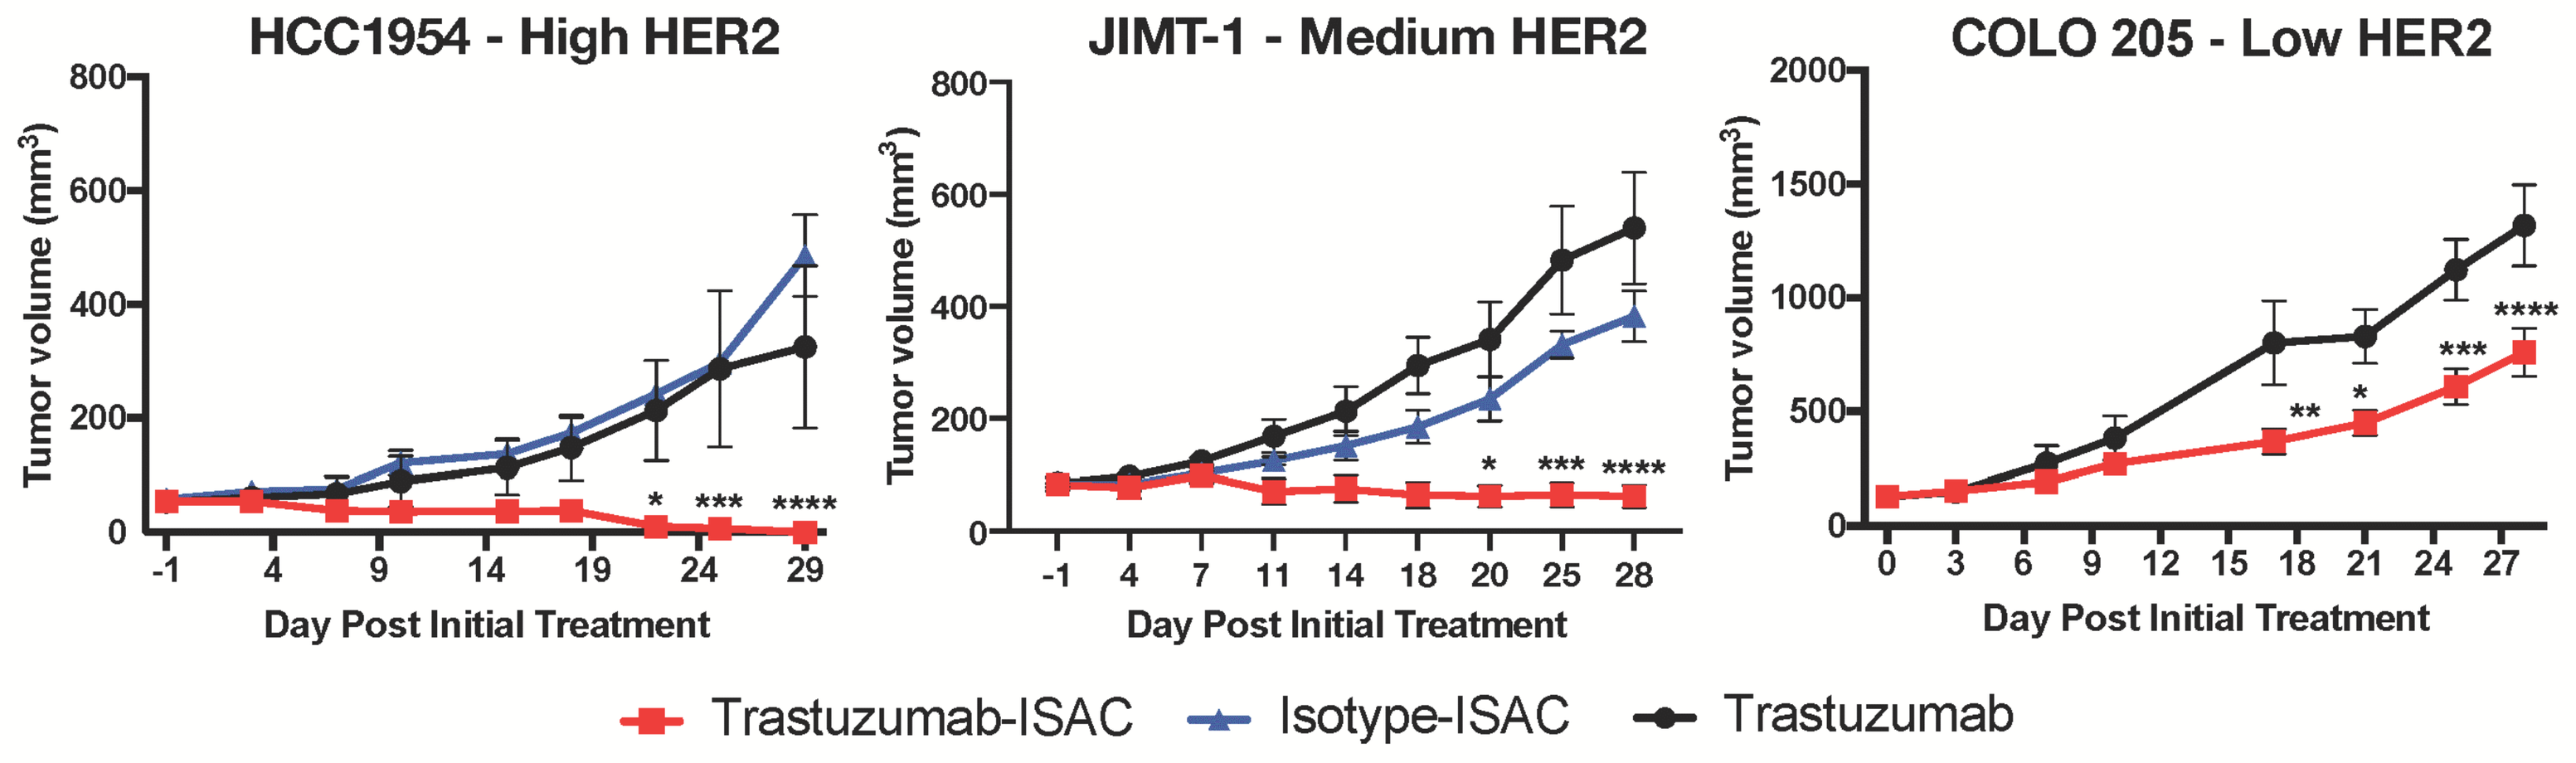
HCC1954 - High HER2
JIMT-1 - Medium HER2
COLO 205 - Low HER2
Tumor volume (mm3)
Day Post Initial Treatment
Trastuzumab-ISAC
Isotype-ISAC
Trastuzumab

