HER2-targeting TLR7/8 immune-stimulating antibody conjugates elicit robust myeloid activation and anti-tumor immune responses in a TLR- and FcR- dependent manner.

Shelley E. Ackerman[1,2], Joseph C. Gonzalez[2], Cecelia I. Pearson[2], Justin A. Kenkel[2,3], Joshua D. Gregorio[2], Po Y. Ho[2], Angela Luo[2], Murray L. Nguyen[2], Jason C. Paik[3], Arthur Lee[2], Richard P. Laura[2], Hai Li[2], Cindy Kreder[2], Karla A. Henning[2], Steve J. Chapin[2], Bruce H. Devens[2], Brian S. Safina[2], David Y. Jackson[2], Edgar G. Engleman[3], David Dornan[2], Michael N. Alonso[2,3]

[1]Stanford University Department of Bioengineering, [2]Bolt Biotherapeutics, [3]Stanford University Department of Pathology

ABSTRACT

Background: Antigen presenting cells (APCs) process tumor antigens and facilitate recruitment of tumor-specific T cells to generate a robust anti-tumor immune response. APCs are present in almost all tumor types, including those that are refractory to checkpoint inhibitors; however, the highly immunosuppressive tumor microenvironment (TME) often renders APCs unable to effectively process and present tumor antigens. To overcome this immunosuppressive TME and rescue the function of tumor-infiltrating APCs, we developed an immune-stimulating antibody conjugate (ISAC) through covalent attachment of a TLR7/8 agonist to a HER2-targeting monoclonal antibody.

Results: In vitro co-cultures with human primary leukocytes and cancer cell lines revealed that ISACs potently activates APCs, leading to increased co-stimulatory molecule expression and secretion of pro-inflammatory cytokines. In myeloid-focused in vivo models utilizing HER2-positive human tumor cell line xenografts, which are resistant to trastuzumab, anti-HER2 ISAC treatment led to tumor regression and clearance. Further mechanistic studies revealed that anti-HER2 ISAC efficacy was TLR- and FcR- dependent. In syngeneic tumor models in which anti-HER2 ISAC treatment led to tumor clearance, animals rechallenged with the parental tumor cell line lacking HER2 antigen expression were protected from tumor growth. In addition, depletion of CD4 and CD8 T cells prior to rechallenge led to loss of immunological memory.

Conclusions: These data provide a strong rationale for pursuing an anti-HER2 TLR7/8 ISAC into the clinic for the potential treatment of HER2-positive cancers.

PROPOSED MECHANISM OF ACTION

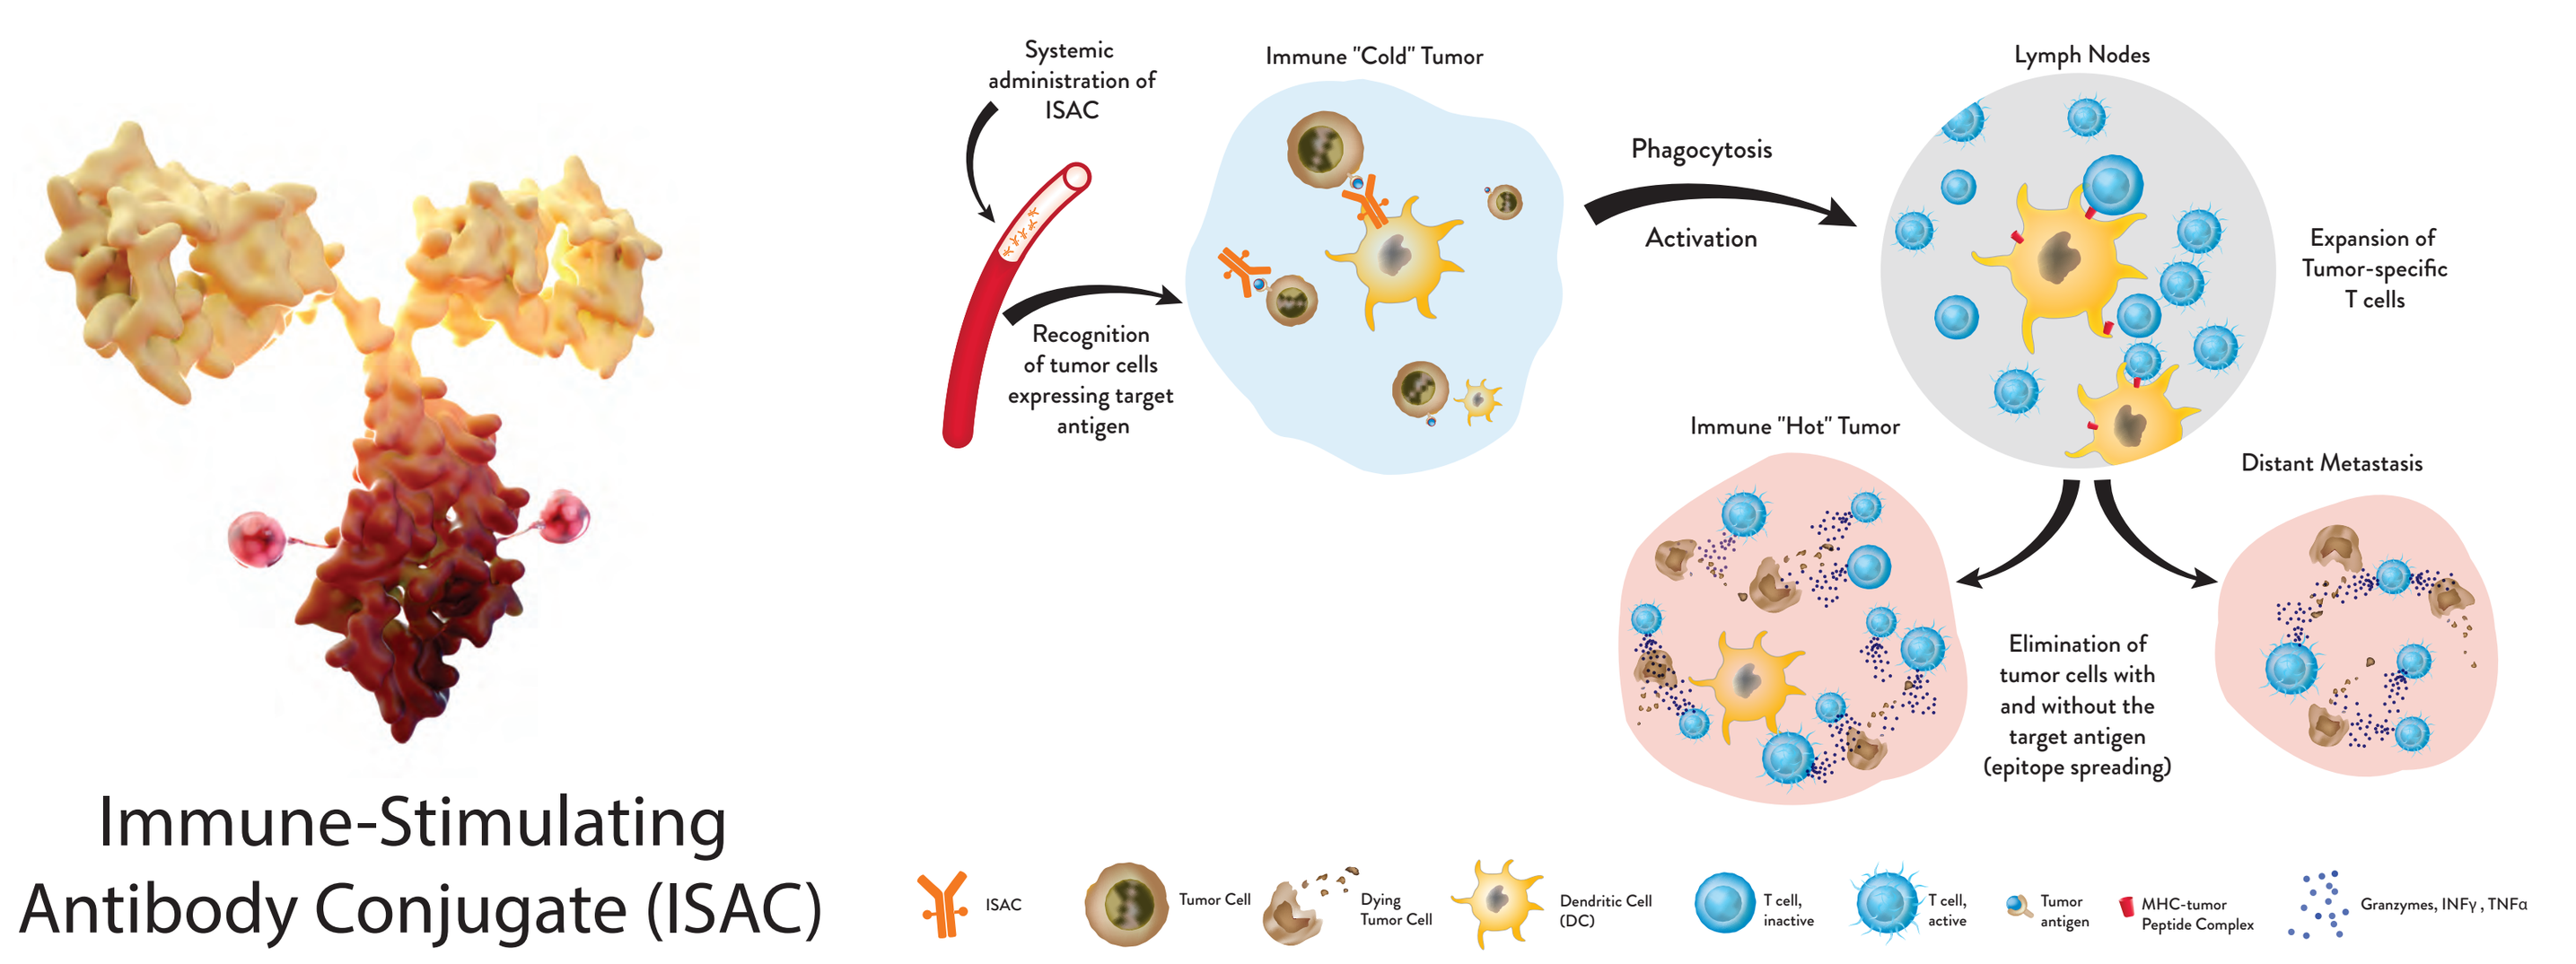
Immune-Stimulating Antibody Conjugate (ISAC)
Systemic administration of ISAC
Immune "Cold" Tumor
Recognition of tumor cells expressing target antigen
Phagocytosis
Activation
Lymph Nodes
Expansion of Tumor-specific T cells
Immune "Hot" Tumor
Distant Metastasis
Elimination of tumor cells with and without the target antigen (epitope spreading)

HUMAN IN VITRO EXPERIMENTAL DESIGN

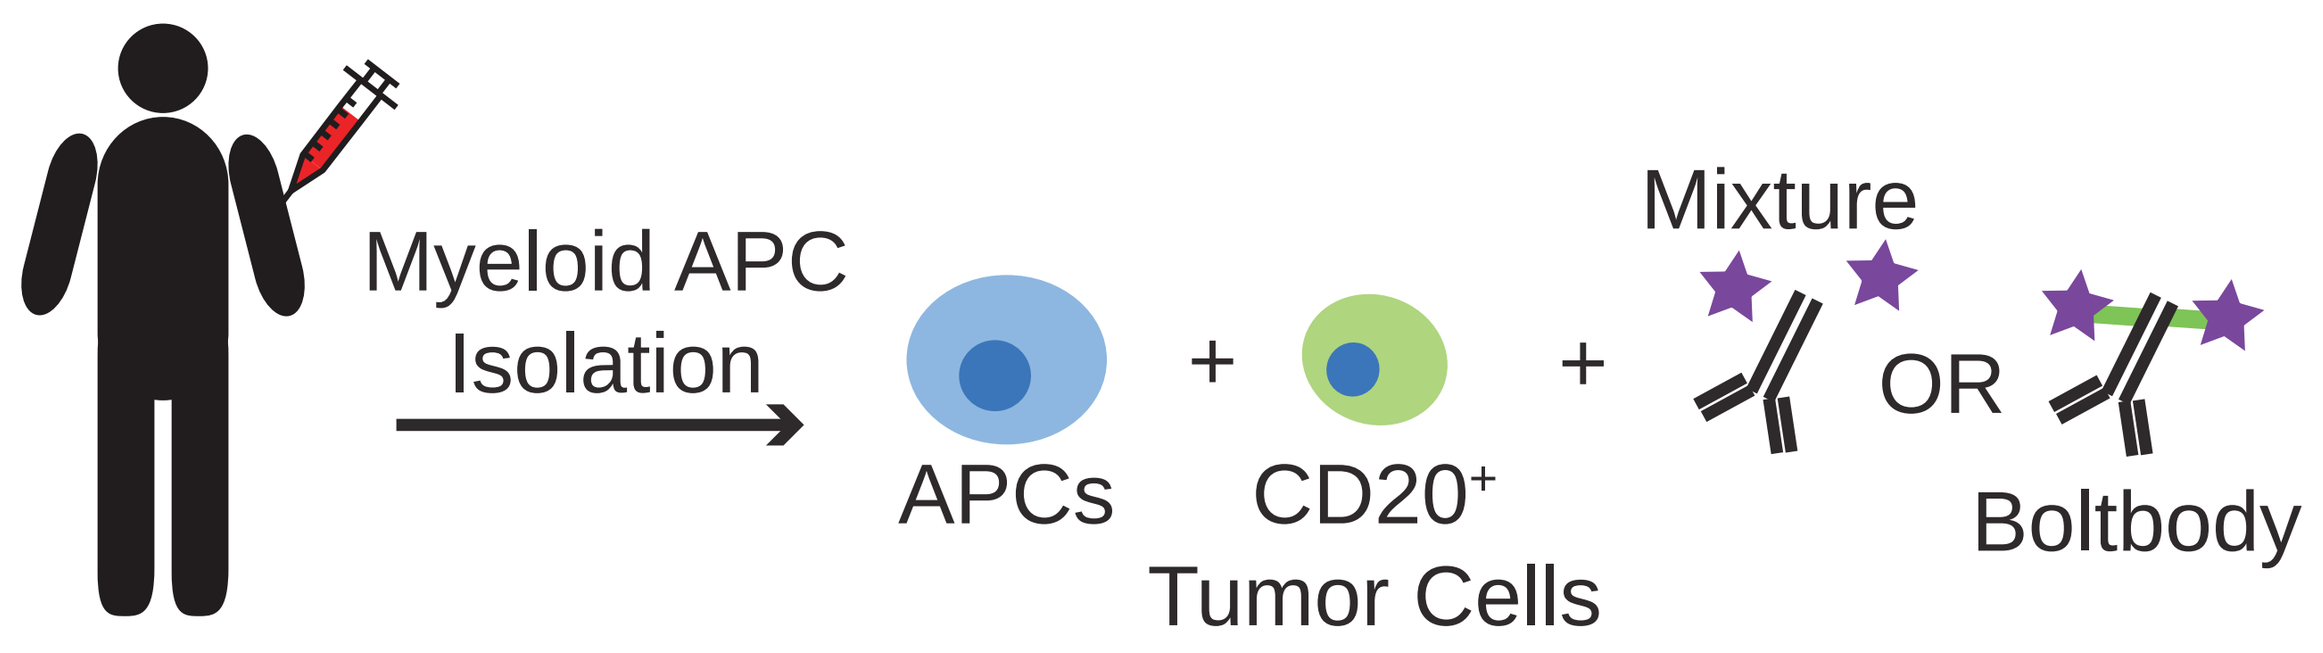
Myeloid APC Isolation
APCs
+
CD20+ Tumor Cells
+
Mixture
OR
Boltbody

Figure 1: Assessment of ISAC Activity. Myeloid APCs were isolated from healthy donor via negative selection and co-cultured with CFSE-labeled CD20+ tumor cells. After 18 hours, cells were assessed for cellular activation/differentiation via microscopy and flow cytometry (Figures 2-5).

HUMAN IN VITRO RESULTS

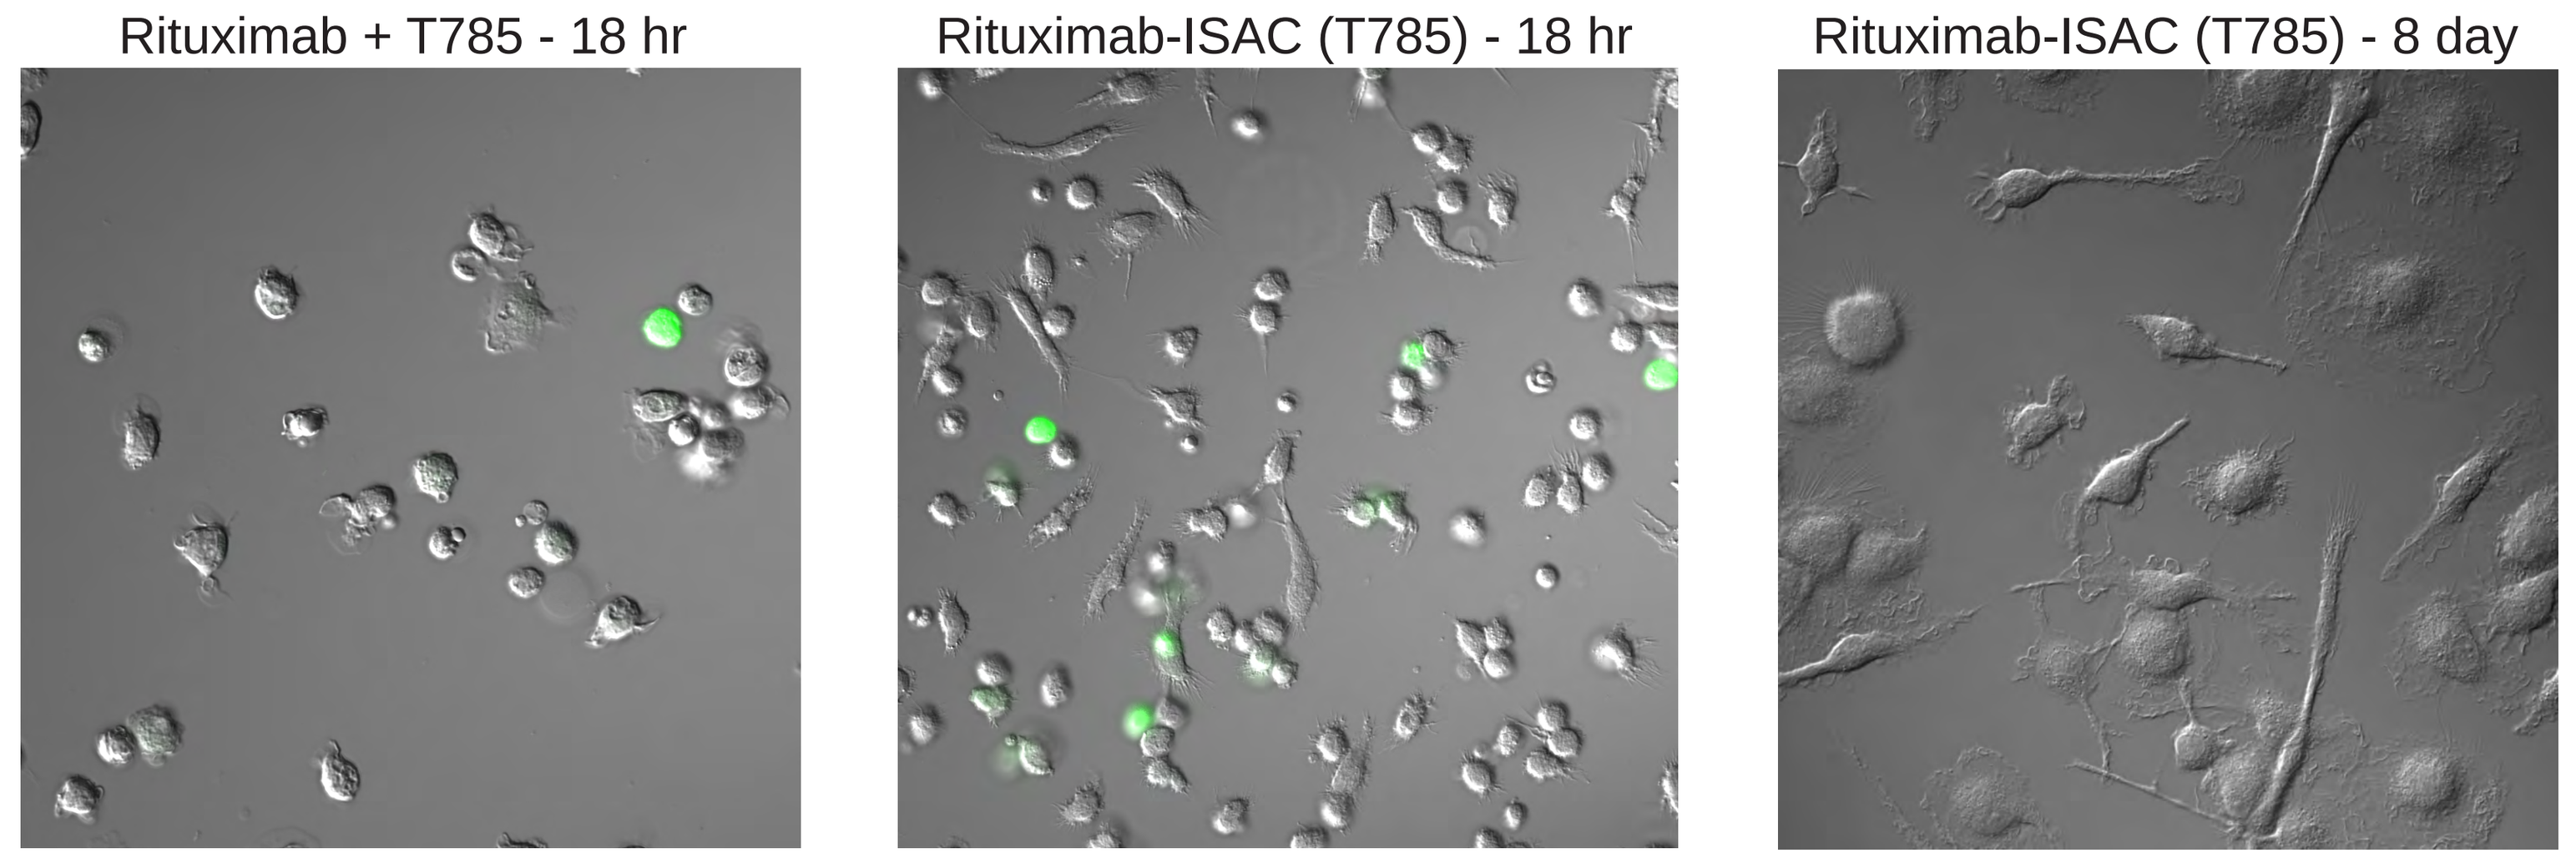
Rituximab + T785 - 18 hr
Rituximab-ISAC (T785) - 18 hr
Rituximab-ISAC (T785) - 8 day