Figure 6: ISACs mediate efficacy in trastuzumab-resistant models.

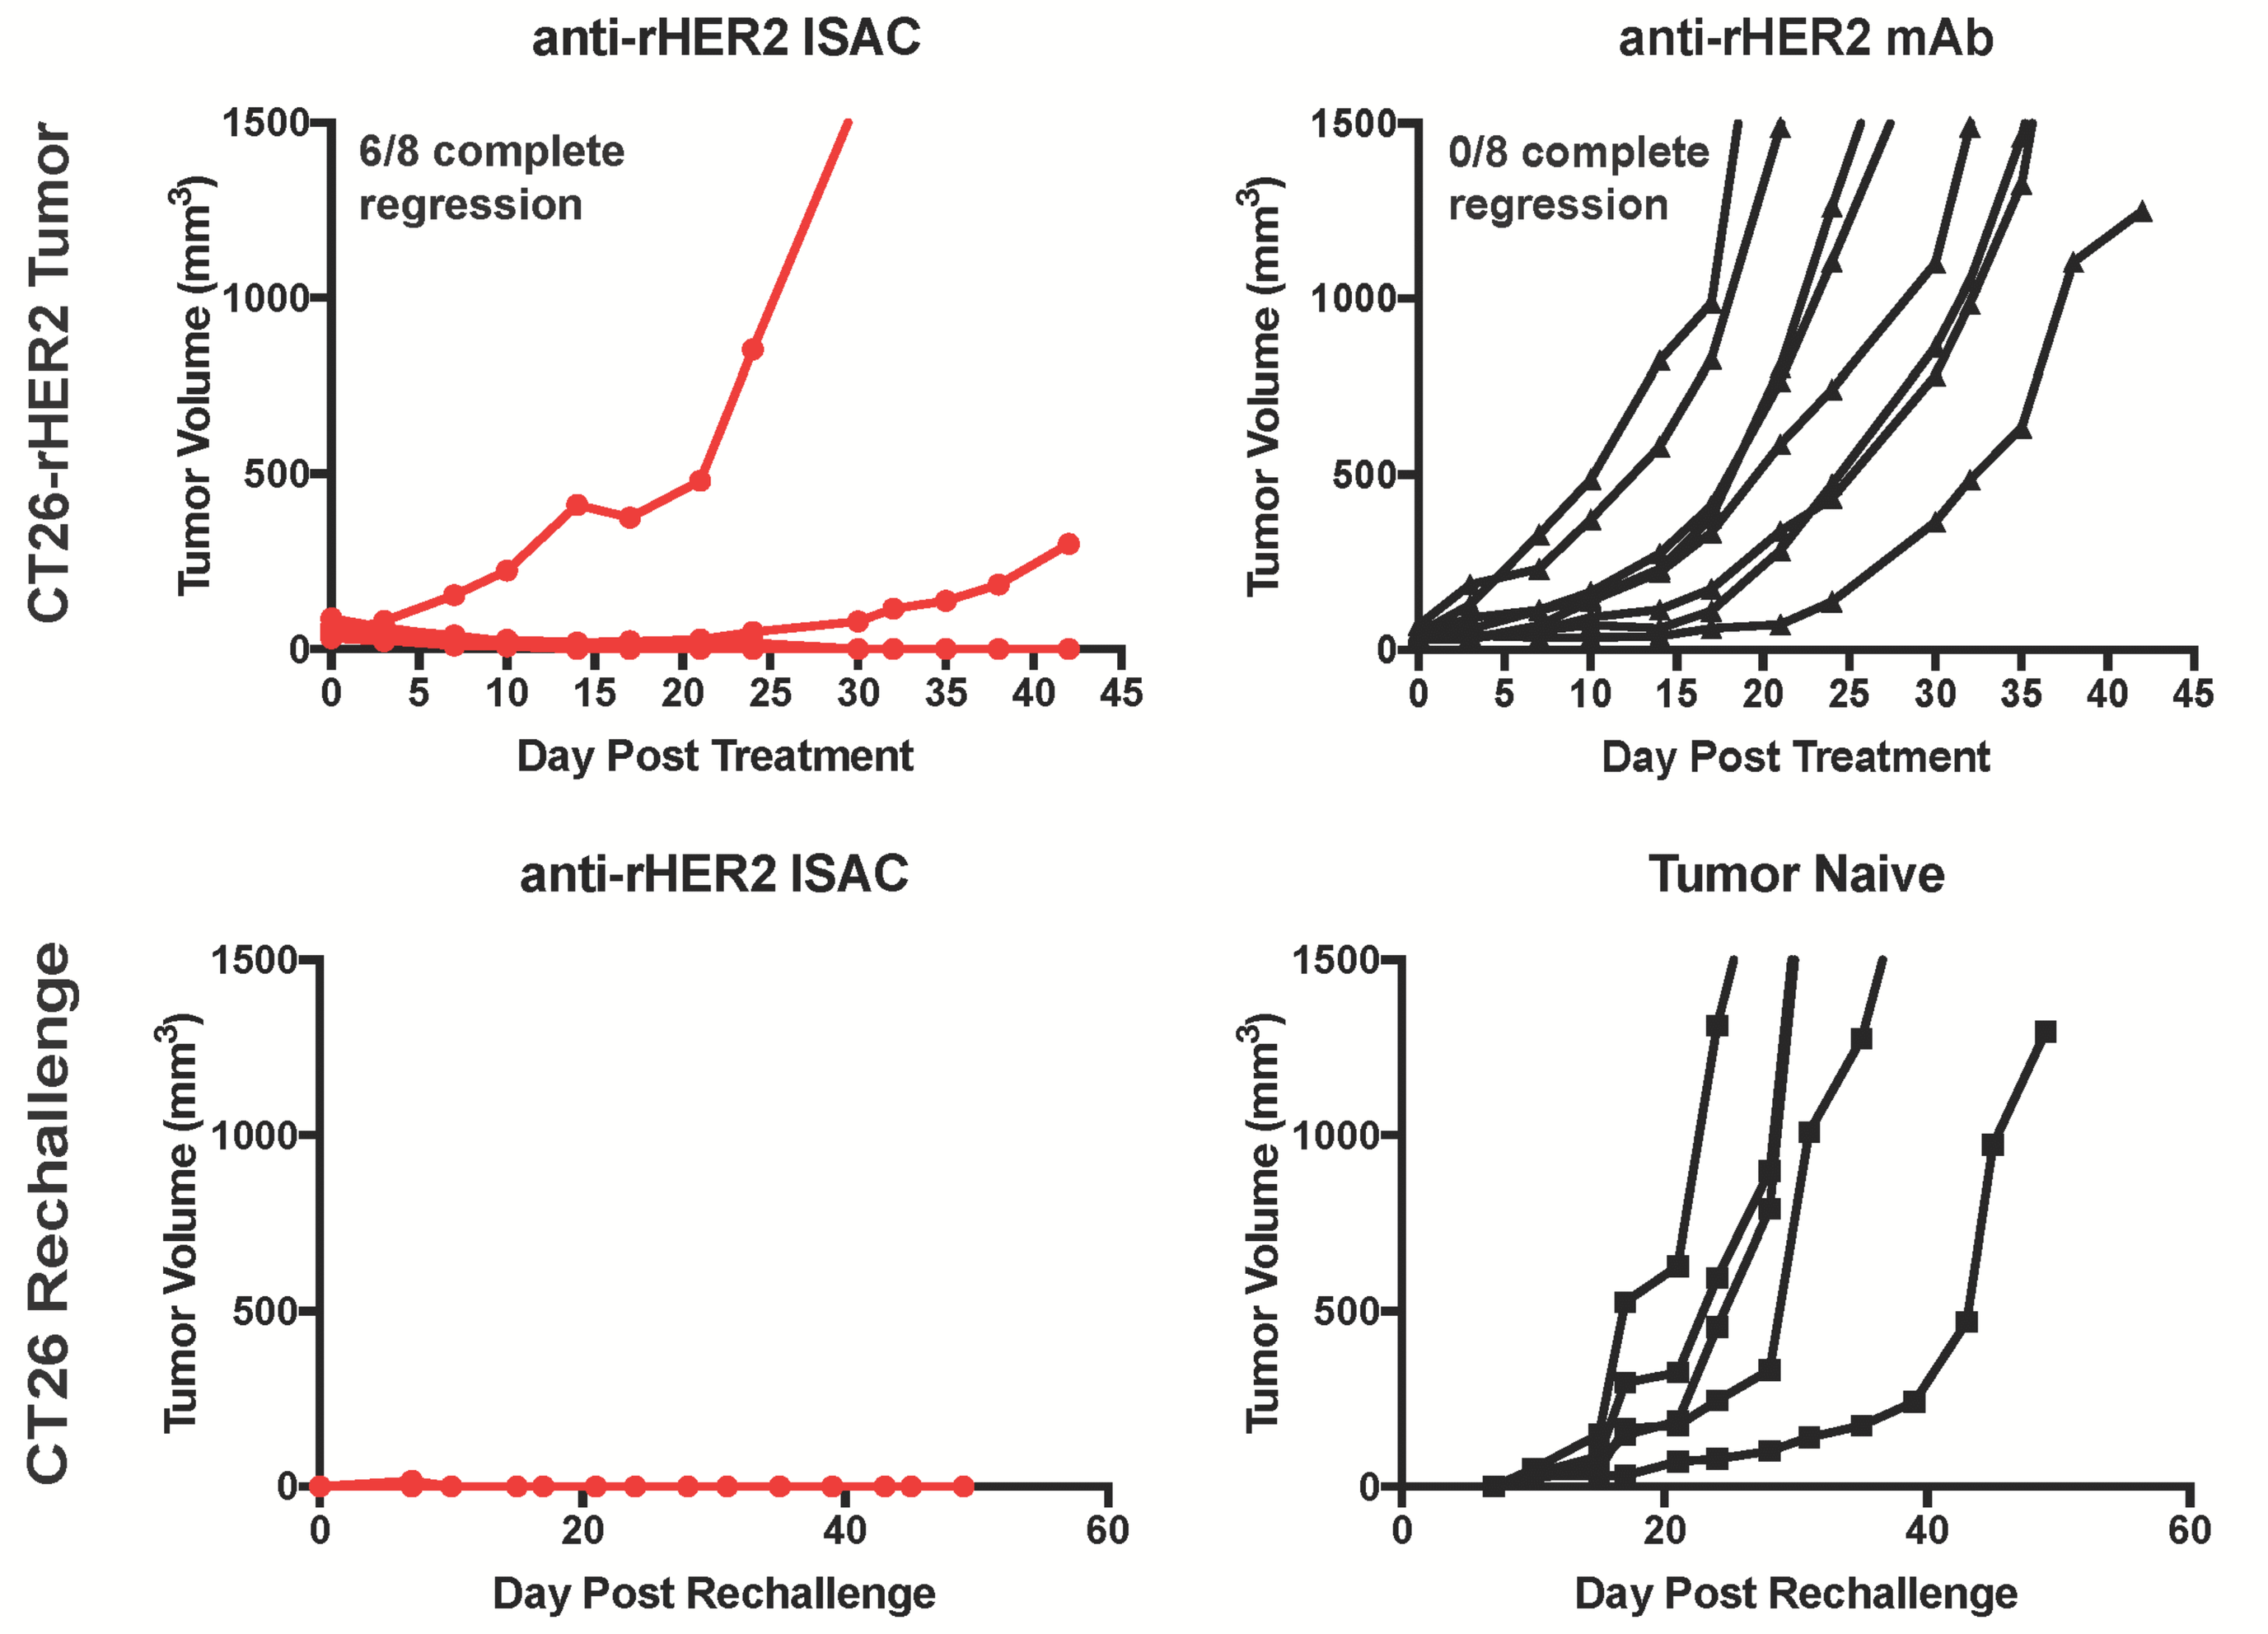
anti-rHER2 ISAC
anti-rHER2 mAb
CT26-rHER2 Tumor
6/8 complete regression
0/8 complete regression
Tumor Volume (mm3)
Day Post Treatment