Figure 2: ISACs elicit distinct changes in cellular morphology.

HUMAN IN VITRO RESULTS

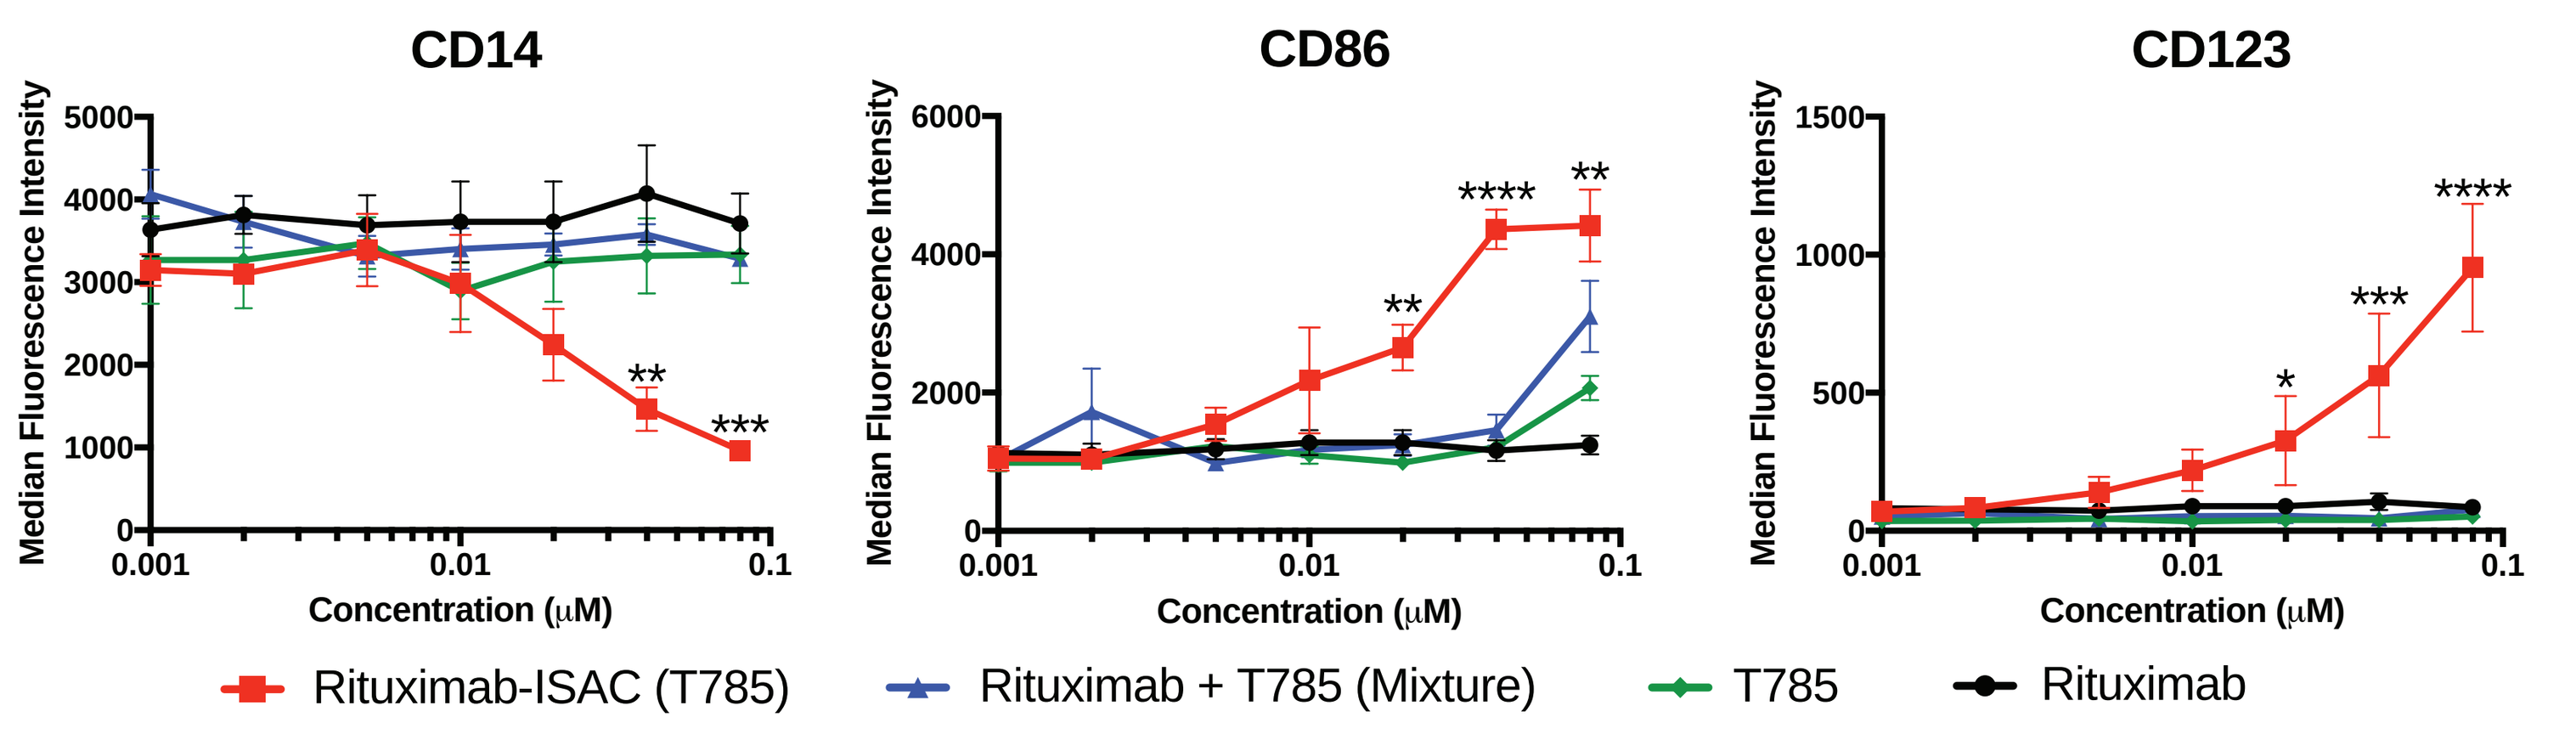
CD14
CD86
CD123
Median Fluorescence Intensity
Concentration (μM)
Rituximab-ISAC (T785)
Rituximab + T785 (Mixture)
T785
Rituximab

Figure 3: ISACs elicit myeloid activation and DC differentiation.

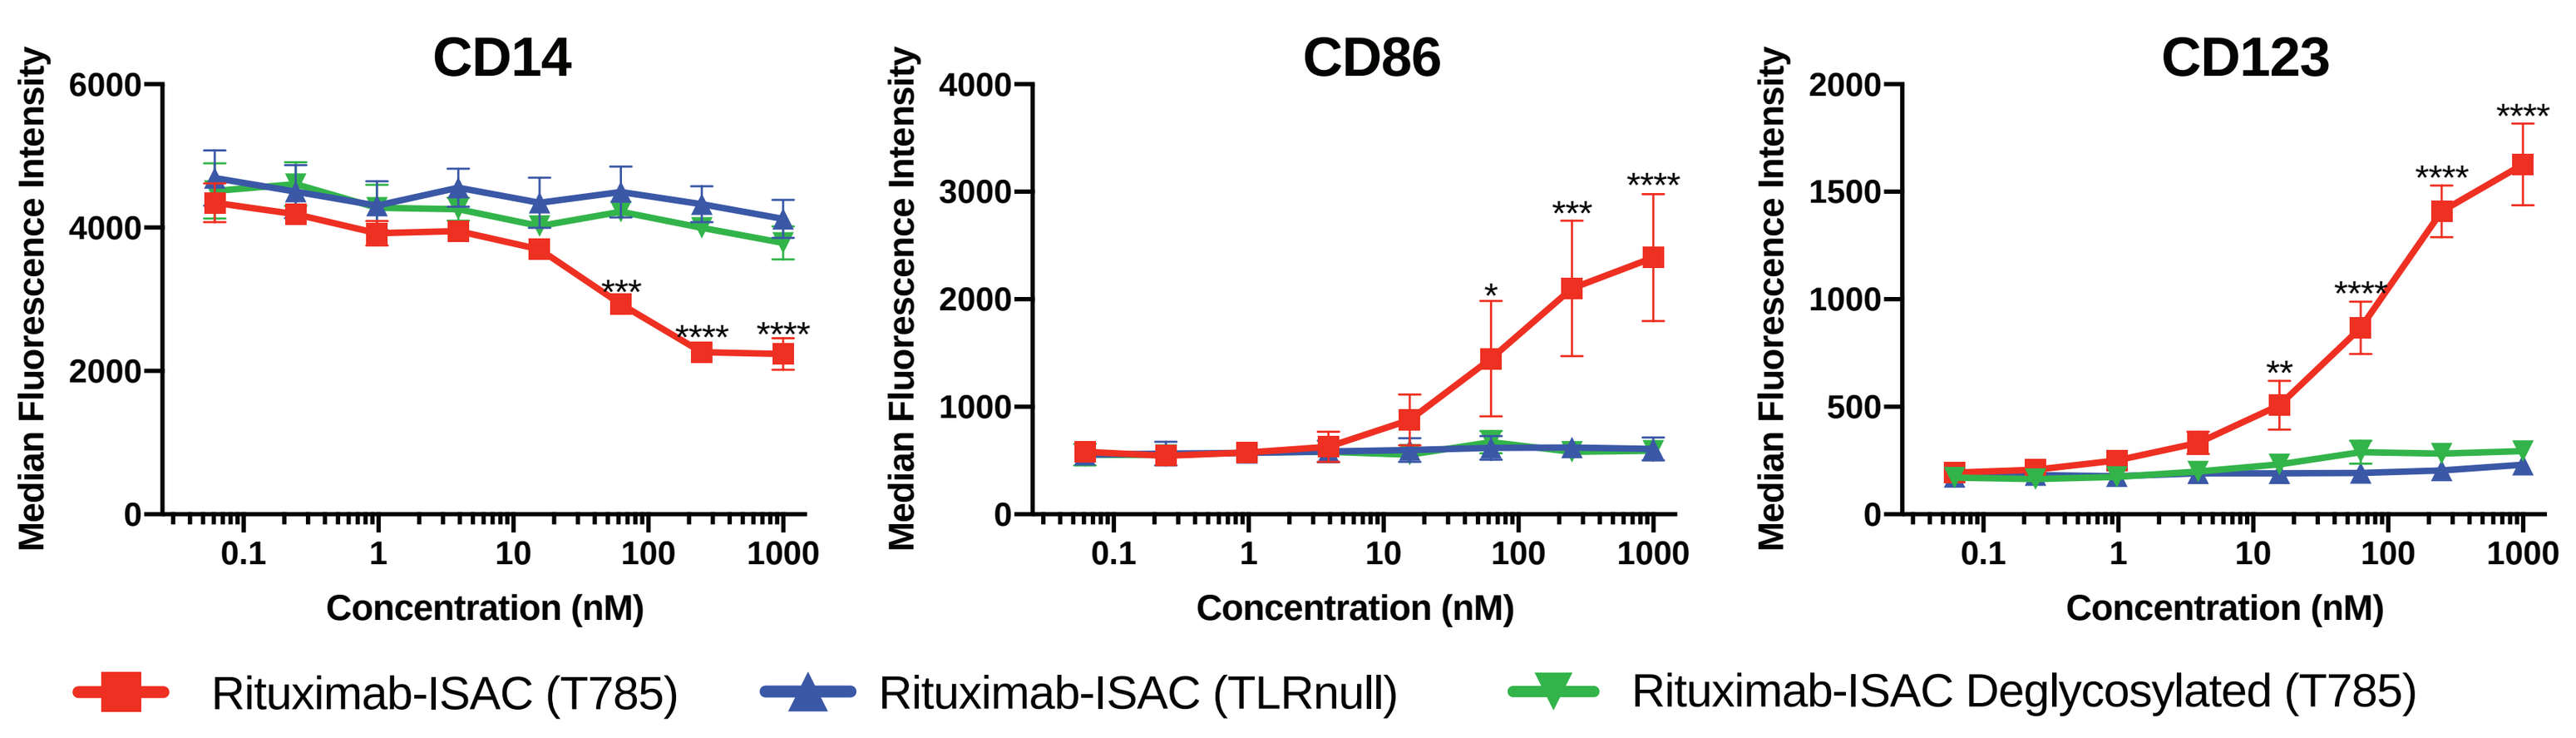
CD14
CD86
CD123
Median Fluorescence Intensity
Concentration (nM)
Rituximab-ISAC (T785)
Rituximab-ISAC (TLRnull)
Rituximab-ISAC Deglycosylated (T785)

Figure 4: ISACs require intact Fc and TLR engagement for in vitro potency.

IN VIVO EXPERIMENTAL DESIGN

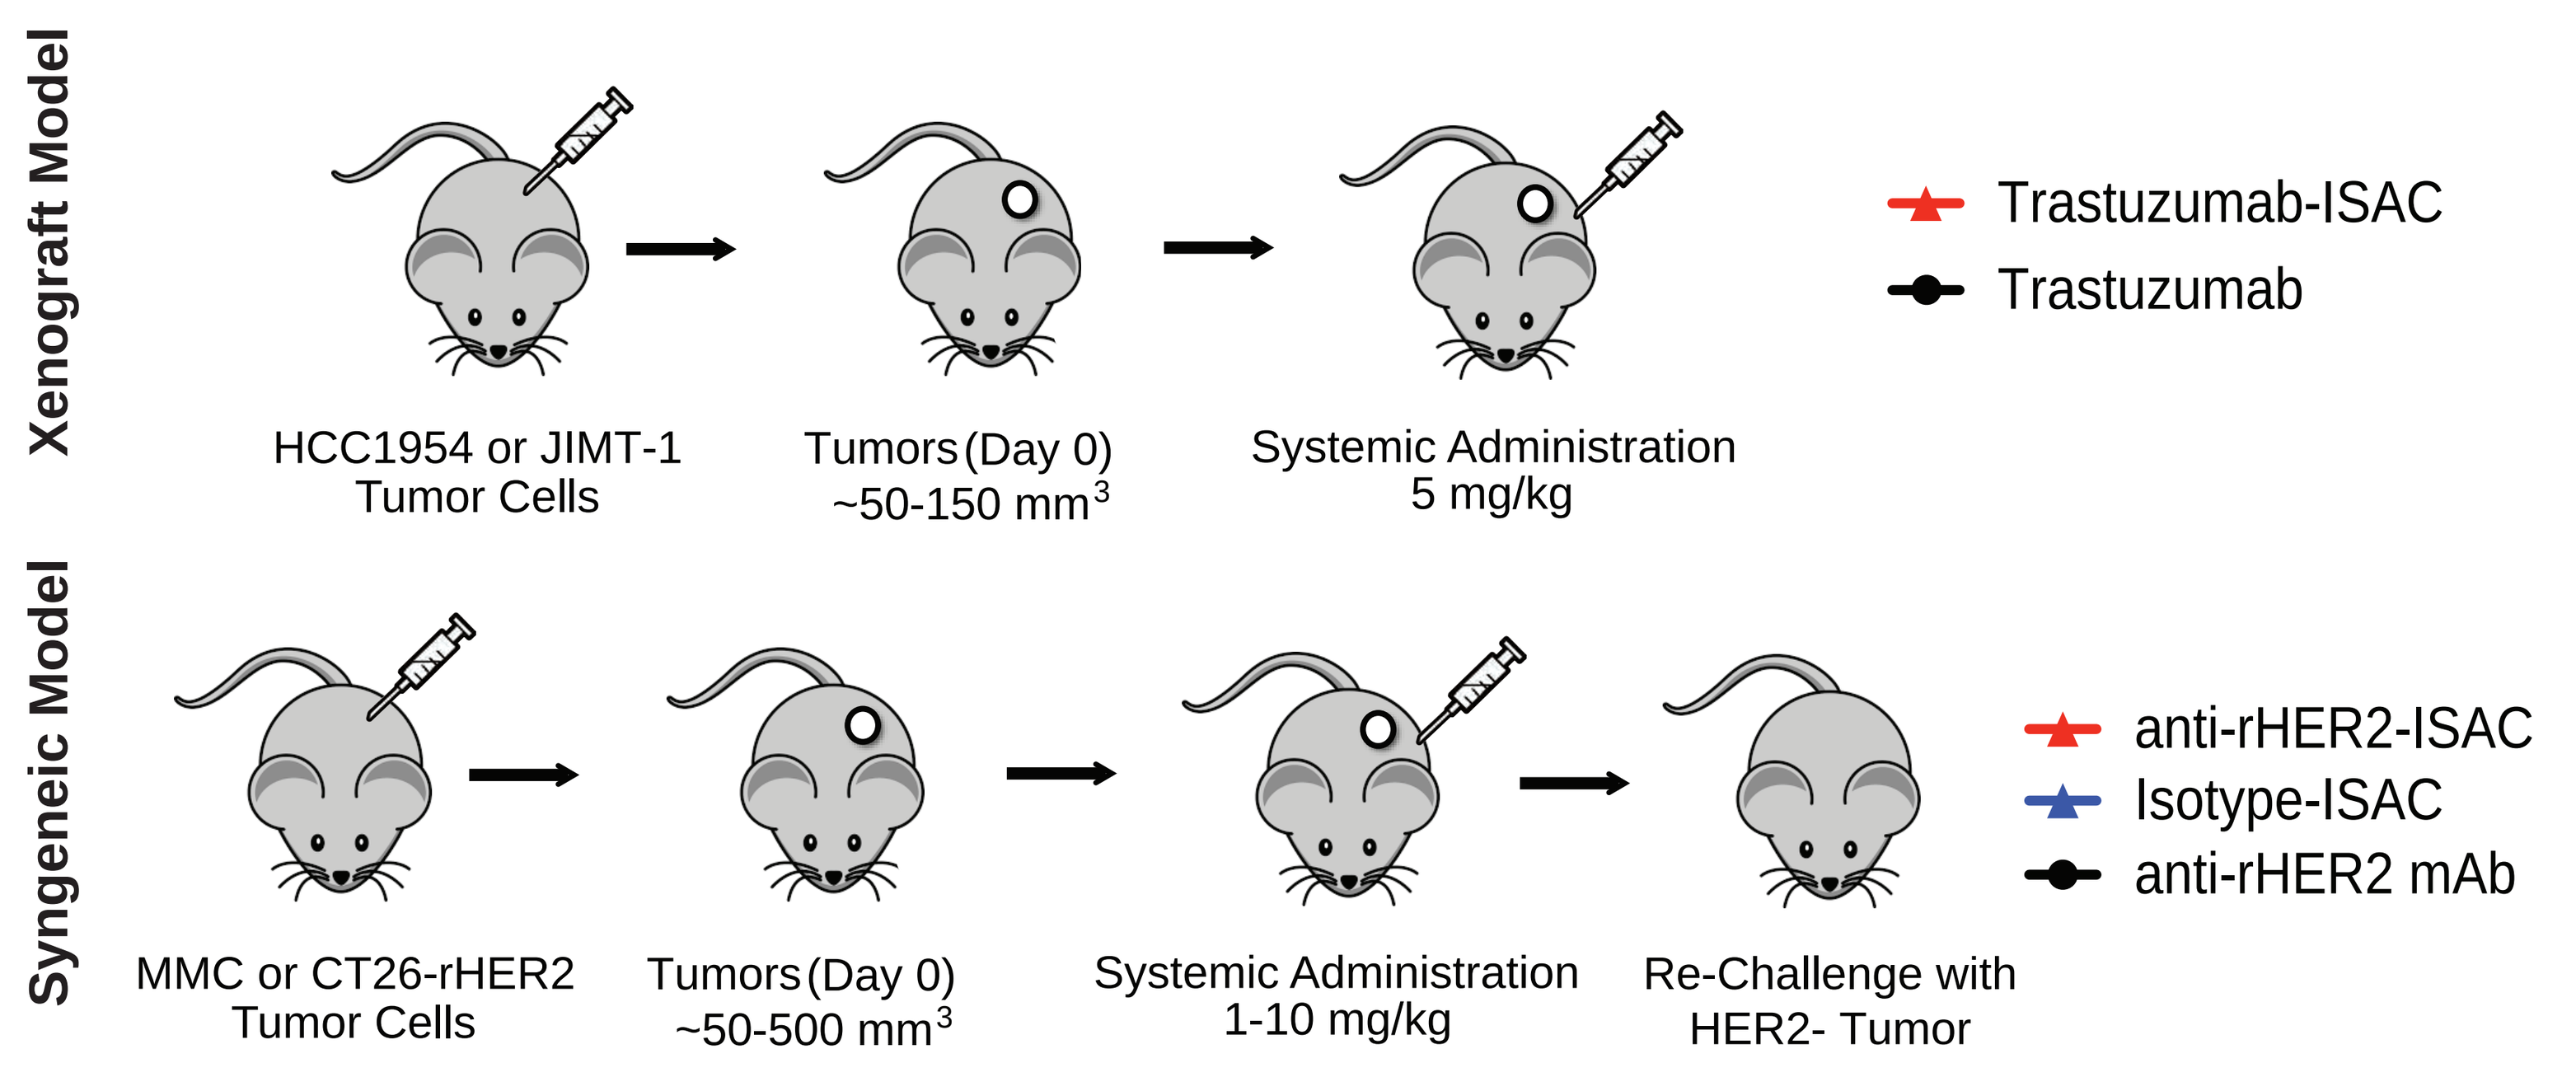
Xenograft Model
HCC1954 or JIMT-1 Tumor Cells
Tumors (Day 0) ~50-150 mm3
Systemic Administration 5 mg/kg
Trastuzumab-ISAC
Trastuzumab
Syngeneic Model
MMC or CT26-rHER2 Tumor Cells
Tumors (Day 0) ~50-500 mm3
Systemic Administration 1-10 mg/kg
Re-Challenge with HER2- Tumor
anti-rHER2-ISAC
Isotype-ISAC
anti-rHER2 mAb