anti-rHER2 ISAC
Tumor Naive
CT26 Rechallenge
Day Post Rechallenge

Figure 7: ISACs lead to tumor clearance, epitope spreading and govern immunologic memory in syngeneic models.

NHP PHARMACOKINETICS

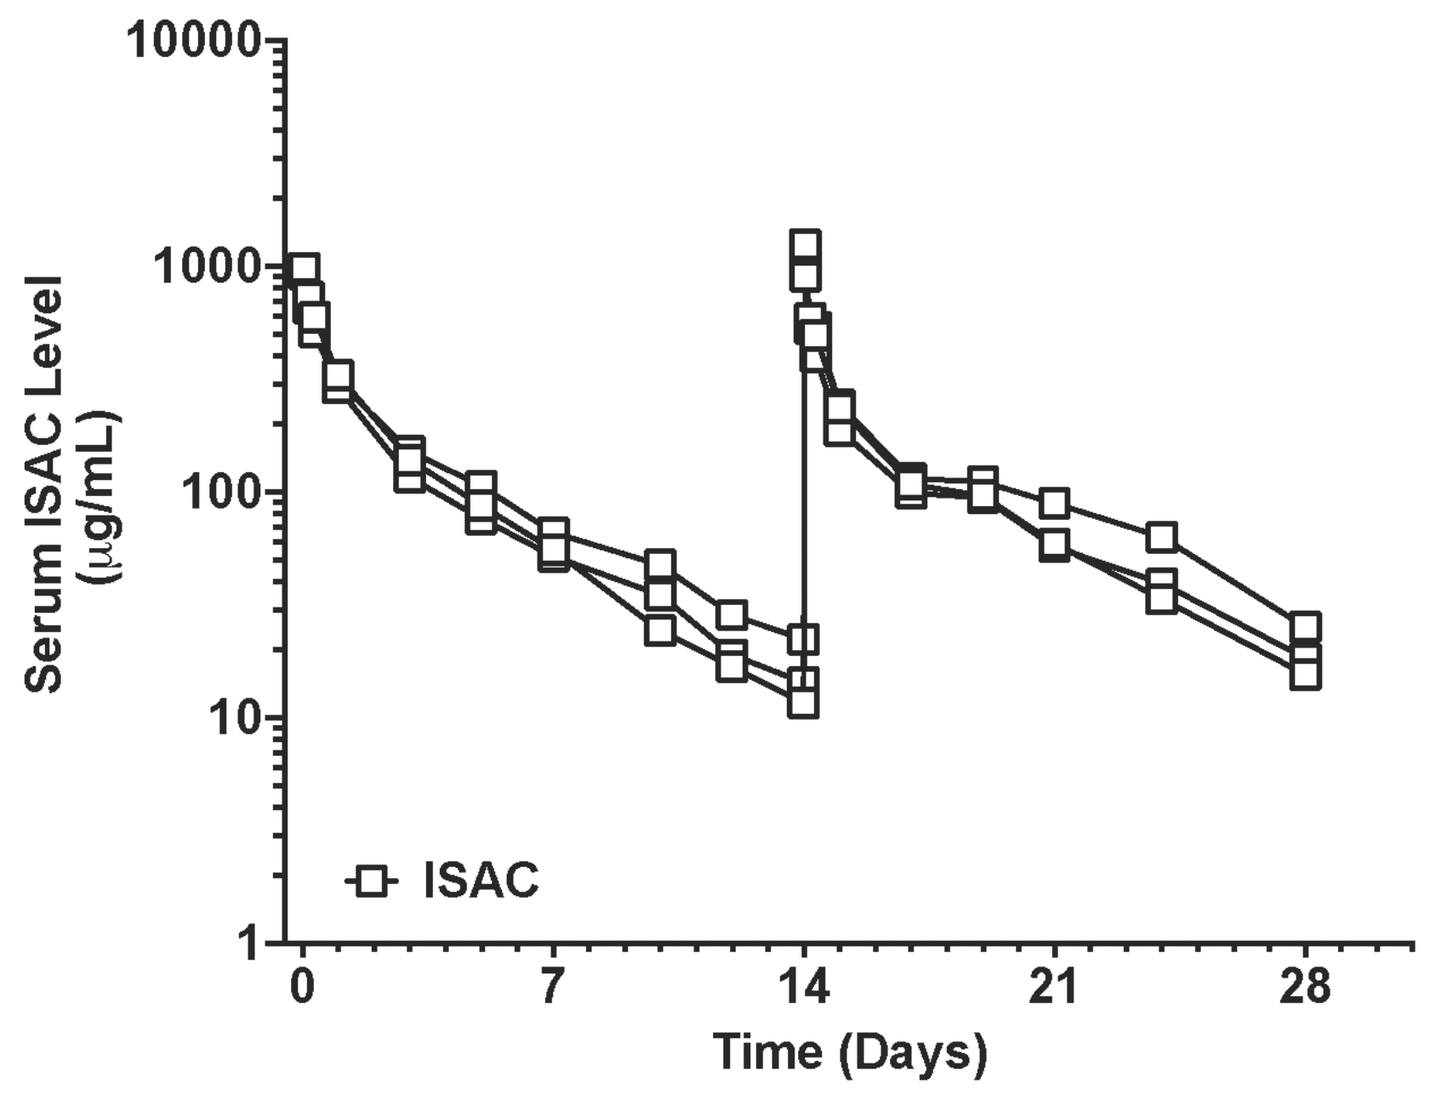
Serum ISAC Level (μg/mL)
Time (Days)
ISAC

3 cynomolgus monkeys dosed every 14 days by intravenous infusion with 30 mg/kg ISAC

Figure 8: ISAC pharmacokinetics in multi-dose NHP study.

KEY FINDINGS

1. ISACs elicit robust myeloid activation and DC differentiation in human cells
2. ISAC treatment leads to anti-tumor efficacy in trastuzumab resistant models
3. ISAC-mediated anti-tumor immunity supports epitope spreading beyond ISAC-target antigen
4. Tumor clearance following ISAC treatment generates immunologic memory
5. ISAC has acceptable pharmacokinetics and is well tolerated in NHP study