Figure 5: Assessment of ISAC Anti-Tumor Activity. Myeloid-mediated anti-tumor efficacy was assessed in mice that lacked B/T/NK cells with huHER2+ tumor cell lines (Q5Dx6, 5 mg/kg - Figure 6-8), or in immunocompotent syngeneic rHER2+ tumor cell lines (Figure 9-10).

IN VIVO RESULTS

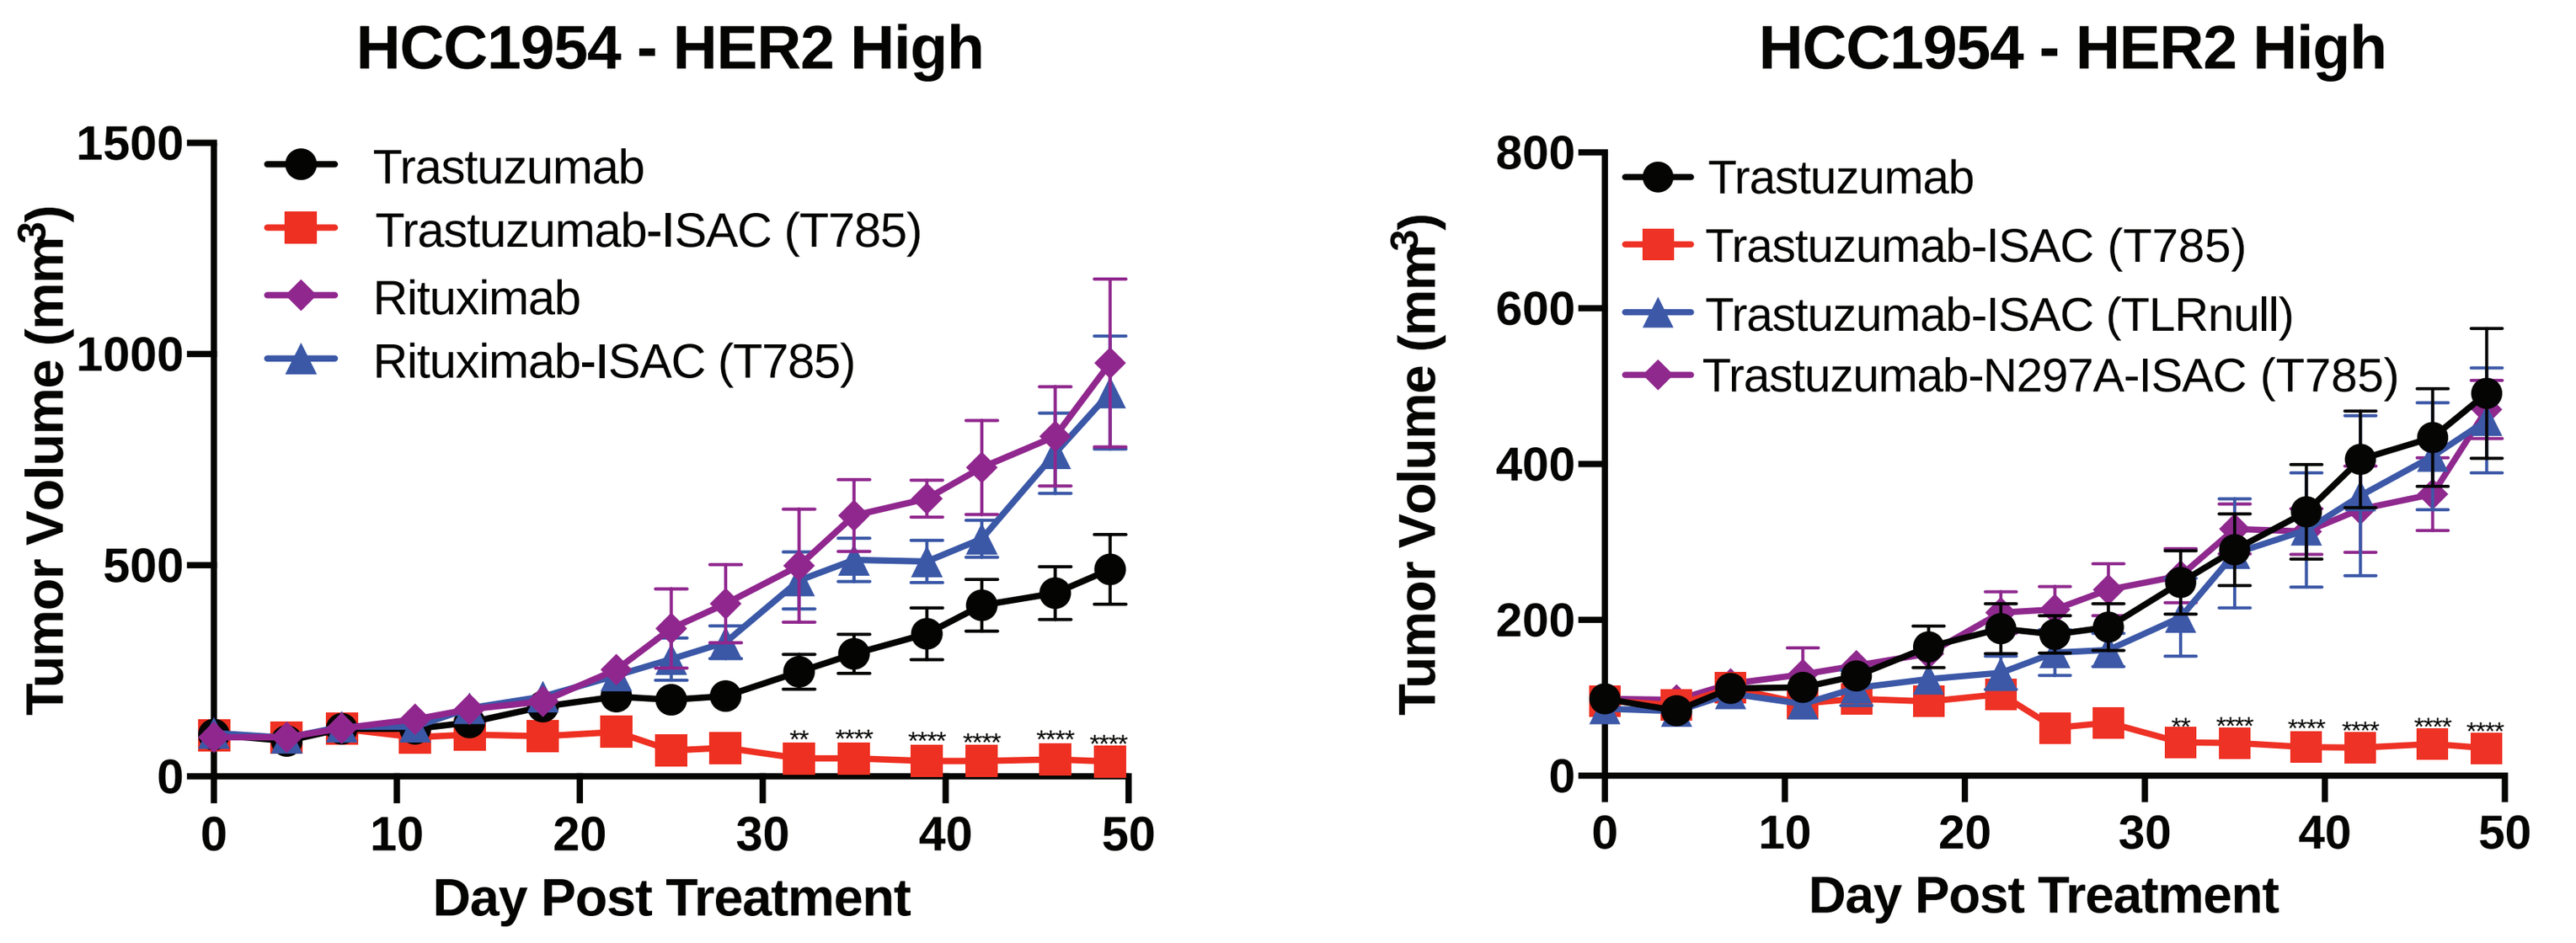
HCC1954 - HER2 High
Tumor Volume (mm3)
Day Post Treatment
Trastuzumab
Trastuzumab-ISAC (T785)
Rituximab
Rituximab-ISAC (T785)
HCC1954 - HER2 High
Trastuzumab
Trastuzumab-ISAC (T785)
Trastuzumab-ISAC (TLRnull)
Trastuzumab-N297A-ISAC (T785)

Figure 6: T785-containing ISACs require TLR- and FcR- engagement for anti-tumor efficacy.

IN VIVO RESULTS

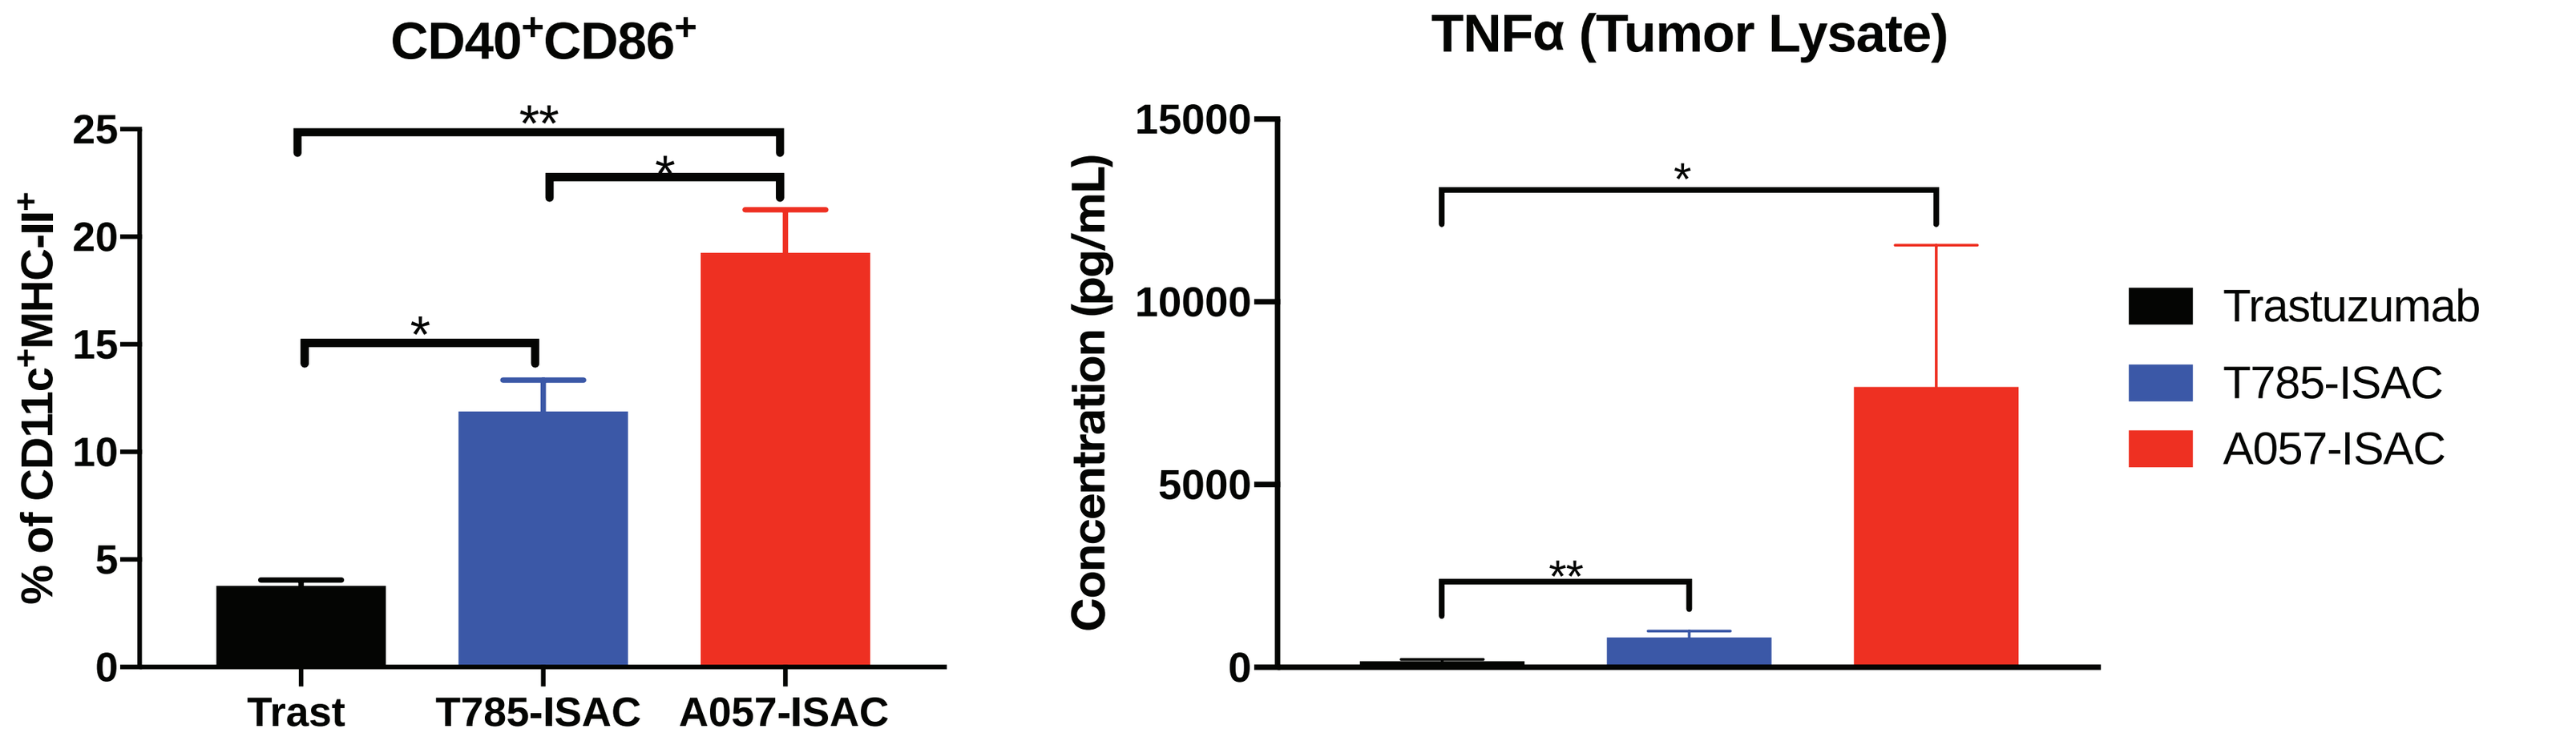
CD40+CD86+
% of CD11c+MHC-II+
Trast
T785-ISAC
A057-ISAC
TNFα (Tumor Lysate)
Concentration (pg/mL)
Trastuzumab
T785-ISAC
A057-ISAC

Figure 7: A057-ISACs exhibit greater potency than T785-ISACs.

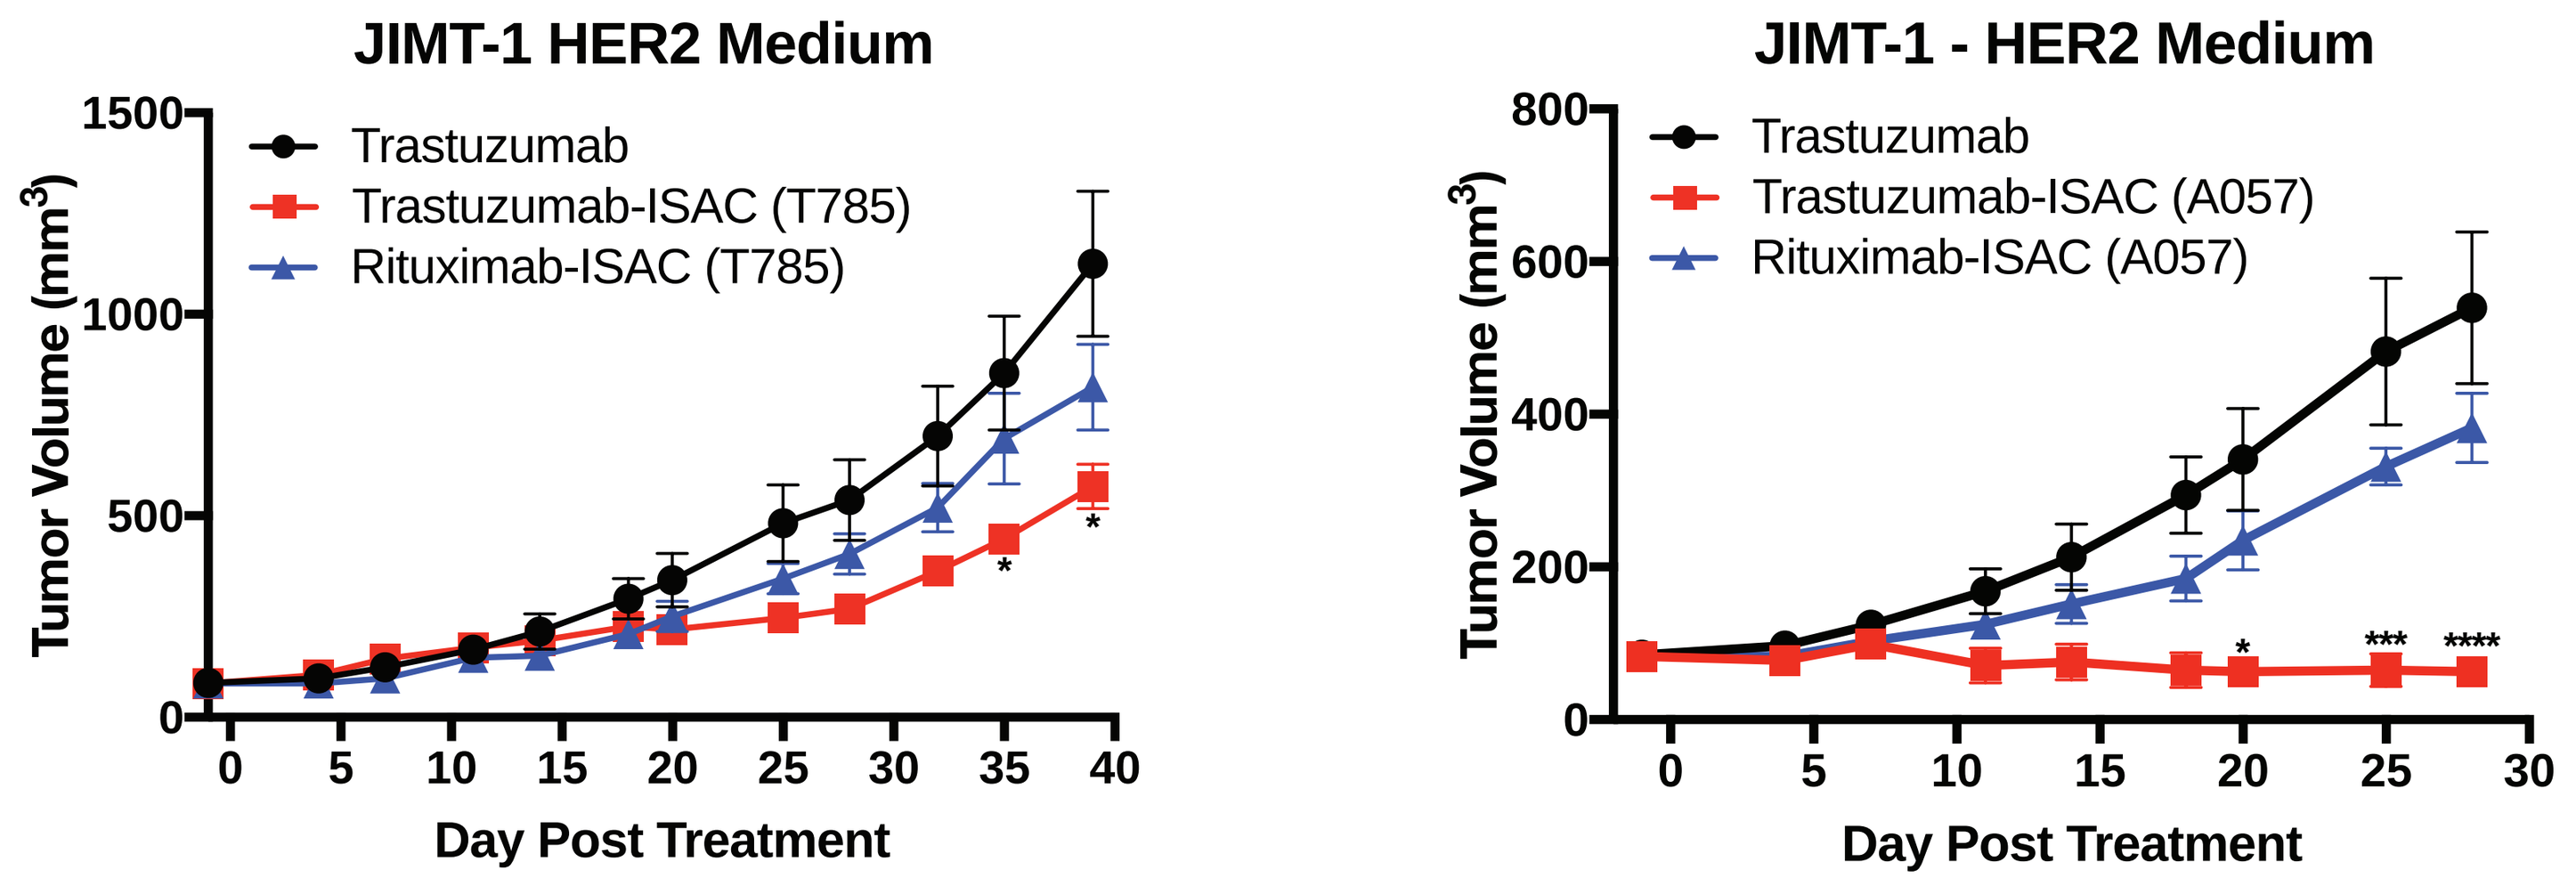
JIMT-1 HER2 Medium
Tumor Volume (mm3)
Day Post Treatment
Trastuzumab
Trastuzumab-ISAC (T785)
Rituximab-ISAC (T785)
JIMT-1 - HER2 Medium
Trastuzumab
Trastuzumab-ISAC (A057)
Rituximab-ISAC (A057)

Figure 8: A057-ISACs mediate enhanced efficacy in trastuzumab-resistant model JIMT-1 as compared to T785-ISACs.

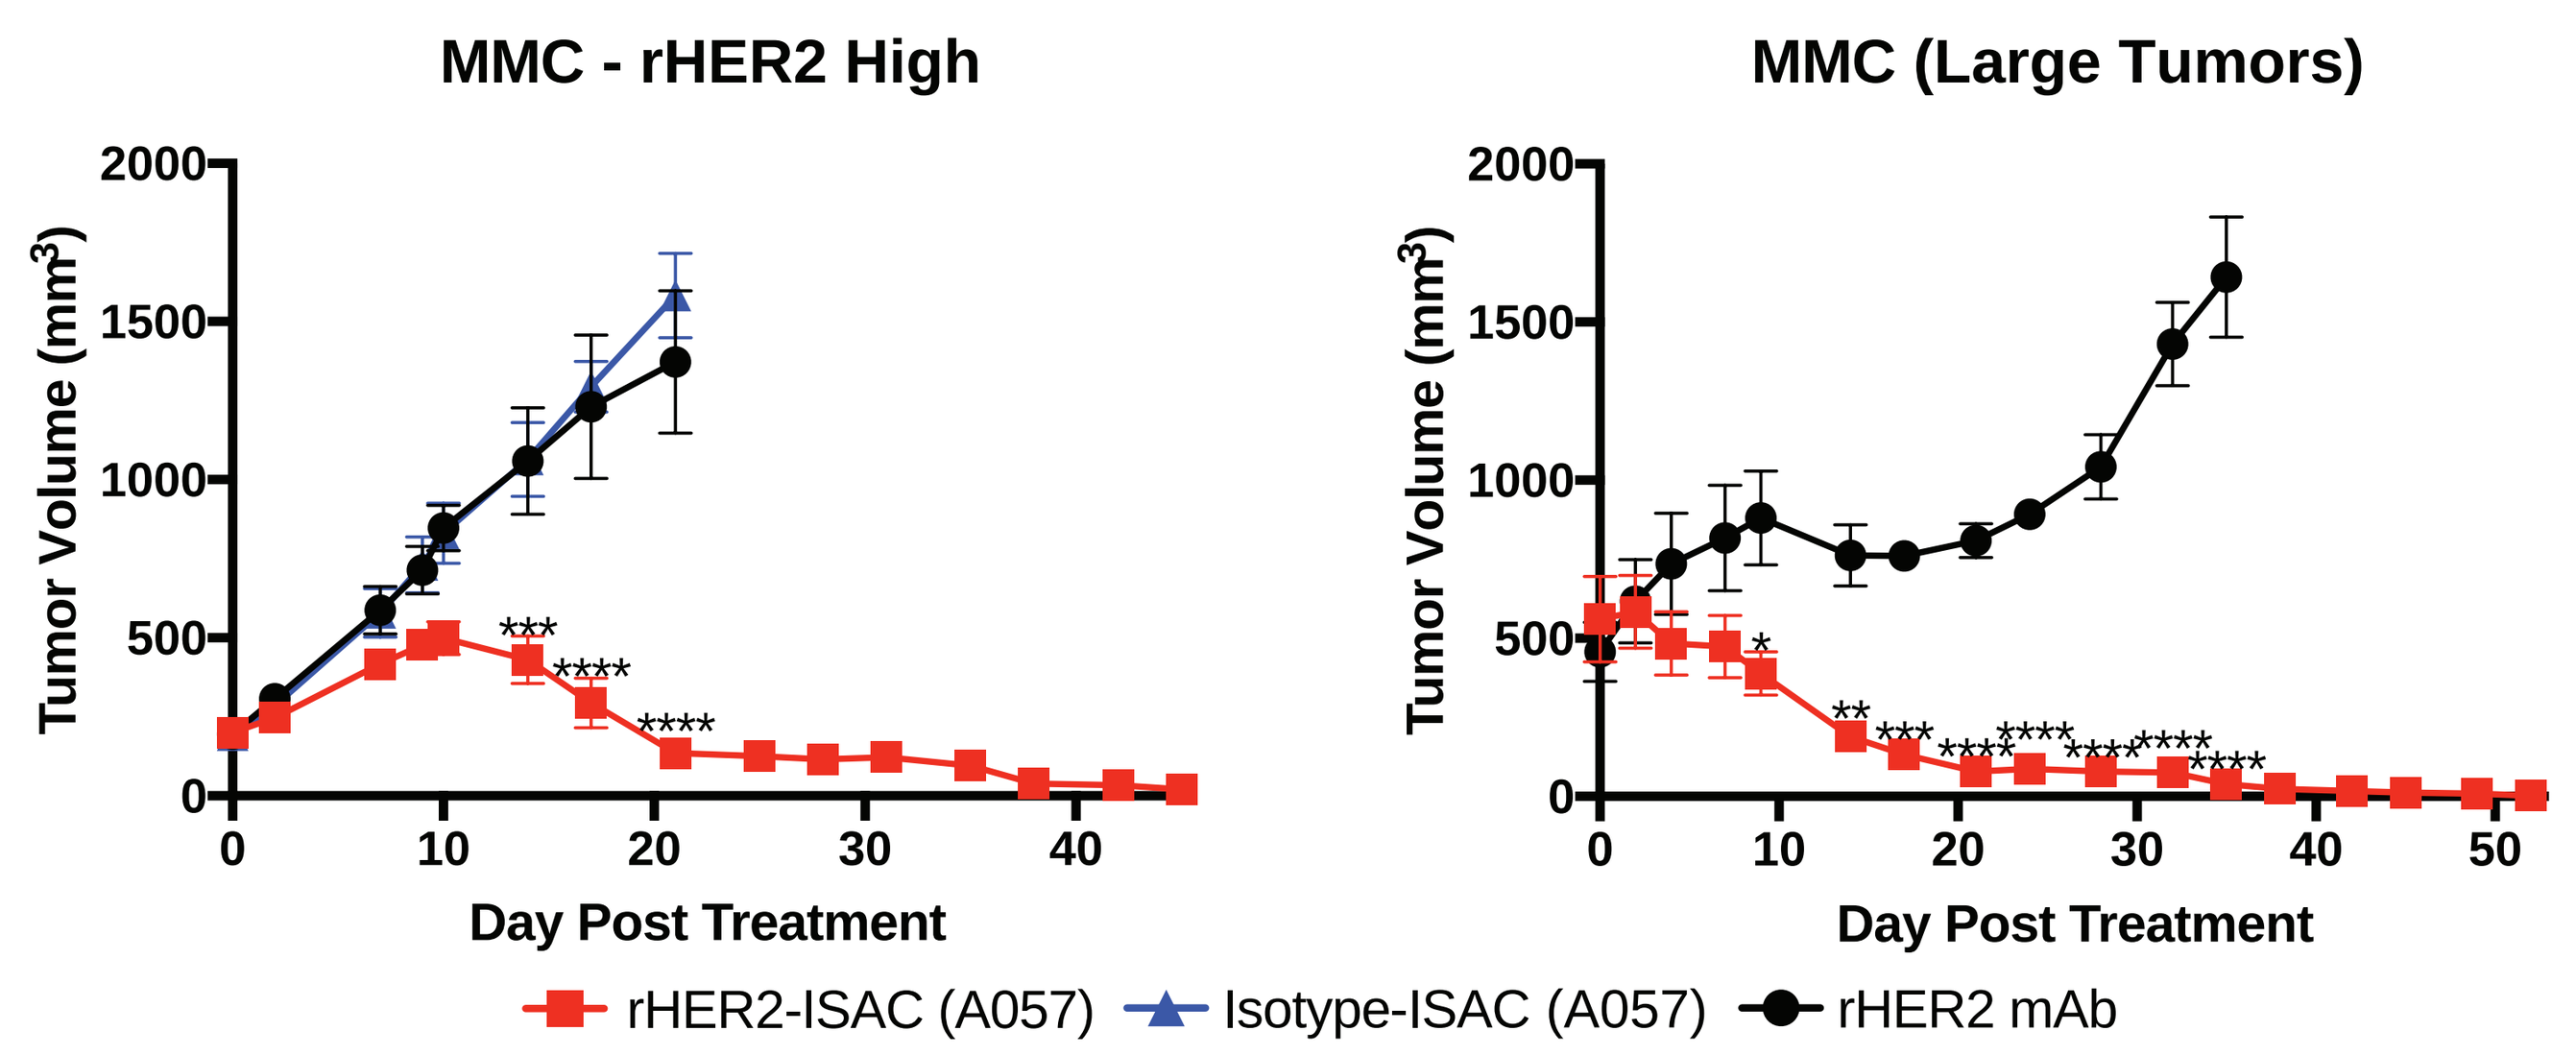
MMC - rHER2 High
MMC (Large Tumors)
Tumor Volume (mm3)
Day Post Treatment
rHER2-ISAC (A057)
Isotype-ISAC (A057)
rHER2 mAb

Figure 9: ISACs lead to tumor clearance of tumors >500 mm³ in syngeneic tumor model.

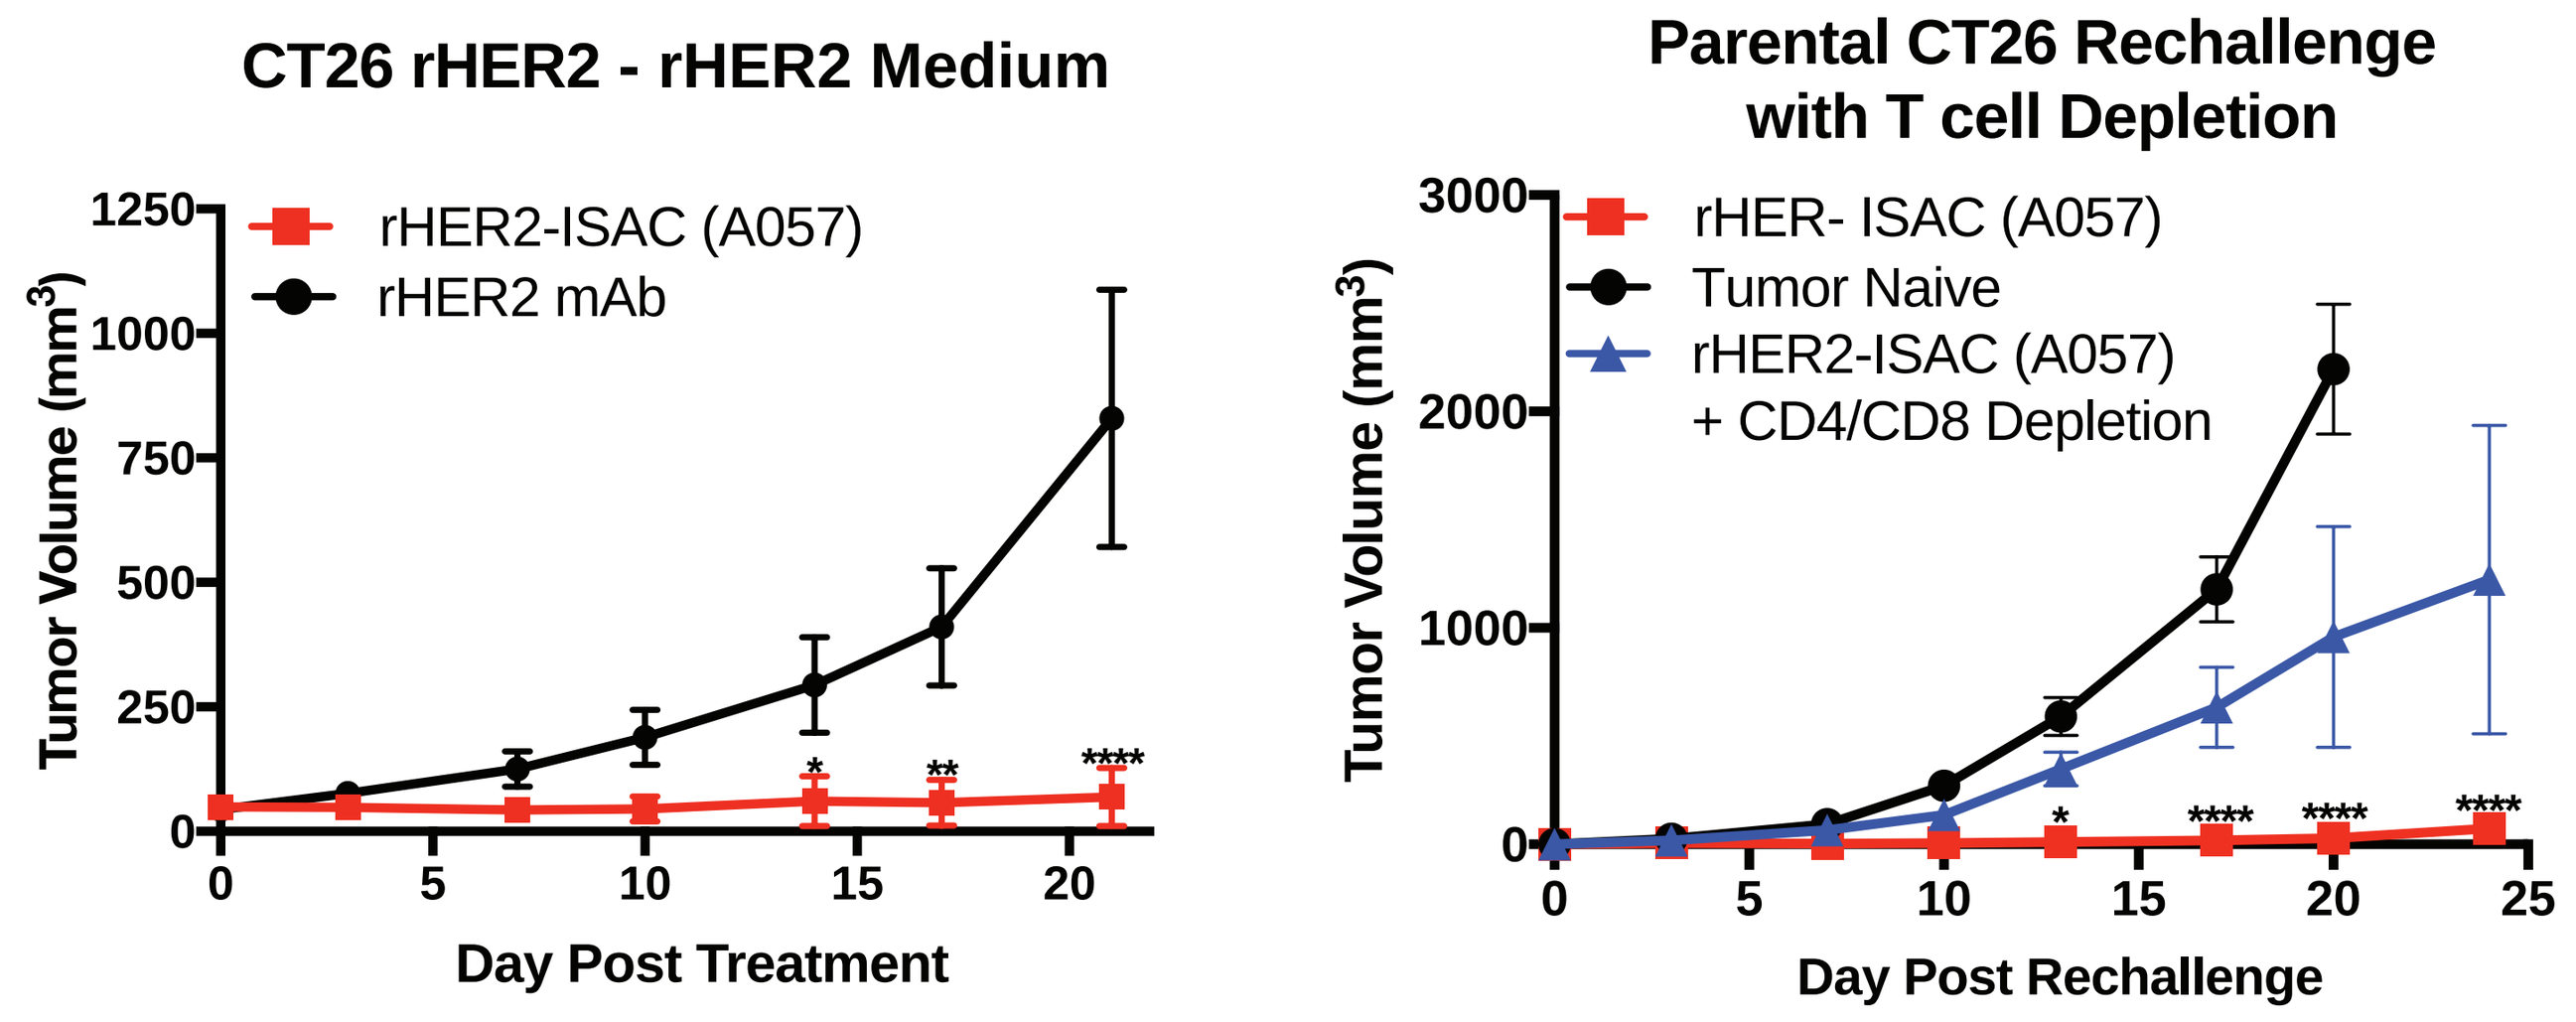
CT26 rHER2 - rHER2 Medium
Tumor Volume (mm3)
Day Post Treatment
rHER2-ISAC (A057)
rHER2 mAb
Parental CT26 Rechallenge with T cell Depletion
rHER- ISAC (A057)
Tumor Naive
rHER2-ISAC (A057) + CD4/CD8 Depletion
Day Post Rechallenge

Figure 10: ISACs lead to tumor clearance, epitope spreading and govern T cell-dependent immunologic memory.

KEY FINDINGS

1. ISACs elicit robust myeloid activation and DC differentiation in human cells
2. ISAC treatment leads to anti-tumor efficacy in trastuzumab resistant models in TLR- and FcR- dependent manner
3. ISACs demonstrate efficacy tumors as large as >500 mm³ in volume
4. Tumor clearance following ISAC treatment leads to T cell-dependent immunologic memory